The GABA$_A$ Receptor Positive Allosteric Modulator Zuranolone in Major Depressive Disorder: A Double-Blind, Randomized, Placebo-Controlled Phase 3 Trial

Handan Gunduz-Bruce, MD, MBA[1], Anita H. Clayton, MD[2], Robert Lasser, MD, MBA[1], Indrani Nandy, PhD[1], Abdul J. Sankoh, PhD[1], Brian Werneburg, PhD[1], Jeffrey Jonas, MD[1], Stephen J. Kanes, MD, PhD[1]

[1]Sage Therapeutics, Inc., Cambridge, MA, USA
[2]University of Virginia, School of Medicine, Charlottesville, VA, USA

Introduction

- Major depressive disorder (MDD) is characterized by discrete major depressive episodes (MDEs) of at least 2 weeks in duration that are associated with changes in affect, cognition, and function [1].
- An estimated 17 million (6.3%) of adults aged 18 and older across five European countries (United Kingdom, Germany, France, Spain, and Italy) experience at least one MDE each year [2].
- MDD is one of the largest contributors to disability worldwide [3-6].
- Gamma-aminobutyric acid (GABA)-ergic neurotransmission regulates the activity of diverse brain networks that may be implicated in depression [7-9].
- Zuranolone (ZRN; SAGE-217) is an investigational oral neuroactive steroid GABA-A receptor positive allosteric modulator [10-12].
- ZRN binds to both synaptic and extrasynaptic GABA-A receptors and has a distinct pharmacological profile from benzodiazepines, which bind to synaptic GABA-A receptors [11,13].
- In a pivotal trial in MDD (NCT03000530), ZRN demonstrated statistically significant improvements in depressive symptoms after two weeks of treatment and was generally well tolerated [13].
- The most common adverse events (≥5%) in any group were headache, dizziness, somnolence, and nausea.
- The efficacy and safety of two-week treatment with ZRN 20 mg or ZRN 30 mg were evaluated in this double-blind, randomized, placebo-controlled Phase 3 trial (MOUNTAIN study; NCT03672175) in adults with MDD.

Conclusions

- While the MOUNTAIN study did not meet its primary endpoint, statistical significance from placebo in the ZRN 30 mg group was noted at measured timepoints leading up to Day 15.
- In post hoc analyses in patients with measurable drug concentration and patients with both measurable drug concentration and a HAMD-17 total score ≥24 at baseline, ZRN 30 mg was associated with a significant mean reduction of HAMD-17 total score compared to placebo at all measured timepoints up to, and including, Day 15.
- ZRN was generally well tolerated and showed a similar safety profile as seen in earlier studies [13].
- The most common adverse events (≥5%) in any group during the two-week treatment period and the four-week follow-up were headache, dizziness, somnolence, fatigue, diarrhea, sedation, and nausea.
- Two patients receiving ZRN 30 mg experienced serious adverse events (SAEs) during treatment while three patients, one in each treatment group, reported SAEs during follow-up.
- Based on the results from this study, the ZRN LANDSCAPE program has been expanded to study a 50 mg dose regimen moving forward.

Methods

- The study enrolled 581 adult (18-65 years old) patients with MDD and of qualifying severity by the 17-item Hamilton Rating Scale for Depression total score (HAMD-17; score ≥22) and the Montgomery-Åsberg Depression Rating Scale total score (MADRS; score ≥32).
- Patients were stratified based on baseline antidepressant use and randomized 1:1:1 to receive ZRN 20 mg, ZRN 30 mg, or placebo. Blinded study drug was administered as identical gelatin capsules on an outpatient basis, daily, in the evening, for two weeks with follow-up through Day 42.
- Key exclusion criteria included attempted suicide associated with the current depressive episode, treatment resistant depression, or medical history of bipolar disorder, schizophrenia, and/or schizoaffective disorder.
- Concomitant antidepressant medication use was permitted at a stable dose from 60 days prior to Day 1 through Day 42.
- No changes in concomitant antidepressant medications were permitted from at least 60 days prior to Day 1 through the Day 42 follow-up.
- The change from baseline (CFB) in HAMD-17 total score at Day 15 was the primary endpoint.
- Secondary endpoints included the CFB in HAMD-17 total score at all other timepoints.
- Post hoc endpoints included CFB in HAMD-17 in patients with measurable drug concentration, and CFB in HAMD-17 total score in patients with measurable drug concentration and a HAMD-17 total score ≥24 at baseline.
- Secondary and post hoc analyses were not adjusted for multiplicity.
- Statistics are from a mixed effect model for repeated measures including the least-squares mean CFB in HAMD-17 at each visit as the dependent variable.
- Safety and tolerability were evaluated throughout the study by treatment emergent adverse event (TEAE) reporting, the Columbia Suicide Severity Rating Scale (C-SSRS), the 20-item Physician Withdrawal Checklist, and standard clinical assessments.

Results

STUDY DESIGN

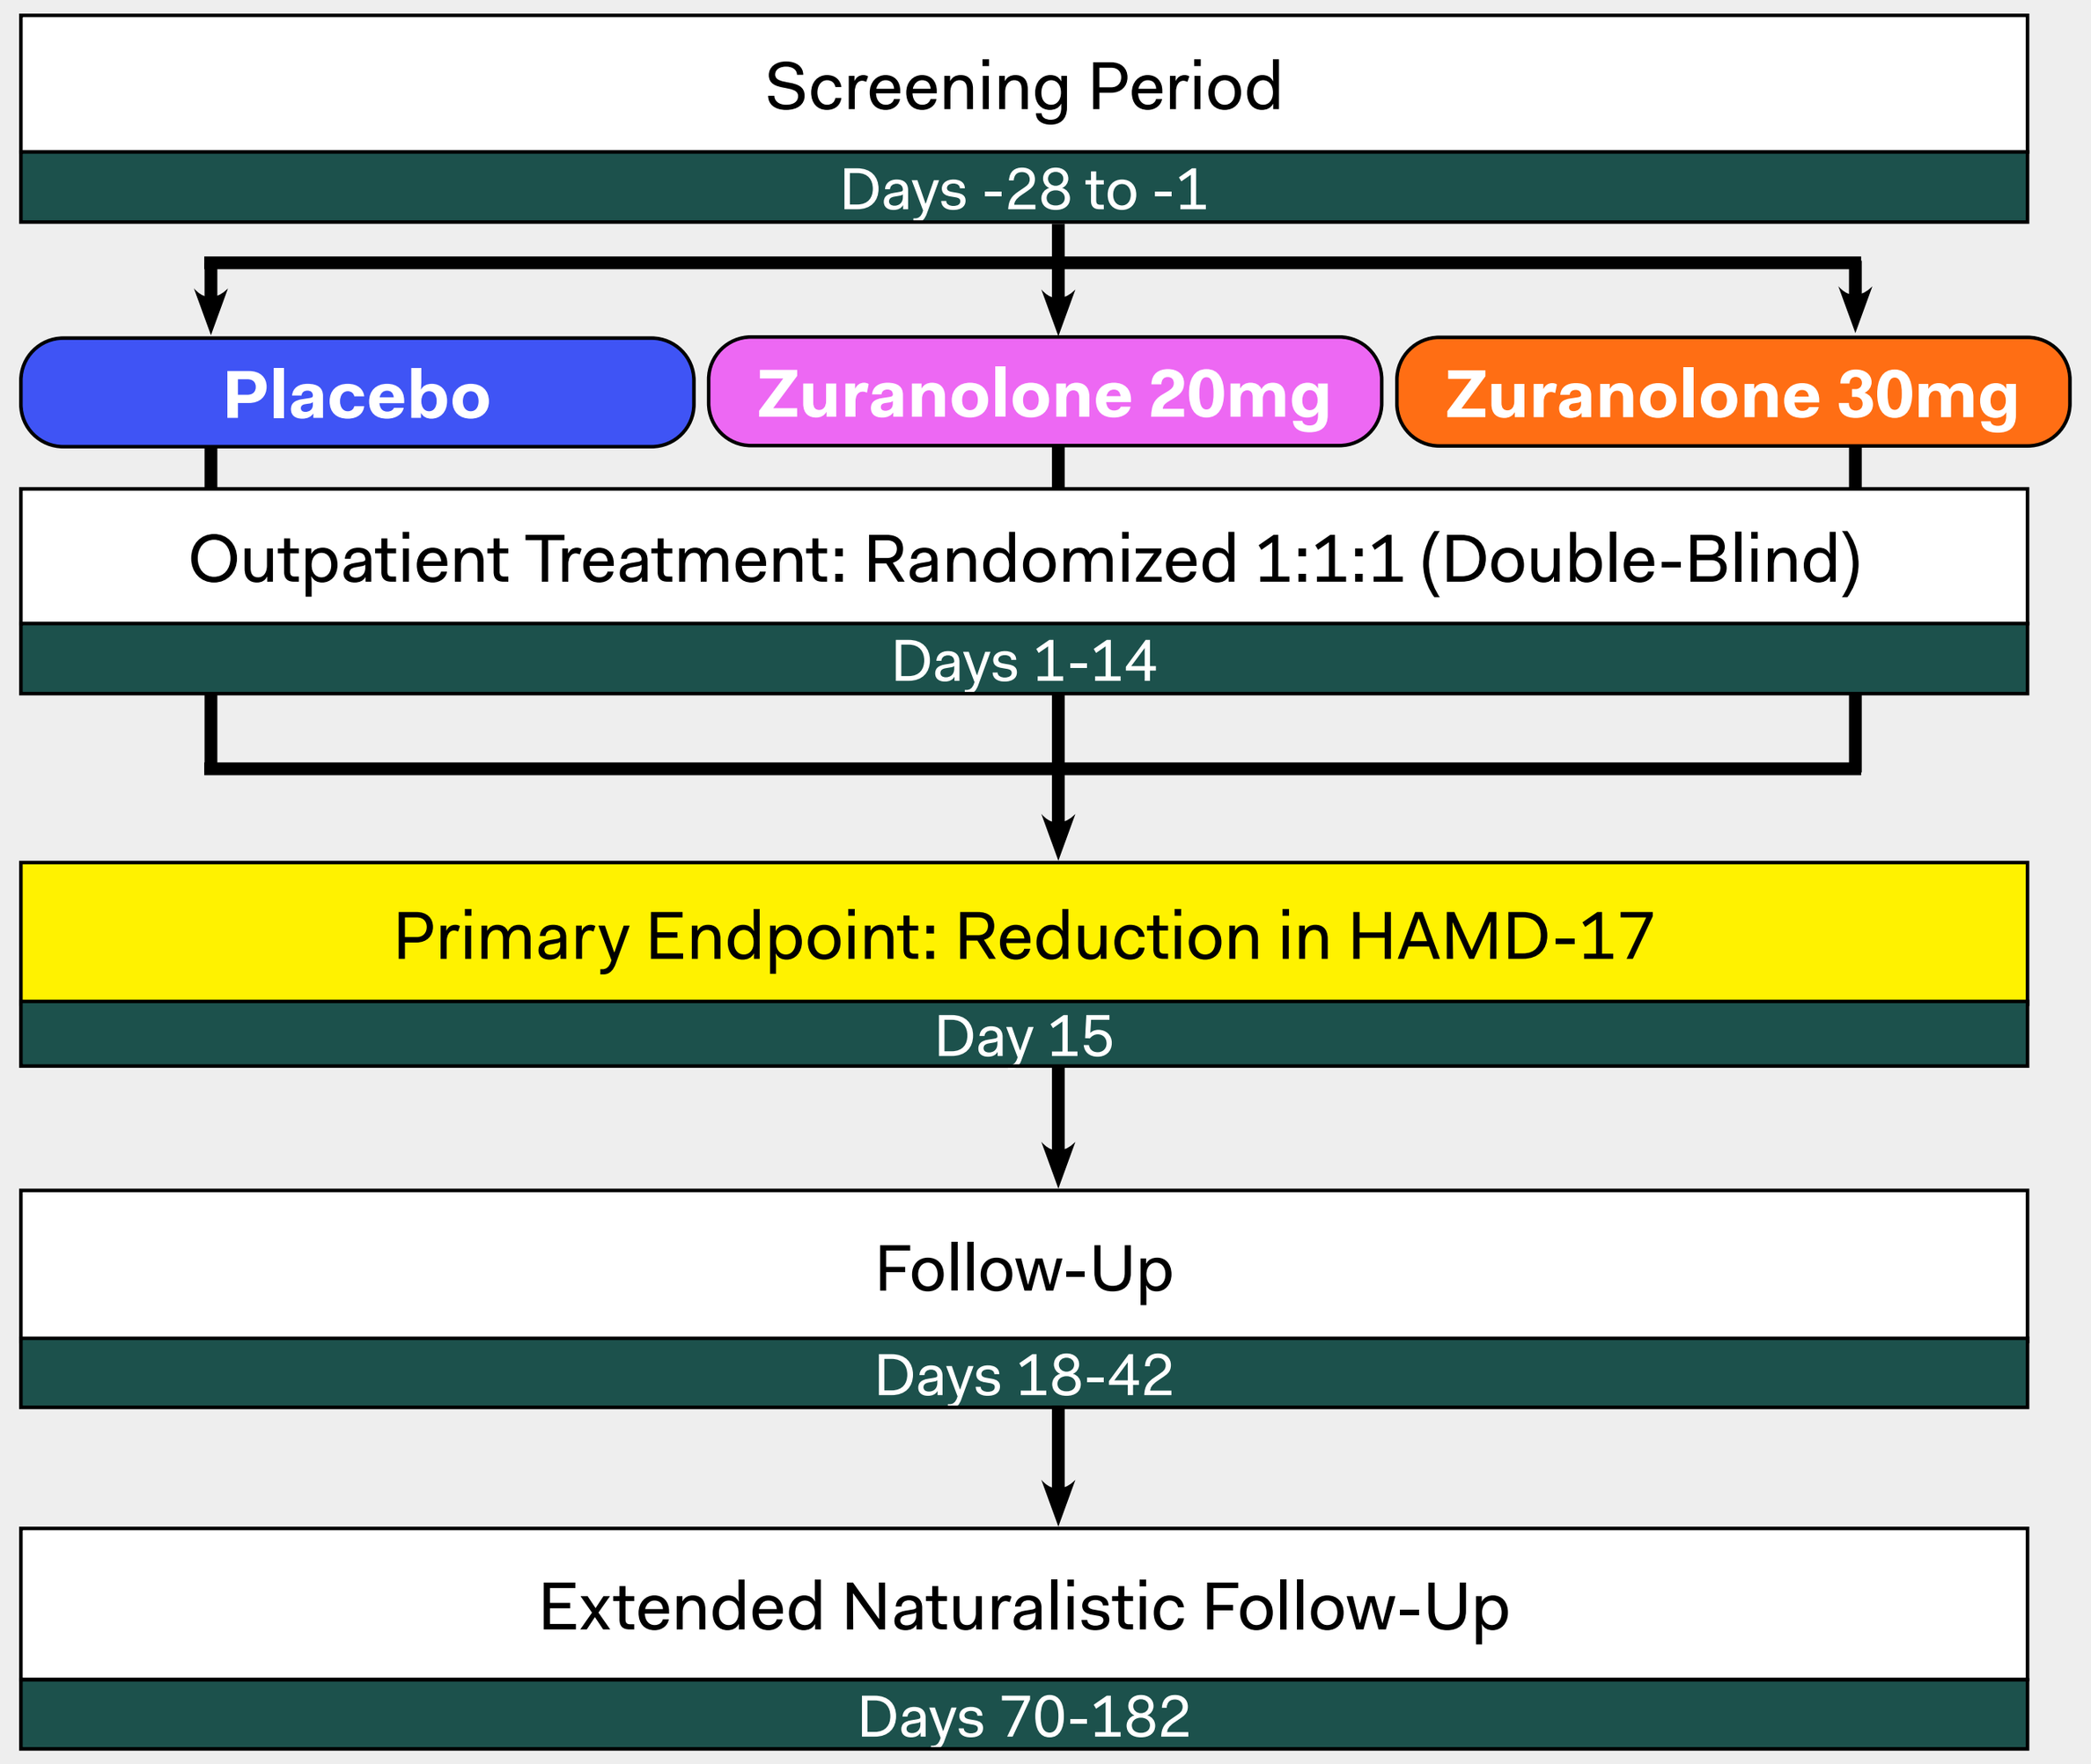
Screening Period
Days -28 to -1
Placebo
Zuranolone 20mg
Zuranolone 30mg
Outpatient Treatment: Randomized 1:1:1 (Double-Blind)
Days 1-14
Primary Endpoint: Reduction in HAMD-17
Day 15
Follow-Up
Days 18-42
Extended Naturalistic Follow-Up
Days 70-182

DEPRESSIVE SYMPTOMS OVER TIME: PRIMARY AND KEY SECONDARY ENDPOINTS

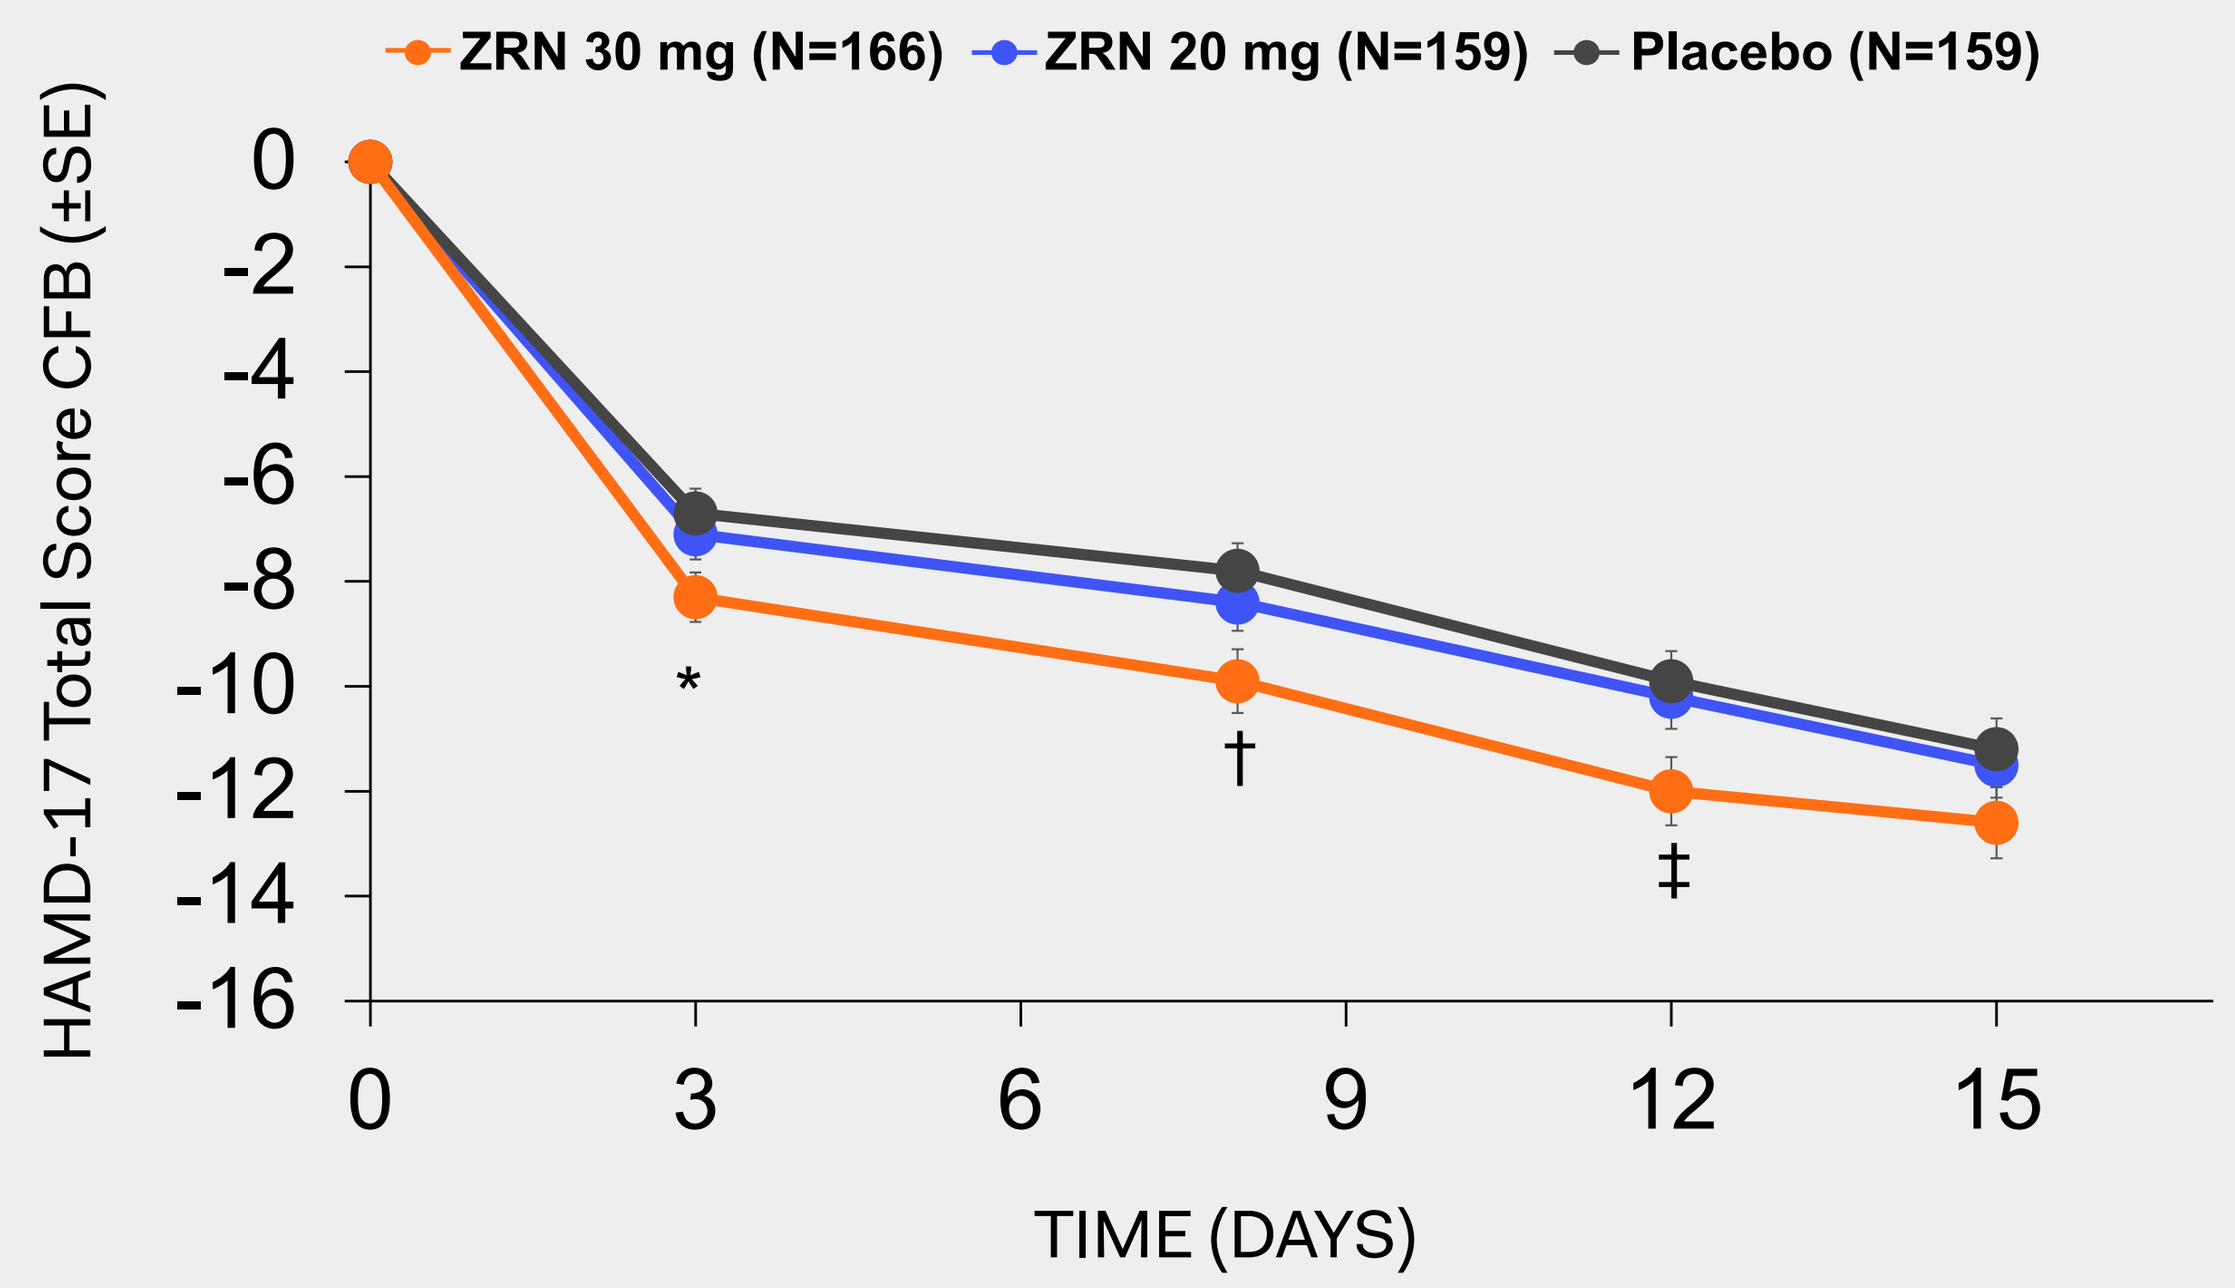
ZRN 30 mg (N=166)
ZRN 20 mg (N=159)
Placebo (N=159)
HAMD-17 Total Score CFB (±SE)
0
-2
-4
-6
-8
-10
-12
-14
-16
0
3
6
9
12
15
TIME (DAYS)
*
†
‡

*p=0.016, †p=0.008, ‡p=0.018 (p-values for ZRN 30mg vs. placebo)

- Primary endpoint was not statistically significant from placebo. HAMD-17 total score CFB at Day 15: ZRN 30 mg (-12.6) vs Placebo (-11.2; p=0.115).
- ZRN 20 mg was not statistically significant from placebo at any measured timepoint.
- Statistical significance from placebo in the ZRN 30 mg group was noted at:
 - Day 3 (p=0.016)
 - Day 8 (p=0.008)
 - Day 12 (p=0.018)

ZRN TREATMENT EMERGENT ADVERSE EVENTS

Total Number of TEAEs*	PLACEBO N = 190	ZURANOLONE 20 MG N=188	ZURANOLONE 30 MG N=192
Any TEAEs	93 (48.9)	94 (50.0)	104 (54.2)
Severe TEAEs	2 (1.1)	3 (1.6)	2 (1.0)
Serious TEAEs	1 (0.5)	1 (0.5)	3 (1.6)
TEAE-Drug Discontinued	6 (3.2)	3 (1.6)	4 (2.1)
Deaths	0 (0)	0 (0)	0 (0)
Most Common TEAEs, ≥5% Patients			
Somnolence	8 (4.2)	11 (5.9)	13 (6.8)
Headache	14 (7.4)	21 (11.2)	12 (6.3)
Dizziness	7 (3.7)	14 (7.4)	11 (5.7)
Fatigue	5 (2.6)	3 (1.6)	13 (6.8)
Diarrhea	10 (5.3)	11 (5.9)	12 (6.3)
Sedation	6 (3.2)	11 (5.9)	9 (4.7)
Nausea	9 (4.7)	10 (5.3)	7 (3.6)

Data are from the safety population and from the double-blind period, reported as N (%).

- Serious Adverse Events (SAEs):
 - Double-blind period: 5 patients overall.
 - Treatment period: 2 patients receiving ZRN 30 mg.
 - 1 suicide attempt (Day 5, patient with a longstanding history of MDD and a previous suicide attempt).
 - 1 bile duct stone (Day 2, requiring removal in a patient with a prior bile duct repair).
- Follow-up period: 3 patients, 1 in each treatment group, all occurring at least 1 week following cessation of treatment.
 - 1 syncope and associated injuries (Day 28, ZRN 30 mg, which occurred with dehydration and orthostatic hypotension during exercise in a patient with a history of bradycardia
 - Multiple SAEs (Day 39, ZRN 20 mg, which were related to medical complications of cocaine ingestion).
 - 1 suicidal ideation (Day 22, placebo).
- There were no adverse events of loss of consciousness reported.
- There was no signal for increased suicidal ideation or suicidal behavior compared to baseline, as measured by the C-SSRS.

SIGNIFICANTLY GREATER IMPROVEMENT IN DEPRESSIVE SYMPTOMS WITH ZRN 30 MG VS PLACEBO: POST HOC ENDPOINTS

Patients With Measurable Drug

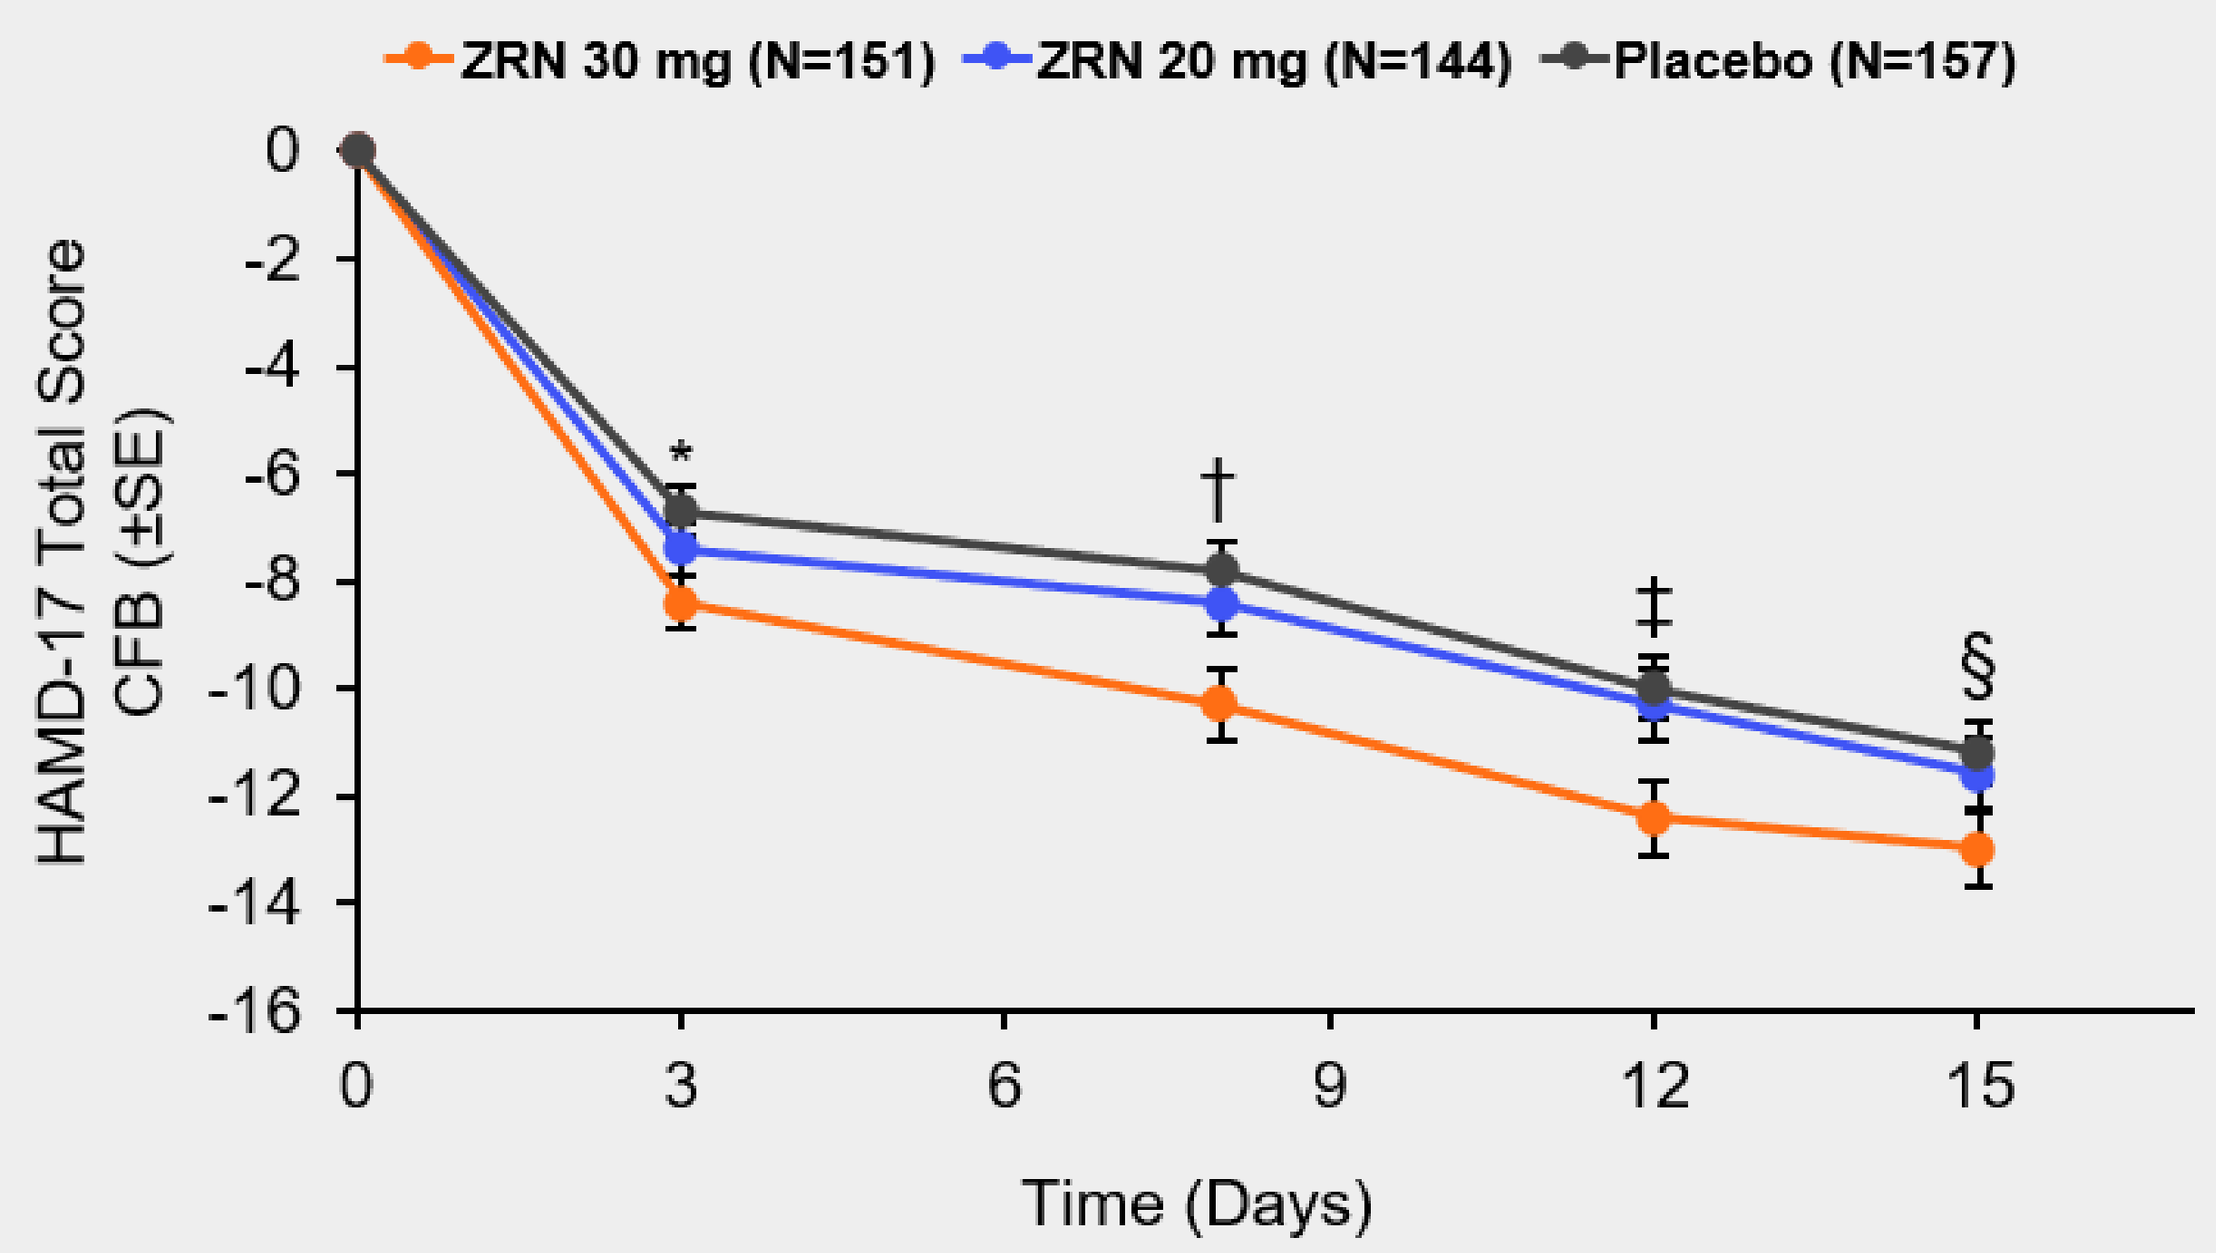
ZRN 30 mg (N=151)
ZRN 20 mg (N=144)
Placebo (N=157)
HAMD-17 Total Score CFB (±SE)
0
-2
-4
-6
-8
-10
-12
-14
-16
0
3
6
9
12
15
Time (Days)
*
†
‡
§

*p=0.011, †p=0.002, ‡p=0.006, §p=0.048; p-values shown for ZRN 30 mg vs placebo.

- 9% of patients in the ZRN 30 mg group had no measurable drug concentration, consistent with non-compliance in taking ZRN.
- Excluding these patients from the primary analysis set (ZRN 30 mg vs placebo), resulted in statistical significance at all timepoints (p<0.048).

Patients With Measurable Drug and HAMD-17 ≥24

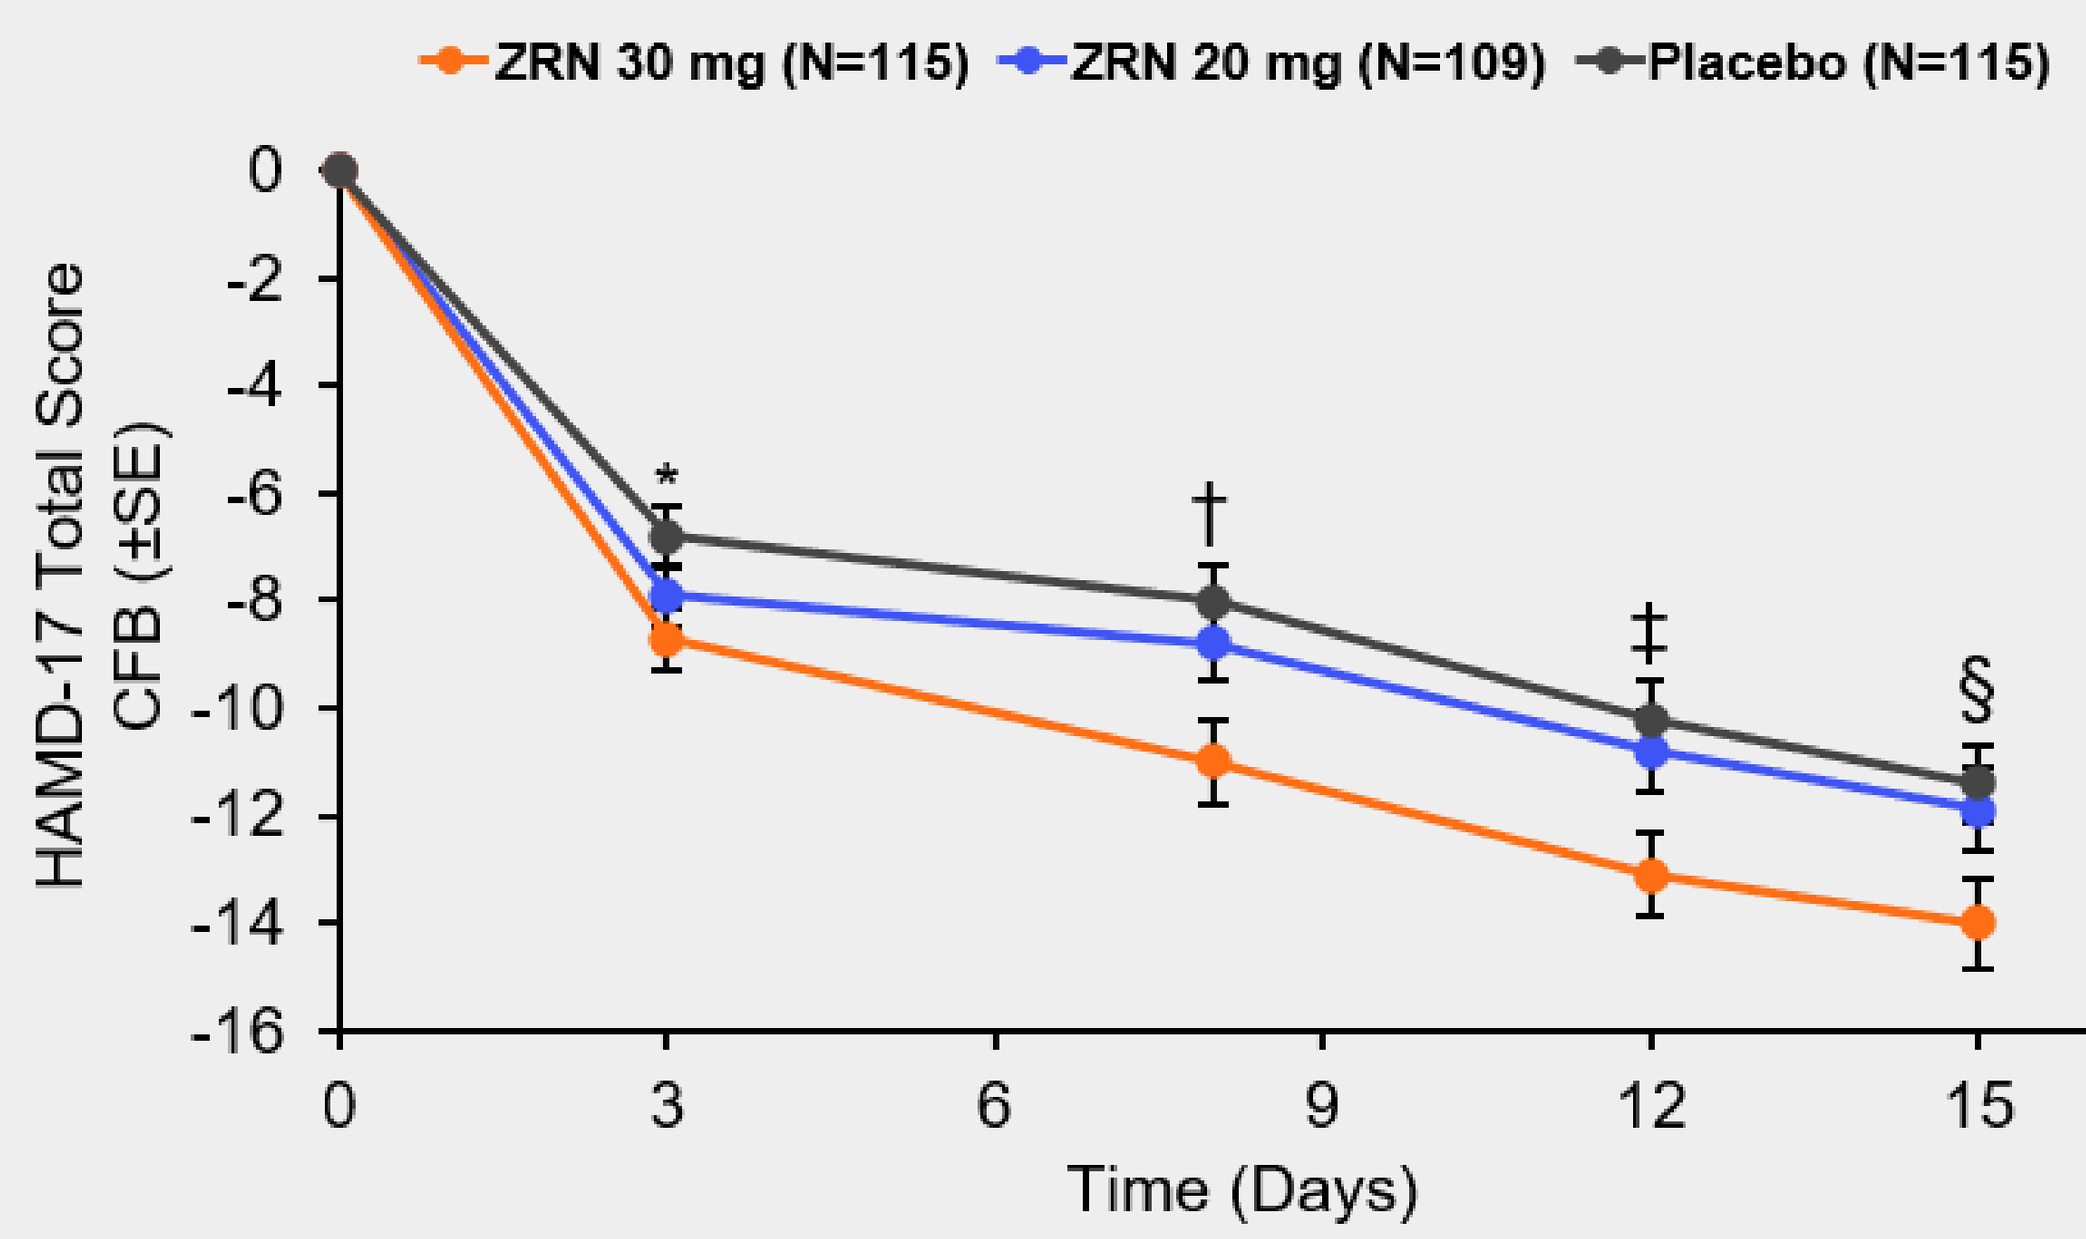
ZRN 30 mg (N=115)
ZRN 20 mg (N=109)
Placebo (N=115)
HAMD-17 Total Score CFB (±SE)
0
-2
-4
-6
-8
-10
-12
-14
-16
0
3
6
9
12
15
Time (Days)
*
†
‡
§

*p=0.015, †p=0.003, ‡p=0.005, §p=0.017; p-values shown for ZRN 30 mg vs placebo.

- More patients with an overall distribution of milder severity of symptoms were enrolled than in previous studies of ZRN.
- Including patients with measurable drug concentration (ZRN 30 mg vs placebo) and a HAMD-17 total score ≥24 at baseline, resulted in statistical significance at all time points (p<0.017).
- ZRN 20 mg was not statistically significant from placebo at any measured timepoint in either analysis.

REFERENCES
1. American Psychiatric Association. Diagnostic and Statistical Manual of Mental Disorders. Text Revision - Fifth. Washington, D.C.: American Psychiatric Association; 2013.
2. Data on File, MLIB-2623, EU-5 MDD Epidemiology. Sage Therapeutics, Inc. [2019].
3. GBD. Global, regional, and national incidence, prevalence, and years lived with disability for 354 diseases and injuries for 195 countries and territories, 1990–2017: a systematic analysis for the Global Burden of Disease Study 2017. Lancet. (2018) 392:1789-1858.
4. Depression and Other Common Mental Disorders: Global Health Estimates. Geneva: World Health Organization; 2017. License: CC BY-NC-SA 3.0 IGO.
5. Global Health Estimates 2016: Disease Burden by Cause, Age, Sex, by Country, and by Region, 2000-2016. Geneva, World Health Organization; 2018.
6. USBD. JAMA. (2018) 319:1444-1472.
7. Luscher B, Möhler H. F1000Research (2019) 8:751.
8. Maguire J. Front Cell Neurosci (2019) 13:83.
9. Northoff and Sibille. Mol. Psychiatry (2014) 19:966–9779.
10. Hoffmann E, et al. Clin Pharmacokinet (2019) 59:111–120.
11. Martinez Botella G, et al. J Med Chem (2017) 60:7810-7819.
12. Reddy DS and Estes WA. Trends in Pharmacological Sciences (2016) 37:543-561.
13. Gunduz-Bruce H, et al. N Eng J Med (2019) 381:903-911.

MOUNTAIN, SAGE THERAPEUTICS and the SAGE THERAPEUTICS logo are trademarks of Sage Therapeutics, Inc. All other trademarks referenced herein are the property of their respective owners.

SUPPORT & DISCLOSURES
Handan Gunduz-Bruce and the authors RL, IN, AJS, BW, JJ, and SJK are employees of Sage Therapeutics, Inc., with stock/stock options. AHC reports funding from Allergan, Endoceutics, Janssen, and Sage Therapeutics, Inc.; consulting fees from Acadia, Alkermes, Allergan, AMAG Pharmaceuticals, Daré Bioscience, Fabre-Kramer, Ovoca Bio, Palatin Technologies, S1 Biopharma, Sage Therapeutics, Inc., Sprout Pharmaceuticals, Takeda, and Lundbeck; royalties from Ballantine Books/Random House, the Changes in Sexual Function Questionnaire, and Guilford Publications; and stock in Euthymics and S1 Biopharma. This study was funded by Sage Therapeutics, Inc. This study and medical writing support were funded by Sage Therapeutics, Inc. Writing and editorial support was provided by Elizabeth Wheatley of Boston Strategic Partners.

European College of Neuropsychopharmacology
ECNP 2020 Annual Meeting
iPoster Session

MRC-217-00641 09/20