Poster #: 001

An Open-Label, Phase 1b Study of the Neuroactive Steroid $GABA_A$ Receptor Positive Allosteric Modulator SAGE-324 in Essential Tremor

Helen Colquhoun, MD[1], David P. Nguyen, PhD[1], Min Qin, PhD[1], Angela Wehr, PhD[1], James Doherty, PhD[1], and Stephen J. Kanes, MD, PhD[1]

[1]Sage Therapeutics, Inc., Cambridge, MA, USA

Introduction

- Essential Tremor (ET) is a chronic neurological disorder characterized by bilateral postural tremor and tremor that occurs during voluntary movement of the upper limbs (kinetic tremor).[1]
- The inhibitory neurotransmitter gamma-aminobutyric acid (GABA) may play a significant role in regulation of cerebellar function, which is postulated to be involved in the genesis of tremor in ET.[2]
- The etiology of ET has been associated with impaired cerebellar GABAergic function, resulting from degeneration in the dentate nucleus and Purkinje cell loss.[2,3]
- Both pre-clinical and clinical data support the role of GABA in ET, including decreased GABA levels in cerebro-spinal fluid from individuals with ET, in vivo neuroimaging and histopathology studies showing changes in GABA receptor binding, and electrophysiological studies measuring GABAergic regulation of deep cerebellar nuclei.[4-9]
- The potential use of $GABA_A$ receptor ($GABA_AR$) positive allosteric modulators (PAMs) in ET was evaluated in early phase clinical trials of brexanolone injection and the novel, investigational, oral, $GABA_AR$ PAM zuranolone (SAGE-217) in patients with ET.[10,11] Brexanolone injection is not approved for ET.
- Data from these exploratory studies demonstrated target engagement and promising activity in reduction of kinetic tremor, supporting the development of the investigational compound SAGE-324 and the $GABA_AR$ PAM mechanism in ET.
- This single dose, open-label, Phase 1b study assessed the safety, tolerability, pharmacokinetics (PK), and pharmacodynamics (PD) of SAGE-324 for otherwise healthy ET patients.

Conclusions

- Promising signals of tremor reduction in patients with ET (N=11) consistent with previous studies of brexanolone injection and zuranolone were observed.
- The long half-life of SAGE-324 has the potential to provide consistent plasma concentrations during the dose period (low peak:trough ratio).
- SAGE-324 was generally well-tolerated at the 45 mg and 60 mg doses, with all AEs being mild or moderate.
- Data suggest that the proposed mechanism of action of SAGE-324 on tremor may be consistent with brexanolone and zuranolone, allowing Sage to leverage total GABA platform knowledge to continue development of SAGE-324 in ET.

STUDY DESIGN

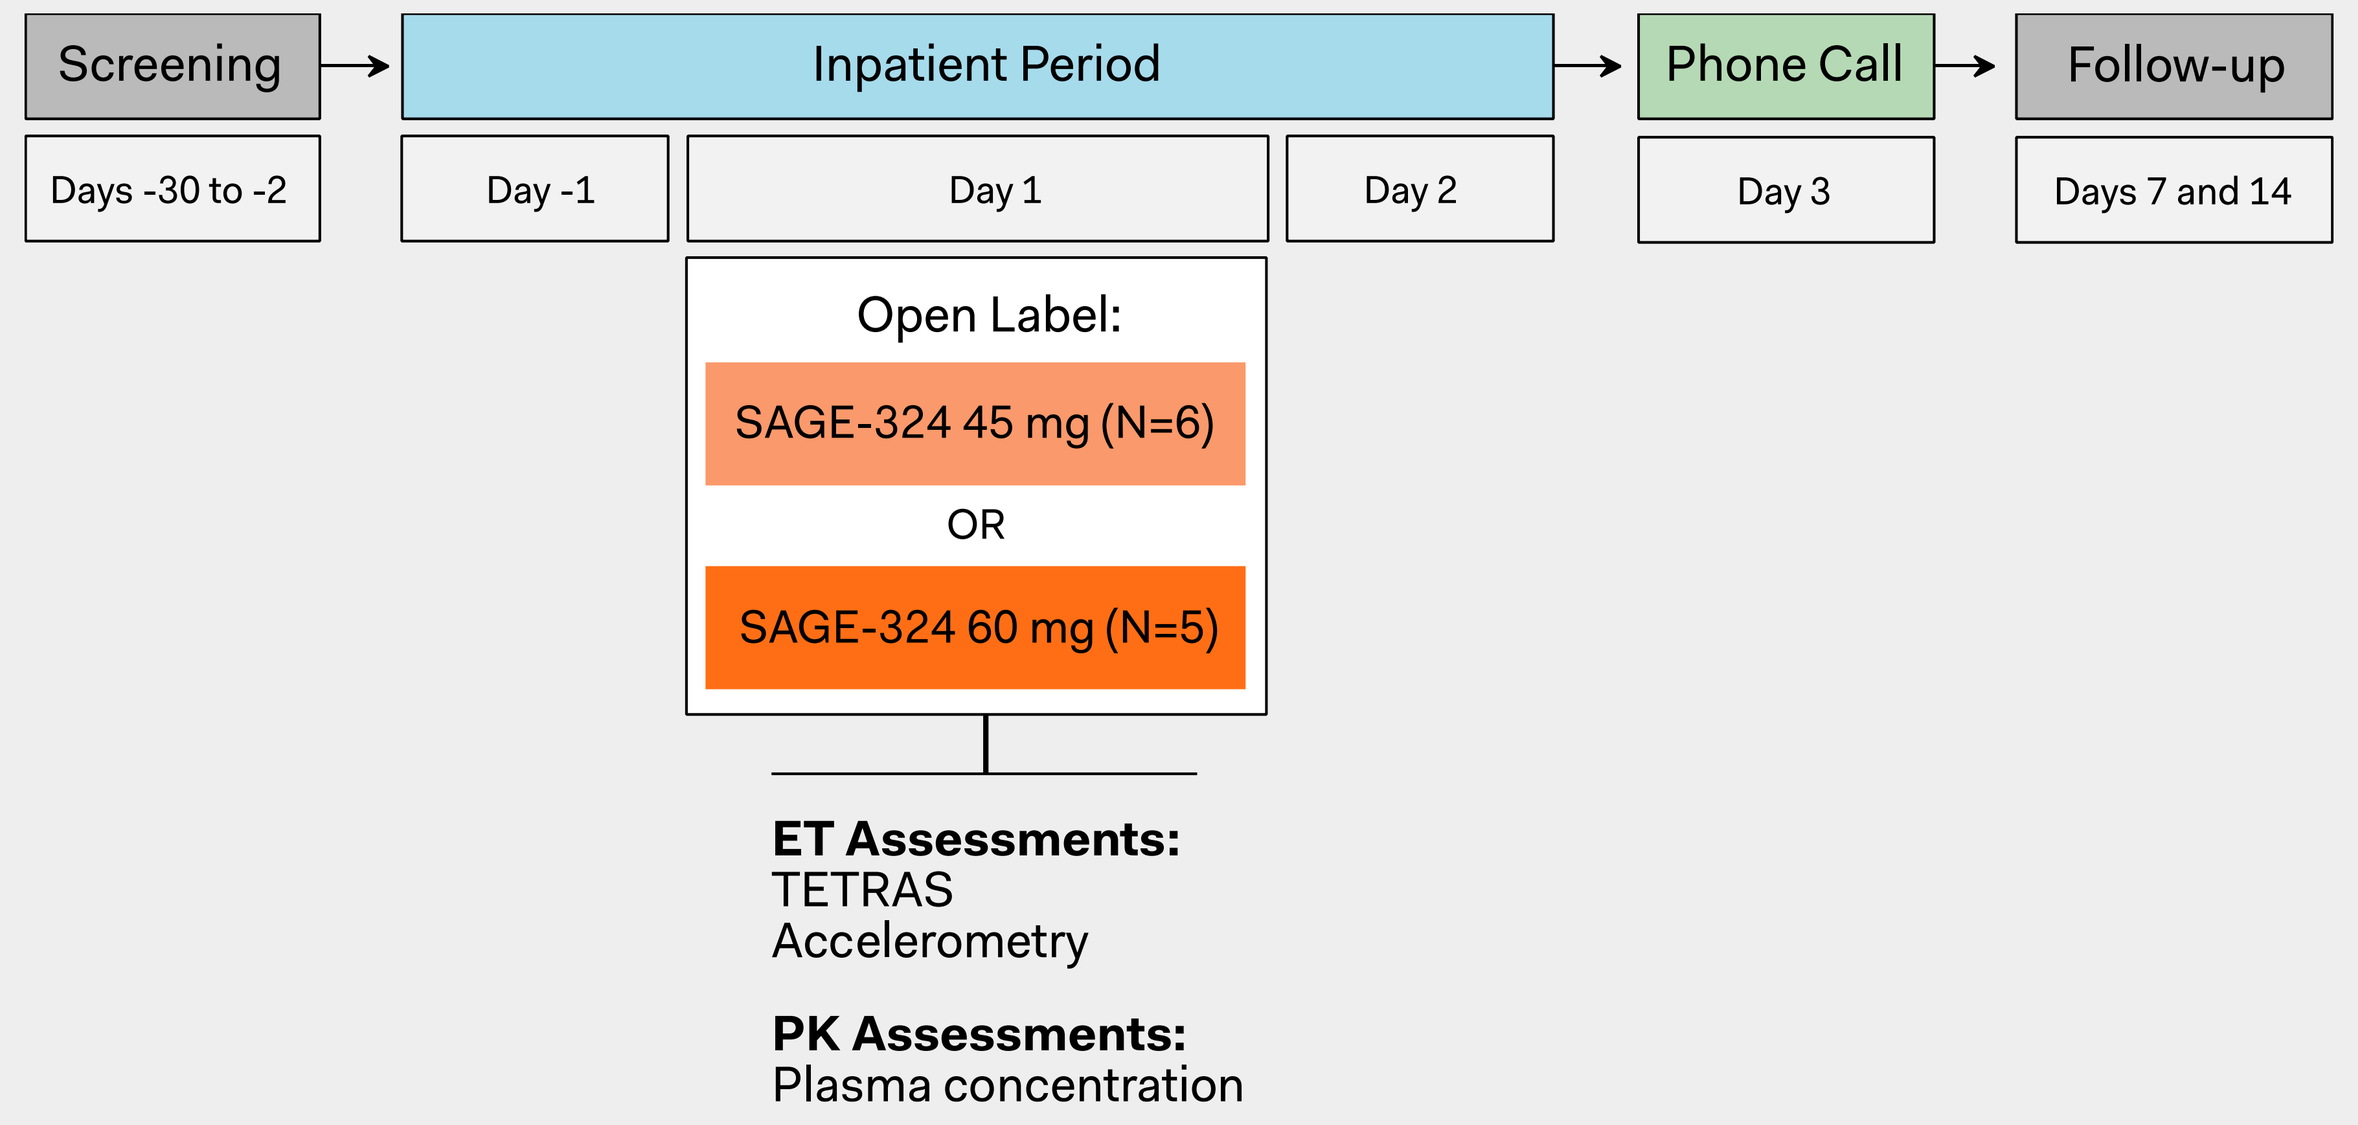
Screening
Days -30 to -2
Inpatient Period
Day -1
Day 1
Day 2
Open Label:
SAGE-324 45 mg (N=6)
OR
SAGE-324 60 mg (N=5)
Phone Call
Day 3
Follow-up
Days 7 and 14
ET Assessments:
TETRAS
Accelerometry
PK Assessments:
Plasma concentration

Results

DEMOGRAPHICS AND BASELINE CHARACTERISTICS

	SAGE-324 45 mg N = 6	SAGE-324 60 mg N = 5
Gender		
Female	3 (50)	2 (40)
Male	3 (50)	3 (60)
Age (years)	62.0 (5.9)	60.2 (13.3)
Ethnicity		
Hispanic/Latino	5 (83)	4 (80)
Race		
White	6 (100)	5 (100)
BMI (kg/m²)	29.4 (4.1)	28.5 (4.5)

Data are mean (SD) or N (%)

TREMOR EFFECTS WITH SAGE-324 PLASMA CONCENTRATIONS: TETRAS COMBINED UPPER LIMB AND ACCELEROMETER ASSESSMENTS

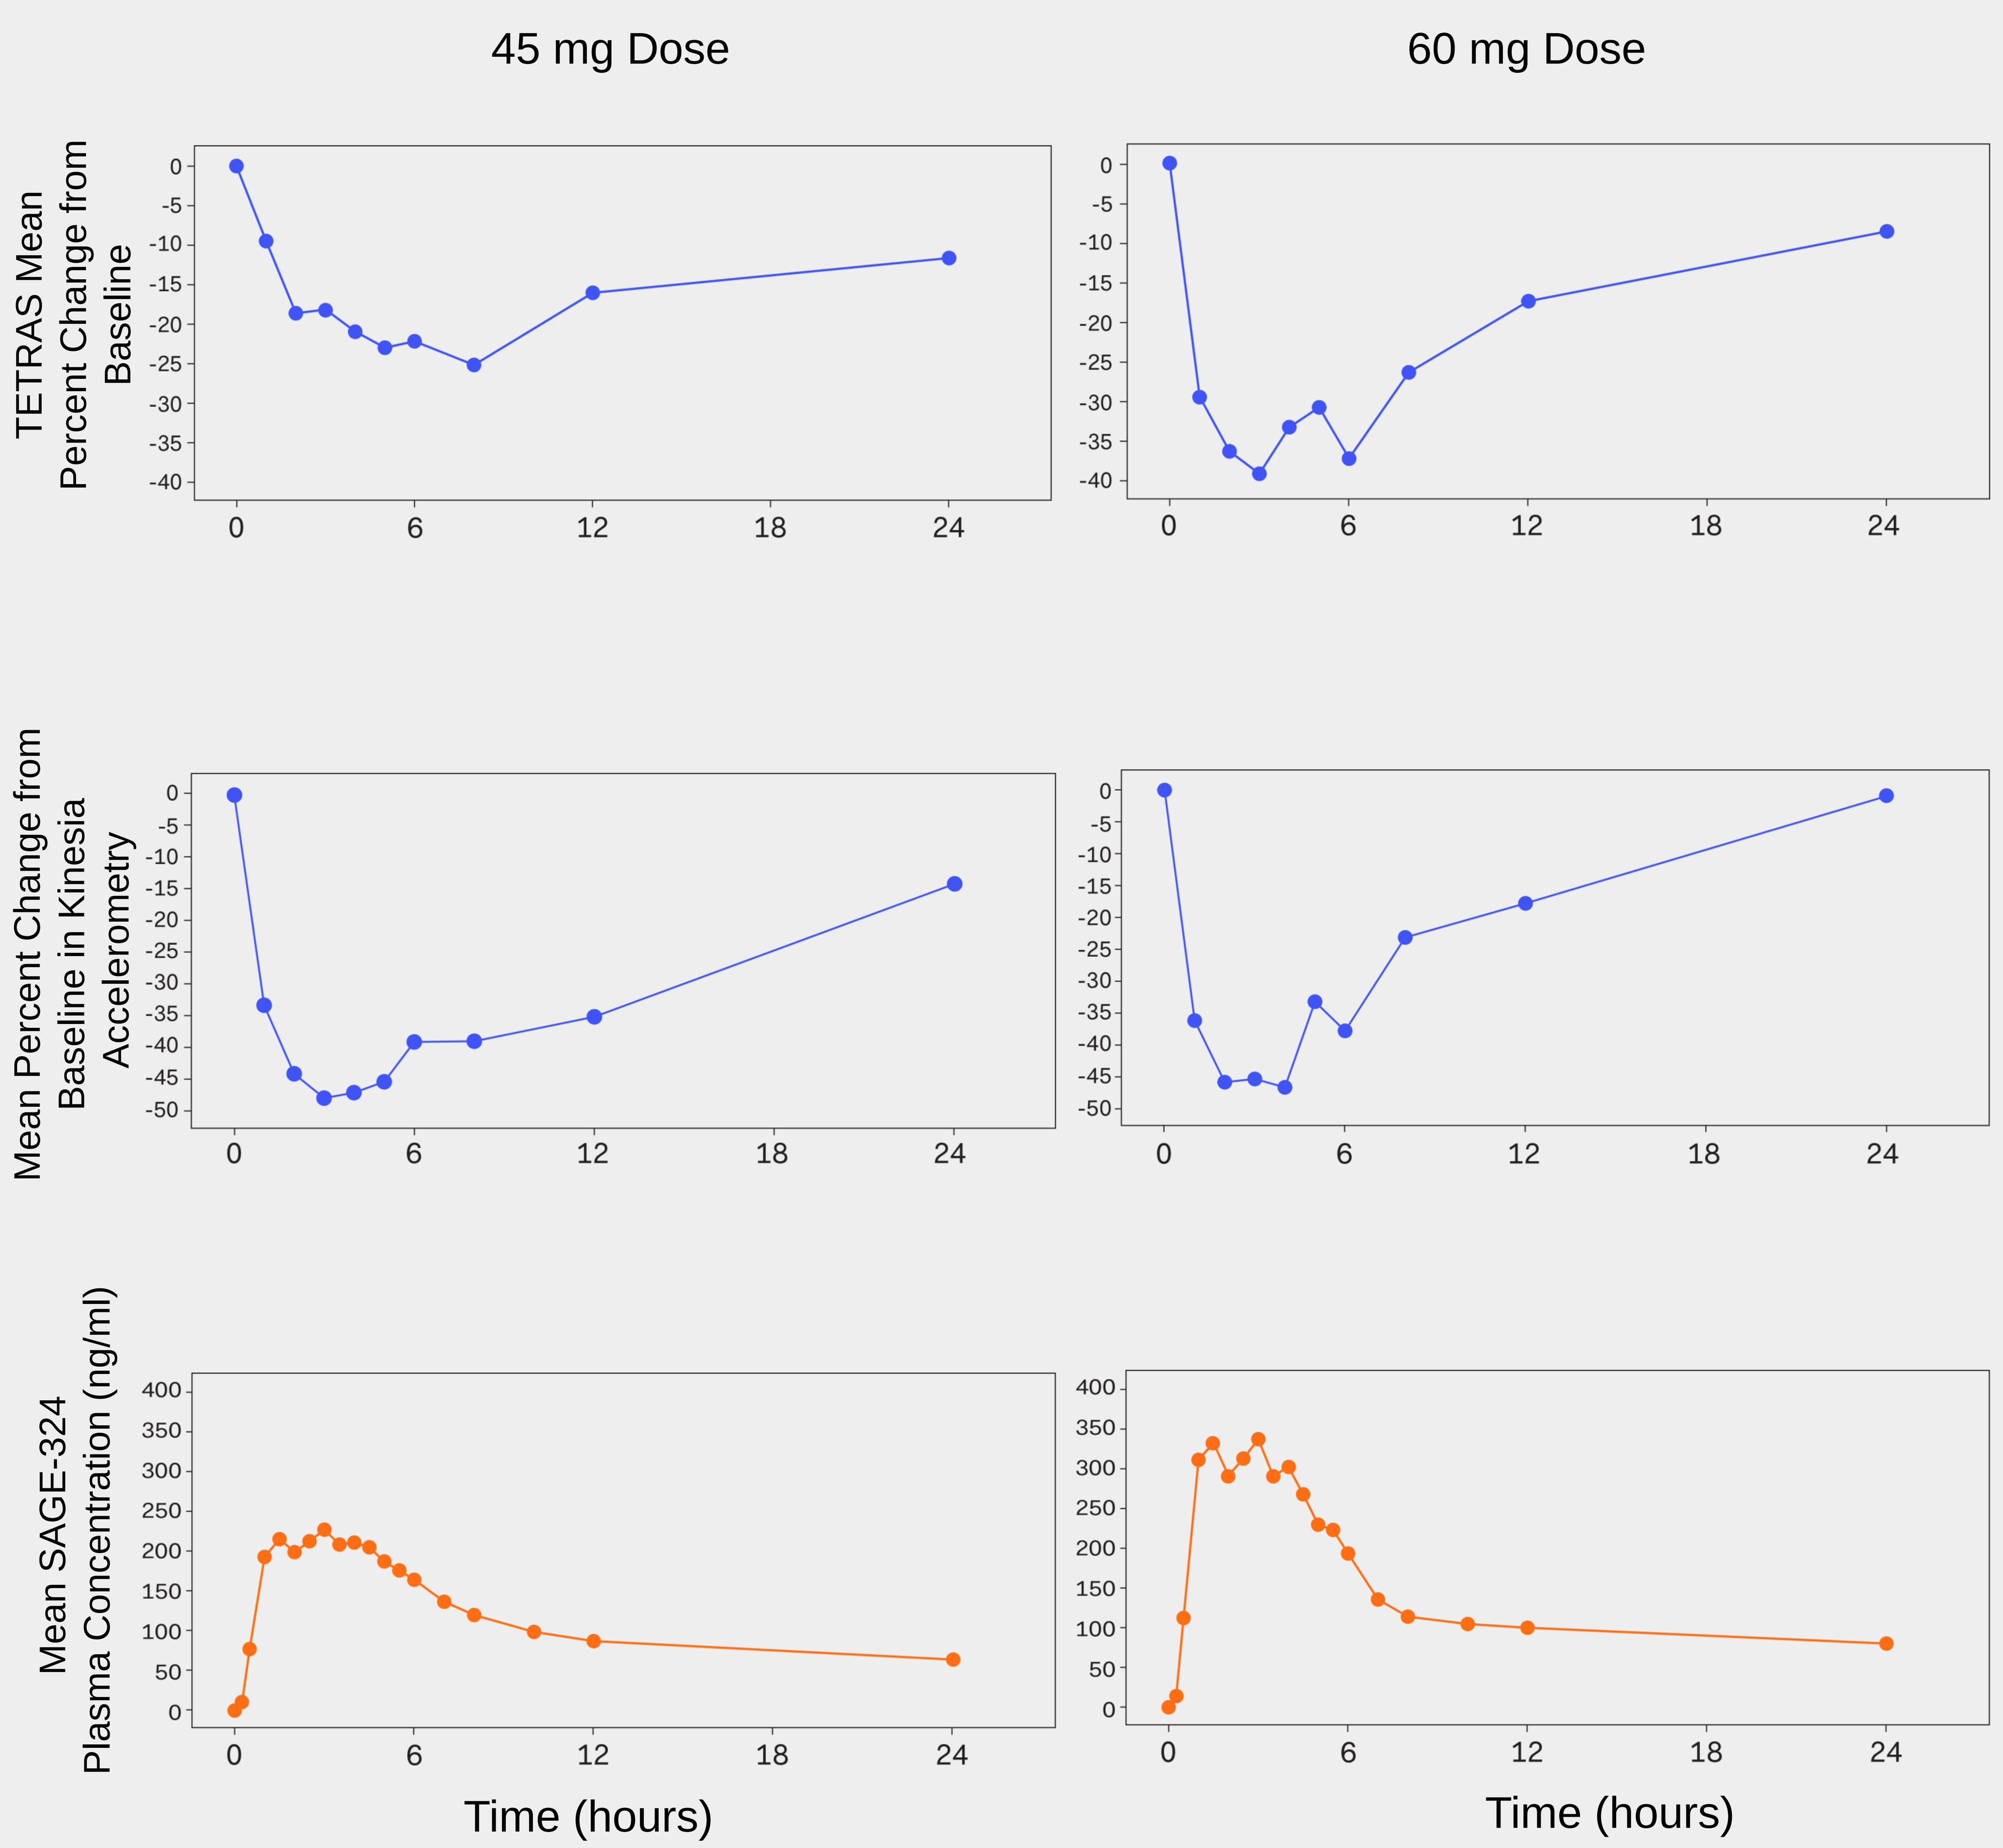
45 mg Dose
60 mg Dose
TETRAS Mean Percent Change from Baseline
Mean Percent Change from Baseline in Kinesia Accelerometry
Mean SAGE-324 Plasma Concentration (ng/ml)
Time (hours)

- In the both the 45 mg and 60 mg dosing conditions, SAGE-324 showed a clear PK/PD relationship as assessed by TETRAS and accelerometry.

TETRAS AND KINESIA ACCELEROMETER SCORES CORRELATION

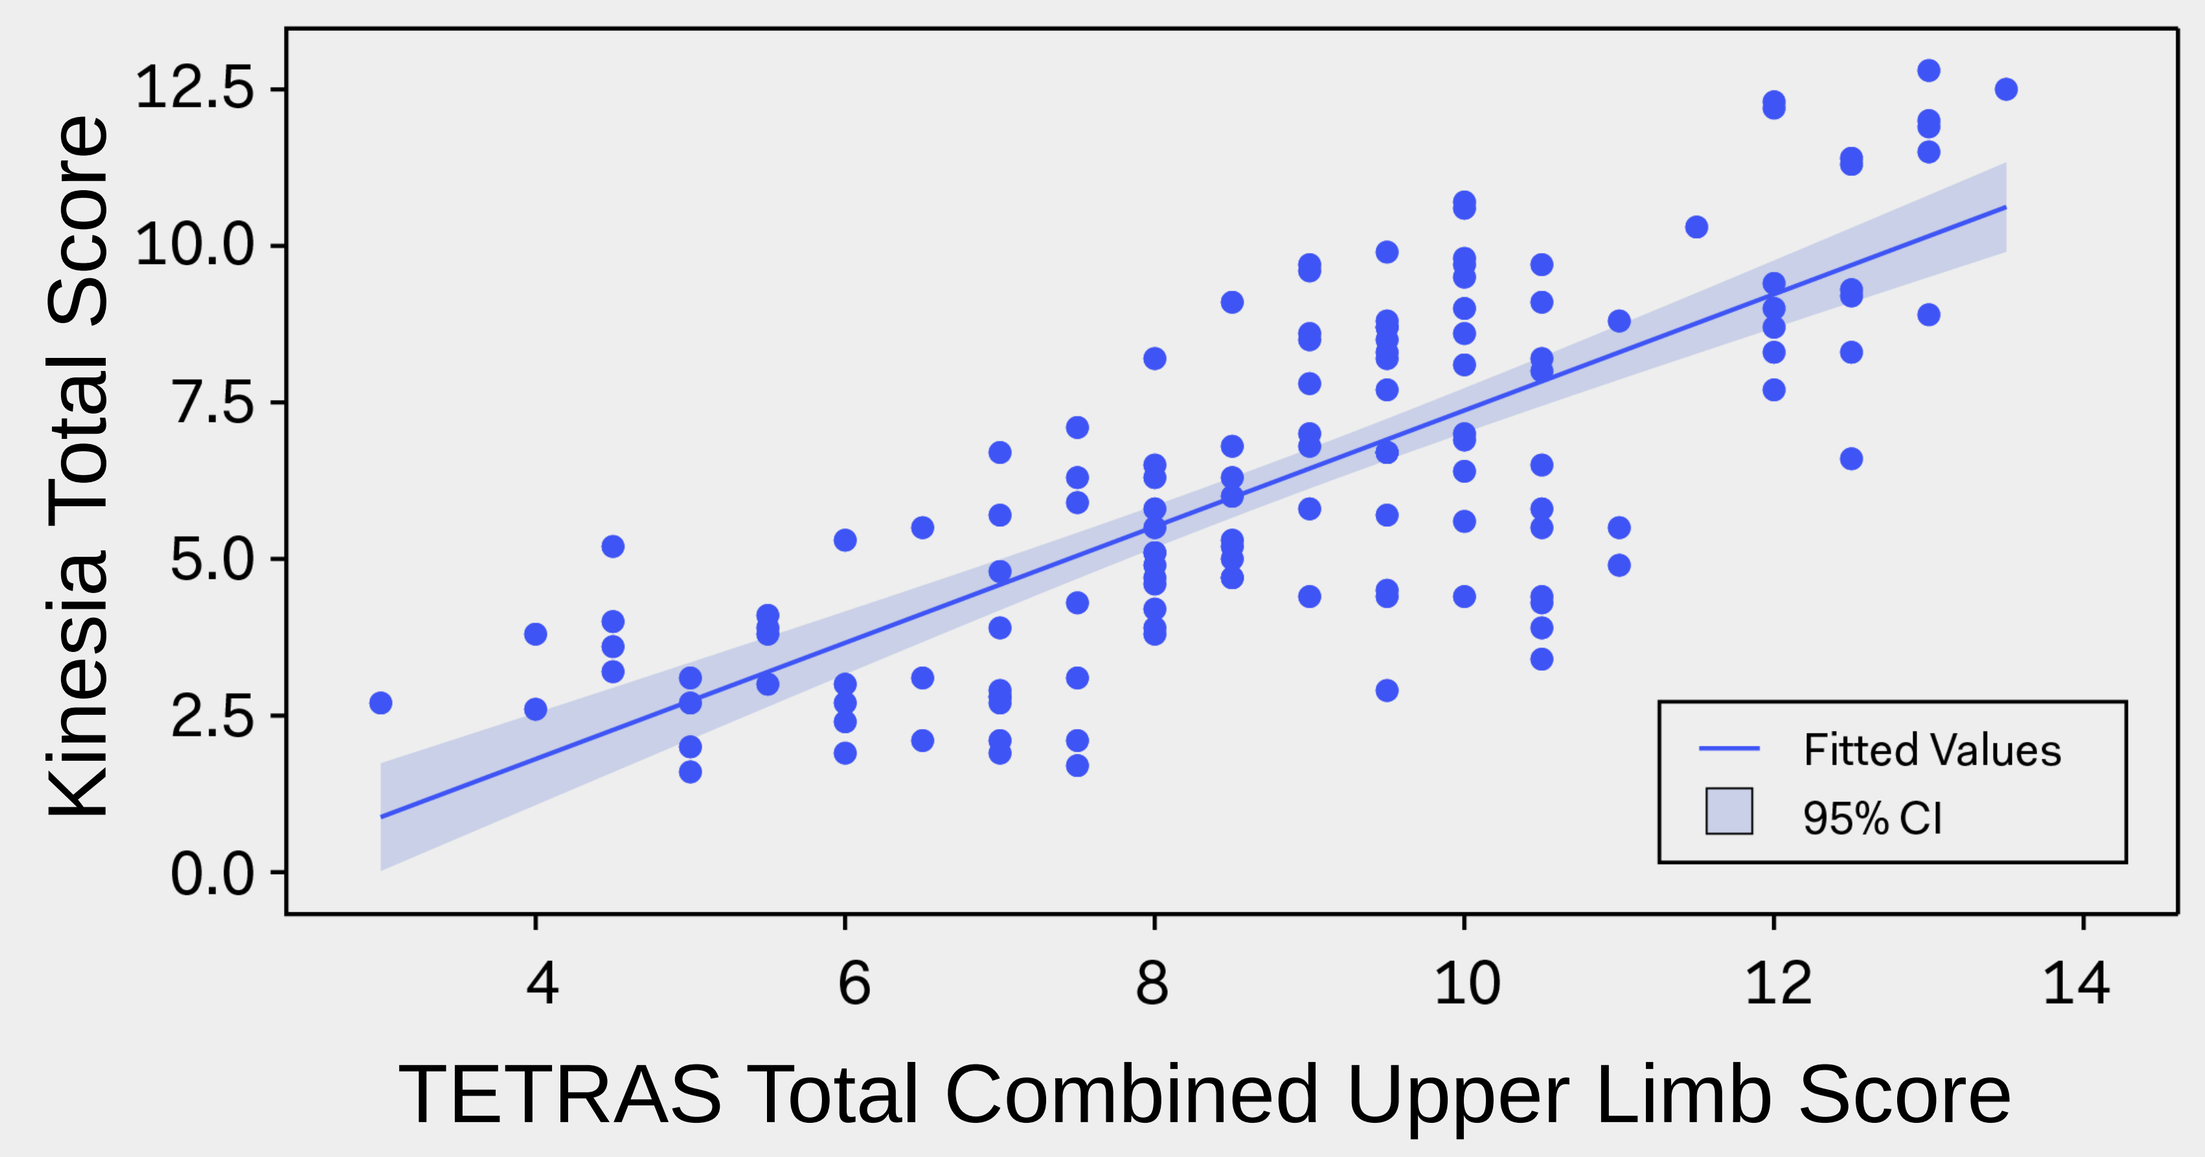
Kinesia Total Score
TETRAS Total Combined Upper Limb Score
Fitted Values
95% CI

- TETRAS total combined upper limb scores and Kinesia total score were well correlated (Pearson correlation r=0.76394, 95% CI 0.682 to 0.827, p≤0.0001, all individual scores shown).
- This correlation indicates that the change in tremor severity following SAGE-324 treatment is well-captured by both the TETRAS and Kinesia accelerometry measures.

TREATMENT EMERGENT ADVERSE EVENTS (TEAEs)

	SAGE-324 45 mg N = 6	SAGE-324 60 mg N = 5
Patients with any TEAE	2 (33)	5 (100)
Patients with mild TEAEs	2 (33)	4 (80)
Patients with moderate TEAEs	2 (33)	1 (20)
Severe TEAEs	0	0
Serious TEAEs	0	0
TEAE-Drug Discontinued	0	0
Deaths	0	0

Data are N (%)

- There were no serious or severe TEAEs in either cohort.
- TEAEs in the 45 mg group were vasovagal symptoms, drowsiness, dry eyes, double vision, dizziness, lightheadedness, bilateral ear fullness, hot flash, headache, feelings of euphoria, and feelings of relaxation, and TEAEs in the 60 mg group were drowsiness, orthostatic hypertension, lightheadedness, and sedation.

Methods

- A single dose of open-label SAGE-324 oral suspension (45 mg or 60 mg) was administered to otherwise healthy ET patients.
- ET was defined by the following criteria: isolated tremor syndrome of bilateral upper limb action tremor, at least 3 years duration, with or without tremor in other locations, and absence of other neurological signs, such as dystonia, ataxia, or parkinsonism.
- Patients with ET (N=11) were 18-80 years old, willing to discontinue ET treatments (for ≥14 days prior to admission and through the course of the study) and benzodiazepines (for 5 half-lives prior to dosing and through the course of the study), and presenting with upper limb symptoms as confirmed by a combined The Essential Tremor Rating Assessment Scale (TETRAS) upper limb total score (sum of left and right kinetic, postural, wing-beating tremor) of ≥8 on the Performance subscale Part 4, at screening and admission.
- ET patients with prior surgery for ET were excluded.
- Tremor severity was evaluated over time using the TETRAS Performance subscale Part 4 and the accelerometer-based Kinesia assessment (Great Lakes Neuro Technologies).
- Plasma concentrations of SAGE-324 were assessed throughout the study.
- Safety and tolerability were assessed by adverse event reporting and standard clinical assessments.

REFERENCES
1. Thanvi B et al. Age and Ageing 2006; 35(4): 344–349.
2. Gironell A. Louis ED, ed. Tremor and Other Hyperkinetic Movements. 2014;4:254.
3. Choe M, et al. Mov Disord. 2016;31(3):393-401
4. Mally J, et al. J Neural Transm. 1996;103:555–560.
5. Mouginot D, Gahwiler BH. J Neurophysiol. 1996; 75: 894–901.
6. Paris-Robidas A, et al. Brain. 2012; 135: 105–116.
7. Uusisaari M, Knopfel T. Neuroscience. 2008; 156: 537–549.
8. Boecker H, et al. J Nucl Med. 2010; 51: 1030–1035.
9. Gironell A, et al. Parkinsonism Relat Disord 2012; 18: 876–880.
10. Kaul I et al. Evaluating SAGE-217 Capsules in Essential Tremor: an open-label, pilot clinical trial. Poster at 2nd Pan American Parkinson's Disease and Movement Disorders Congress. June 22-24, 2019. Miami, FL, USA.
11. Ellenbogen AE et al. Exploratory Trial Results for SAGE-547 in Essential Tremor. Poster at the American Academy of Neurology 2016 Annual Meeting. April 15-21, 2016. Vancouver, BC, Canada

SAGE THERAPEUTICS is a trademark of Sage Therapeutics, Inc. All other trademarks referenced herein are the property of their respective owners.

SUPPORT & DISCLOSURES
HC, DPN, MQ, AW, JD, and SJK are employees of Sage Therapeutics, Inc., with stock/stock options.This study was funded by Sage Therapeutics, Inc. This poster was assembled with assistance from Boston Strategic Partners.

American Academy of Neurology Annual Meeting
April 25 – May 1, 2020
Toronto, Canada
MRC-324-00133 05/20