Cognitive deficits in Huntington's Disease and altered 24(S)-hydroxycholesterol

Michael Lewis, PhD[1], Jing Dai, PhD[1], Amrita Mohan, PhD[2], Sarah Tabrizi, PhD[3], James Doherty, PhD[1], Albert J Robichaud, PhD[1], Michael Quirk, PhD[1]

[1]Sage Therapeutics, Inc., Cambridge, MA, USA
[2]CHDI Foundation, New York, NY, USA
[3]UCL Institute of Neurology, University College London, London, UK

Introduction

- Nearly 25% of total body cholesterol resides in the central nervous system (CNS), where it is the principal component of myelin.[1]
- Lack of blood brain barrier permeability requires the conversion of cholesterol to 24(S)-hydroxycholesterol (HC) by the brain specific enzyme CYP46A1.[2,3]
- 24(S)-HC is a potent endogenous positive allosteric modulator (PAM) of the N-methyl-D-aspartate (NMDA) receptor.[4]
- 24(S)-HC is reduced in both plasma and brain tissue across Huntington disease (HD) stages.[5,6]
- Importantly, SAGE-718, an investigational, proprietary NMDA positive allosteric modulator, previously demonstrated an improvement in cognitive performance in a Phase 1 healthy volunteer study (NCT03844906).[7]
- We examined correlations between plasma levels of 24(S)-HC and cognitive performance across the years of TRACK-HD, a longitudinal observational study of biological and clinical manifestations of HD.[8]

Conclusions

- Our data support a role for 24(S)-HC in cognitive processes in HD and suggest that NMDA hypofunction may contribute to cognitive impairment in HD.
- The associations do not appear to be driven solely by neurodegenerative processes, and the associations are specific to 24(S)-HC (No associations were observed with 25-HC or 27-HC; data not shown).
- SAGE-718, an investigational NMDA positive allosteric modulator, is currently in clinical development for HD (Phase 1).

Results

TRACK-HD: A MULTI-CENTER PROSPECTIVE LONGITUDINAL OBSERVATIONAL STUDY[8]

Primary Goal: Determine biological and clinical measures sensitive to change over the course of pre-manifest and early HD stages.

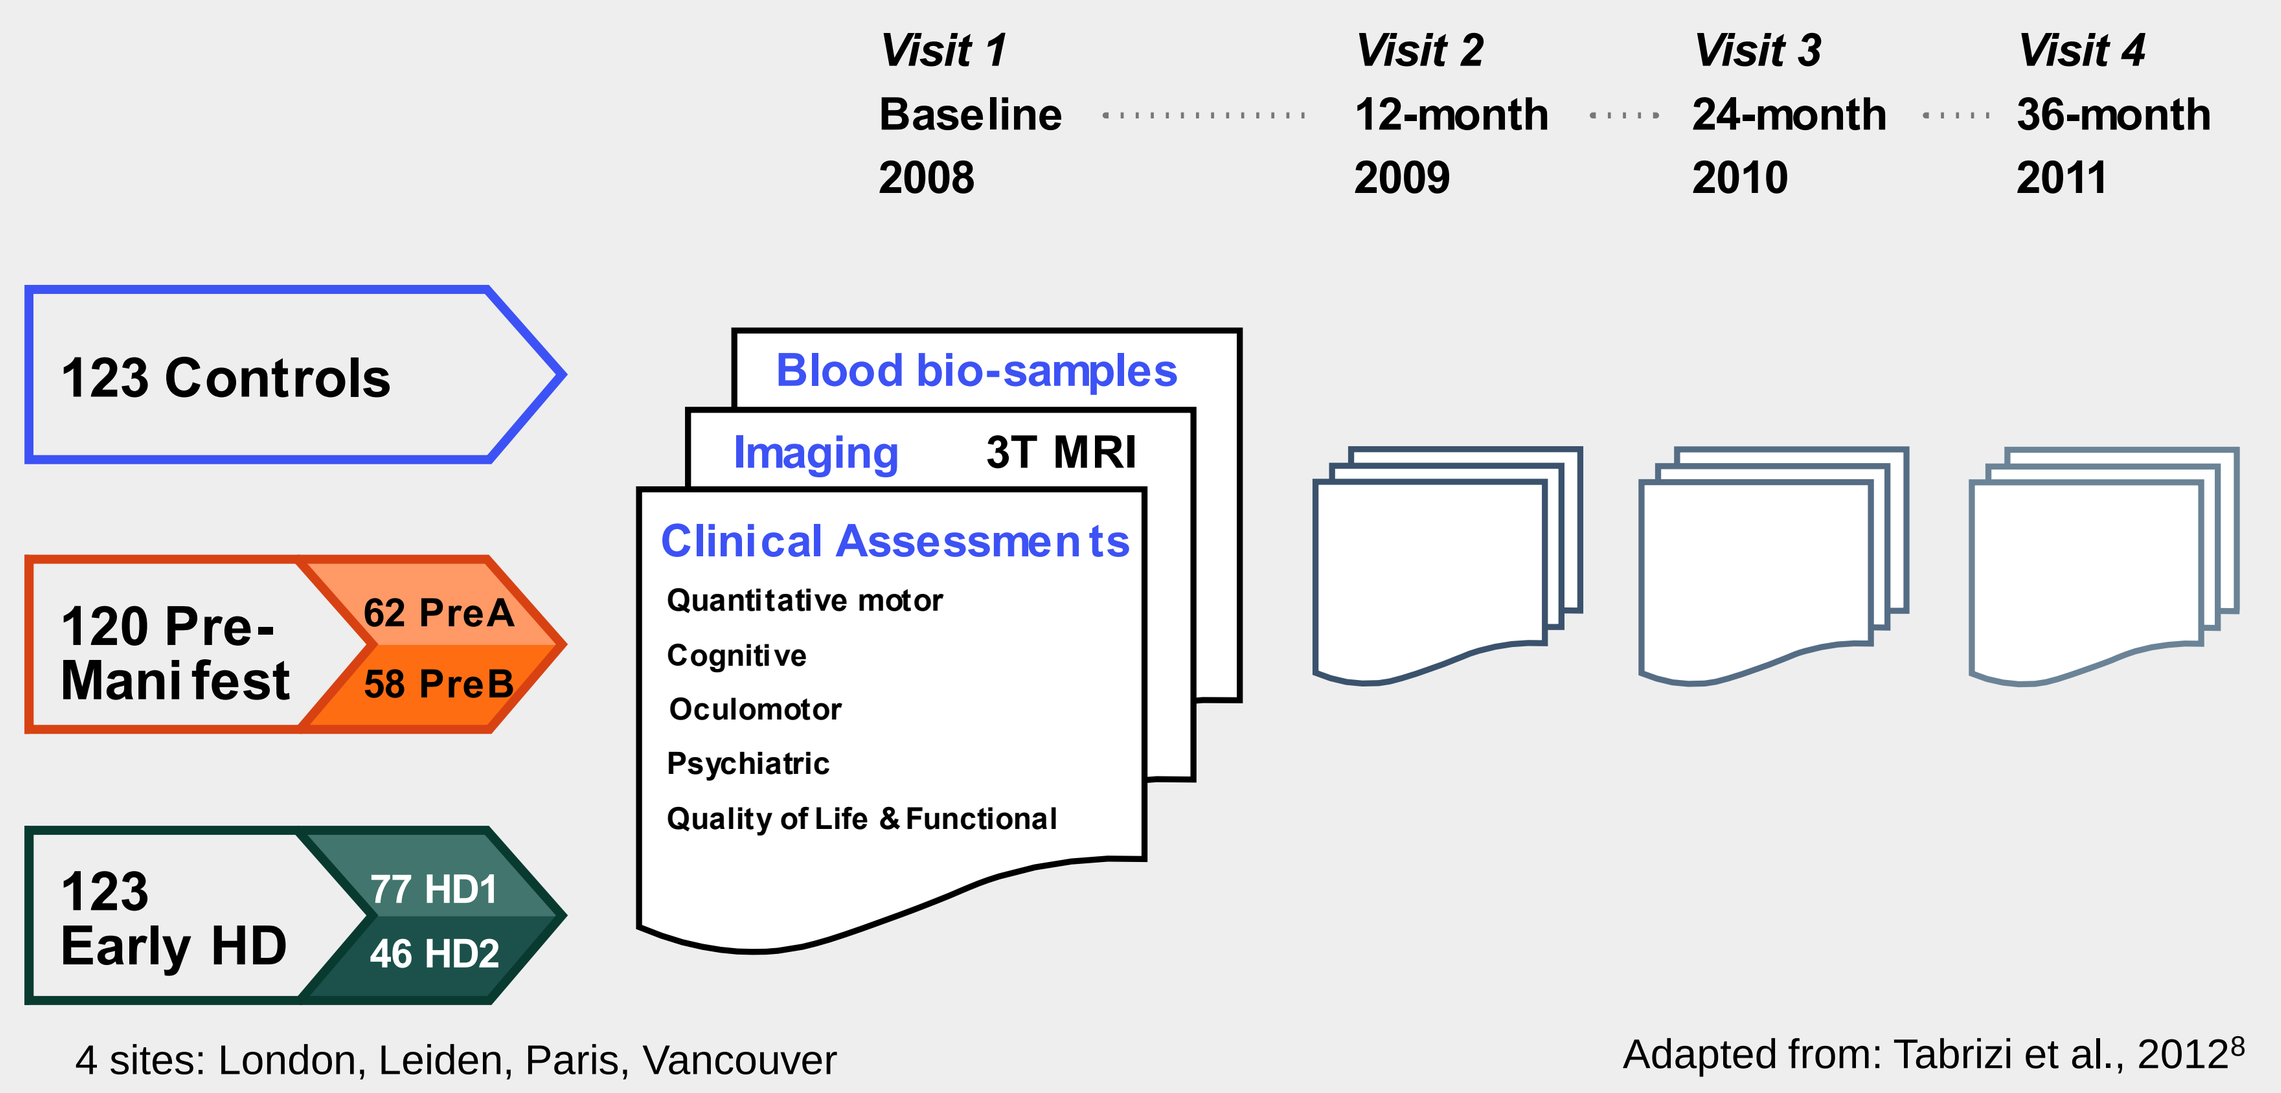
Visit 1 Baseline 2008
Visit 2 12-month 2009
Visit 3 24-month 2010
Visit 4 36-month 2011
123 Controls
120 Pre-Manifest
62 PreA
58 PreB
123 Early HD
77 HD1
46 HD2
Blood bio-samples
Imaging 3T MRI
Clinical Assessments
Quantitative motor
Cognitive
Oculomotor
Psychiatric
Quality of Life & Functional
4 sites: London, Leiden, Paris, Vancouver
Adapted from: Tabrizi et al., 2012[8]

- TRACK-HD was a longitudinal study of biological and clinical manifestations of HD.
- We collaborated with Cure Huntington's Disease Initiative (CHDI) to determine plasma levels of 24(S)-HC and correlate levels with performance on clinically relevant endpoints.

24(S)-HC CORRELATED WITH COGNITION IN TRACK-HD

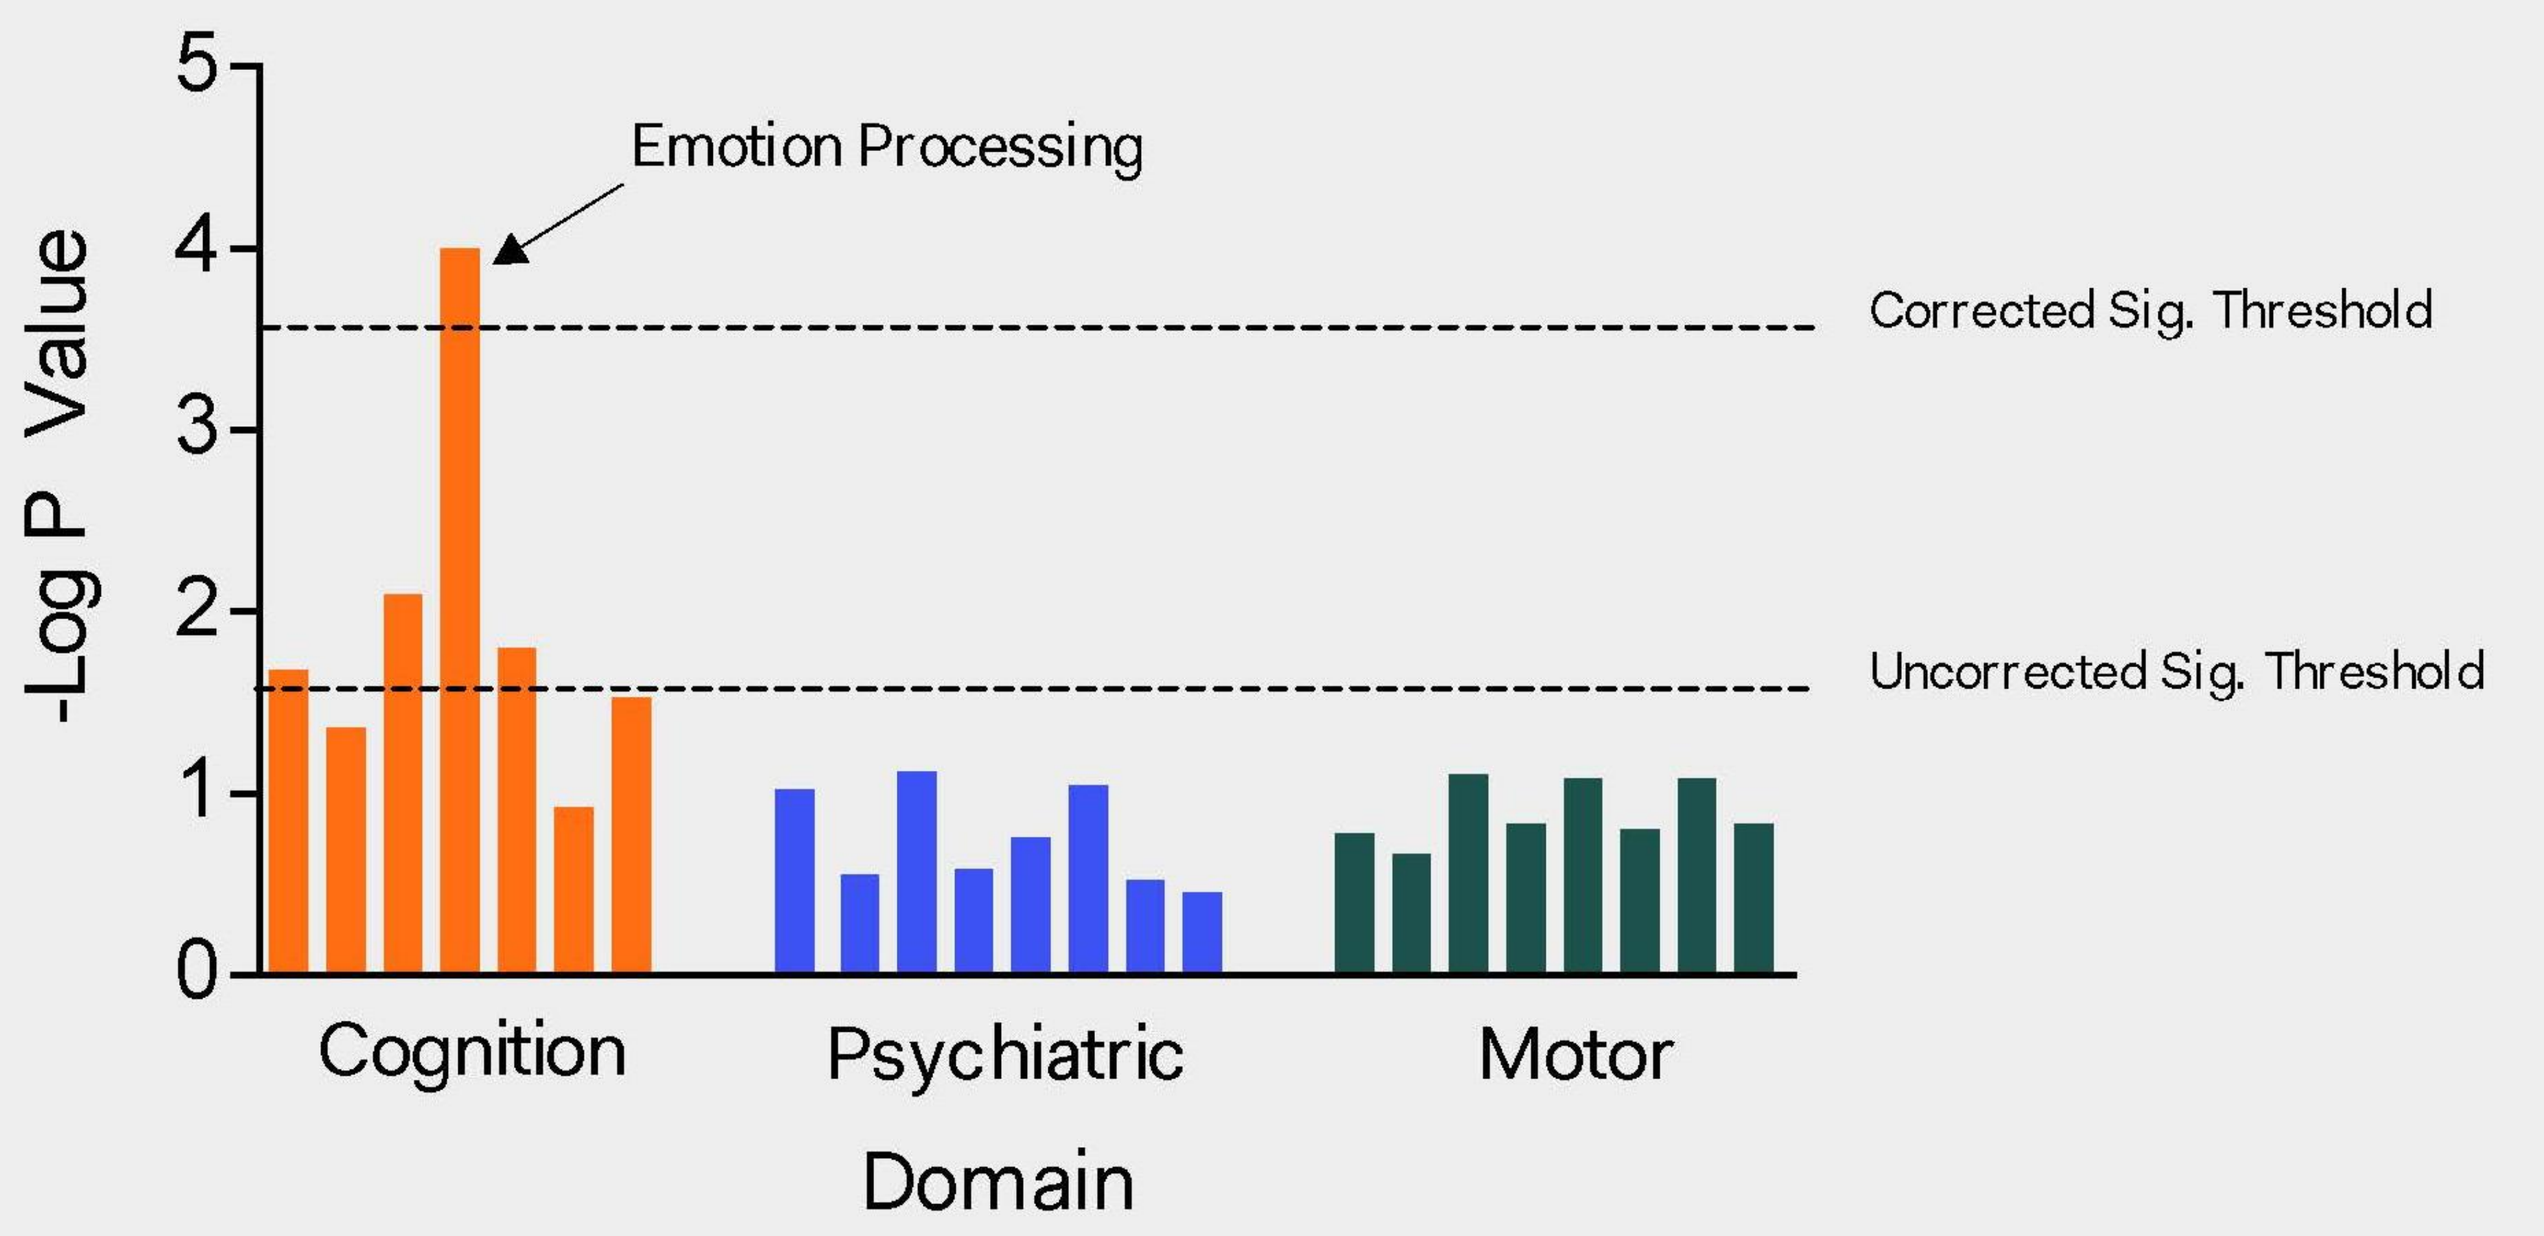
-Log P Value
Emotion Processing
Corrected Sig. Threshold
Uncorrected Sig. Threshold
Cognition
Psychiatric
Motor
Domain

- 24(S)-HC correlations across endpoints in TRACK-HD. 24(S)-HC levels were significantly correlated with cognitive endpoints in year 2 of the TRACK-HD study.
- After correcting for multiple comparisons, performance on the negative emotion (Eckman Faces) task remained significant.
- Data are represented as –Log P value of regression P value.

24(S)-HC CORRELATIONS AFTER CELL LOSS CORRECTION

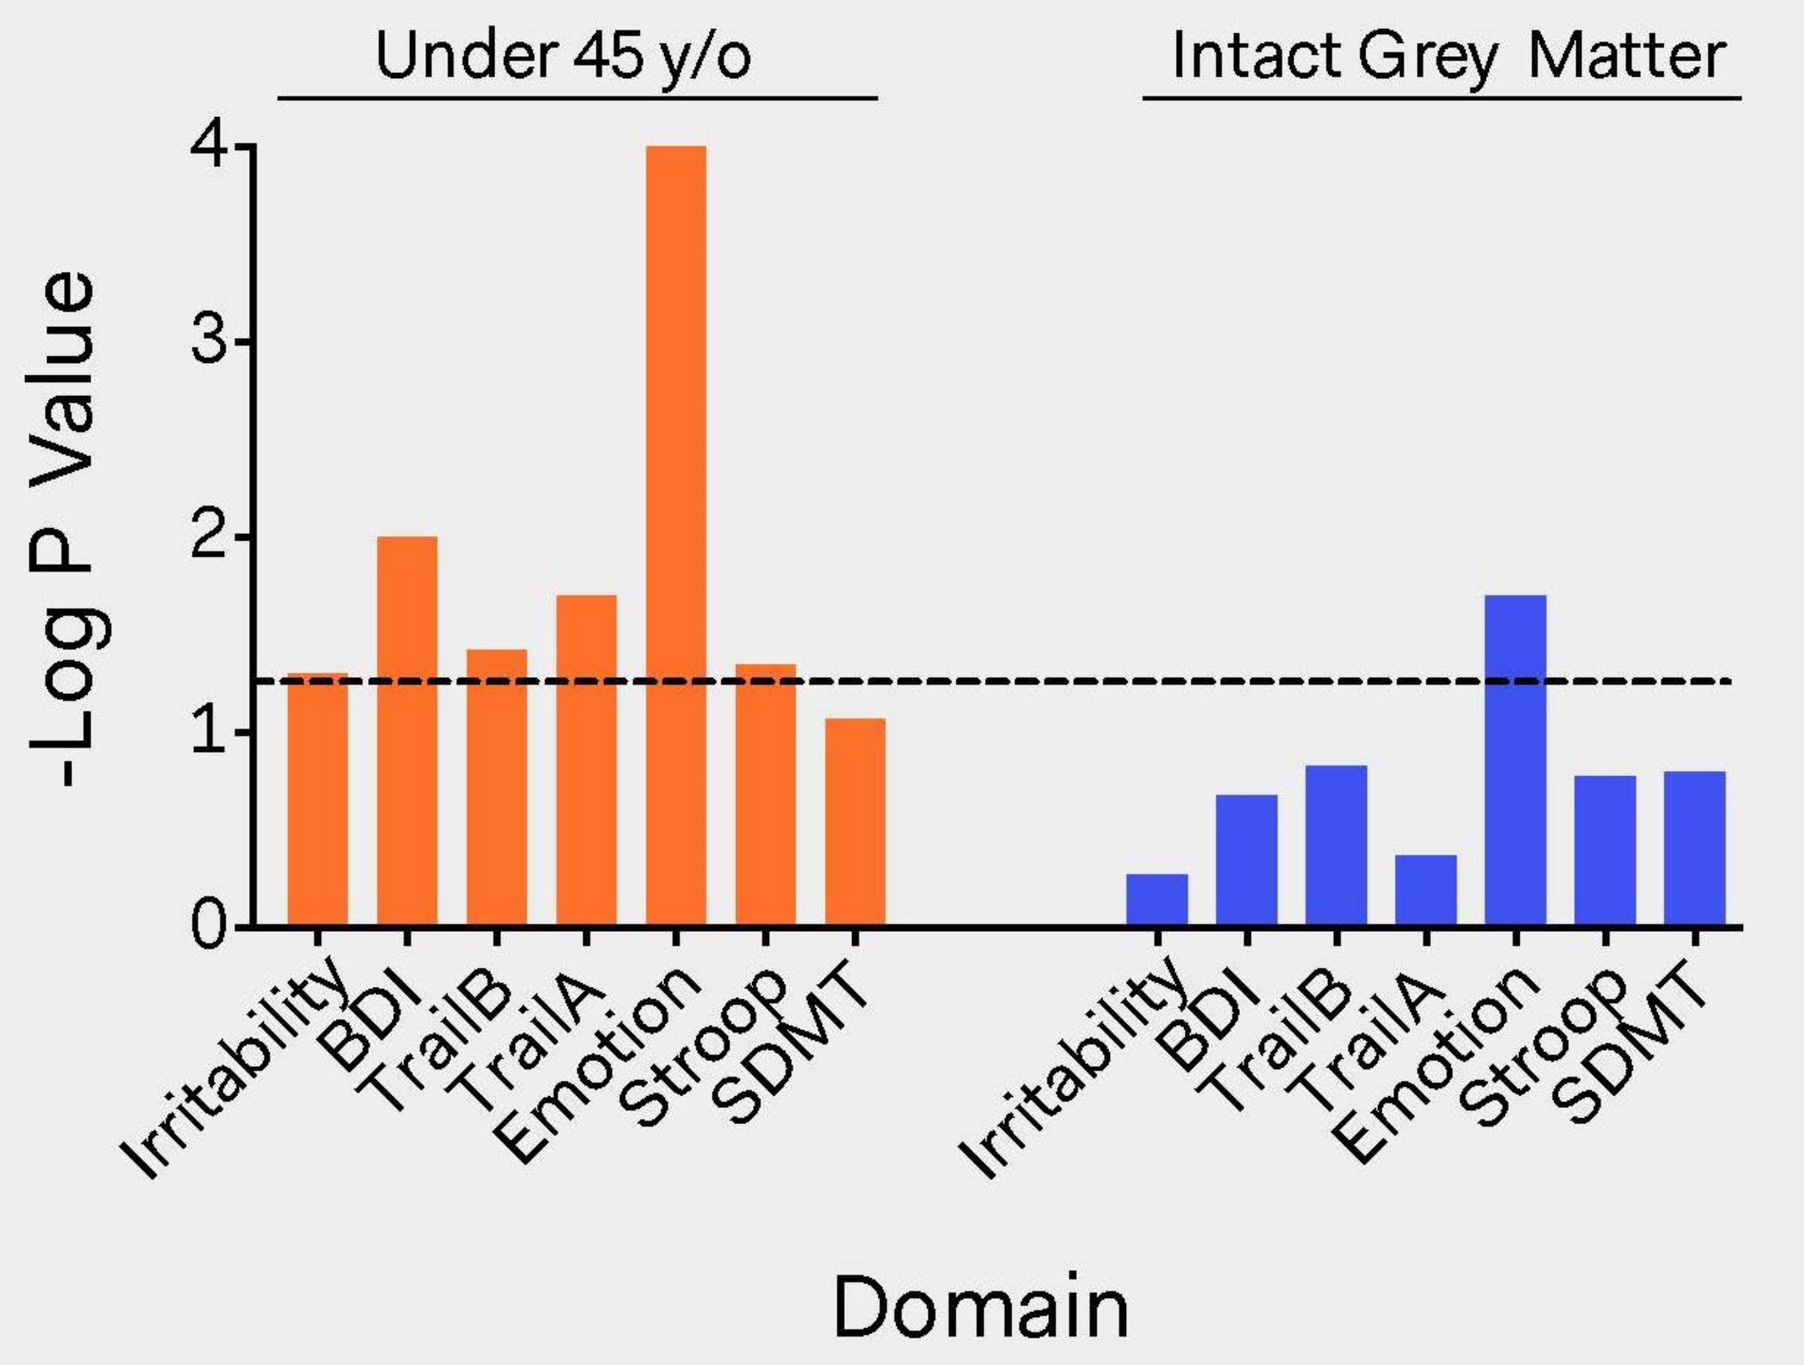
Under 45 y/o
Intact Grey Matter
-Log P Value
Irritability
BDI
TrailB
TrailA
Emotion
Stroop
SDMT
Domain

- 24(S)-HC correlations across endpoints after correcting for potential cell loss. 24(S)-HC levels remain robust after segregating the data on median age and grey matter volume.
- Data are represented as –Log P value of regression.

24(S)-HC ASSOCIATIONS ACROSS TRACK-HD

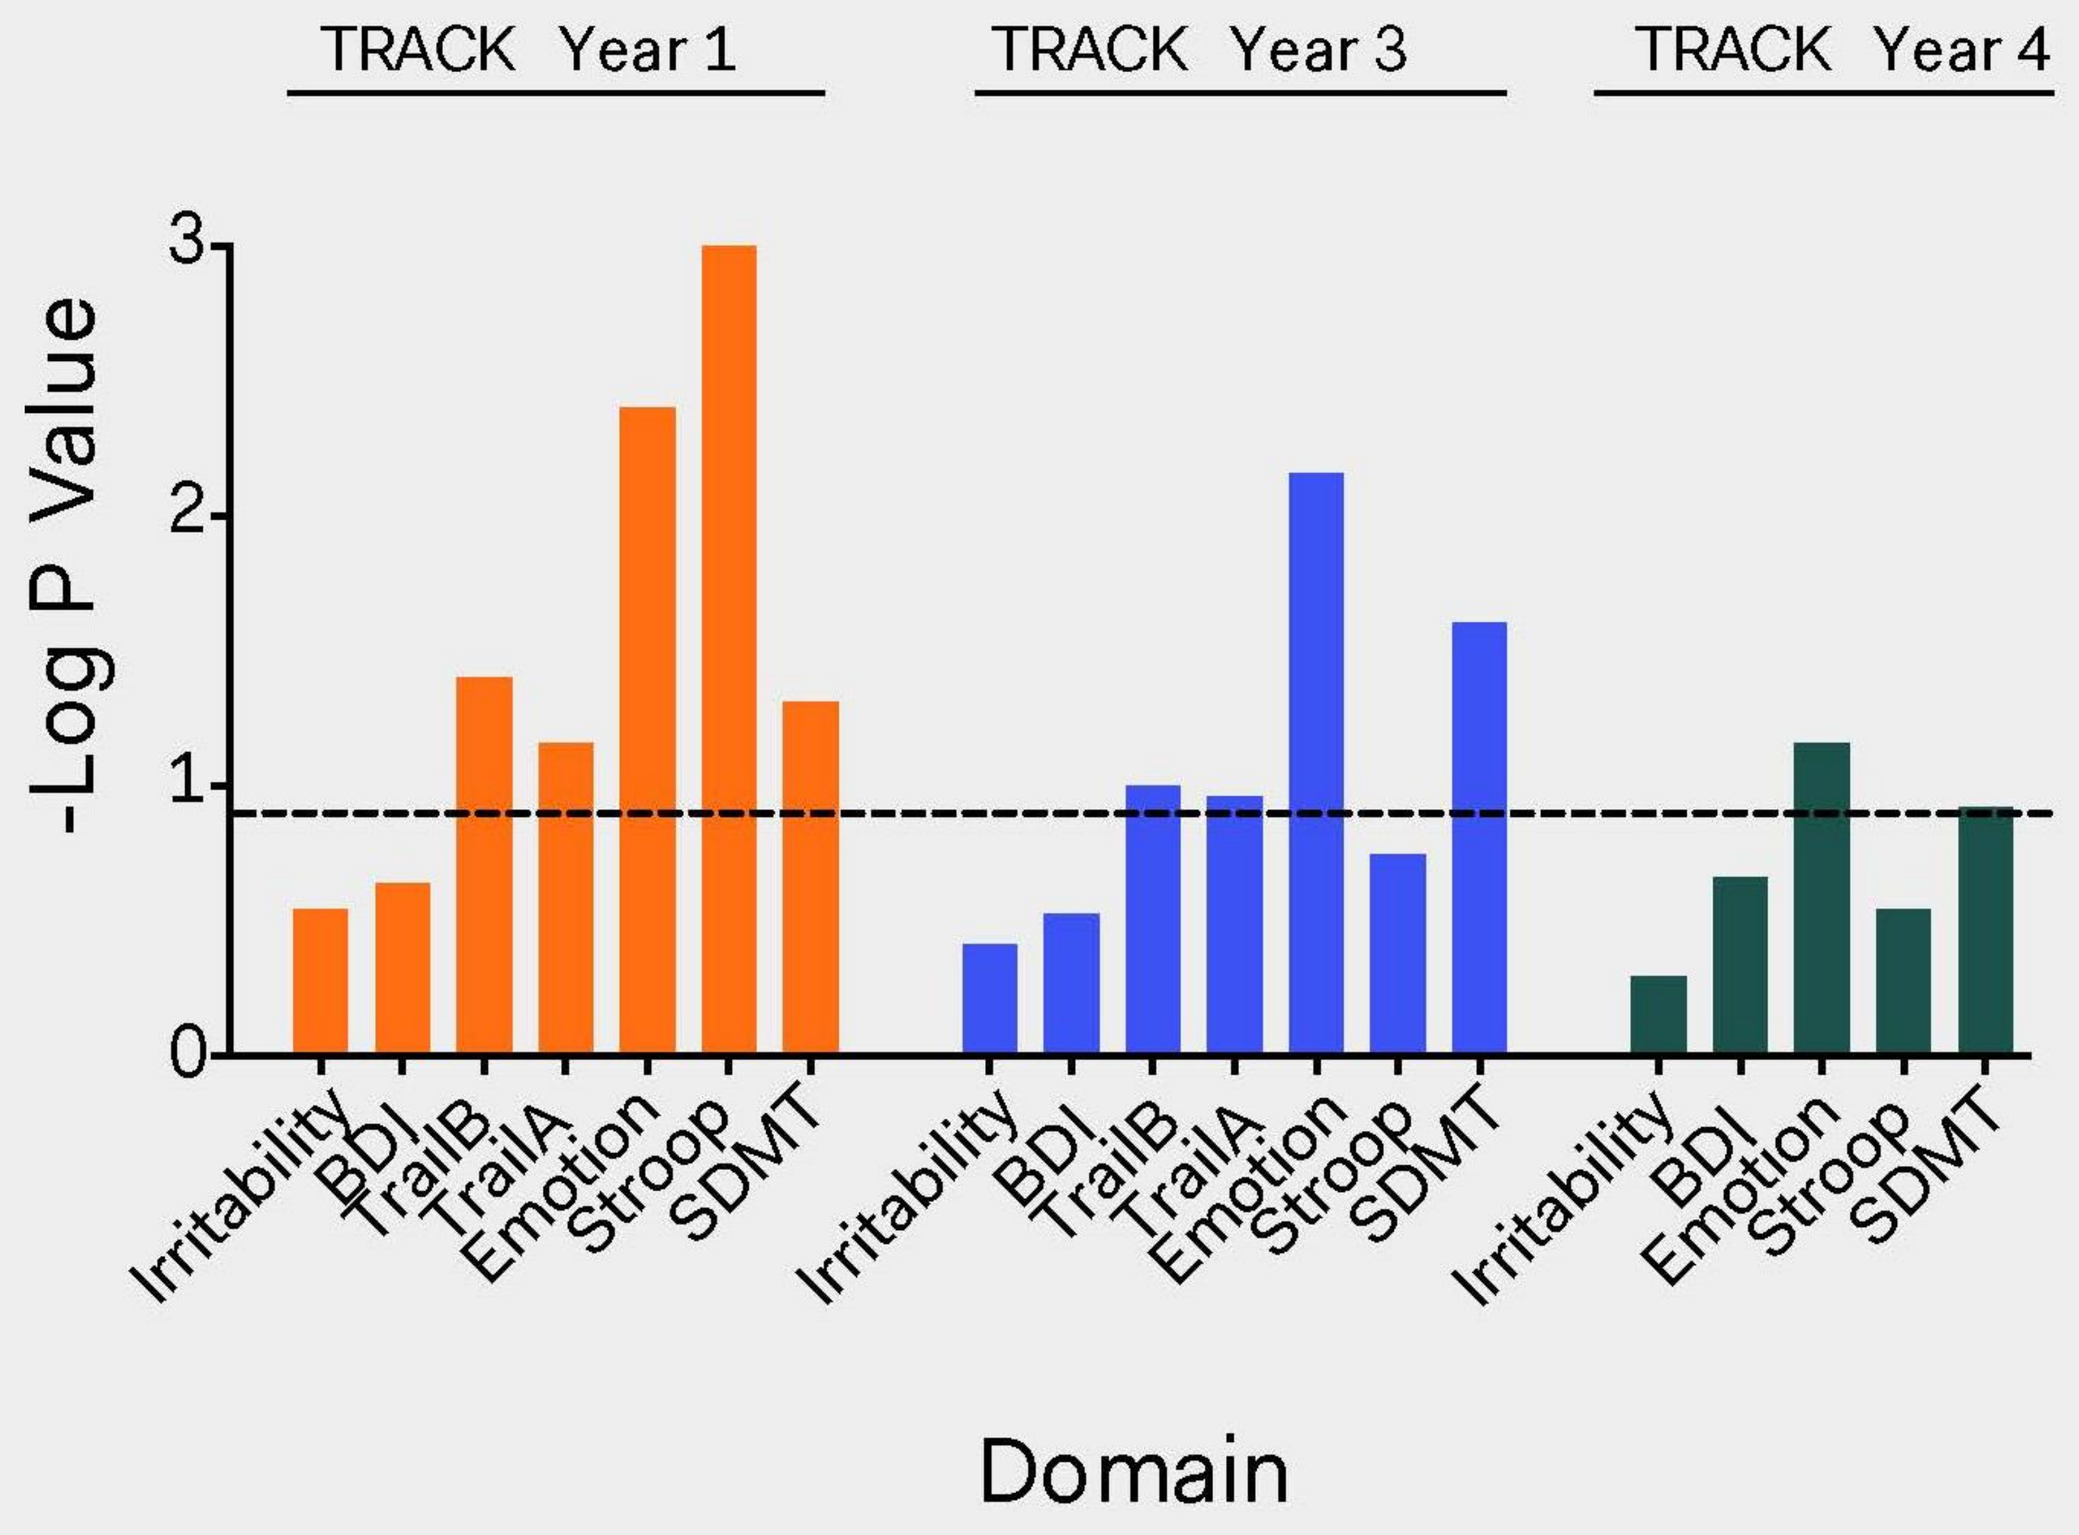
TRACK Year 1
TRACK Year 3
TRACK Year 4
-Log P Value
Irritability
BDI
TrailB
TrailA
Emotion
Stroop
SDMT
Domain

- 24(S)-HC correlations in years one, three, and four of TRACK-HD.
- Strong relationships between 24(S)-HC and cognitive measures were present across the years of TRACK-HD.
- Data are represented as –Log P value of regression.

EFFECT ON COGNITION: WORKING MEMORY AND PROBLEM SOLVING

TWO-BACK TEST (WORKING MEMORY)

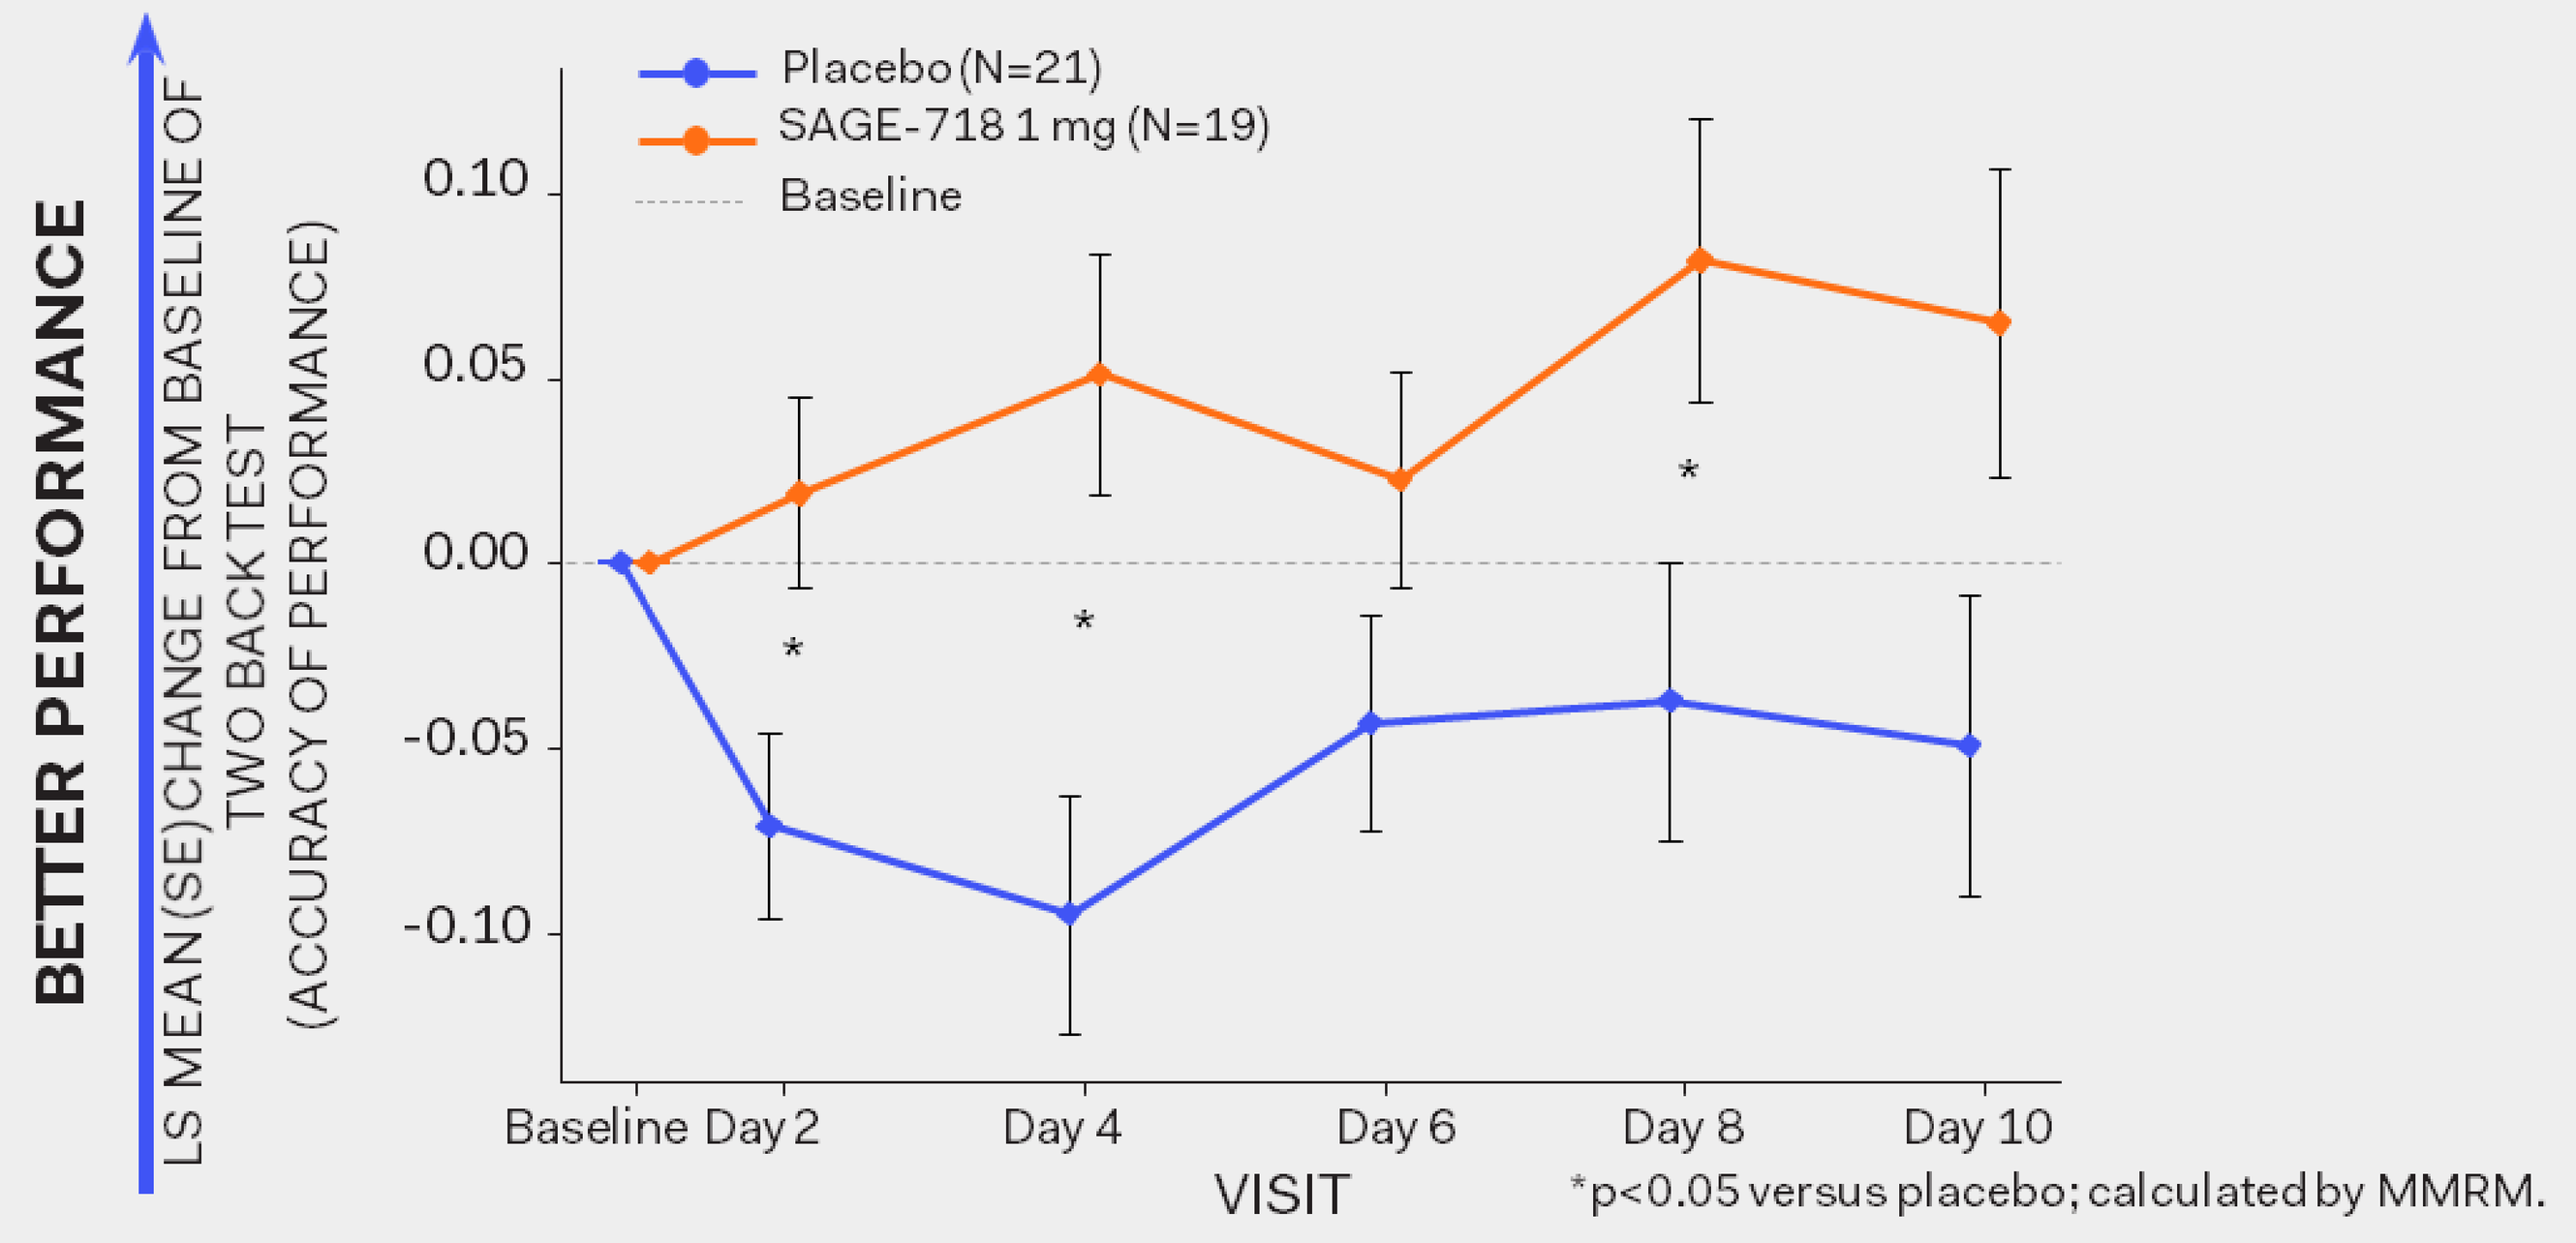
BETTER PERFORMANCE
LS MEAN (SE) CHANGE FROM BASELINE OF TWO BACK TEST (ACCURACY OF PERFORMANCE)
Placebo (N=21)
SAGE-718 1 mg (N=19)
Baseline
Baseline
Day 2
Day 4
Day 6
Day 8
Day 10
VISIT
*p<0.05 versus placebo; calculated by MMRM.

GROTON MAZE TEST (EXECUTIVE FUNCTION)

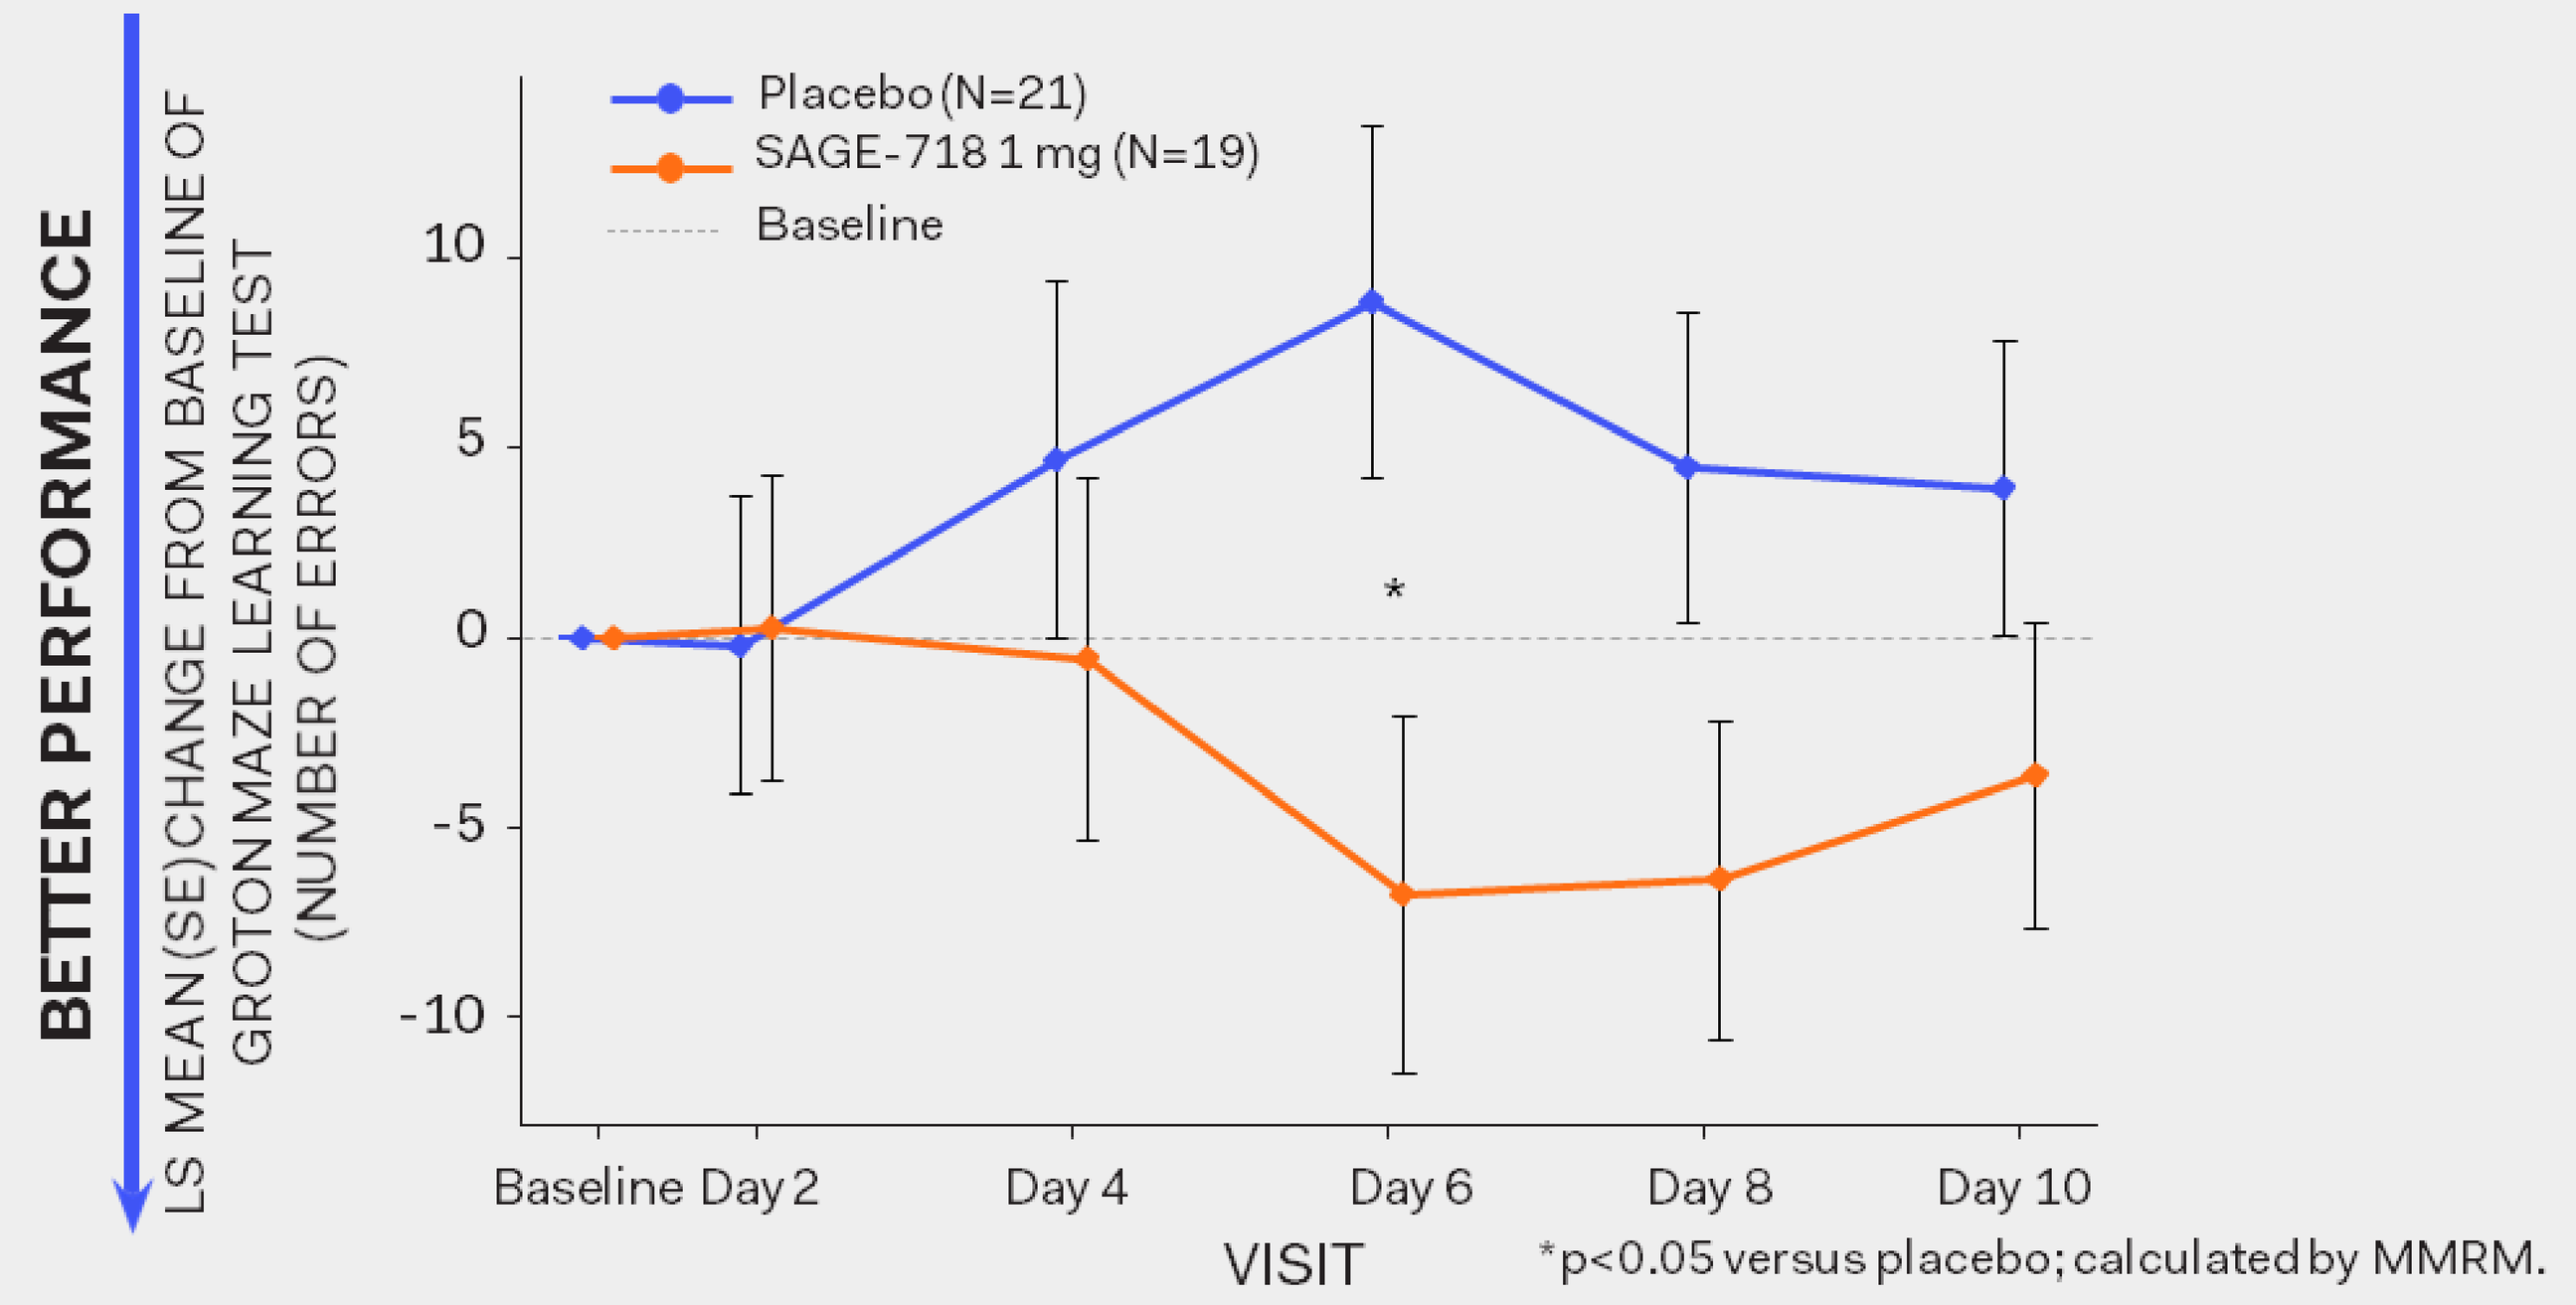
BETTER PERFORMANCE
LS MEAN (SE) CHANGE FROM BASELINE OF GROTON MAZE LEARNING TEST (NUMBER OF ERRORS)
Placebo (N=21)
SAGE-718 1 mg (N=19)
Baseline
Baseline
Day 2
Day 4
Day 6
Day 8
Day 10
VISIT
*p<0.05 versus placebo; calculated by MMRM.

- SAGE-718 significantly improved working memory at Days 2, 4 and 8, and executive functioning at Day 6 in a Phase 1, multiple ascending dose study in healthy volunteers.

Methods

- Plasma samples were collected over 4 years as part of the TRACK-HD study.
- Plasma samples were extracted using liquid-liquid extraction (oxysterols) or protein precipitation (cholesterol) and analyzed via liquid chromatography tandem mass spectrometry.
- 24(S)-HC, 25-HC, and 27-HC were profiled and normalized (for 25-HC and 27-HC) to total cholesterol.
- Each oxysterol was then correlated across 19 cognitive and 23 neuropsychiatric endpoints within the year 2 samples (hypothesis generation).
- Those endpoints that were significantly correlated with oxysterol levels within year 2 were then tested against samples from years 1, 3, and 4 (hypothesis testing).
- A mixed model for repeated measures was applied, with change from baseline in each cognitive assessment test score as the response variable and treatment, visit, visit by-treatment interaction as fixed effect, baseline as covariate, and measurements within the same subject as repeated measures. Unstructured covariance structure was applied for the repeated measure.

REFERENCES
1. Martin M, et al. EMBO Reports. 2014; 15 (10):1036–1052.
2. Saeed AA, et al. J Biol Chem. 2014 Aug 22; 289(34): 23712–23722.
3. Boussicault L, et al. Brain. 2016 Mar;139(Pt 3):953-70.
4. Paul SM, et al. J Neurosci. 2013; 33(44):17290-300.
5. Kreilaus F, et al. Neuropathol Appl Neurobiol. 2016; 42(6):535-46.
6. Leoni V, et al. Neurosci Lett. 2011; 494(3):245-9.
7. Koenig et al. Cognitive-Enhancing Effects of the Novel NMDA Positive Allosteric Modulator SAGE-718 in Healthy Volunteers. ACNP 2019.
8. Tabrizi SJ, et al. Lancet Neurol. 2012; 11(1):42-53

SAGE THERAPEUTICS is a trademark of Sage Therapeutics, Inc. All other trademarks referenced herein are the property of their respective owners.

SUPPORT & DISCLOSURES
ML, JD, JD, AJR and MQ are employees of Sage Therapeutics, Inc. with stock/stock options. AM is is employed by the CHDI Foundation. ST reports personal fees and other support from Ionis Pharmaceuticals and personal fees from F. Hoffmann La Roche Ltd, and received grant funding from the Medical Research Council UK, the Wellcome Trust, the Rosetrees Trust, Takeda Pharmaceuticals, Cantervale Limited, the NIHR North Thames Local Clinical Research Network, the UK Dementia Research Institute, the Wolfson Foundation for Neurodegeneration and CHDI Foundation. This study was funded by Sage Therapeutics, Inc. This poster was assembled with assistance from Boston Strategic Partners.

American Academy of Neurology Annual Meeting
April 25 – May 1, 2020
Toronto, Canada
MRC-718-00111 05/20