Poster #: 009

Using a Multimodal Biomarker Approach to Identify Functional Target Engagement of the Novel NMDA Positive Allosteric Modulator SAGE-718

Aaron Koenig, MD[1], Jason Berlin, MS[1], Yingchun Luo, PhD[1], Sigui Li, MS[1], Ludmila Kryzhanovskaya, MD, PhD[1], Brandon Farley, PhD[1], David Nguyen, PhD[1], Irena Webster, MPH[1], Michael Quirk, PhD[1], Stephen J. Kanes, MD, PhD[1], James Doherty, PhD[1]

[1]Sage Therapeutics, Inc., Cambridge, MA, USA

Introduction

- N-methyl-D-aspartate (NMDA) receptor hypoactivity has been linked to a range of clinical phenomena, including cognitive dysfunction and alterations in social behavior.[1-3]
- Potential treatments for conditions characterized by NMDA hypofunction may involve enhancement of NMDA receptor-related neurotransmission by regulation of synaptic strength, health, and plasticity.[1-3]
- A glutamate agonist compound was previously shown to demonstrate target engagement of NMDA receptors by attenuating or reversing electrophysiological effects of ketamine, a non-competitive NMDA receptor antagonist.[4]
 - Non-competitive NMDA receptor antagonists, such as ketamine, may cause auditory-gating and cognitive deficits.[4,5]
- SAGE-718 is an investigational, oral, neuroactive steroid, NMDA receptor positive allosteric modulator (PAM) and is hypothesized to attenuate ketamine-induced electrophysiological effects.
- In these randomized double-blind, placebo-controlled studies, target engagement of SAGE-718 was assessed by its impact on ketamine-induced effects in healthy volunteers by electrophysiological parameters (EPs; NCT03771586) and functional magnetic resonance imaging (fMRI; NCT03770780).

Conclusions

- SAGE-718 administration altered electrophysiological measures and ketamine-induced changes in functional neuroimaging, indicating pharmacological engagement of NMDA receptors.
- SAGE-718 decreased the standard N100 auditory evoked potential measured during the auditory oddball paradigm.
- SAGE-718 modulated the effects of ketamine on regional and global measures of resting brain activity on fMRI.
- SAGE-718 with ketamine challenge was generally well tolerated. There were no serious AEs or TEAEs leading to study withdrawal or discontinuation.
- The observed effects support the characterization of the proposed mechanism of action of SAGE-718 as an NMDA receptor PAM and functional engagement of the NMDA receptor. Further development of SAGE-718 in disorders characterized by NMDA receptor hypoactivity are ongoing.

Results

DEMOGRAPHICS AND BASELINE CHARACTERISTICS

	718-EXM-101 (N = 20)	718-EXM-102 (N = 19)
Age (years)	39.7 (11.9)	42.2 (12.5)
Weight (kg)	78.2 (10.7)	77.8 (12.6)
Height (cm)	176.1 (11.2)	170.2 (8.6)
BMI (kg/m²)	25.2 (2.8)	26.7 (2.7)
Gender		
Female	2 (10.0)	7 (36.8)
Male	18 (90.0)	12 (63.2)
Ethnicity		
Hispanic	2 (10.0)	3 (15.8)
Race		
Black/African-American	15 (75.0)	10 (52.6)
White	4 (20.0)	8 (42.1)
Other	1 (5.0)	1 (5.3)

Data are mean (SD) or n (%), from the safety set.

STUDY DESIGN

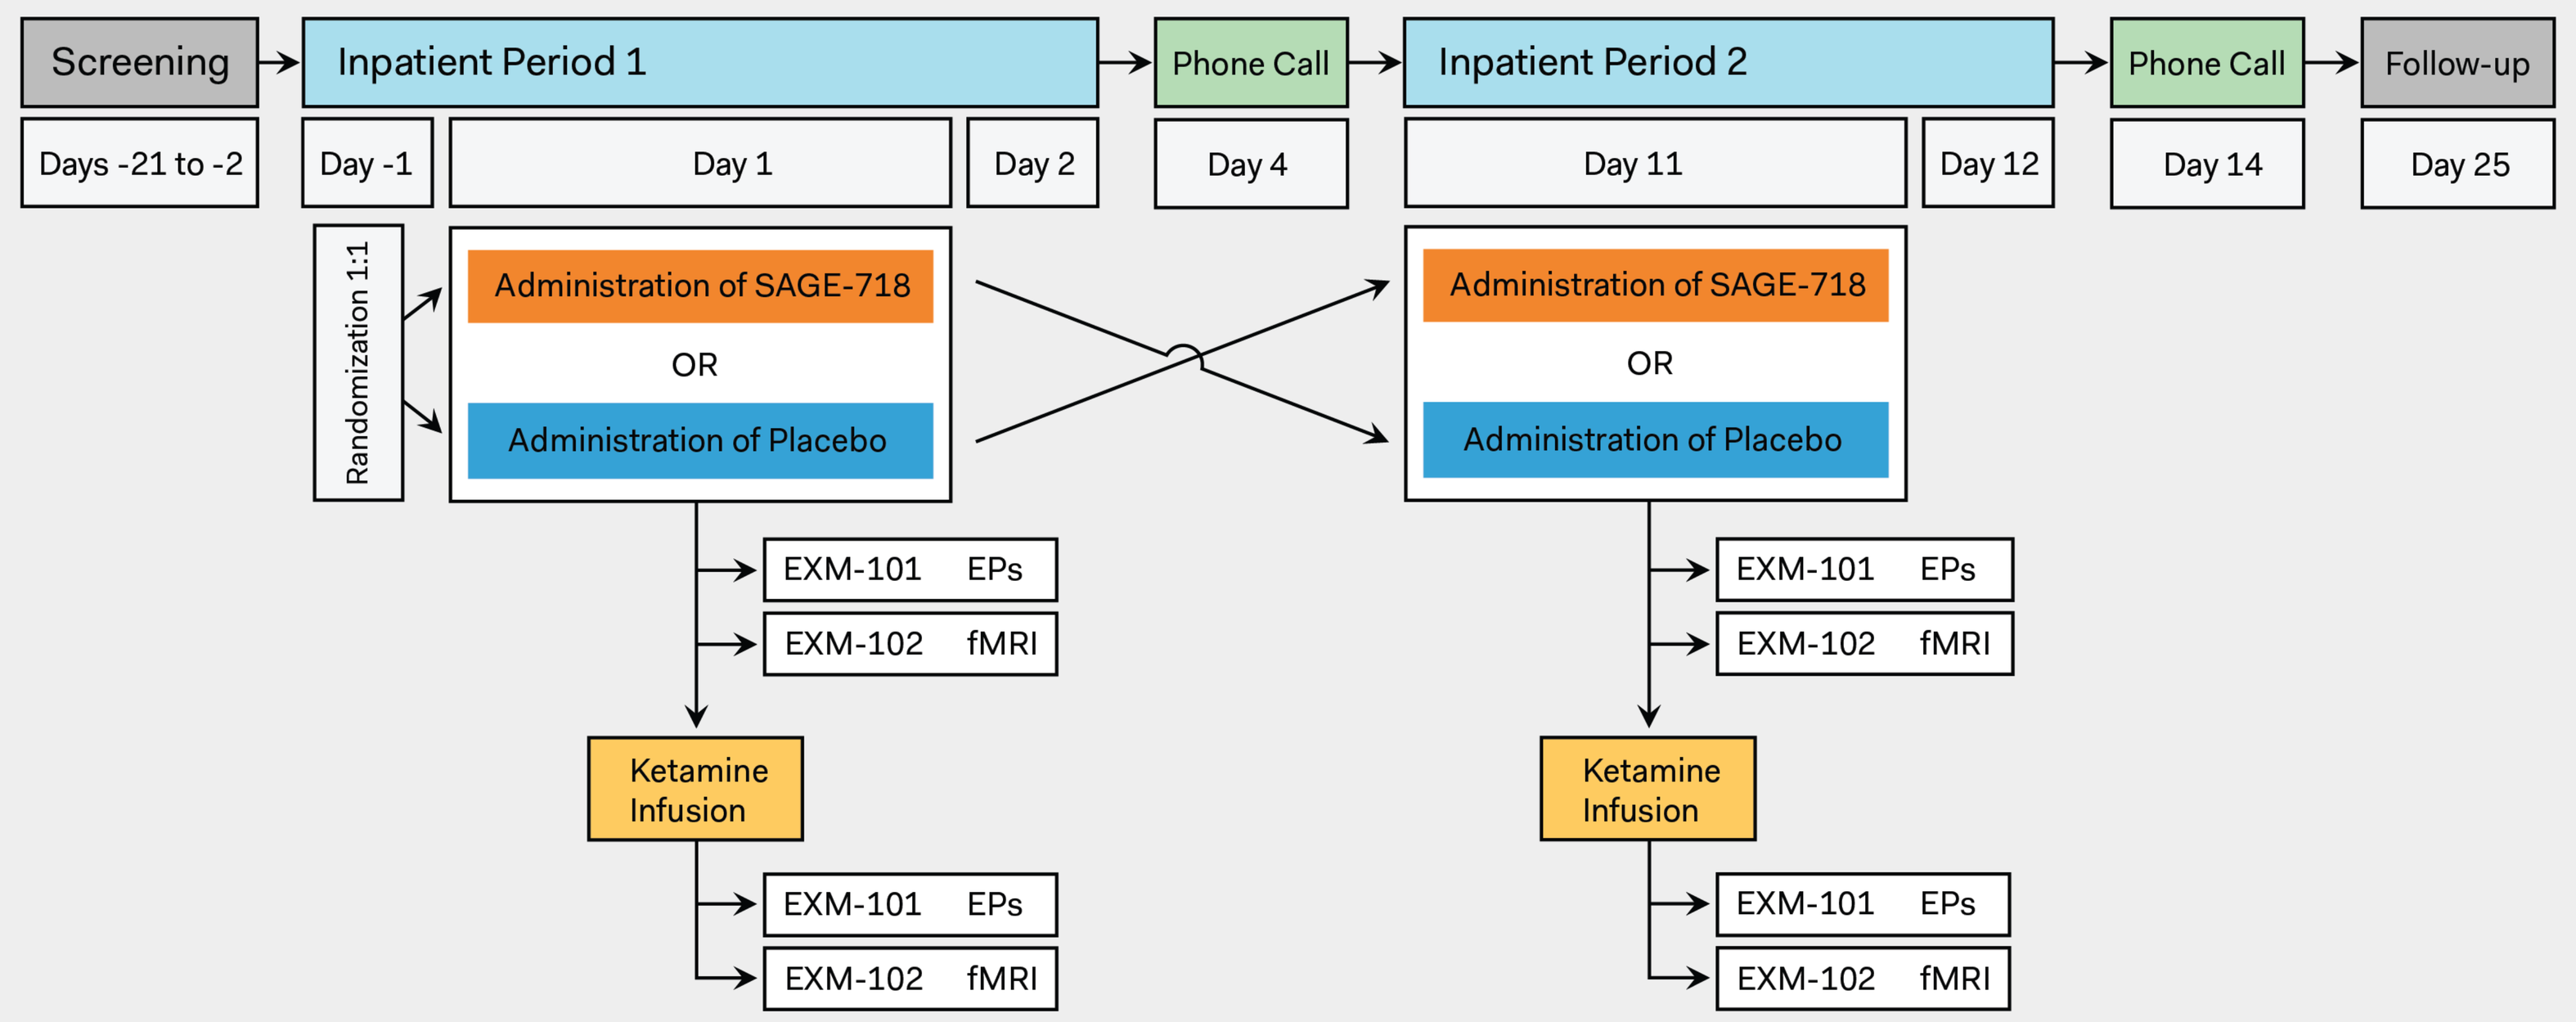
Screening
Inpatient Period 1
Phone Call
Inpatient Period 2
Phone Call
Follow-up
Days -21 to -2
Day -1
Day 1
Day 2
Day 4
Day 11
Day 12
Day 14
Day 25
Randomization 1:1
Administration of SAGE-718
OR
Administration of Placebo
EXM-101 EPs
EXM-102 fMRI
Ketamine Infusion

TREATMENT EMERGENT ADVERSE EVENTS (TEAEs)

	718-EXM-101				718-EXM-102			
	Placebo (n=20)		SAGE-718 (n=22)		Placebo (n=17)		SAGE-718 (n=17)	
Ketamine Infusion	**Pre**	**Post**	**Pre**	**Post**	**Pre**	**Post**	**Pre**	**Post**
Subjects with any TEAE	2 (10.0)	8 (40.0)	1 (4.5)	8 (36.4)	1 (5.9)	3 (17.6)	1 (5.9)	2 (11.8)
Dizziness	0 (0)	4 (20.0)	0 (0)	2 (9.1)	1 (5.9)	1 (5.9)	0 (0)	1 (5.9)
Headache	1 (5.0)	1 (5.0)	1 (4.5)	1 (4.5)	0 (0)	1 (5.9)	1 (5.9)	1 (5.9)
Nausea	0 (0)	1 (5.0)	0 (0)	2 (9.1)	0 (0)	1 (5.9)	0 (0)	1 (5.9)
Vomiting	0 (0)	0 (0)	0 (0)	2 (9.1)	0 (0)	0 (0)	0 (0)	0 (0)
Bradykinesia	0 (0)	0 (0)	0 (0)	1 (4.5)	0 (0)	0 (0)	0 (0)	0 (0)
Dissociation	0 (0)	0 (0)	0 (0)	1 (4.5)	0 (0)	0 (0)	0 (0)	0 (0)
Fatigue	1 (5.0)	0 (0)	1 (4.5)	0 (0)	0 (0)	0 (0)	0 (0)	0 (0)
Feeling drunk	0 (0)	1 (5.0)	0 (0)	1 (4.5)	0 (0)	0 (0)	0 (0)	0 (0)
Skin abrasion	0 (0)	0 (0)	0 (0)	1 (4.5)	0 (0)	0 (0)	0 (0)	0 (0)
Somnolence	1 (5.0)	3 (15.0)	0 (0)	1 (4.5)	0 (0)	0 (0)	0 (0)	0 (0)
Upper respiratory tract infection	0 (0)	0 (0)	0 (0)	1 (4.5)	0 (0)	0 (0)	0 (0)	0 (0)
Feeling abnormal	0 (0)	1 (5.0)	0 (0)	0 (0)	0 (0)	1 (5.9)	0 (0)	0 (0)
Visual impairment	0 (0)	0 (0)	0 (0)	0 (0)	0 (0)	2 (11.8)	0 (0)	0 (0)

Data are n (%) from the safety set.

- SAGE-718 was generally well tolerated with no serious AEs or deaths.
- No TEAEs resulted in discontinuation or dose reduction of SAGE-718.
- Compared with pre-ketamine dosing, a greater number of AEs were observed post-ketamine dosing in all groups across studies.

ATTENUATION OF KETAMINE EFFECTS ON BOLD fMRI

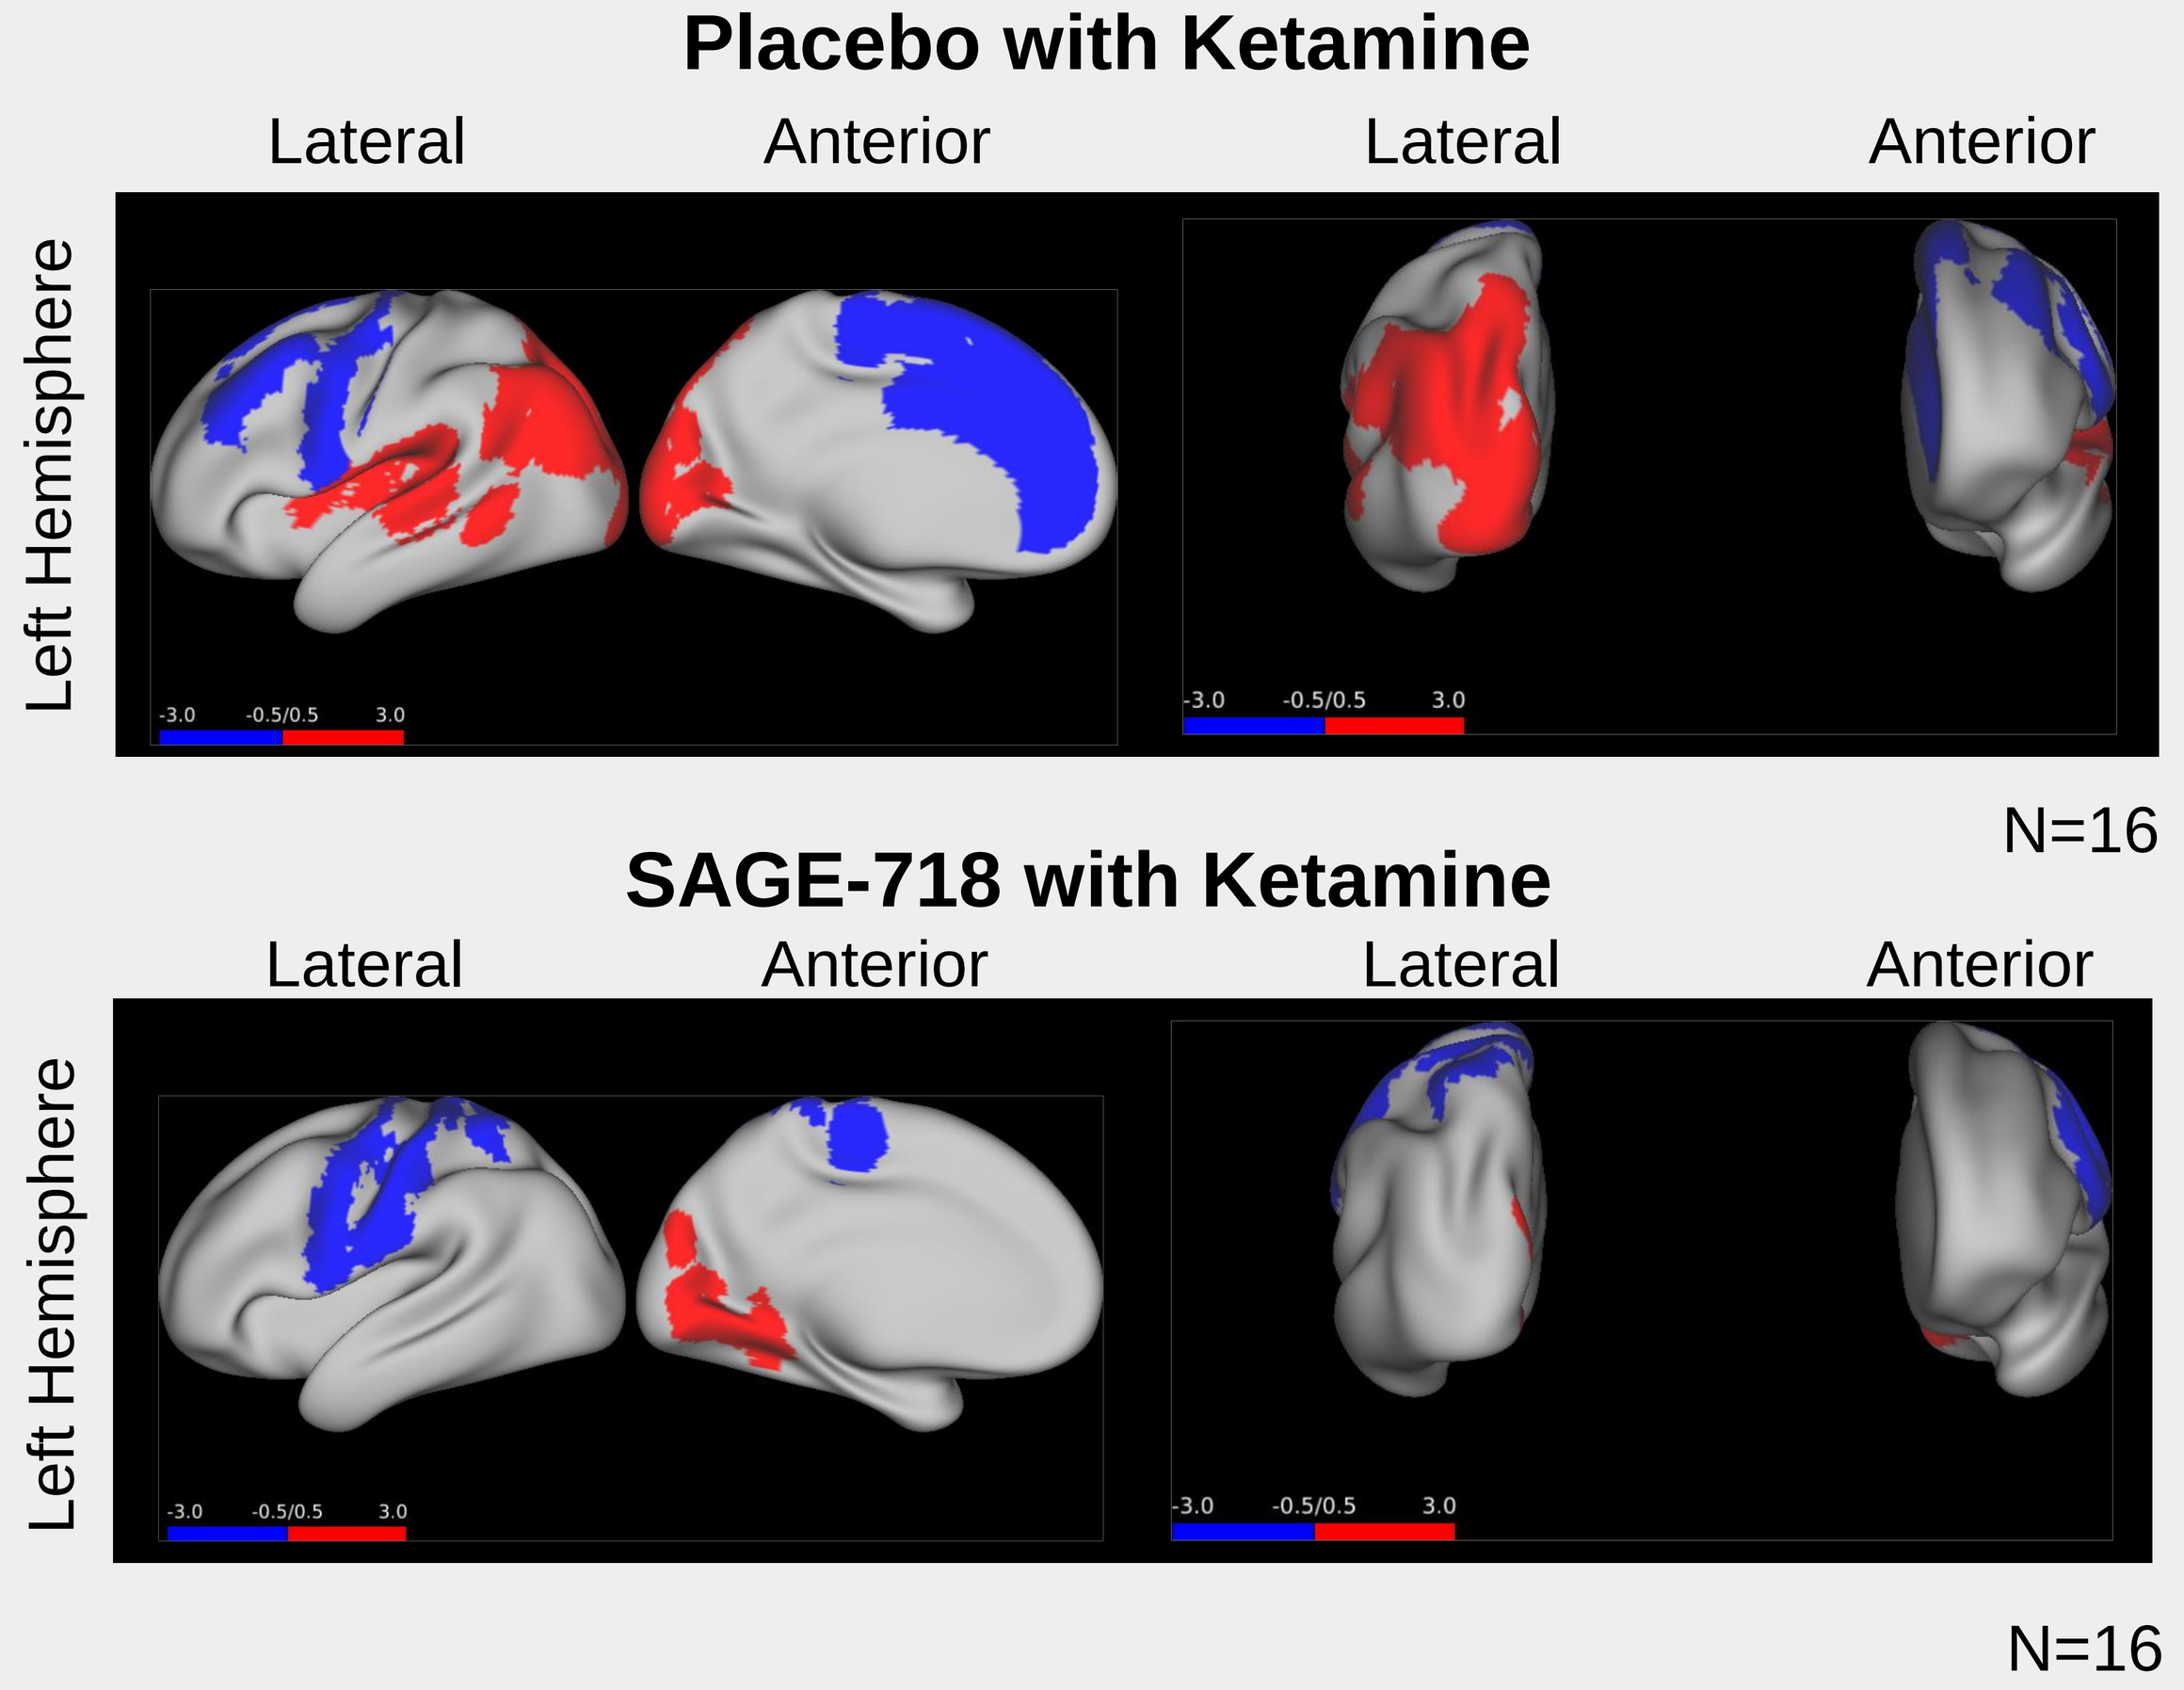
Placebo with Ketamine
Lateral
Anterior
Lateral
Anterior
Left Hemisphere
-3.0 -0.5/0.5 3.0
N=16
SAGE-718 with Ketamine
Lateral
Anterior
Lateral
Anterior
Left Hemisphere
-3.0 -0.5/0.5 3.0
N=16

- Ketamine-induced BOLD changes were broad, including robust increases (RED) in posterior brain regions and decreases (BLUE) in anterior brain regions, in particular the pre-specified dorsal anterior cingulate cortex.
- SAGE-718 attenuated ketamine-induced BOLD alterations independent of their directionality.

AUDITORY-EVOKED POTENTIAL RESPONSE TO STANDARD TONE DURING ODDBALL PARADIGM

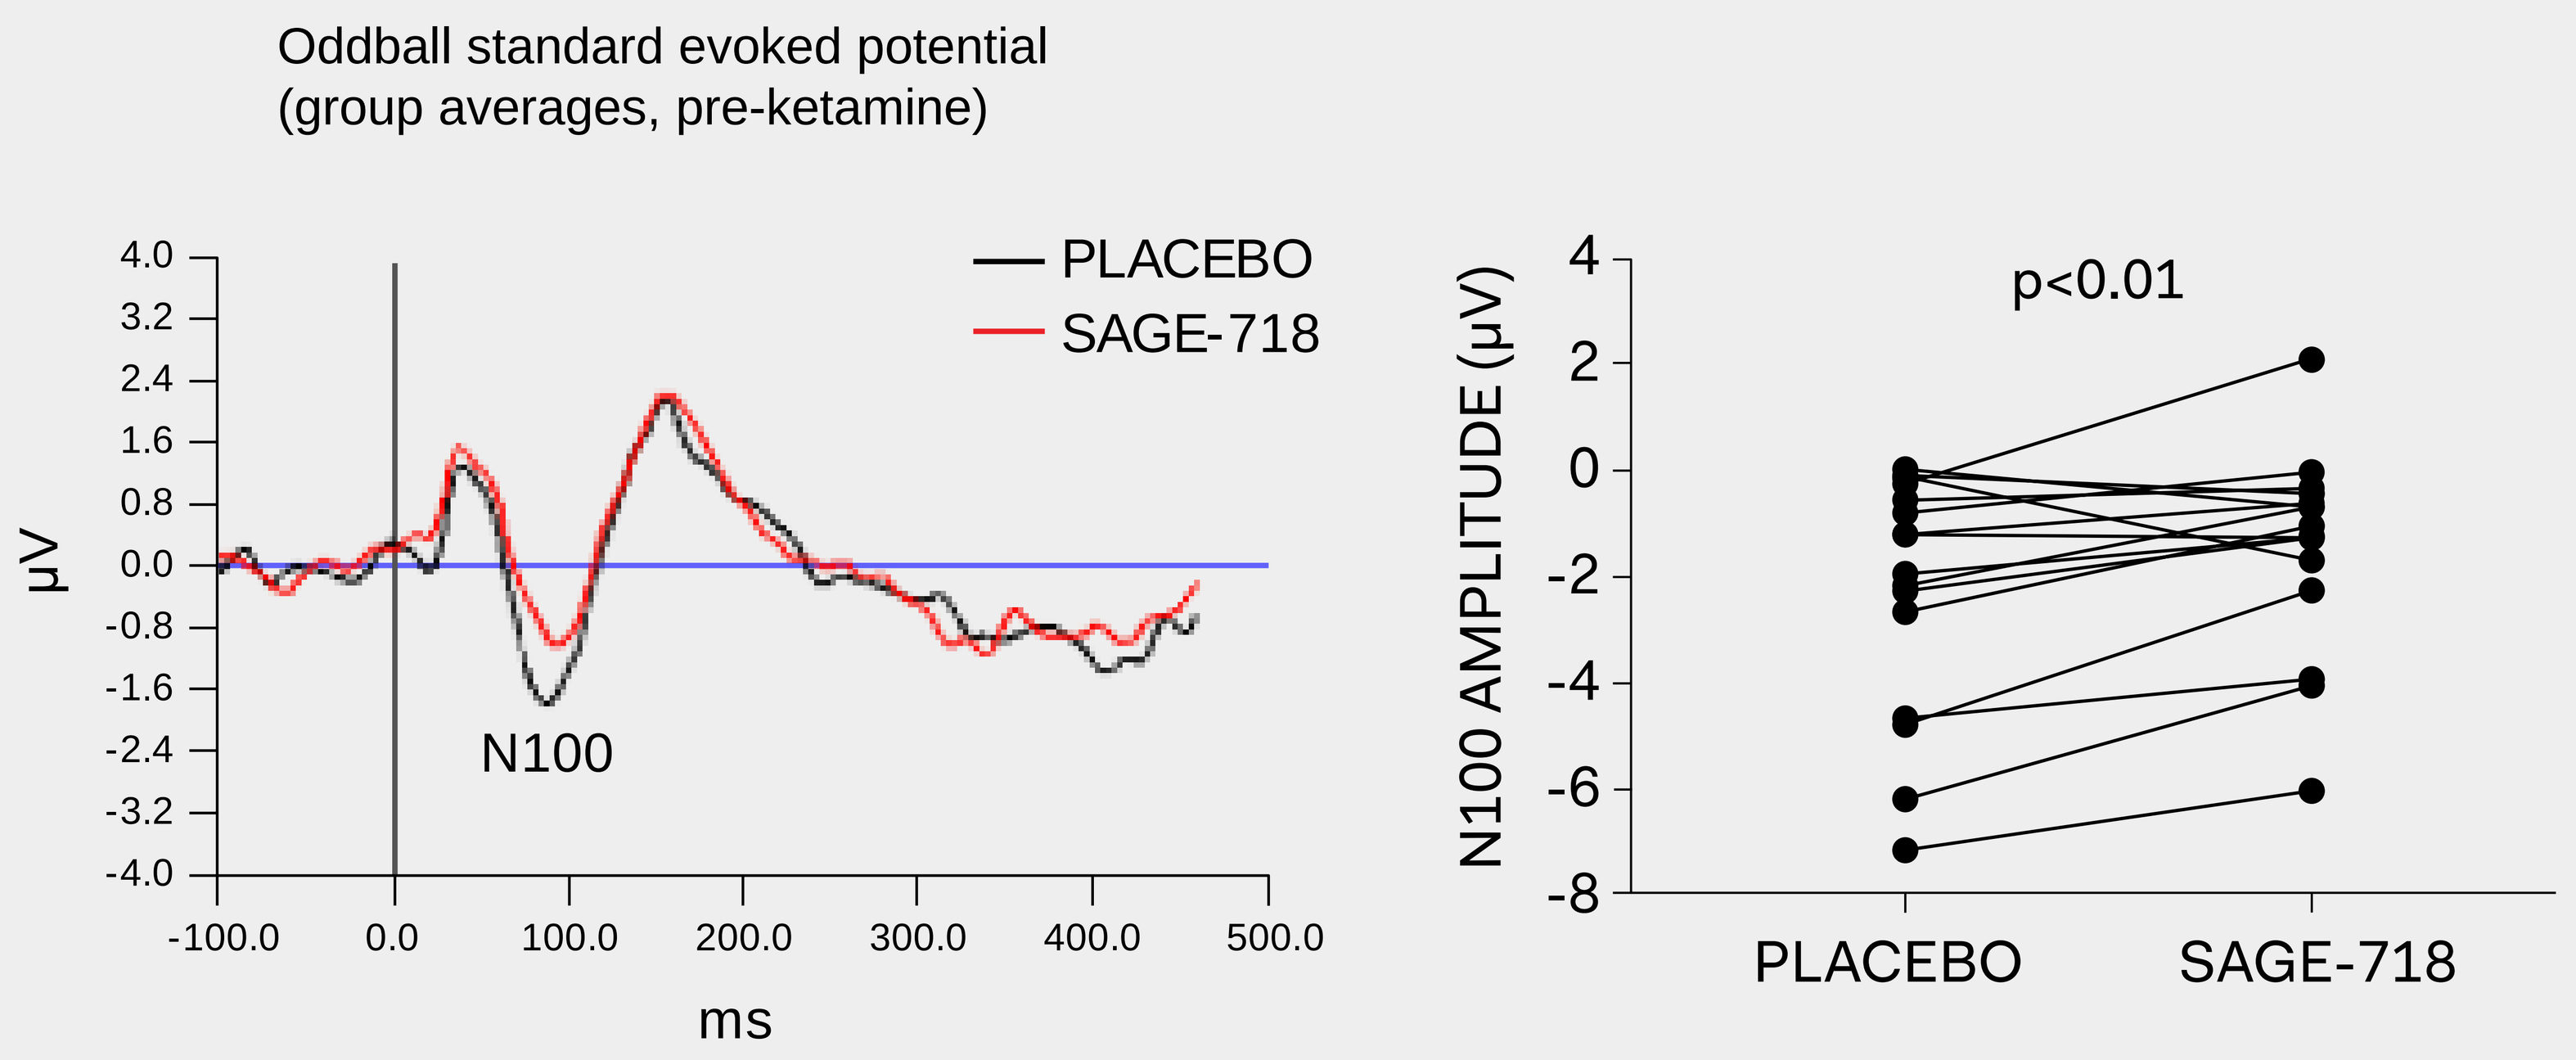
Oddball standard evoked potential
(group averages, pre-ketamine)
PLACEBO
SAGE-718
µV
4.0
3.2
2.4
1.6
0.8
0.0
-0.8
-1.6
-2.4
-3.2
-4.0
N100
-100.0
0.0
100.0
200.0
300.0
400.0
500.0
ms
N100 AMPLITUDE (µV)
4
2
0
-2
-4
-6
-8
p<0.01
PLACEBO
SAGE-718

- Plotting the standard evoked potential waveforms in a post hoc analysis highlighted a decreased N100 in the SAGE-718 group.
- Statistical analysis (paired t-test) confirmed a significant decrease (p<0.01) in the SAGE-718 condition.

Methods

718-EXM-101

- Healthy volunteers (n=2 sentinel dosing, unblinded, n=20 double-blinded, randomized 1:1 to SAGE-718 3 mg or placebo), ages 18-65, body weight ≥50 kg, and a body mass index of 18.0-30.0 kg/m² were enrolled in a two period, cross-over study (see study design figure).
- Exclusion criteria included a heart rate <50 or >100 beats per minute, systolic blood pressure <90 or >140 mmHg, or diastolic blood pressure <60 or >90 mmHg.
- On Day 1, subjects received an oral solution of study drug 7 hours before a 60-minute infusion of ketamine (0.25 mg/kg).
- After 10 days, subjects returned for the same protocol to receive the study drug not given in the first period, with follow-up through Day 25.
- EPs were assessed approximately 6 hours after the administration of study drug, and again approximately 30 minutes after ketamine infusion was initiated.
- The primary endpoint was a change in the following EPs: ketamine-induced changes in single-tone auditory evoked potential (AEP; N100-P200 amplitude), mismatch negativity, and auditory steady state response.
- Safety and tolerability were evaluated using the frequency and severity of reported adverse events (AEs) and standard clinical assessments.

718-EXM-102

- Healthy subjects (n=19) were enrolled with inclusion criteria similar to study 718-EXM-101, and were randomized 1:1 to SAGE-718 3 mg or placebo in a two period, cross-over study (see study design figure).
- Exclusion criteria were similar to study 718-EXM-101.
- On Day 1, subjects underwent fMRI approximately 6.5 hours after study drug dosing to obtain a baseline assessment.
- Two fMRI assessments were also completed at 15 and 45 minutes after ketamine infusion was initiated.
- fMRI-derived changes of the blood oxygenation level (BOLD) response and functional connectivity between regions of interest were recorded.
- Safety and tolerability were evaluated using the frequency and severity of reported adverse events (AEs) and standard clinical assessments.

REFERENCES

1. Kantrowitz JT and Javitt DC. Brain Res Bull. 2010 Sep 30; 83(3-4): 108-121.
2. Lin C, et al. Curr Pharm Des. 2014;20(32):5169-79.
3. Newcomer JW and Krystal JH. Hippocampus. 2001;11(5):529-42.
4. Murck H, et al. Biological Psychiatry. 2016; 59: 440-445.
5. Kozak R, et al. J Neurosci. 2014 Aug 6;34(32):10592-602.

SAGE THERAPEUTICS is a trademark of Sage Therapeutics, Inc. All other trademarks referenced herein are the property of their respective owners.

SUPPORT & DISCLOSURES

AK, JB, YL, SL, LK, BF, DN, IW, MQ, SJK, and JD are employees of Sage Therapeutics, Inc. with stock/stock options. This study was funded by Sage Therapeutics, Inc. This poster was assembled with assistance from Boston Strategic Partners.

American Academy of Neurology Annual Meeting
April 25 – May 1, 2020
Toronto, Canada

MRC-718-00109 05/20