Poster #: 003

Cognitive Performance After Repeated Administration of the NMDA Positive Allosteric Modulator SAGE-718 in Healthy Volunteers

Aaron Koenig, MD[1], Jason Berlin, MS[1], Yingchun Luo, PhD[1], Sigui Li, MS[1], Ludmila Kryzhanovskaya, MD, PhD[1], Brandon Farley, PhD[1], David Nguyen, PhD[1], Anne C. Smith, PhD[1], Irena Webster, MPH[1], Michael Quirk, PhD [1], Stephen J. Kanes, MD, PhD[1], James Doherty, PhD[1]

[1]Sage Therapeutics, Inc., Cambridge, MA, USA

Introduction

– N-methyl-D-aspartate receptors (NMDARs) are ionotropic glutamate receptors that play a key role in a host of cognitive and behavioral processes.[1-3]

– Based on the discovery of an endogenous NMDAR modulator, 24-(s)-hydroxycholesterol,[4] a research effort was initiated to discover novel NMDAR modulators.

– A library of investigational oxysterol-based NMDAR modulators with unique profiles has been built by Sage Therapeutics, the first of these being SAGE-718.

– Potential treatments for conditions characterized by NMDA hypofunction may involve enhancement of NMDAR-related neurotransmission by regulating synaptic strength, health, and plasticity.[1-3]

– In this exploratory analysis, following a ketamine infusion, the effects of 10-day repeated exposure to SAGE-718 on a core cognitive battery were investigated in healthy volunteers (NCT03844906).

Conclusions

– In this exploratory analysis, healthy volunteers dosed with SAGE-718 exhibited improvement in performance on tests of working memory and complex problem solving.

– Healthy volunteers receiving SAGE-718 exhibited improvement on tests of higher-order working memory (Two-Back Test) and complex problem solving (Groton Maze Test), which at times reached the threshold for statistical significance as compared with healthy volunteers receiving placebo.

– Healthy volunteers who received SAGE-718 did not perform better than placebo on tests of lower order functioning (psychomotor function, attention, simple working memory, and inhibition).

– The observed effects were not directly attributable to changes in reaction time or attention.

– SAGE-718 was generally well tolerated. There were no SAEs, severe TEAEs, or TEAEs leading to study withdrawal or discontinuation of oral study drug.

– These results suggest that SAGE-718 is potentially distinct from other cognitive-enhancing compounds and that further investigation of SAGE-718 is warranted in conditions characterized by states of relative NMDA hypofunction, particularly those manifesting with executive deficits.

Results

DEMOGRAPHICS AND BASELINE CHARACTERISTICS

	PBO (n=21)	SAGE-718 (n=19)
Age (years)	44.8 (12.2)	42.7 (12.0)
Weight (kg)	79.4 (13.0)	79.0 (11.6)
Height (cm)	173.9 (8.7)	173.3 (10.5)
BMI (kg/m²)	26.2 (3.0)	26.3 (2.8)
Gender		
Female	5 (23.8)	3 (15.8)
Male	16 (76.2)	16 (84.2)
Ethnicity		
Hispanic	7 (33.3)	6 (31.6)
Race		
Black/African-American	9 (42.9)	9 (47.4)
White	12 (57.1)	9 (47.4)
Other	0	1 (5.3)

Data are mean (SD) or n (%), from the safety set.

– There were no differences between SAGE-718 and placebo on ketamine-induced changes in electrophysiology parameters (primary endpoints).

COGNITIVE TESTS

TEST	Cognitive Domain and Instructions	Outcome Measure
Two Back Test[1]	Working Memory "Is the card the same as that shown 2 cards ago?"	Accuracy of performance*
Groton Maze Learning Test[2]	Executive Function "Find the hidden pathway."	Number of errors*
Continuous Paired Associative Learning[2]	Paired Associative Learning (Spatial Learning/Recall) "In what locations do these pictures belong?"	Total number of errors across 7 rounds in the test
Detection Test[2]	Psychomotor Function "Has the card turned over?"	Speed of performance*
Identification Test[2]	Attention "Is the card red?"	Speed of performance*
One Back Test[2]	Working Memory "Is the card the same as the previous card?"	Speed of performance*
STOP Test[2]	Inhibition (Executive Function) "Don't respond after a beep."	Reaction Time*

Test score interpretation: 1: Higher score = better performance; 2: Lower score = better performance
*Outcome measure is standardized change from baseline

EFFECTS ON COGNITIVE PERFORMANCE

TWO-BACK TEST (WORKING MEMORY)

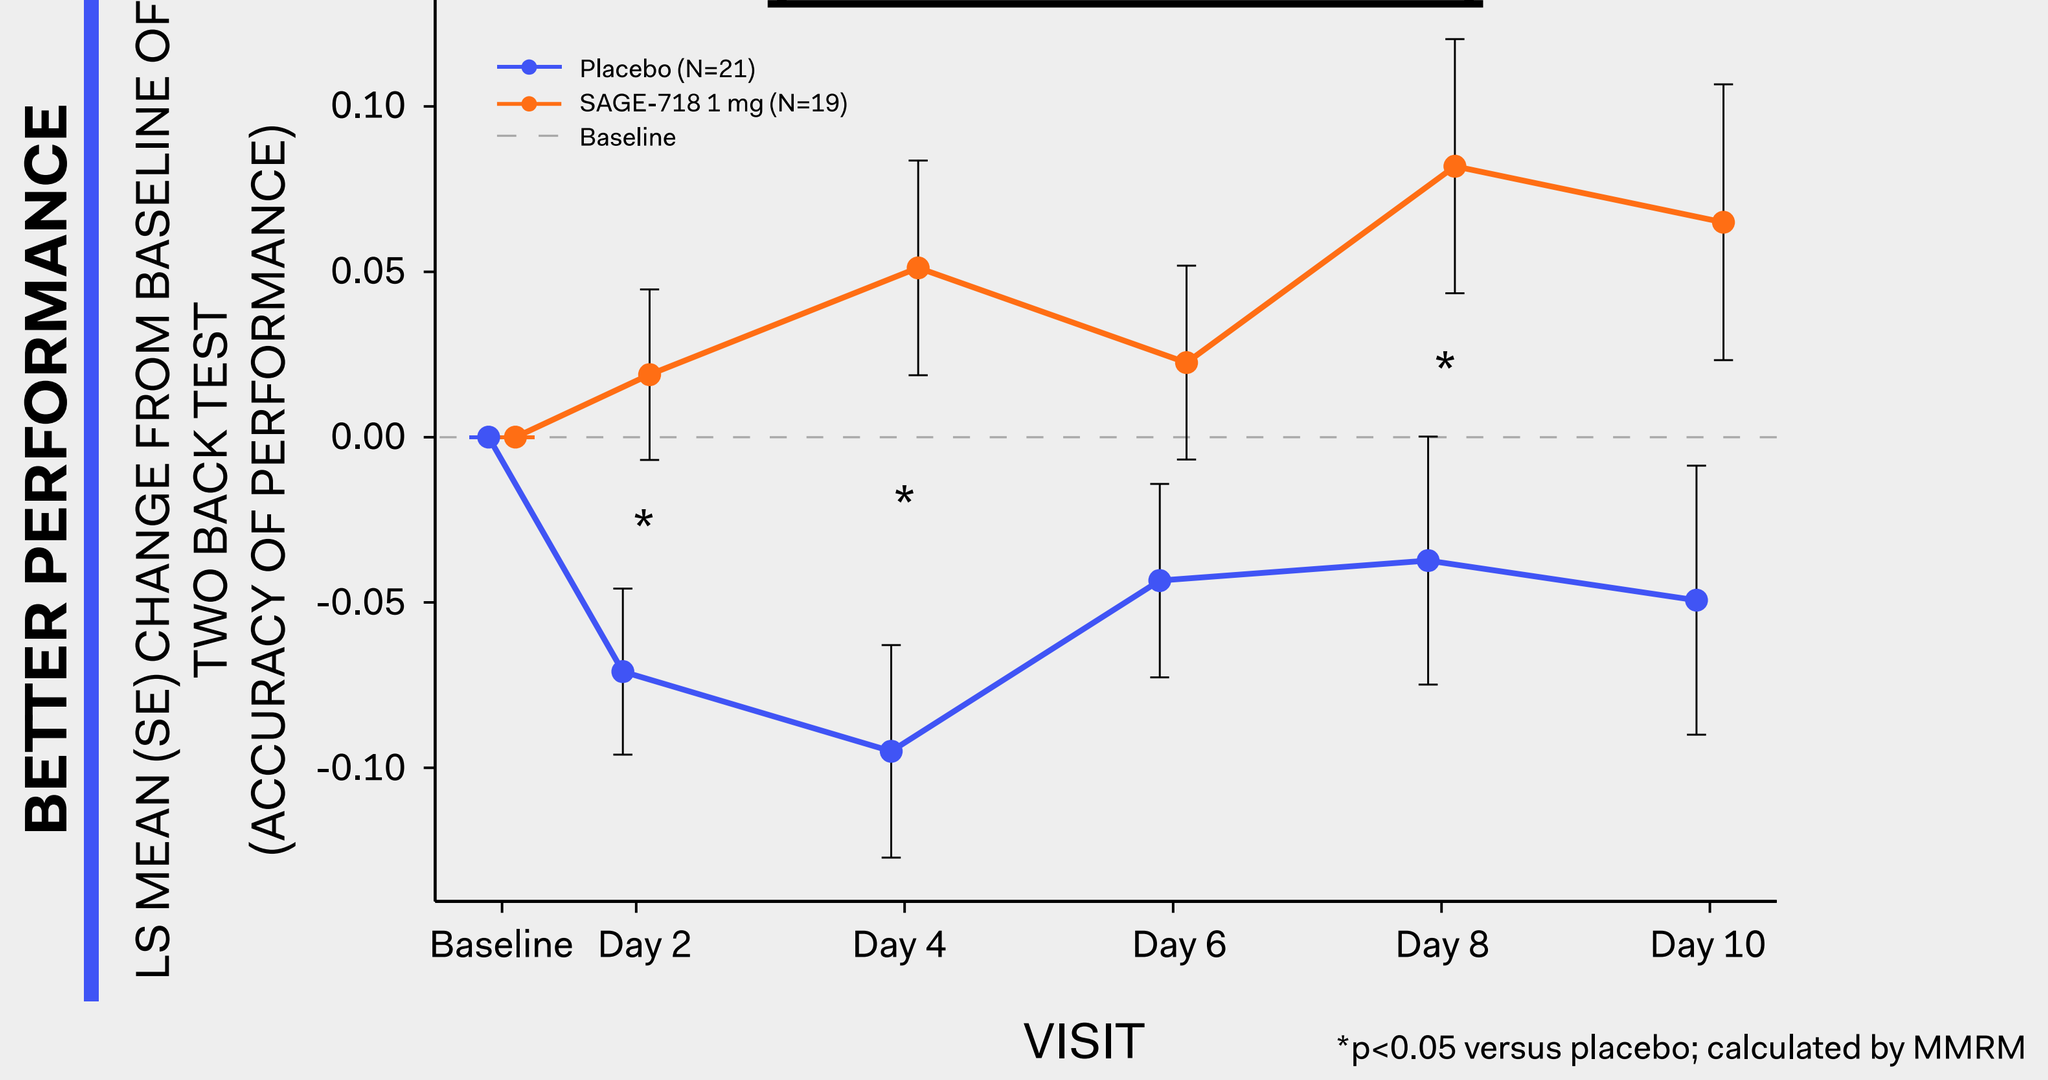
BETTER PERFORMANCE
LS MEAN (SE) CHANGE FROM BASELINE OF TWO BACK TEST (ACCURACY OF PERFORMANCE)
Placebo (N=21)
SAGE-718 1 mg (N=19)
Baseline
VISIT
*p<0.05 versus placebo; calculated by MMRM

GROTON MAZE LEARNING TEST (EXECUTIVE FUNCTION)

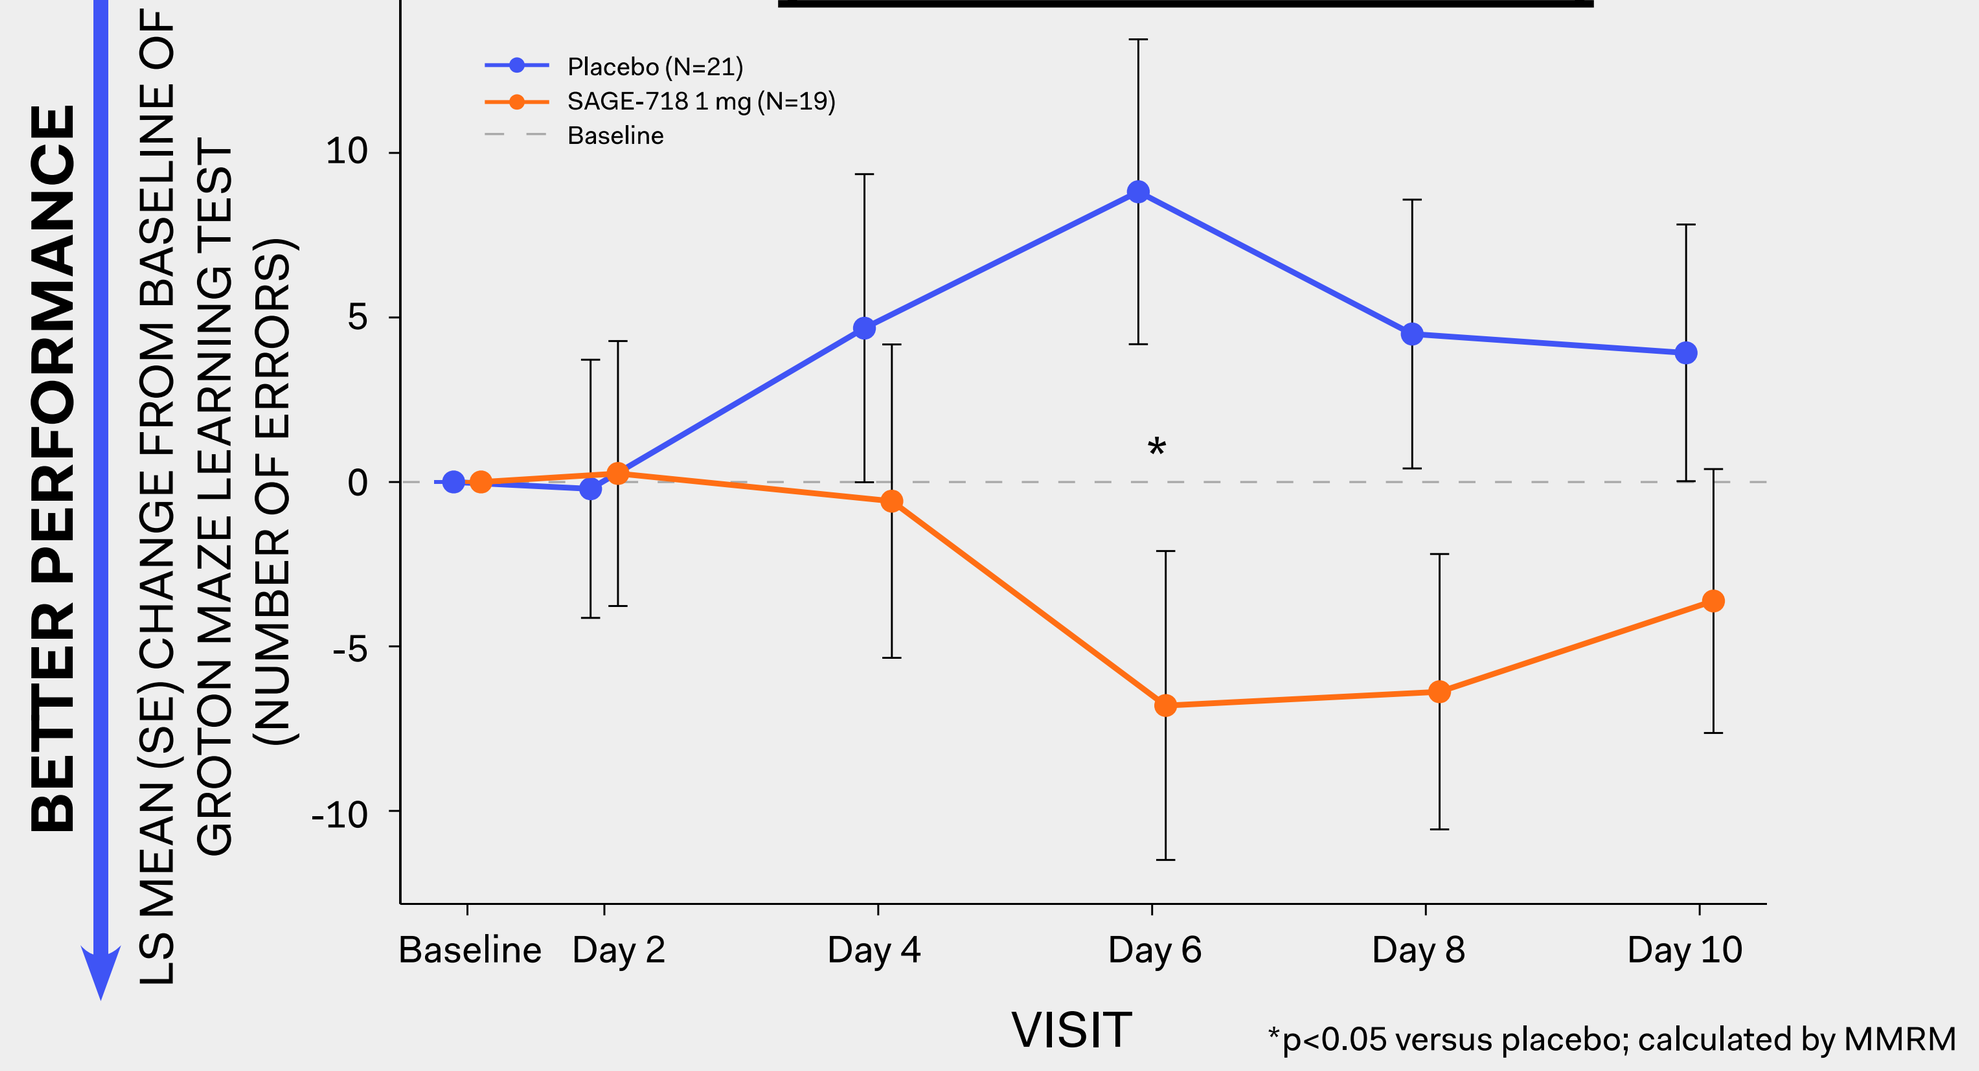
BETTER PERFORMANCE
LS MEAN (SE) CHANGE FROM BASELINE OF GROTON MAZE LEARNING TEST (NUMBER OF ERRORS)
Placebo (N=21)
SAGE-718 1 mg (N=19)
Baseline
VISIT
*p<0.05 versus placebo; calculated by MMRM

CONTINUOUS PAIRED ASSOCIATIVE LEARNING (SPATIAL LEARNING/RECALL)

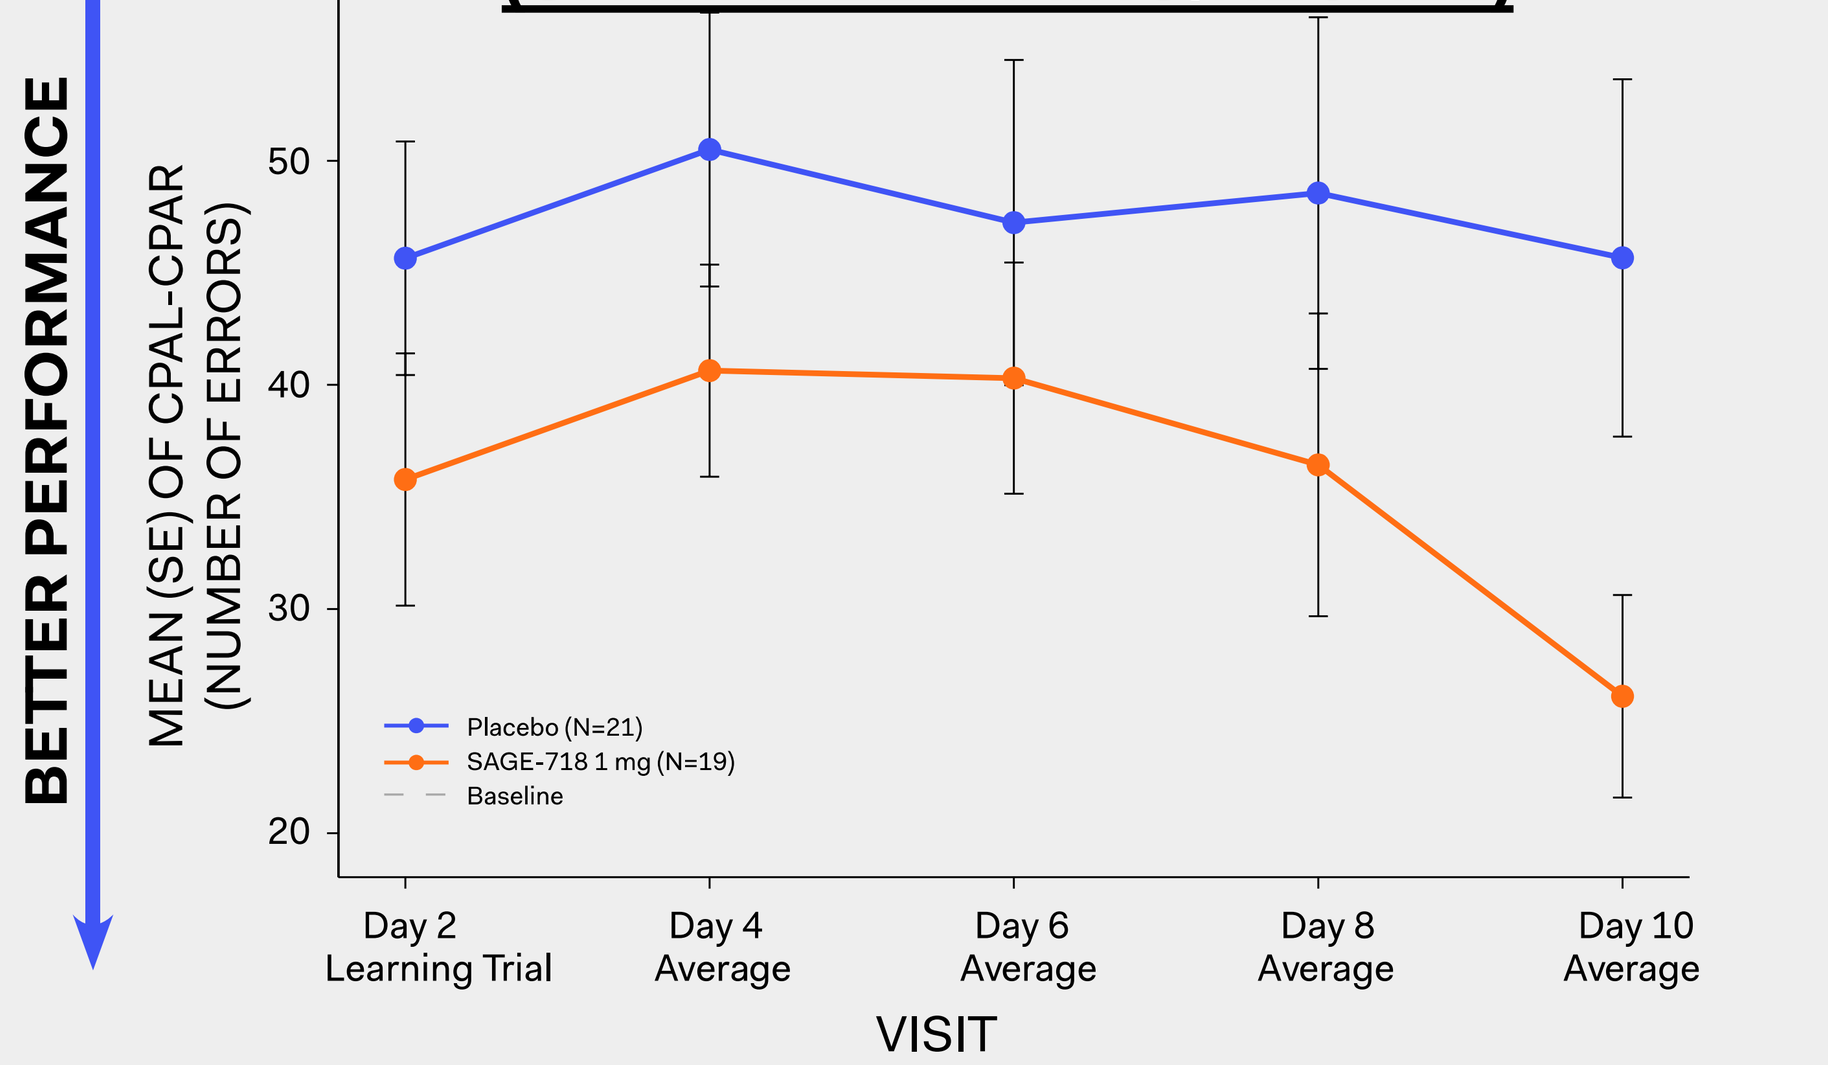
BETTER PERFORMANCE
MEAN (SE) OF CPAL-CPAR (NUMBER OF ERRORS)
Placebo (N=21)
SAGE-718 1 mg (N=19)
Baseline
Day 2 Learning Trial
Day 4 Average
Day 6 Average
Day 8 Average
Day 10 Average
VISIT

– Healthy volunteers in the SAGE-718 group demonstrated statistically significant improvements vs placebo on Day 2 (0.019 vs. -0.071, 0.090 diff, 95% confidence interval [CI] 0.017 to 0.163, p=0.018), Day 4 (0.051 vs. -0.095, 0.146 diff, 95% CI 0.053 to 0.239, p=0.003), and Day 8 (0.082 vs. -0.037, 0.119 diff, 95% CI 0.010 to 0.228 p=0.033).

– Healthy volunteers in the SAGE-718 group demonstrated statistically significant improvement vs placebo on Day 6 (-6.797 vs. 8.821, -15.62 diff, 95% CI -29.06 to -2.170, p=0.024).

– Healthy volunteers in the SAGE-718 group demonstrated numerically better performance and made fewer errors in paired associative learning than placebo, from Days 2 through 10.

DETECTION TEST (PSYCHOMOTOR FUNCTION)

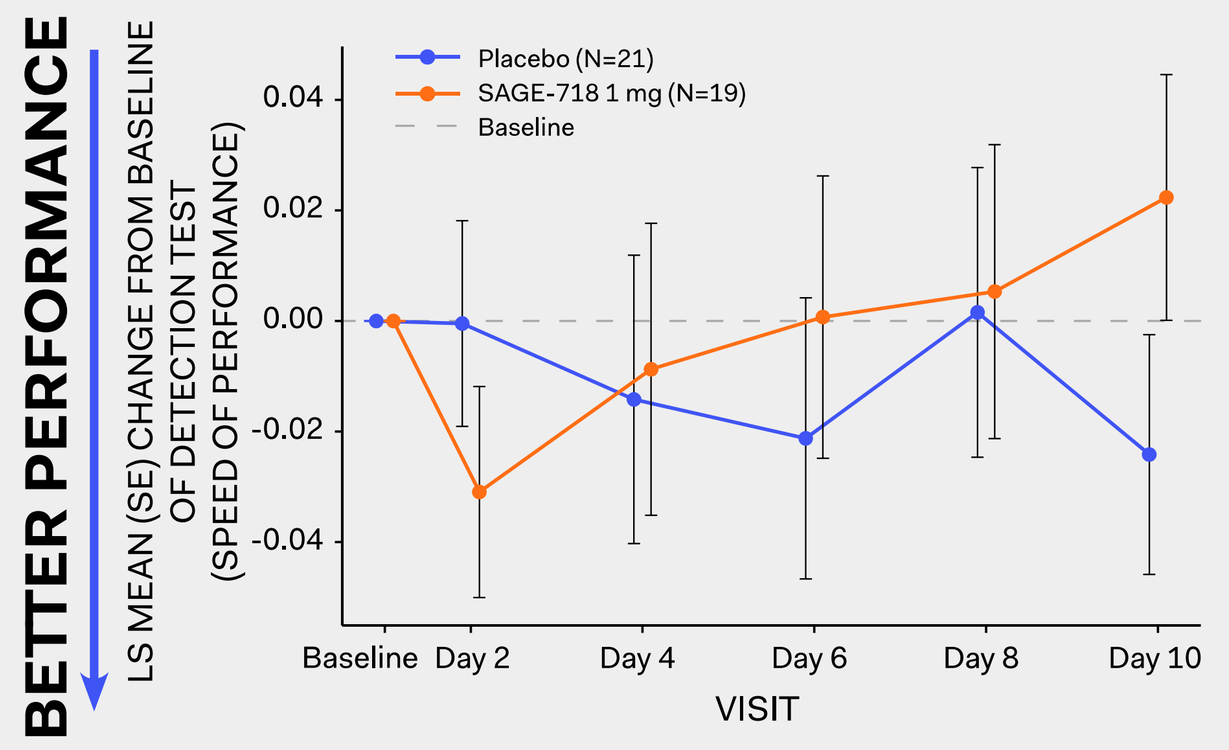
BETTER PERFORMANCE
LS MEAN (SE) CHANGE FROM BASELINE OF DETECTION TEST (SPEED OF PERFORMANCE)
Placebo (N=21)
SAGE-718 1 mg (N=19)
Baseline
VISIT

IDENTIFICATION TEST (ATTENTION)

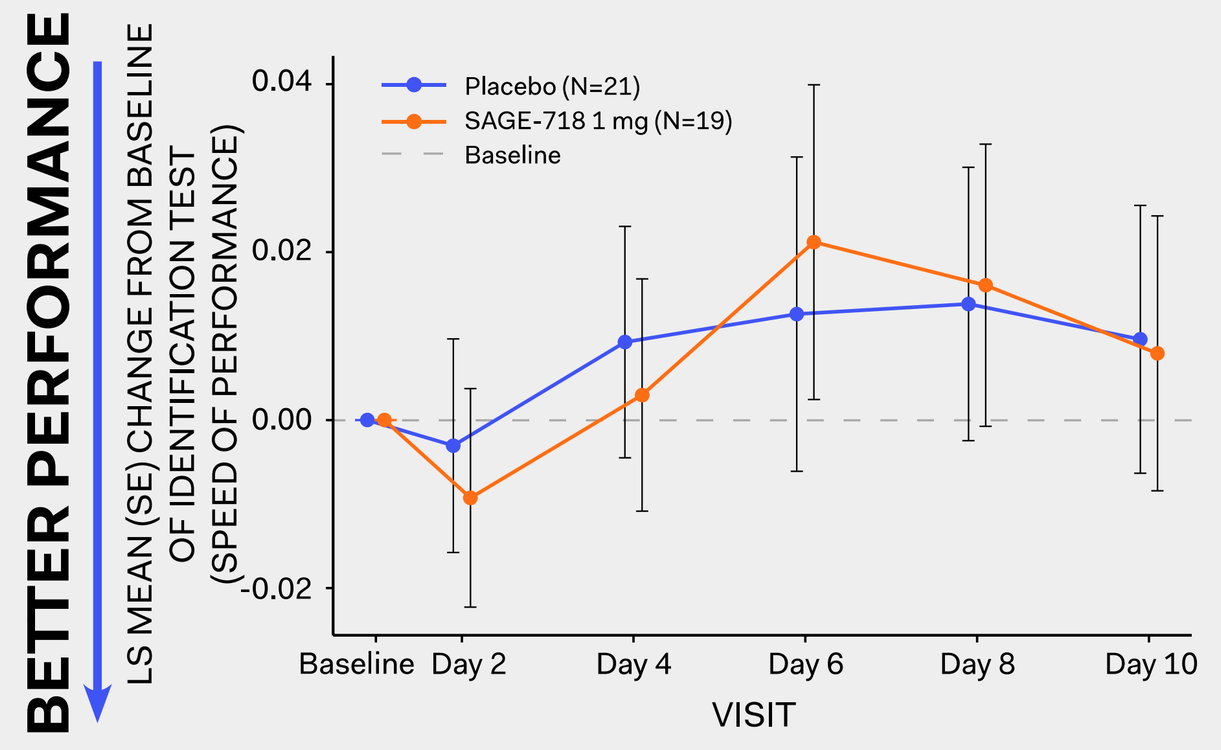
BETTER PERFORMANCE
LS MEAN (SE) CHANGE FROM BASELINE OF IDENTIFICATION TEST (SPEED OF PERFORMANCE)
Placebo (N=21)
SAGE-718 1 mg (N=19)
Baseline
VISIT

ONE-BACK TEST (SIMPLE WORKING MEMORY)

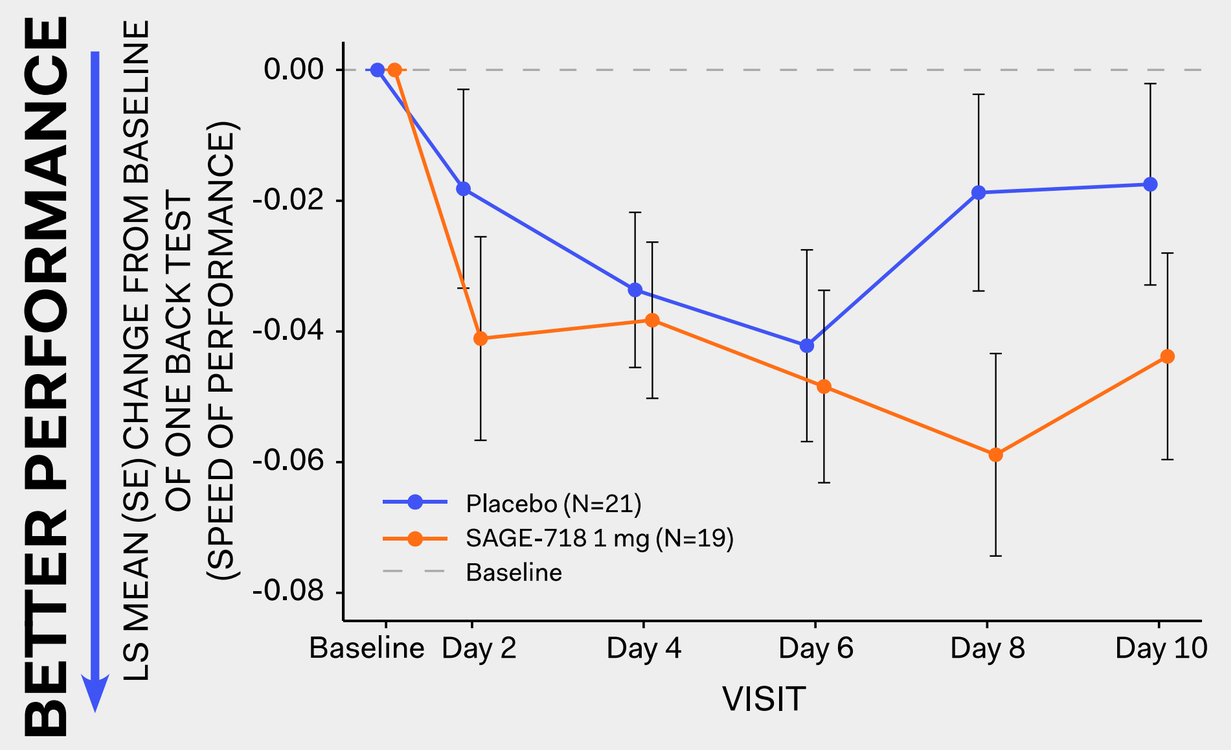
BETTER PERFORMANCE
LS MEAN (SE) CHANGE FROM BASELINE OF ONE BACK TEST (SPEED OF PERFORMANCE)
Placebo (N=21)
SAGE-718 1 mg (N=19)
Baseline
VISIT

STOP TEST [INHIBITION (EXECUTIVE FUNCTION)]

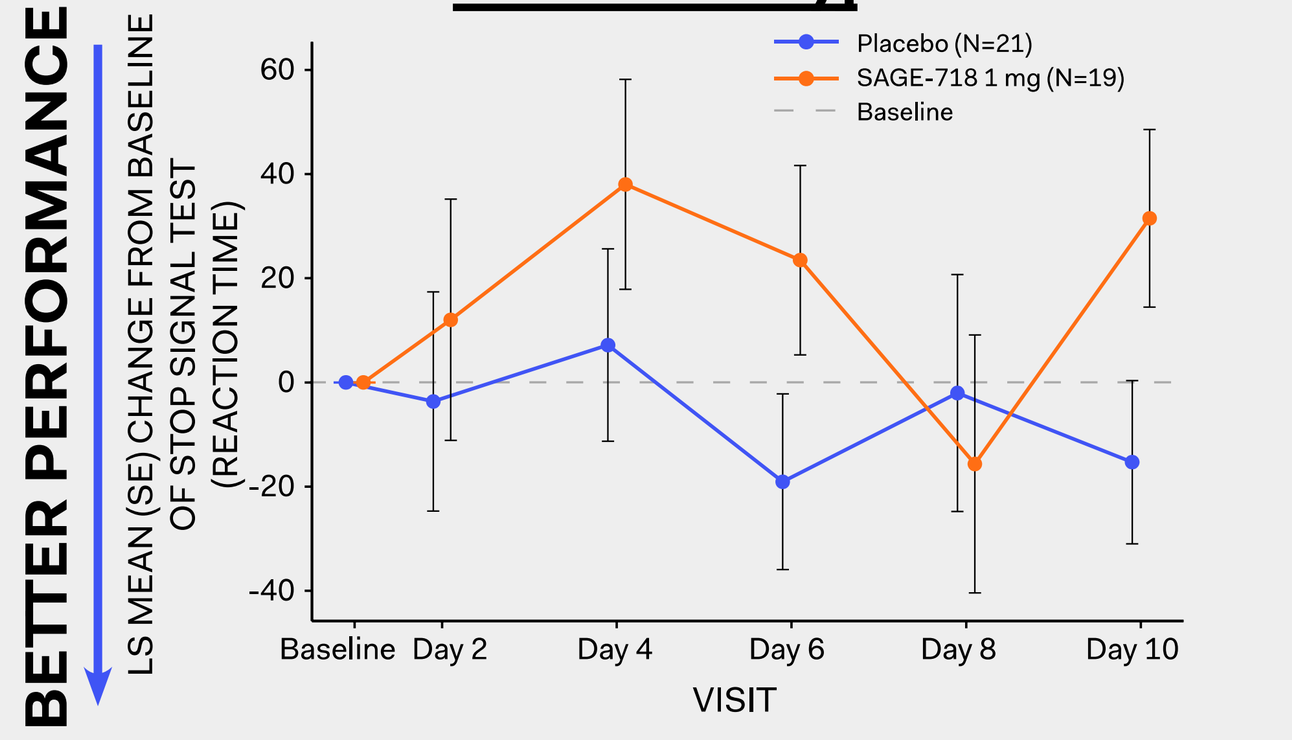
BETTER PERFORMANCE
LS MEAN (SE) CHANGE FROM BASELINE OF STOP SIGNAL TEST (REACTION TIME)
Placebo (N=21)
SAGE-718 1 mg (N=19)
Baseline
VISIT

– Additional measures of cognition (Detection, Identification, One-Back, Stop Test) demonstrated comparable performance in the SAGE-718 group and placebo.

RELATIONSHIP BETWEEN COGNITIVE PERFORMANCE AND CMAX AT DAY 10

TWO BACK TEST

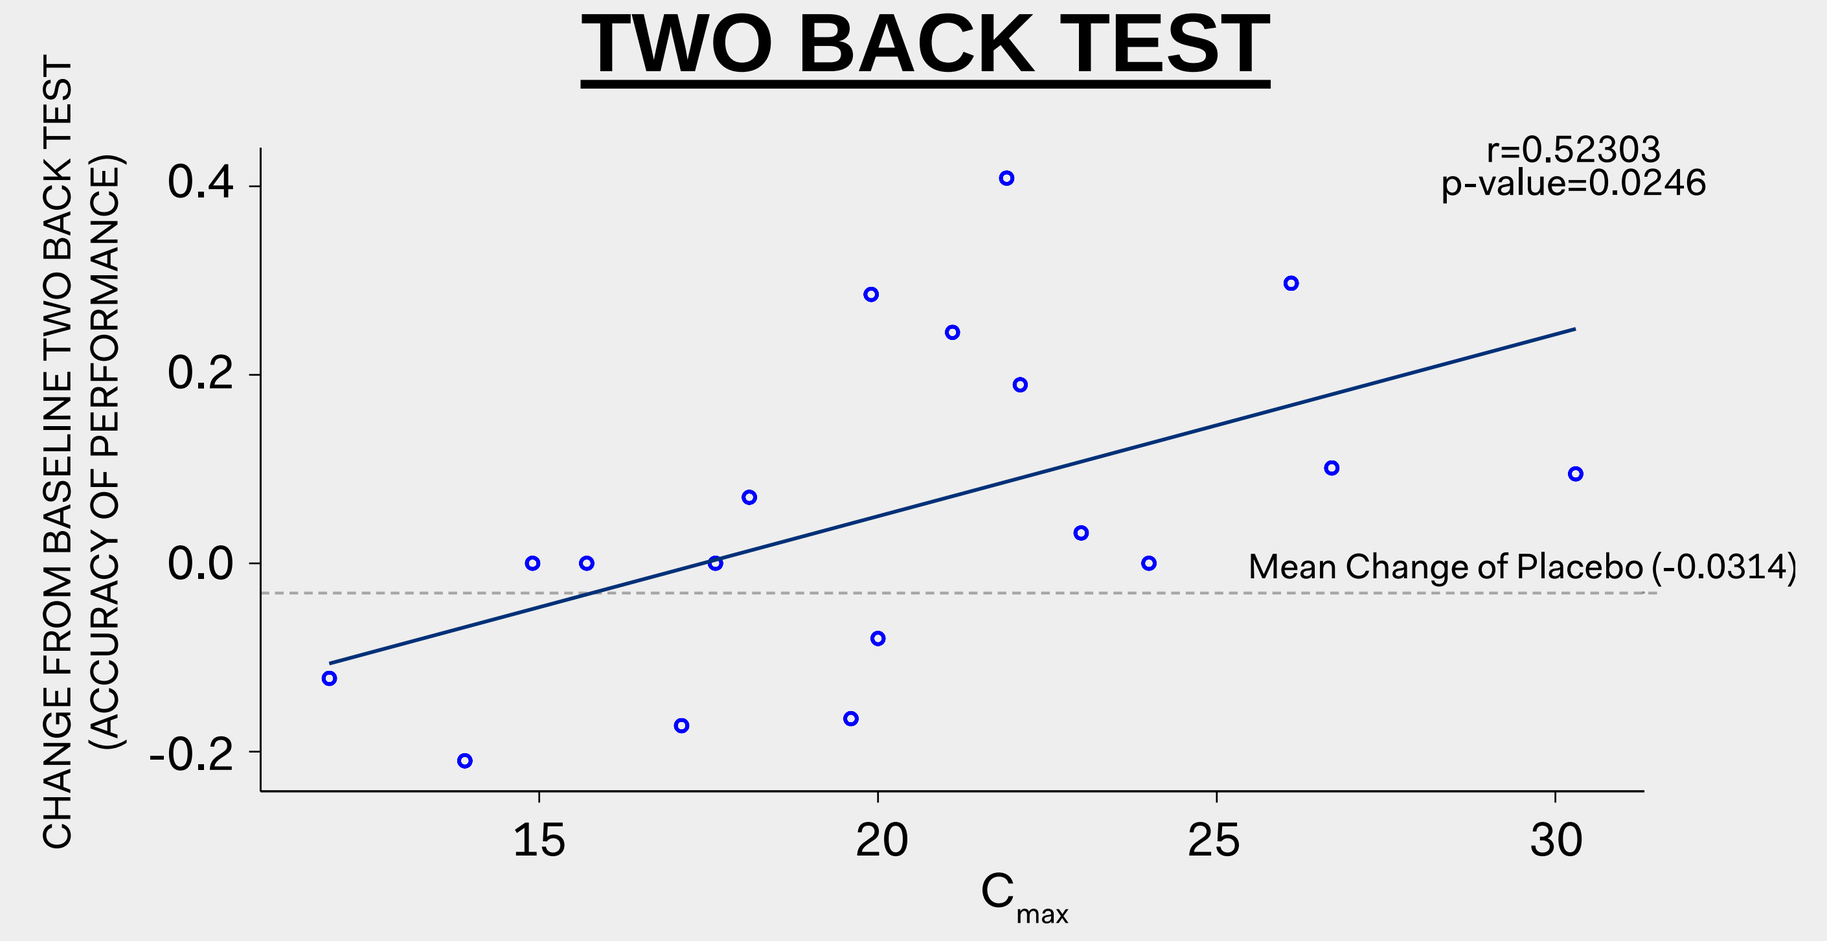
r=0.52303
p-value=0.0246
Mean Change of Placebo (-0.0314)
CHANGE FROM BASELINE TWO BACK TEST (ACCURACY OF PERFORMANCE)
Cmax

– Two Back Test performance showed a positive correlation with SAGE-718 plasma concentration at Day 10 (r=0.52303, p=0.0246).

GROTON MAZE LEARNNG TEST

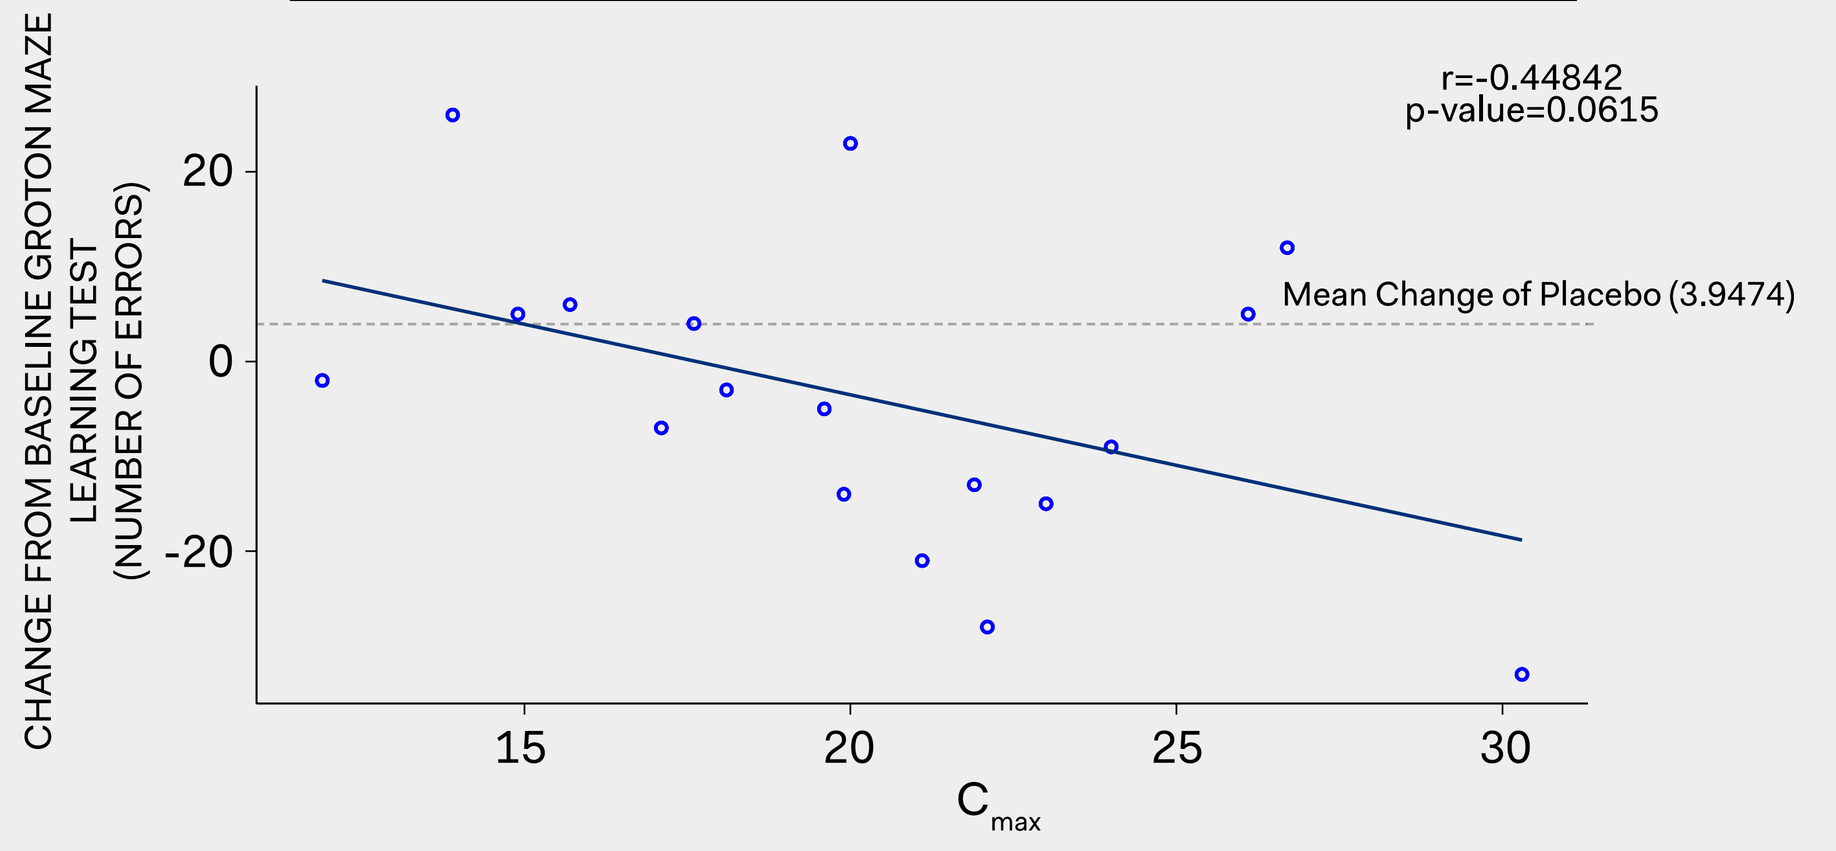
r=-0.44842
p-value=0.0615
Mean Change of Placebo (3.9474)
CHANGE FROM BASELINE GROTON MAZE LEARNING TEST (NUMBER OF ERRORS)
Cmax

– A negative correlation was observed between SAGE-718 plasma levels and number of errors in the Groton Maze Learning test at Day 10 (r=0.44842, p=0.0615).

TREATMENT EMERGENT ADVERSE EVENTS (TEAEs)

	PBO (n=21)	SAGE-718 (n=19)
Subjects with any TEAE	7 (33.3%)	6 (31.6%)
Gastrointestinal	2 (9.5%)	1 (5.3%)
Infectious	2 (9.5%)	0 (0%)
Laboratory	0 (0%)	2 (10.5%)
Nervous system	5 (23.8%)*	3 (15.8%)†
Psychiatric	2 (9.5%)	1 (5.3%)

Data are n (%) from the safety set.
*PBO nervous system TEAEs by preferred term: headache n=2 (9.5%), head discomfort n=1 (4.8%), myoclonus n=1 (4.8%), paraesthesia n=1 (4.8%).
†SAGE-718 nervous system TEAEs by preferred term: dizziness n=1 (5.3%), headache n=1 (5.3%), head discomfort n=1 (5.3%), balance disorder n=1 (5.3%), restless legs syndrome n=1 (5.3%).

– Most treatment-emergent adverse events were mild in severity; no serious or severe AEs were reported.

– No TEAEs resulted in study drug discontinuation or dose reduction.

– No clinically relevant mean changes from baseline were observed in clinical laboratory parameters, vital signs, ECGs, or safety EEG results.

Methods

– Healthy volunteers, ages 18-65, body weight ≥50 kg, and a body mass index of 18.0-30.0 kg/m² who were willing to comply with all study procedures and restrictions (n=40) were enrolled and randomized (1:1) to either multiple oral doses of SAGE-718 plus ketamine (SAGE-718) or placebo plus ketamine (PBO).

– A 60-minute IV infusion of 0.25 mg/kg of ketamine was administered on Days 1 and 11, to investigate other modalities of target engagement.

– On Day 2, healthy volunteers received double-blind, first oral doses of either SAGE-718 1.0 mg or placebo, with daily dosing through Day 11.

– Cognitive testing was performed on Days 2, 4, 6, 8 and 10 when no ketamine was being administered.

– Exclusion criteria included clinically significant findings during physical examination, heart rate <50 or >100 beats per minute, systolic blood pressure <90 or >140 mmHg, or diastolic blood pressure <60 or >90 mmHg.

– Plasma samples were used to calculate C_{max} as well as other pharmacokinetic (PK) parameters.

– Primary endpoints were related to auditory evoked potential parameters.

– Exploratory endpoints of cognitive effects were evaluated by a battery of tests (see Table).

– A mixed effect model for repeated measurements was applied, with change from baseline in each cognitive test score as the response variable, and treatment, visit, and visit-by-treatment interaction as fixed effects, with baseline as a covariate, and measurements within the same subject as repeated measures.

– Safety and tolerability were evaluated using the frequency and severity of reported adverse events (AEs) and standard clinical assessments.

REFERENCES
1. Kantrowitz JT and Javitt DC. Brain Res Bull. 2010 Sep 30; 83(3-4): 108-121.
2. Lin C, et al. Curr Pharm Des. 2014;20(32):5169-79.
3. Newcomer JW and Krystal JH. Hippocampus. 2001;11(5):529-42.
4. Paul SM, et al. J Neurosci. 2013 Oct 30;33(44):17290-300.

SAGE THERAPEUTICS is a trademark of Sage Therapeutics, Inc. All other trademarks referenced herein are the property of their respective owners.

SUPPORT & DISCLOSURES
AK, JB, YL, SL, LK, BF, DN, AS, IW, MQ, SJK, and JD are employees of Sage Therapeutics, Inc. with stock/stock options. This study was funded by Sage Therapeutics, Inc. This poster was assembled with assistance from Boston Strategic Partners.
American Academy of Neurology Annual Meeting
April 25 – May 1, 2020
Toronto, Canada
MRC-718-00110 05/20