Poster #: 010

Zuranolone in Major Depressive Disorder: A Phase 3, Multicenter, Double-Blind, Randomized, Placebo-Controlled Trial

Akanksha Mittal, PhD[1], Anita Clayton, MD[2], Robert Lasser, MD, MBA[1], Indrani Nandy, PhD[1], Abdul J. Sankoh, PhD[1] Andrew D. Campbell, PhD[1], Brian Werneburg, PhD[1], Christopher Silber, MD[1], Jeffrey Jonas, MD[1], Stephen J. Kanes, MD, PhD[1], Handan Gunduz-Bruce, MD, MBA[1]

[1]Sage Therapeutics, Inc., Cambridge, MA, USA; [2]University of Virginia, School of Medicine, Charlottesville, VA, USA

Introduction

- In the United States, 6.8% of adults (or 1 in 14) experience at least one depressive episode each year.[1]
 - MDD is one of the largest contributors to disability in the United States.[2-4]
- MDD is characterized by a period of depressive symptoms lasting at least 2 weeks and is associated with changes in affect, cognition, and function.[5]
- Gamma-aminobutyric acid (GABA)-ergic neurotransmission regulates the activity of diverse brain networks that may be implicated in depression.[6-8]
- Zuranolone (SAGE-217) is an investigational oral neuroactive steroid $GABA_A$ receptor positive allosteric modulator.[9, 10]
- Zuranolone binds to both synaptic and extrasynaptic $GABA_A$ receptors receptors and has a distinct pharmacological profile from benzodiazepines which bind to synaptic $GABA_A$ receptors.[10,11]
- In a pivotal trial in MDD (NCT03000530), zuranolone demonstrated a clinically meaningful and statistically significant improvement in depressive symptoms after two weeks of treatment and was generally well tolerated.[12]
 - The most common adverse events (≥5%) in any group were headache, dizziness, somnolence, and nausea.
- The efficacy and safety of two-week treatment with zuranolone 20 mg or 30 mg were evaluated in this double-blind, randomized, placebo-controlled Phase 3 trial (MOUNTAIN study; NCT03672175) in adults with MDD.

Conclusions

- The MOUNTAIN study did not meet its primary endpoint of a statistically significant reduction from baseline compared to placebo in HAMD-17 total score at Day 15.
- Statistical significance from placebo in the zuranolone 30 mg group was noted at measured timepoints during the treatment period leading up to Day 15.
- The 20 mg dose of zuranolone did not separate from placebo in this dose-ranging study.
- In a post-hoc analysis, excluding approximately 9% of patients in the zuranolone 30 mg group with no measurable drug from the primarily analysis set (zuranolone 30 mg vs placebo) resulted in statistical significance at all measured timepoints up to, and including, Day 15.
- In patients with both a measurable drug concentration of zuranolone and a HAMD-17 ≥24, zuranolone was associated with a significant mean reduction of HAMD-17 compared to placebo at all measured timepoints up to, and including, Day 15.
- Zuranolone was generally well-tolerated and showed a similar safety profile as seen in earlier studies.[13]
 - The most common adverse events (≥5%) in any group during the two-week treatment period and the four-week follow-up were headache, dizziness, somnolence, fatigue, diarrhea, sedation, and nausea.
 - Two patients receiving zuranolone 30 mg experienced serious adverse events (SAEs) during treatment while three patients, one in each treatment group, reported SAEs during follow-up.

Methods

- The study enrolled 581 adult (18-65 years old) patients with MDD and of qualifying severity by the 17-item Hamilton Rating Scale for Depression total score (HAMD-17; score ≥22) and the Montgomery-Åsberg Depression Rating Scale total score (MADRS; score ≥32).
- Patients were stratified based on baseline antidepressant use and randomized 1:1:1 to receive zuranolone 20 mg, zuranolone 30 mg, or placebo. Blinded study drug was administered as identical gelatin capsules on an outpatient basis, daily, in the evening, for two weeks with follow-up through Day 42.
- Exclusion criteria included attempted suicide associated with the current depressive episode, treatment resistant depression, or medical history of bipolar disorder, schizophrenia, and/or schizoaffective disorder.
- Concomitant antidepressant medication use was permitted at a stable dose from 60 days prior to Day 1 through Day 42.
 - No changes in concomitant antidepressant medications were permitted from at least 60 days prior to Day 1 through the Day 42 follow-up.
- The change from baseline in HAMD-17 total score at Day 15 was the primary endpoint.
- Secondary endpoints included the change from baseline in HAMD-17 total score at all other timepoints.
- Post-hoc endpoints included change from baseline in HAMD-17 in patients with measurable drug, and change from baseline in HAMD-17 in patients with measurable drug and a baseline HAMD-17 ≥24.
- Secondary and post-hoc analyses were not adjusted for multiplicity.
- Statistics are from a mixed effect model for repeated measures including the least-squares mean (LSM) change from baseline in HAMD-17 at each visit as the dependent variable.
- Safety and tolerability were evaluated throughout the study by adverse event reporting, the Columbia Suicide Severity Rating Scale (C-SSRS), the 20-item Physician Withdrawal Checklist, and standard clinical assessments.

STUDY DESIGN

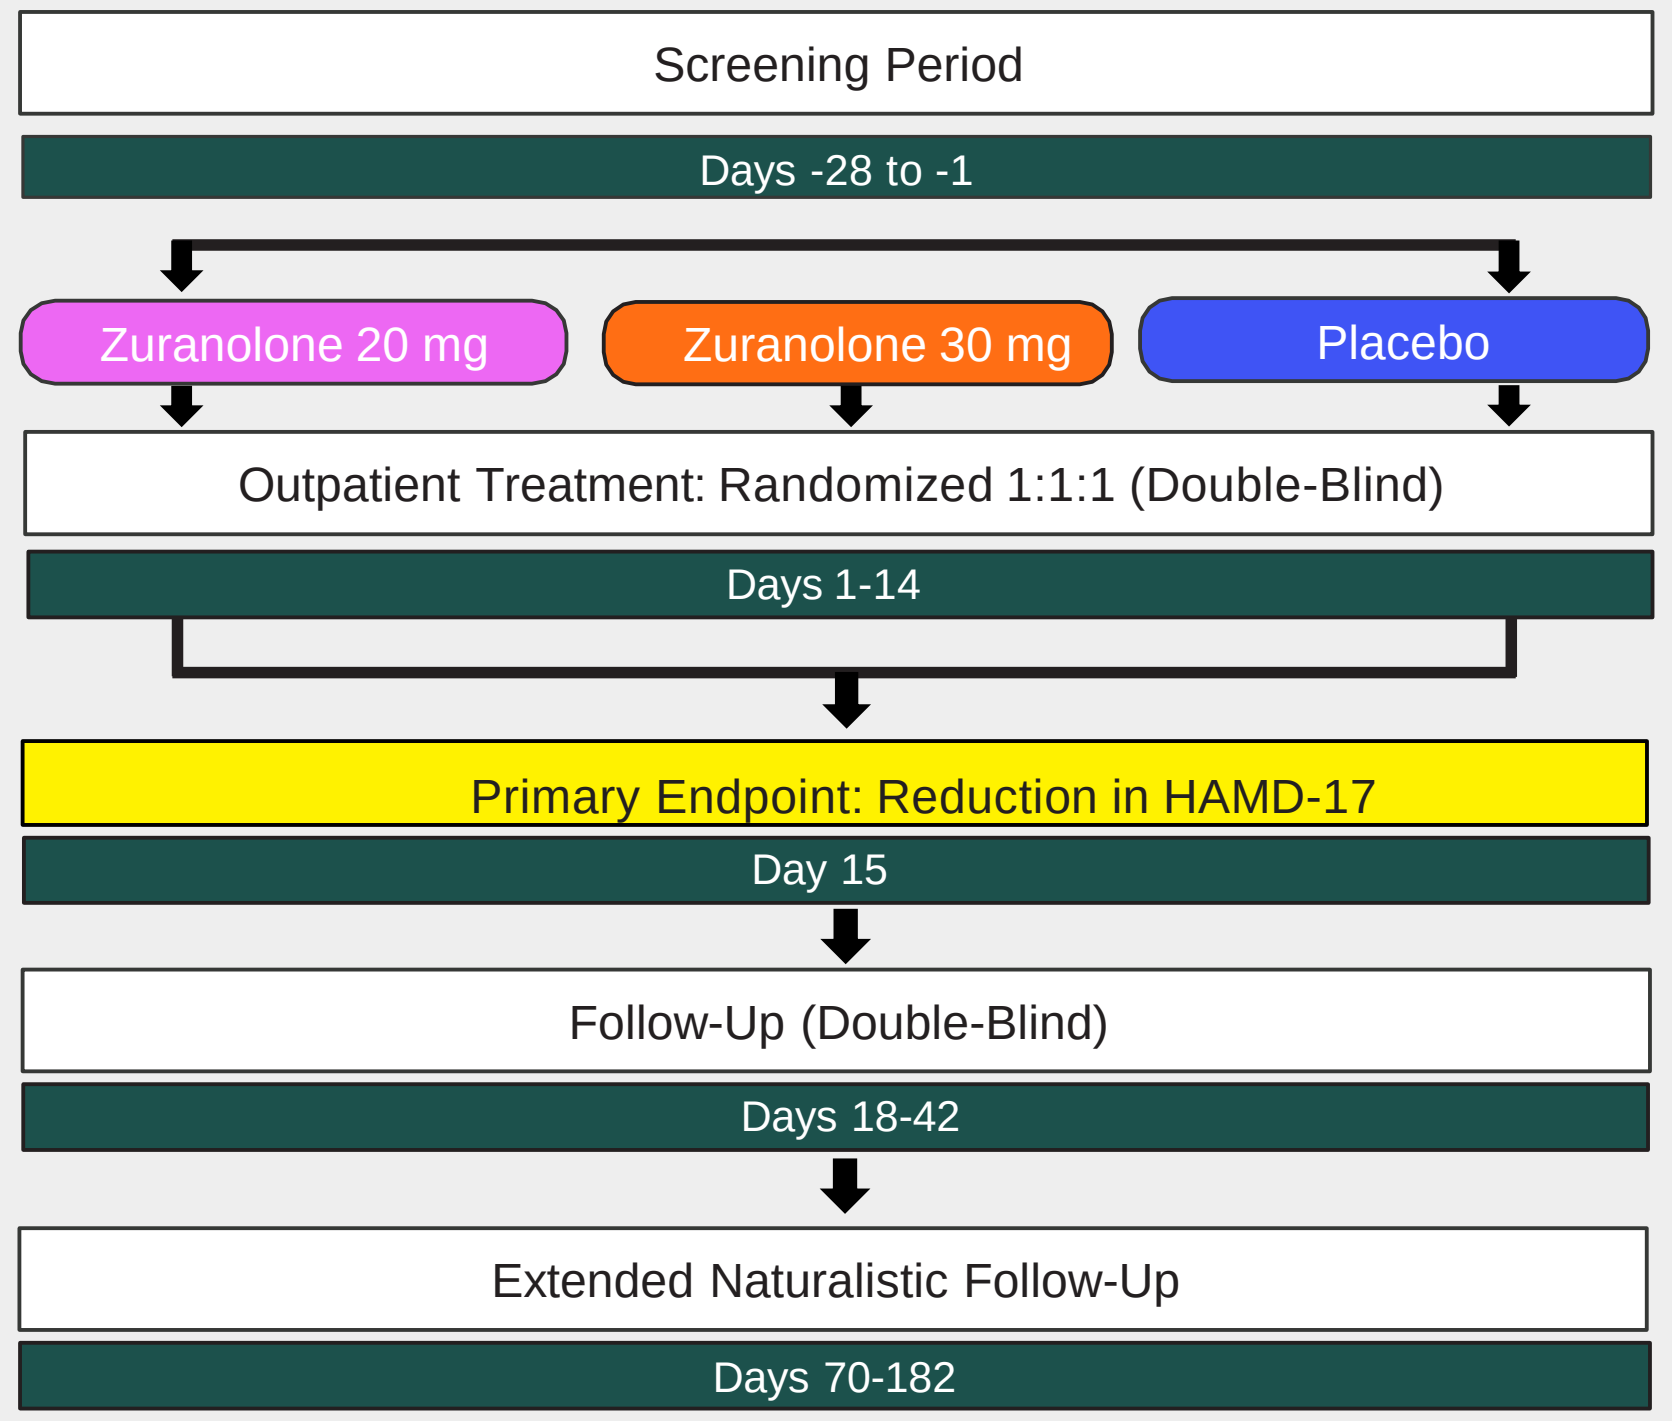
Screening Period
Days -28 to -1
Zuranolone 20 mg
Zuranolone 30 mg
Placebo
Outpatient Treatment: Randomized 1:1:1 (Double-Blind)
Days 1-14
Primary Endpoint: Reduction in HAMD-17
Day 15
Follow-Up (Double-Blind)
Days 18-42
Extended Naturalistic Follow-Up
Days 70-182

Results

DEMOGRAPHICS AND BASELINE CHARACTERISTICS

	ZURANOLONE 20 MG N=159	ZURANOLONE 30 MG N=166	PLACEBO N=157
Age – years	41.9 (12.0)	42.3 (12.0)	41.4 (12.0)
Sex			
Female	112 (70.4)	121 (72.9)	106 (67.5)
Male	47 (29.6)	45 (27.1)	51 (32.5)
Race or ethnic group			
Asian	3 (1.9)	2 (1.2)	3 (1.9)
Black/African-American	56 (35.2)	64 (38.6)	54 (34.4)
Multiple	1 (0.6)	4 (2.4)	3 (1.9)
White	99 (62.3)	94 (56.6)	96 (61.1)
Other	0 (0.0)	2 (1.2)	1 (0.6)
Ethnicity			
Hispanic/Latino	31 (19.5)	27 (16.3)	26 (16.6)
Weight – mean, kg	87.3 (20.2)	89.7 (22.4)	89.5 (22.9)
Baseline HAMD-17	25.8 (2.8)	25.9 (2.9)	25.8 (3.1)
Use of antidepressants at baseline	46 (28.9)	47 (28.3)	49 (31.2)

Data are from the efficacy population, reported as mean (SD) or N (%).

TREATMENT-RELATED ADVERSE EVENTS

Total Number of TEAEs	ZURANOLONE 20 MG N=188	ZURANOLONE 30 MG N=192	PLACEBO N=190
Any TEAEs	94 (50.0)	104 (54.2)	93 (48.9)
Severe TEAEs	3 (1.6)	2 (1.0)	2 (1.1)
Serious TEAEs	1 (0.5)	3 (1.6)	1 (0.5)
TEAE-Drug Discontinued	3 (1.6)	4 (2.1)	6 (3.2)
Deaths	0 (0.0)	0 (0.0)	0 (0.0)
Most Common TEAEs, ≥5% Patients			
Somnolence	11 (5.9)	13 (6.8)	8 (4.2)
Headache	21 (11.2)	12 (6.3)	14 (7.4)
Dizziness	14 (7.4)	11 (5.7)	7 (3.7)
Fatigue	3 (1.6)	13 (6.8)	5 (2.6)
Diarrhea	11 (5.9)	12 (6.3)	10 (5.3)
Sedation	11 (5.9)	9 (4.7)	6 (3.2)
Nausea	10 (5.3)	7 (3.6)	9 (4.7)

Data are from the safety population and from the double-blind period, reported as N (%).

- There were no adverse events of loss of consciousness reported.
- There was no signal for increased suicidal ideation or suicidal behavior compared to baseline, as measured by the C-SSRS.

CHANGE FROM BASELINE IN HAMD-17 TOTAL SCORE

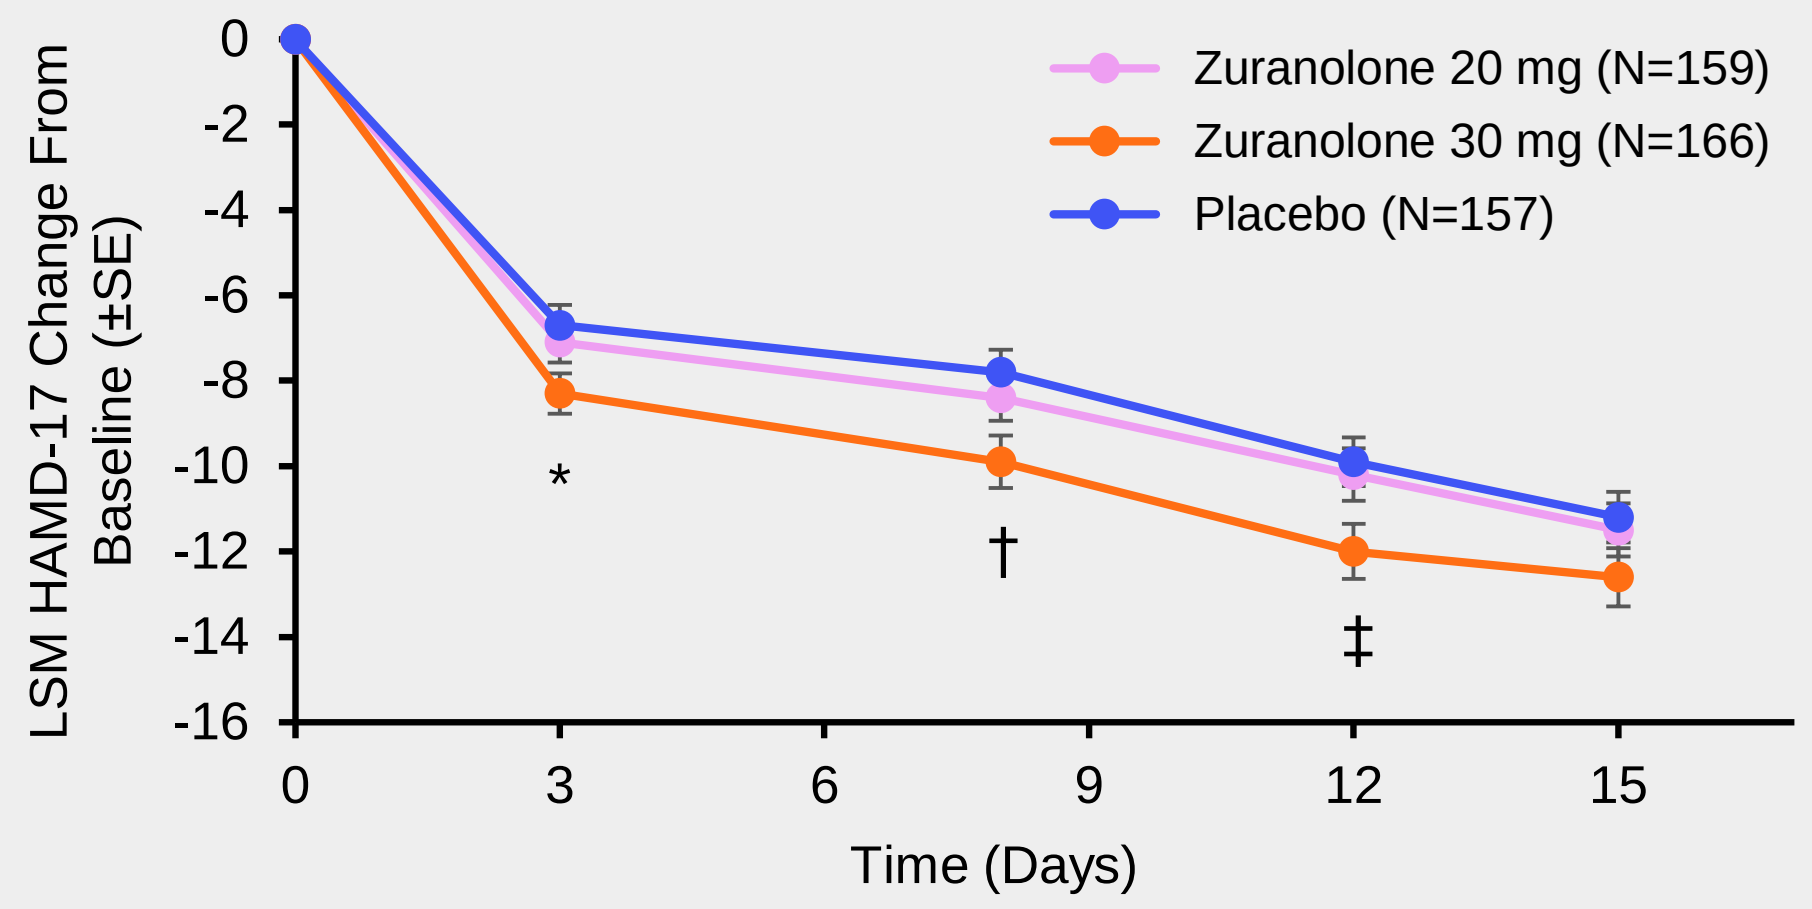
LSM HAMD-17 Change From Baseline (±SE)
0
-2
-4
-6
-8
-10
-12
-14
-16
0
3
6
9
12
15
Time (Days)
Zuranolone 20 mg (N=159)
Zuranolone 30 mg (N=166)
Placebo (N=157)
*
†
‡

*p=0.016, †p=0.008, ‡p=0.018; p-values shown for zuranolone 30 mg vs placebo.

- The MOUNTAIN study did not meet its primary endpoint of a statistically significant reduction from baseline compared to placebo in HAMD-17 total score at Day 15. Zuranolone 30 mg was associated with a mean reduction of 12.6 in HAMD-17 compared to 11.2 for placebo (LSM Difference -1.4, p=0.115).
- Statistical significance from placebo in zuranolone 30 mg was noted beginning at Day 3 and statistical significance from placebo was noted at all measured timepoints during the treatment period leading up to Day 15 (LSM Difference from placebo, p-value): Day 3 (-1.6, p=0.016), Day 8 (-2.1, p=0.008), and Day 12 (-2.1, p=0.018).
- Zuranolone 20 mg was not statistically significant from placebo at any measured timepoint.

CHANGE FROM BASELINE IN HAMD-17 TOTAL SCORE IN PATIENTS WITH MEASURABLE DRUG: POST-HOC ENDPOINT

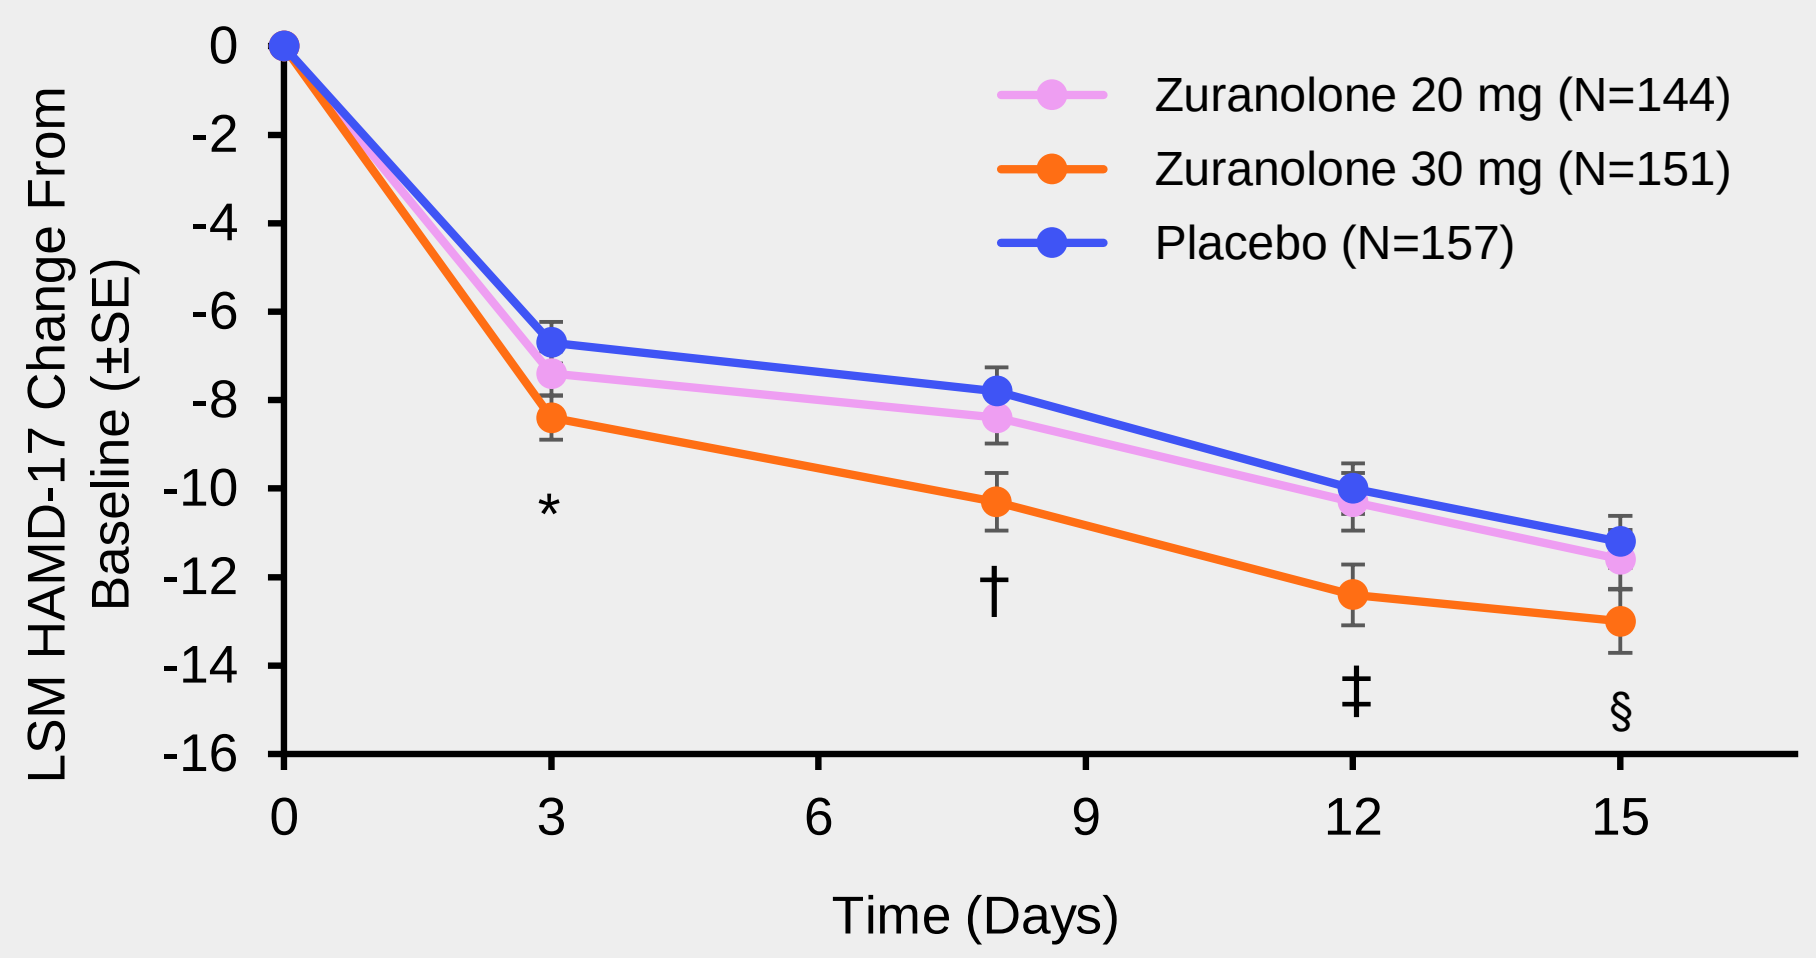
LSM HAMD-17 Change From Baseline (±SE)
0
-2
-4
-6
-8
-10
-12
-14
-16
0
3
6
9
12
15
Time (Days)
Zuranolone 20 mg (N=144)
Zuranolone 30 mg (N=151)
Placebo (N=157)
*
†
‡
§

*p=0.011, †p=0.002, ‡p=0.006, §p=0.048; p-values shown for zuranolone 30 mg vs placebo.

- Post-hoc analyses revealed that approximately 9% of patients in the zuranolone 30 mg group had no measurable drug concentration, consistent with non-compliance in taking zuranolone.
- Excluding these patients from the primary analysis set (zuranolone 30mg vs placebo), resulted in statistical significance at all timepoints measured through and including Day 15 (p<0.048).
- Zuranolone 20 mg was not statistically significant from placebo at any measured timepoint.

CHANGE FROM BASELINE IN HAMD-17 TOTAL SCORE IN PATIENTS WITH MEASURABLE DRUG AND HAMD-17 ≥ 24: POST-HOC ENDPOINT

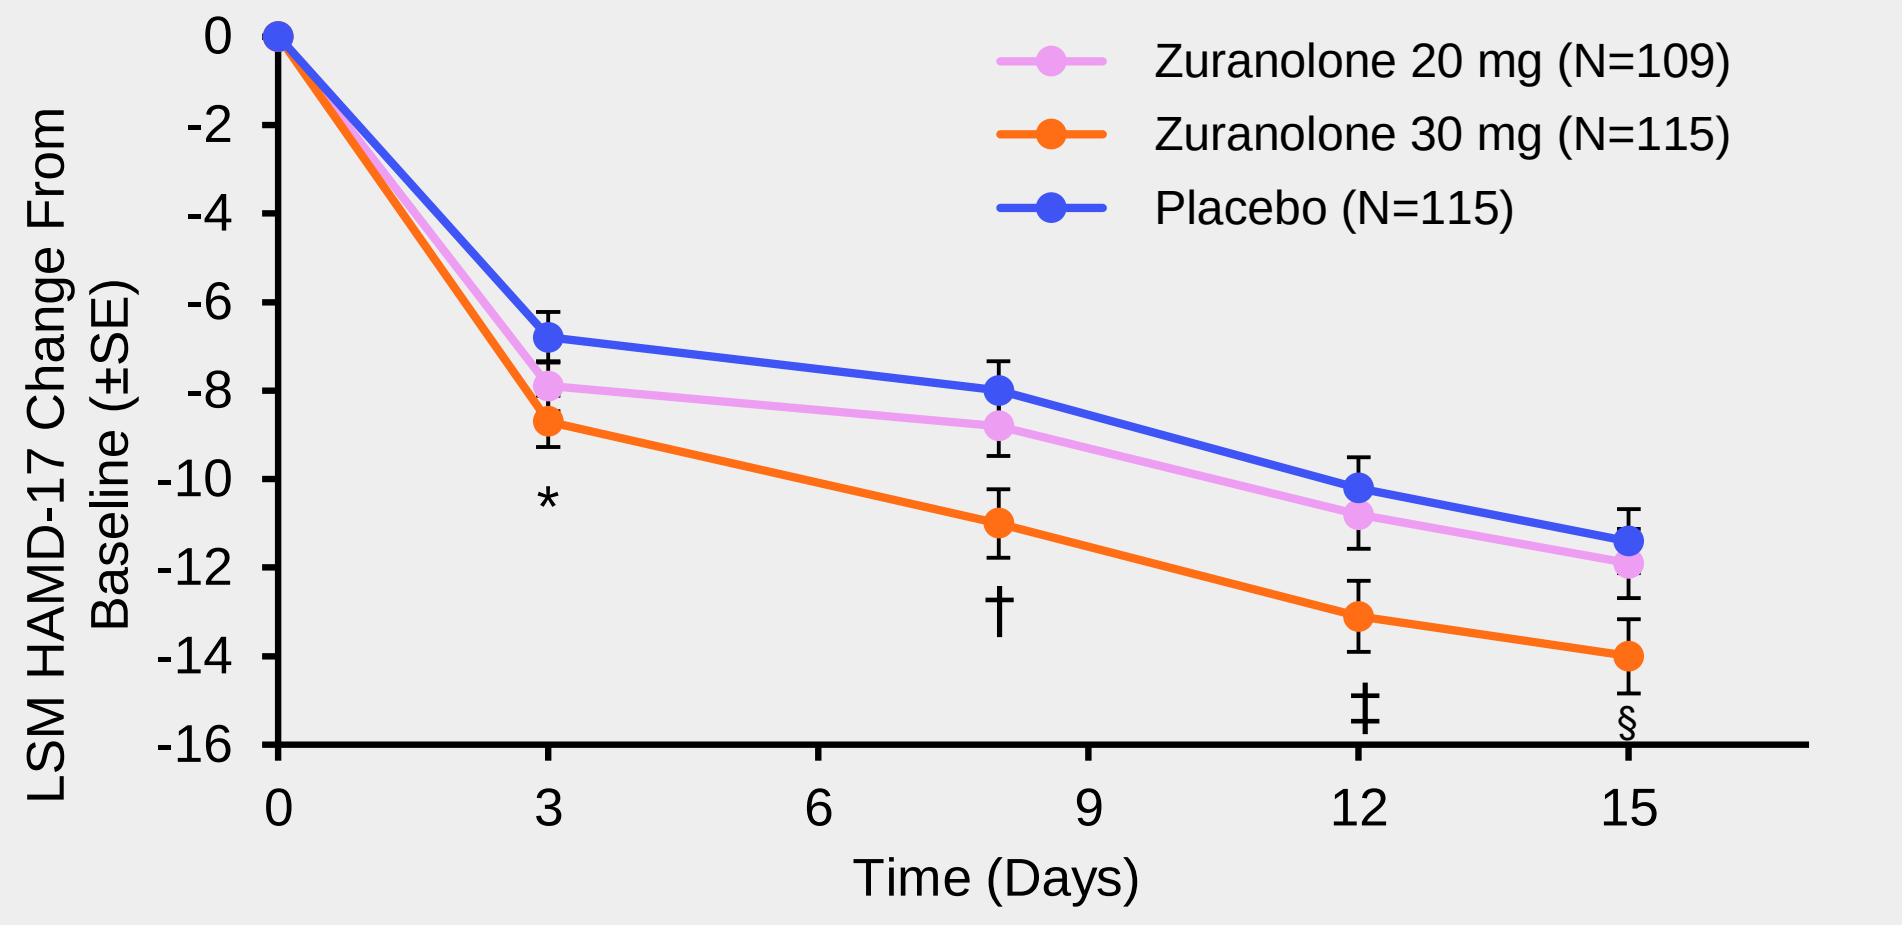
LSM HAMD-17 Change From Baseline (±SE)
0
-2
-4
-6
-8
-10
-12
-14
-16
0
3
6
9
12
15
Time (Days)
Zuranolone 20 mg (N=109)
Zuranolone 30 mg (N=115)
Placebo (N=115)
*
†
‡
§

*p=0.015, †p=0.003, ‡p=0.005, §p=0.017; p-values shown for zuranolone 30 mg vs placebo.

- More patients with an overall distribution of milder severity of symptoms were enrolled than in previous studies of zuranolone.
- Post-hoc analyses in patients with both a measurable drug concentration of zuranolone and a HAMD-17 ≥24, zuranolone 30 mg was associated with a mean reduction of 14.0 in HAMD-17 total score compared to 11.4 for placebo (LSM Difference-2.6, p=0.017)
- Zuranolone 20 mg was not statistically significant from placebo at any measured timepoint.

REFERENCES
1. 12-month prevalence of DSM-IV/WMH-CIDI disorders by sex and cohort (n=9,282). https://www.hcp.med.harvard.edu/ncs/ftpdir/NCS-R_12-month_Prevalence_Estimates.pdf. Updated July 19, 2007. Accessed June 8, 2020.
2. USBD. JAMA. 2018;319(14):1444-14724.
3. Marcus M, Yasamy MT, van Ommeren M, Chisholm D, Saxena S. Depression: a global public health concern. WHO Department of Mental Health and Substance Abuse. 2012.
4. Global Health Estimates 2016: Disease Burden by Cause, Age, Sex, by Country, and by Region, 2000-2016. Geneva, World Health Organization; 2018. https://www.who.int/healthinfo/global_burden_disease/estimates/en/index1.html.
5. American Psychiatric Association. Diagnostic and Statistical Manual of Mental Disorders. Text Revision - Fifth. Washington, D.C.: American Psychiatric Association; 2013.
6. Luscher B, Möhler H. F1000Research. 2019;8(F1000 Faculty Rev):751.
7. Maguire J. Front Cell Neurosci. 2019;13:83.
8. Northoff and Sibille. Mol. Psychiatry, 2014; doi:10.1038/mp.2014.68.
9. Hoffmann E, et al. Clin Pharmacokinet. 2019; doi:10.1007/s40262-019-00801-0.
10. Martinez Botella G, et al. Neuroactive steroids. J Med Chem. 2017;60:7810-7819.
11. Reddy DS and Estes WA. Trends in Pharmacological Sciences. 2016;37:543-561.
12. Gunduz-Bruce H, et al. N Eng J Med. 2019;381(10):903-911.
13. Deligiannidis KM et al. Poster presented at ACNP. Orlando, FL. December 8-11, 2019.

SUPPORT & DISCLOSURES
AM, RL, IN, AJS, ADC, BW, CS, JJ, SJK and HG-B are employees of Sage Therapeutics, Inc., with stock/stock options. AC reports funding from Allergan, Endoceutics, Janssen, and Sage Therapeutics, Inc.; consulting fees from Acadia, Alkermes, Allergan, AMAG Pharmaceuticals, Daré Bioscience, Fabre-Kramer, Ovoca Bio, Palatin Technologies, S1 Biopharma, Sage Therapeutics, Inc., Sprout Pharmaceuticals, Takeda, and Lundbeck; royalties from Ballantine Books/Random House, the Changes in Sexual Function Questionnaire, and Guilford Publications; and stock in Euthymics and S1 Biopharma. This study was funded by Sage Therapeutics, Inc. This poster was assembled with assistance from Boston Strategic Partners.

American Academy of Neurology Annual Meeting 2020
Toronto, Canada
MRC- 217-00612 06/2020

SAGE THERAPEUTICS is a trademark of Sage Therapeutics, Inc. All other trademarks referenced herein are the property of their respective owners.