Anxiety Symptom Response in Patients with Postpartum Depression Treated with the Neuroactive Steroid Brexanolone Injection

Sage Therapeutics™

C. Neill Epperson, MD[1], Brian Werneburg, PhD[2], David R. Rubinow, MD[3], Kristina M. Deligiannidis, MD[4], Robert Riesenberg, MD[5], Kemi Bankole, MD[2], Ming-Yi Huang[2], Haihong Li, PhD[2], Stephen J. Kanes, MD, PhD[2], Samantha Meltzer-Brody, MD, MPH[3]

[1] Department of Psychiatry, University of Colorado School of Medicine, Aurora, CO, USA
[2] Sage Therapeutics, Inc., Cambridge, MA, USA
[3] Department of Psychiatry, University of North Carolina School of Medicine, Chapel Hill, NC, USA
[4] Department of Psychiatry, Women's Behavioral Health, Zucker Hillside Hospital, Glen Oaks, NY, USA
[5] Atlanta Center for Medical Research, Atlanta, GA, USA

Introduction

- Postpartum depression (PPD) is one of the most common medical complications during and after pregnancy.[1-7]
- Anxiety is a prominent symptom of PPD and has been associated with more severe disease.[8-10]
- Brexanolone injection (BRX),[11] a neuroactive steroid $GABA_A$ receptor positive allosteric modulator with a pharmacological profile distinct from benzodiazepines[12],is approved by the FDA for the treatment of PPD in adults.[11]
- The safety, tolerability, and efficacy of BRX in PPD were evaluated in three double-blind randomized placebo-controlled trials (RCTs), where patients treated with BRX90 met the primary efficacy endpoint at Hour 60, by achieving significantly greater 17-item Hamilton Rating Scale for Depression total score (HAMD-17)[13] change from baseline (CFB) vs patients receiving placebo (p<0.0001).[14]
- In addition, improvement from baseline in the core symptoms of depression using the predefined Bech-6 subscale of HAMD-17 favored BRX90 compared with placebo beginning at Hour 24 (p=0.0173) and at all measured timepoints to Day 30 (p=0.0301).[14,15]
- Post hoc analyses from an integrated dataset of the three pivotal BRX RCTs assessed the effect on anxiety symptoms using the HAMD-17 anxiety/somatization subscale (A/S).

Conclusions

- BRX treatment in women with PPD has previously been shown to provide rapid (Hour 24 for HAMD-17 total score and Bech-6 score) and sustained improvement in depressive symptoms (HAMD-17 total score and Bech-6 score at all subsequent measured time points up to Day 30) compared with placebo.[14,15]
- In addition to its effects on core symptoms of depression in this trial, these post hoc analyses showed that BRX treatment also resulted in significantly greater improvement in reported symptoms of anxiety from the HAMD-17-A/S, and higher proportions of patients who achieved HAMD-17-A/S response compared with placebo.

Methods

- The integrated dataset of three pivotal studies in PPD was used for these analyses, with the pooled dataset referring to all patients receiving BRX in the first 24 hours regardless of final maximum dose received.
- Women (N=247) ages 18-45, ≤6 months postpartum, with PPD (defined as a major depressive episode with onset in the 3rd trimester or ≤4 weeks postpartum) and a qualifying HAMD-17 total score (Studies A and B: HAMD-17≥26; Study C: HAMD-17 20-25) were enrolled.
- Studies A and C used 1:1 randomization to placebo and BRX 90 µg/kg/h (BRX90), and Study B used 1:1:1 randomization to placebo, BRX 60 µg/kg/h (BRX60), and BRX90.
- All BRX patients received 30 µg/kg/h for the first 4 hours titrated to 60 µg/kg/h for the next 20 hours. For the next 28 hours, BRX60 patients received 60 µg/kg/h and BRX90 patients received 90µg/kg/h. All BRX patients were then titrated down to 60 µg/kg/h for 4 hours and 30mµg/kg/h for 4 hours until the infusion was completed.
- Patients received a 60-hour continuous infusion of placebo or BRX in a monitored setting, with four weeks follow-up.
- The primary endpoint in all studies was the least-squares mean change from baseline (CFB) in mean HAMD-17 total score at Hour 60.
- HAMD-17 CFB data was analyzed using a mixed effects model for repeated measures.
- Treatment emergent adverse events (TEAEs) were assessed throughout the study.
- Post hoc analyses included the CFB in the HAMD-17 anxiety/somatization subscale (HAMD-17-A/S, which includes six items from HAMD-17: anxiety (psychic), anxiety (somatic), somatic symptoms (gastrointestinal), somatic symptoms (general), hypochondriasis, and insight) and HAMD-17-A/S response (≥50% reduction from baseline score).
- Response assessments were analyzed using a generalized estimating equation approach.
- Secondary endpoints and post hoc analyses were not adjusted for multiplicity.

Results

SIGNIFICANTLY GREATER IMPROVEMENT IN DEPRESSIVE SYMPTOMS ACHIEVED WITH BRX VS PLACEBO

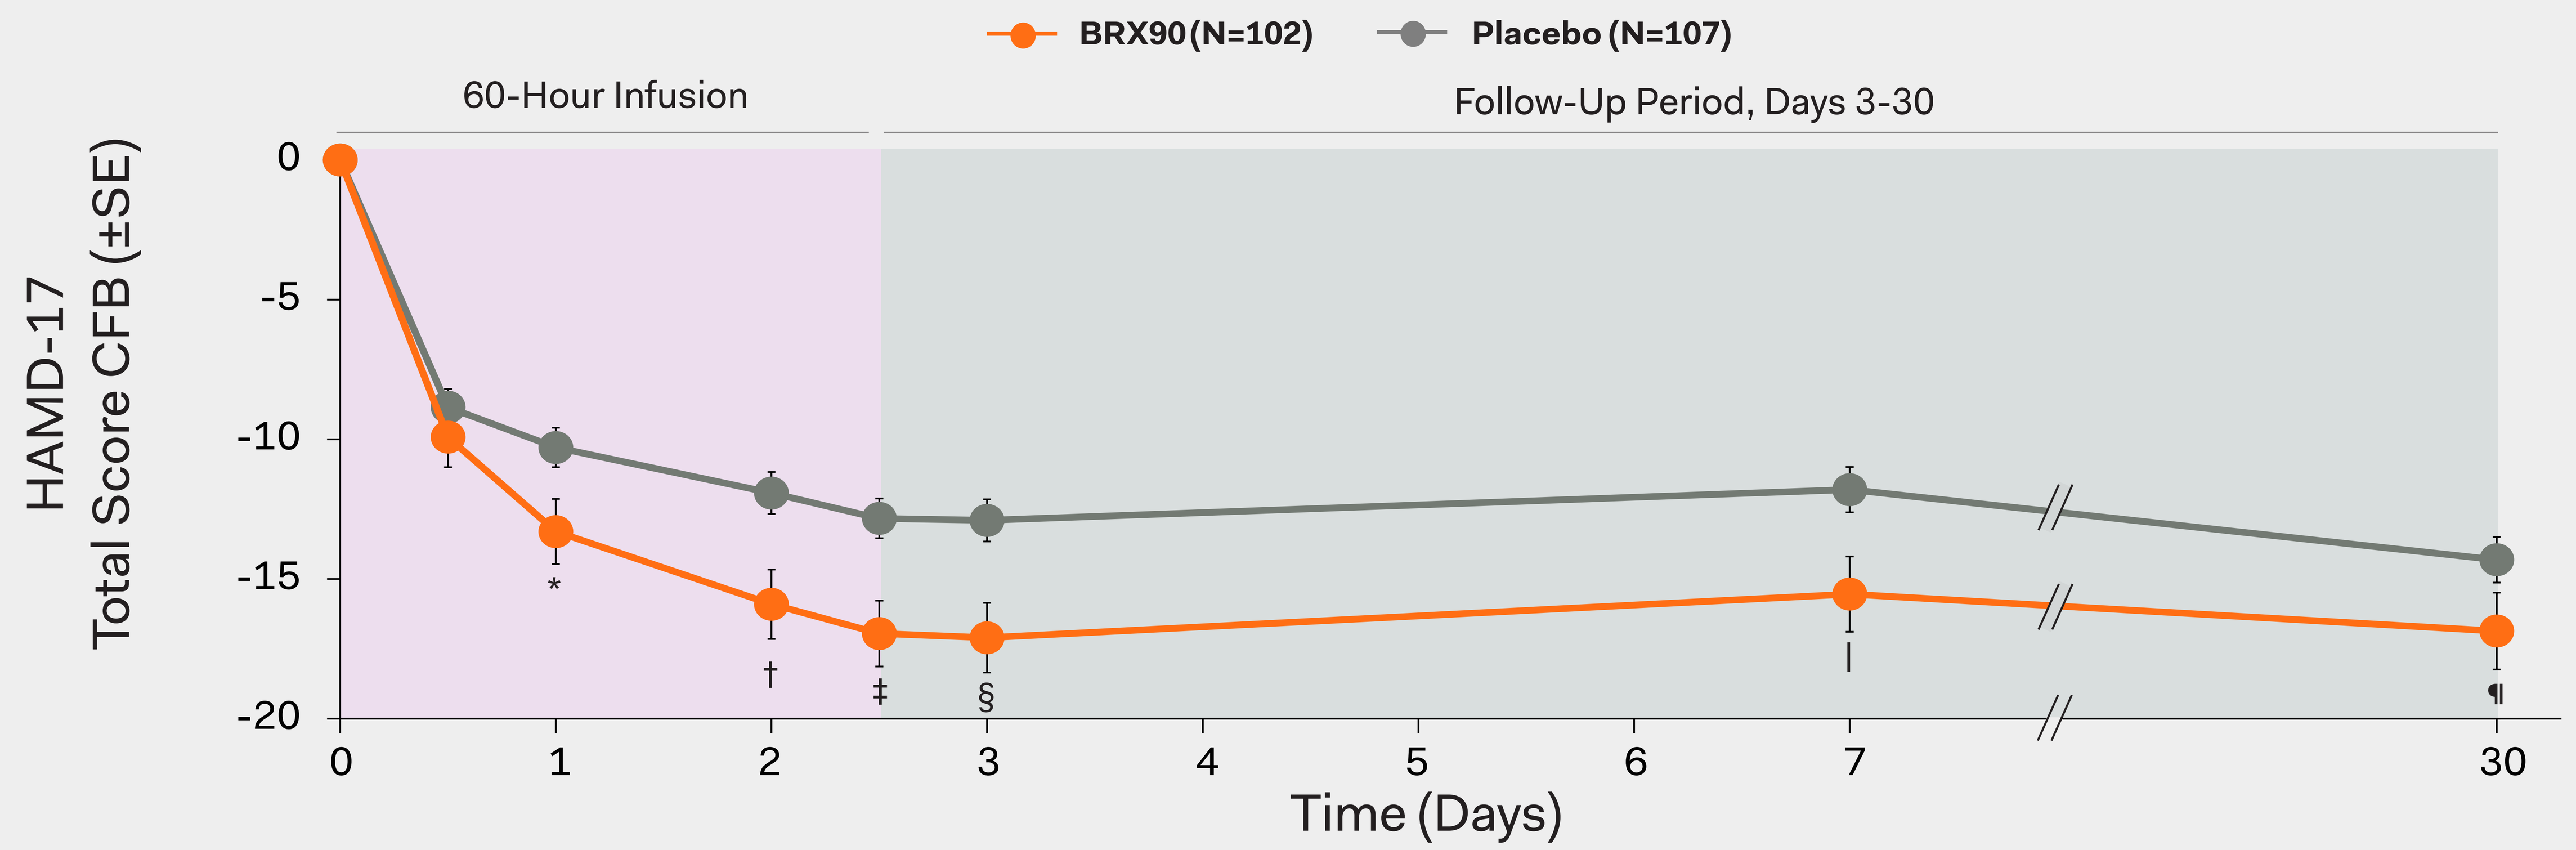
BRX90 (N=102)
Placebo (N=107)
60-Hour Infusion
Follow-Up Period, Days 3-30
HAMD-17
Total Score CFB (±SE)
0
-5
-10
-15
-20
*
†
‡
§
|
¶
0
1
2
3
4
5
6
7
30
Time (Days)

*p=0.0012; †p<0.0001; ‡p<0.0001; §p=0.0001; |p=0.0007; ¶p=0.0213 vs placebo.

- Baseline demographics and patient characteristics were well balanced between the treatment arms and have been described in detail previously.[14,15]
- As previously presented, patients treated with BRX90 met the primary efficacy endpoint at Hour 60, by achieving significantly greater HAMD-17 total score CFB vs patients receiving placebo (p<0.0001).[14,15]
- Significant differences vs placebo in HAMD-17 CFB favoring BRX90 were observed at Hour 24 (p=0.0012) and were sustained at all measured timepoints until Day 30 (p=0.0213).

SAFETY

- BRX common TEAEs (≥5% of BRX and ≥2x placebo rate) included sedation and/or somnolence, dry mouth, loss of consciousness and flushing/hot flush.[14,15]
- Detailed adverse event rates including events of sedation/somnolence and loss of consciousness have been described in detail previously.[14,15]
- BRX caused sedation/somnolence requiring dose interruption/reduction in some patients (5% of BRX compared with 0% of placebo-treated patients). Some patients also had loss/altered state of consciousness (4% of BRX compared with 0% of placebo-treated patients). Time to recovery from loss/altered state of consciousness, after dose interruption, ranged from 15 to 60 minutes. All patients recovered with dose interruption.[14,15]

SIGNIFICANTLY GREATER IMPROVEMENT IN ANXIETY-RELATED SYMPTOMS WITH BRX VS PLACEBO

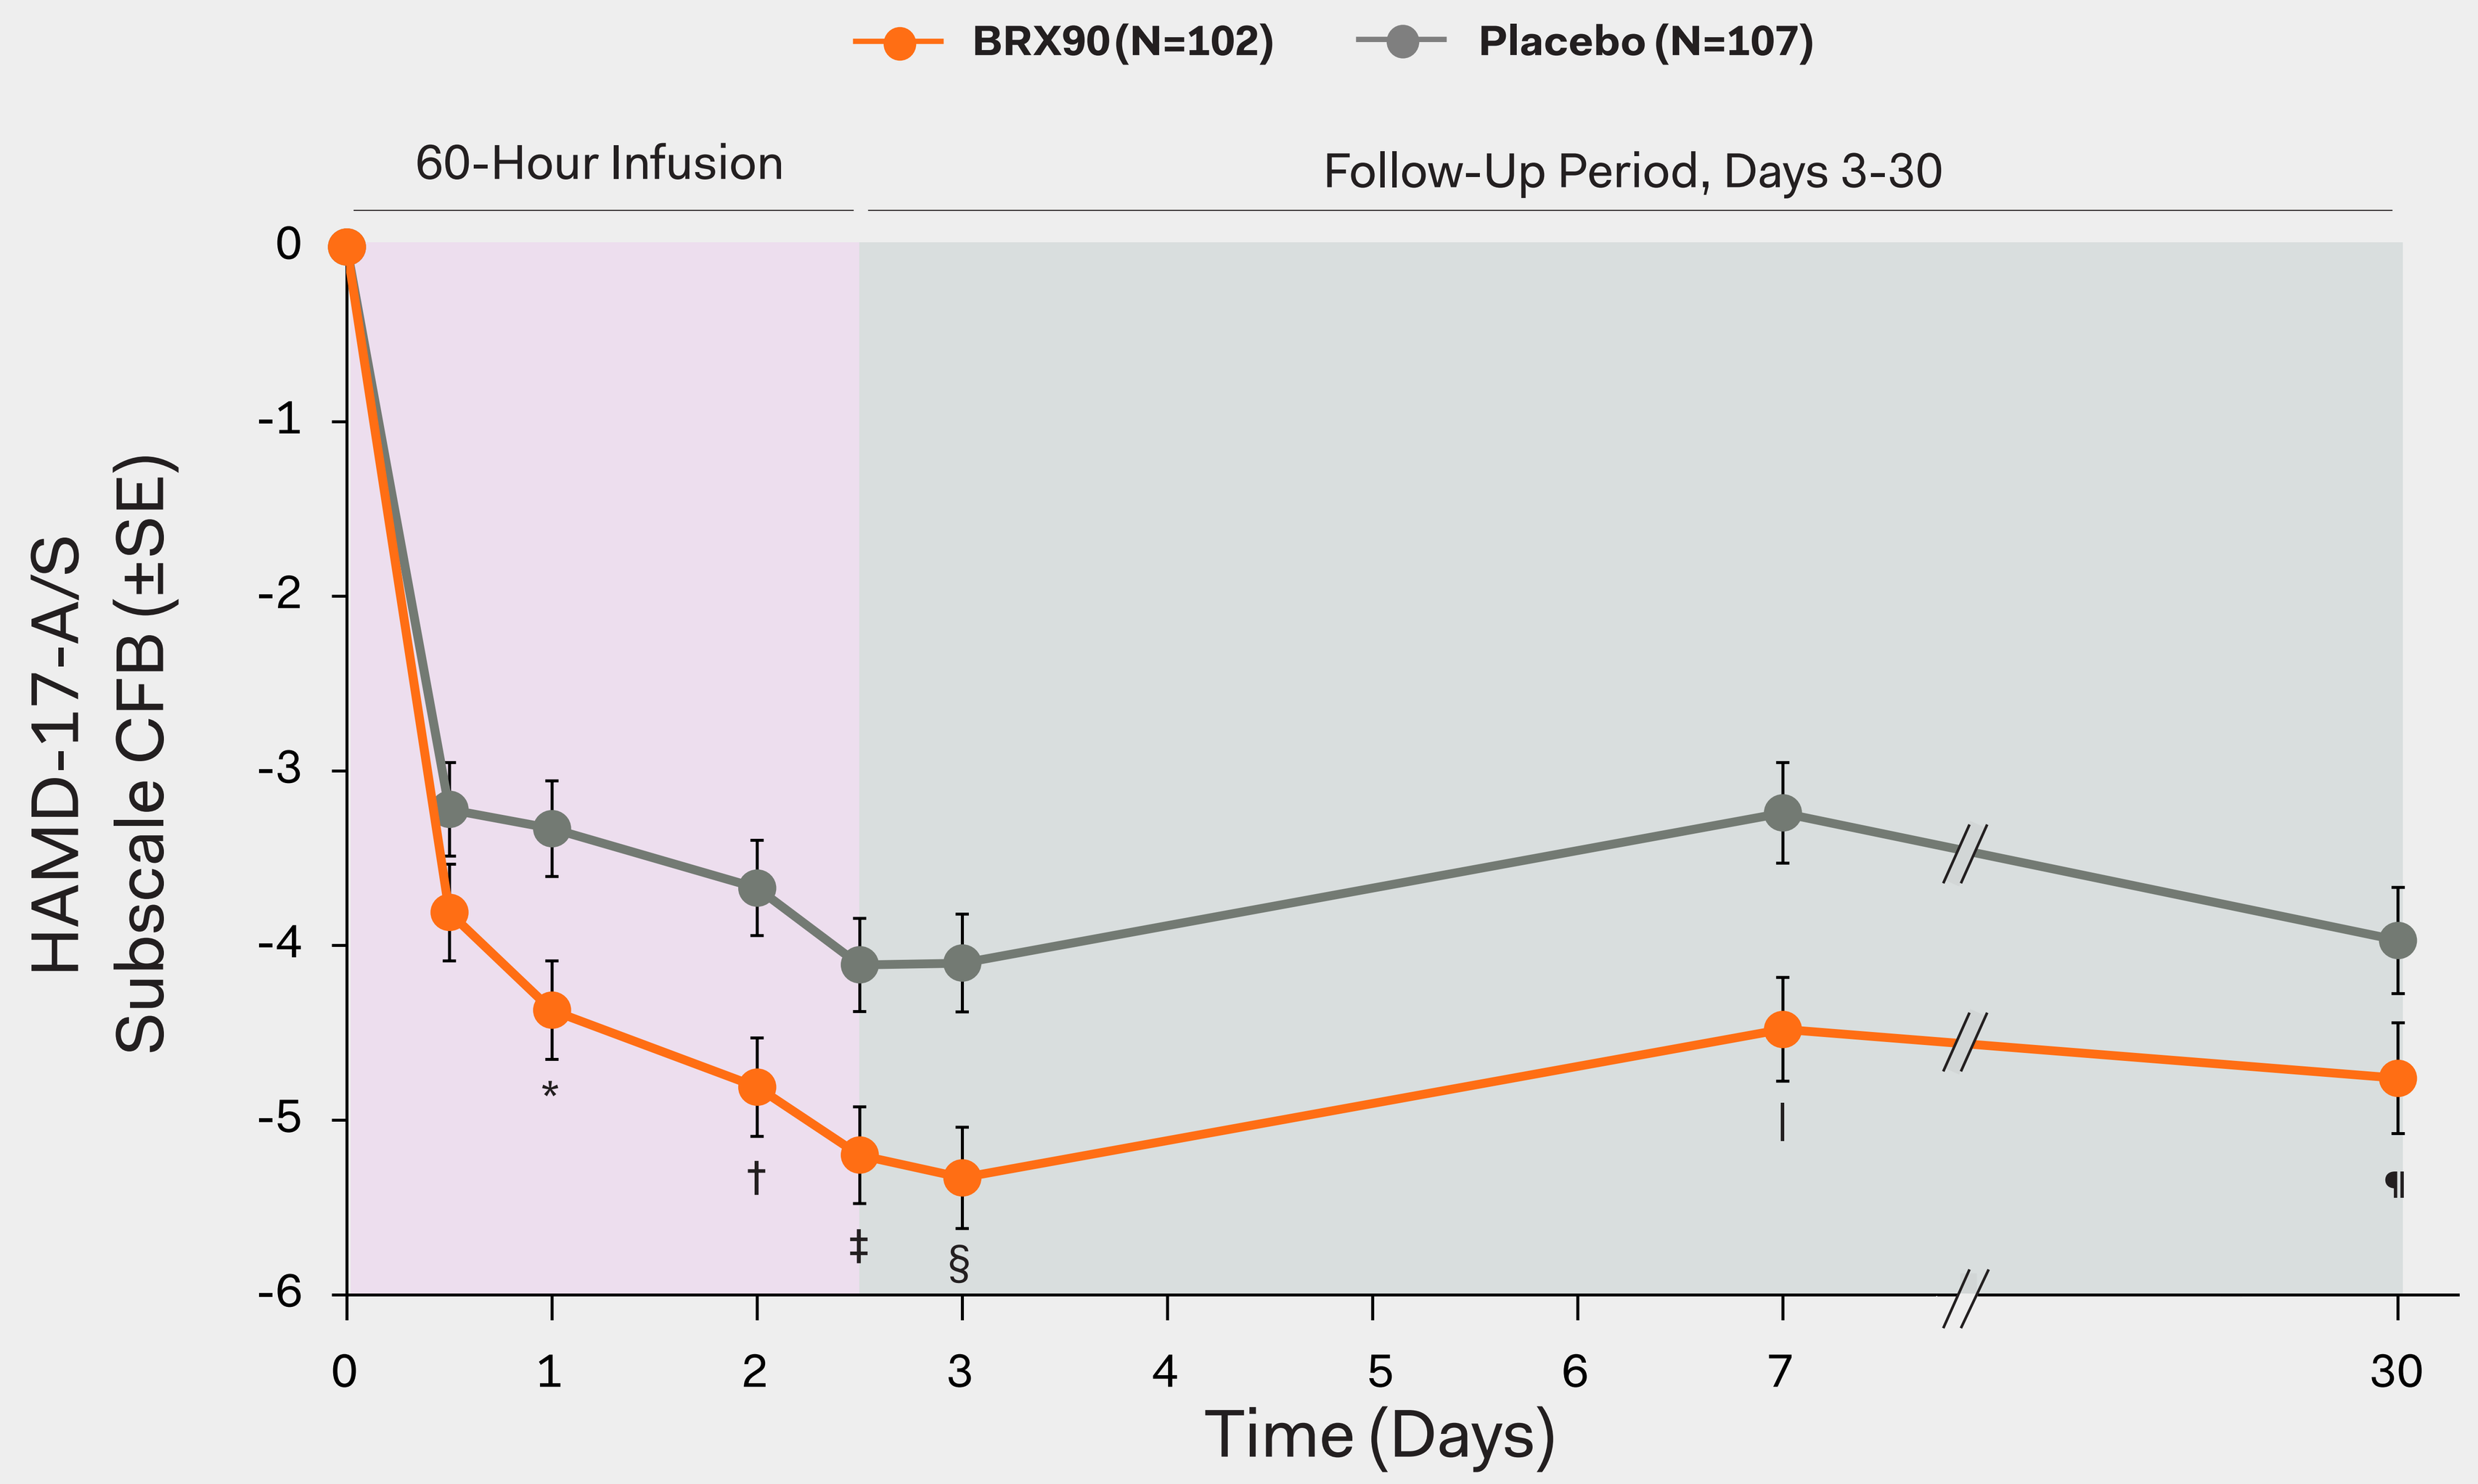
BRX90 (N=102)
Placebo (N=107)
60-Hour Infusion
Follow-Up Period, Days 3-30
HAMD-17-A/S
Subscale CFB (±SE)
0
-1
-2
-3
-4
-5
-6
*
†
‡
§
|
¶
0
1
2
3
4
5
6
7
30
Time (Days)

*p=0.0030; †p=0.0011; ‡p=0.0014; §p=0.0007; |p=0.0009; ¶p=0.00490 vs placebo.

- Baseline mean (±SD) HAMD-17-A/S scores were high in the BRX90 group (7.78±2.01) and in the placebo group (8.05±2.06).
- Significant differences vs placebo in HAMD-17-A/S CFB favoring BRX90 were observed at Hour 24 (p=0.0030) and were sustained at all measured timepoints until Day 30 (p=0.0490).

SIGNIFICANTLY GREATER IMPROVEMENT IN HAMD-17-A/S RESPONSE WITH BRX VS PLACEBO

HAMD-17-A/S RESPONSE
≥50% REDUCTION FROM BASELINE

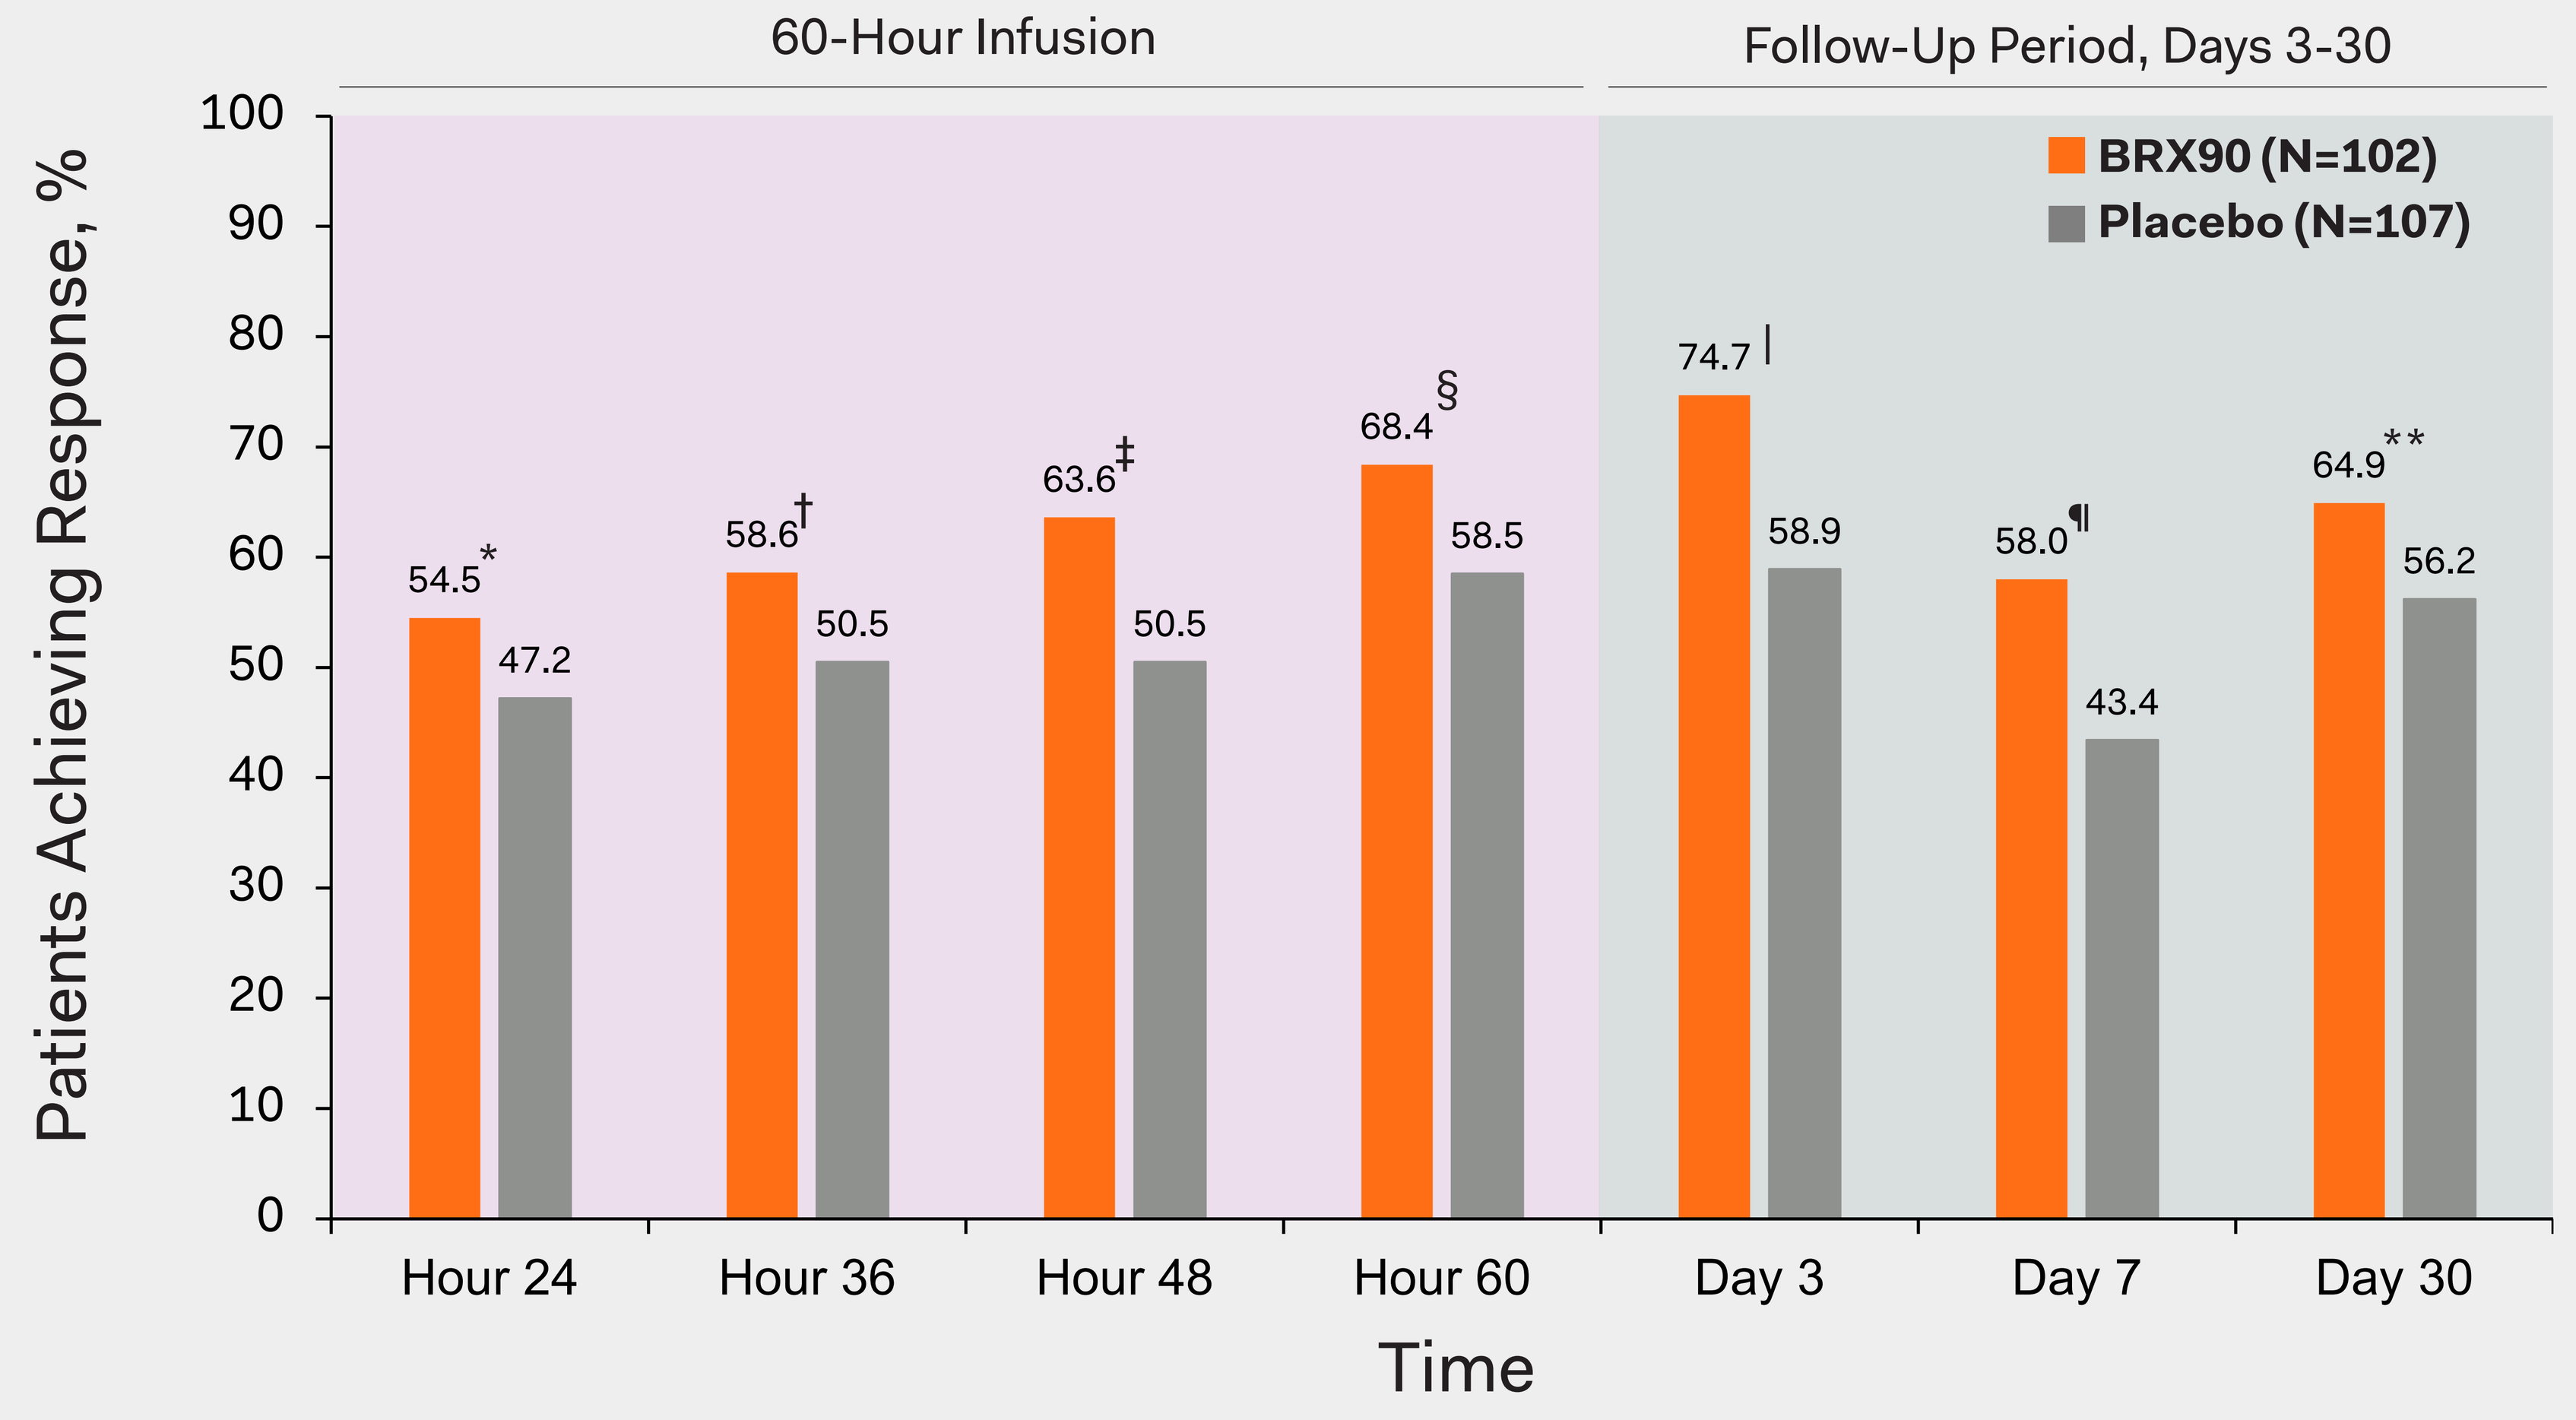
60-Hour Infusion
Follow-Up Period, Days 3-30
BRX90 (N=102)
Placebo (N=107)
Patients Achieving Response, %
100
90
80
70
60
50
40
30
20
10
0
54.5*
47.2
58.6†
50.5
63.6‡
50.5
68.4§
58.5
74.7|
58.9
58.0¶
43.4
64.9**
56.2
Hour 24
Hour 36
Hour 48
Hour 60
Day 3
Day 7
Day 30
Time

*p=0.0394; †p=0.0461; ‡p=0.0053; §p=0.0222; |p=0.0012; ¶p=0.0045; **p=0.0425 vs placebo.

- A significantly greater proportion of BRX90 patients achieved HAMD-17-A/S response compared with placebo at Hour 24 (p=0.0394) and were sustained at all measured time points until Day 30 (p=0.0425).

REFERENCES
1. Bauman BL et al. *MMWR Morb Mortal Wkly Rep* (2020) 69:575-581.
2. Hamilton BE, et al. National Center for Health Statistics. Births: Provisional data for 2018. Vital Statistics Rapid Release; no 7. https://www.cdc.gov/nchs/data/vsrr/vsrr-007-508.pdf. Published May 2019. Accessed November 10, 2020.
3. DeSisto CL, et al. *Prev Chronic Dis* (2014) 11:E10.
4. Centers for Disease Control and Prevention. https://www.cdc.gov/reproductivehealthmaternalinfanthealth/diabetes-during-pregnancy.htm. Updated June 12, 2018 Accessed November 10, 2020.
5. Centers for Disease Control and Prevention. https://www.cdc.gov/reproductivehealth/maternalinfanthealth/pregnancy-complications-data.htm. Updated February 28, 2019. Accessed November 10, 2020.
6. Roberts JM, et al. American College of Obstetricians and Gynecologists Task Force on Hypertension in Pregnancy. https://www.acog.org/~/media/Task%20Force%20and%20Work%20Group%20Reportspublic/HypertensioninPregnancy.pdf. Published 2013. Accessed November 10, 2020.
7. Callaghan WM, et al. *Am J Obstet Gynecol* (2010) 202:353.e1-6.
8. Hendrick V, et al. *Depress Anxiety* (2000) 11:66-72.
9. Fox M, et al. *Depress Anxiety* (2018) 35:292-304.
10. Postpartum Depression: Action Towards Causes and Treatment (PACT) Consortium. Heterogeneity of postpartum depression: a latent class analysis. *Lancet Psychiatry* (2015) 2:59-67.
11. ZULRESSO™ (brexanolone) injection, for intravenous use. Sage Therapeutics, Inc., Cambridge, MA. Revised: 06/2019.
12. Martinez Botella G, et al. *J Med Chem* (2015) 58:3500-3511.
13. Hamilton M. *J Neurol Neurosurg Psychiatry* (1960) 23:56-62.
14. Meltzer-Brody S, et al. *Lancet* (2018) 392:1058-1070.
15. Meltzer-Brody S, et al. Poster presented at ASCP 2020.

SAGE THERAPEUTICS and the SAGE THERAPEUTICS logo are trademarks of Sage Therapeutics, Inc. All other trademarks referenced herein are the property of their respective owners.

SUPPORT & DISCLOSURES
BW, KB, MYH, HL, and SJK are employees of Sage Therapeutics, Inc., with stock/stock options. CNE has received consultation fees from Sage Therapeutics, Inc. DRR is a member of the Sage Therapeutics, Inc. Clinical Advisory Board and has stock/stock options. KMD serves as a consultant to Sage Therapeutics, Inc., receives NIMH support and royalties from an NIH employee invention, and reports grants from Sage Therapeutics, Inc. awarded to Zucker Hillside Hospital during the conduct of the brexanolone injection clinical trials. RR has nothing to disclose. SM-B reports personal fees from MedScape and grants from Sage Therapeutics, Inc. awarded to the University of North Carolina (Chapel Hill, NC, USA) during the conduct of the brexanolone injection clinical trials and grants from Janssen, PCORI, and the NIH outside of the submitted work. This study and medical writing support were funded by Sage Therapeutics, Inc. Writing and editorial support was provided by Bonnie L. Bullock, PhD and Kathryn P. Wall, PhD of Boston Strategic Partners, Inc.

American College of Neuropsychopharmacology 2020
Virtual Annual Meeting
December 6-9, 2020

MRC-ZUL-00148 11/2020