Sage Therapeutics™

Concurrent Improvement of Depressive and Anxiety Symptoms in Patients with Postpartum Depression Treated with the Oral Neuroactive Steroid Zuranolone

Kristina M. Deligiannidis, MD[1], Brian Werneburg, PhD[2], Ming-Yi Huang, PhD[2], Ellison Suthoff, BS, MBA[2], Robert Lasser, MD, MBA[2], Handan Gunduz-Bruce, MD, MBA[2], Sarah Acaster, MSc[3], Moshe Fridman, PhD[4], Vijayveer Bonthapally, PhD[2], Stephen J. Kanes, MD, PhD[2]

[1] Department of Psychiatry, Women's Behavioral Health, Zucker Hillside Hospital, Glen Oaks, NY
[2] Sage Therapeutics, Inc., Cambridge, MA
[3] Acaster Lloyd Consulting Ltd., London, UK
[4] AMF Consulting, Los Angeles, CA

Introduction

- Postpartum depression (PPD) is one of the most common medical complications during and after pregnancy.[1-7]
- In the United States, estimates of new mothers experiencing PPD[8] symptoms each year vary by state from 9.7% to 23.5%, with an average of 13.2%.[1]
- PPD is associated with a significant impairments in mother-infant bonding[9] and maternal function,[10, 11] including breastfeeding[12] and caring for the child,[11,13] with implications for the child's health and development.[14-17]
- Multiple environmental and biological risk factors have been proposed to play a role in the development of PPD.[18-20]
- Zuranolone (SAGE-217) is an investigational oral neuroactive steroid $GABA_A$ receptor positive allosteric modulator, that binds to both synaptic and extrasynaptic $GABA_A$ receptors, distinct from benzodiazepines, which bind to synaptic $GABA_A$ receptors.[21-23]
- In a double-blind, randomized, placebo-controlled Phase 3 trial in adult women with PPD (NCT02978326), zuranolone achieved the primary endpoint of a statistically significant reduction in symptoms of depression compared to placebo assessed by the 17-item clinician-rated Hamilton Rating Scale for Depression (HAMD-17) at Day 15.[24,25]
- These post hoc analyses examined concurrent improvement of depressive and anxiety symptoms at Day 15 and Day 45 (trial follow-up).

Conclusions

- Zuranolone treatment in patients with PPD has previously been shown to provide rapid (HAMD-17 total score by Day 3) and sustained (HAMD-17 total score at all measured timepoints through Day 45) improvement in depressive symptoms.[25]
- Post hoc analyses showed that a higher proportion of zuranolone-treated patients achieved concurrent improvement in core symptoms of depression and anxiety symptoms compared with placebo at both Day 15 and at Day 45, as defined by HAMD-17/HAM-A or MADRS/HAM-A scores.

Methods

- Women (N=151), ages 18-45, ≤6 months postpartum, diagnosed with PPD (defined as a major depressive episode with onset in the 3rd trimester or ≤4 weeks postpartum), and a baseline HAMD-17 ≥26, were administered study drug in an outpatient setting.
- Patients were randomized 1:1 to receive zuranolone 30 mg or placebo once daily for 14 days, with 4 weeks follow-up.
- The change from baseline (CFB) in HAMD-17 at Day 15 was the primary endpoint.
- Secondary endpoints included the CFB in the HAMD-17 at all other measured timepoints aside from the primary endpoint on Day 15, CFB in the Hamilton Rating Scale for Anxiety (HAM-A), and CFB in the Montgomery–Åsberg Depression Rating Scale (MADRS).
- Treatment emergent adverse events (TEAEs) were assessed throughout the study.
- Post hoc analyses explored concurrent improvement of depressive and anxiety symptoms using a combination of scales: HAMD-17 and HAM-A, or MADRS and HAM-A, at Days 15 and 45.
- Depression improvement was defined as either a HAMD-17 total score ≤7, or a MADRS total score ≤10. Improvement in anxiety symptoms was defined as a HAM-A total score ≤7.
- Concurrent improvement was defined using two scale combinations: a HAMD-17 total score ≤7 and a HAM-A total score ≤7, or a MADRS total score ≤10 and a HAM-A total score ≤7.
- CFB in HAMD-17, HAM-A and MADRS total scores were evaluated using the least-squares mean from a mixed-effect model for repeated measures. Concurrent improvement rates were assessed using Fisher's exact test, while estimates for odds ratios (ORs) and 95% confidence intervals (CIs) for ORs were derived using generalized estimating equations models for repeated measures, adjusting for baseline covariates.
- Secondary endpoints and post hoc analyses were not adjusted for multiplicity.

Results

- The zuranolone and placebo arms included 76 and 74 patients, respectively, who were randomized and included in the efficacy analyses.
- Baseline demographics and patient characteristics were well balanced between the two treatment arms and have been described in detail previously.[25]
- As previously presented, patients treated with zuranolone met the primary efficacy endpoint at Day 15 by achieving a significantly greater HAMD-17 total score CFB vs those who received placebo (p=0.0028).[25]
- Zuranolone was generally well tolerated, as previously described.[25] The most common TEAEs occurring in ≥5% of patients who received zuranolone were somnolence, headache, dizziness, upper respiratory tract infection, diarrhea, and sedation.[25]
- One subject experienced a serious adverse event (SAE) in the zuranolone arm that resolved after dose reduction, and one subject experienced an SAE in the placebo arm.[27] There were no reports of loss of consciousness or syncope in either arm.[28]

IMPROVEMENT IN DEPRESSIVE AND ANXIETY SYMPTOMS ACHIEVED WITH ZURANOLONE VS PLACEBO

HAMD-17 TOTAL SCORE

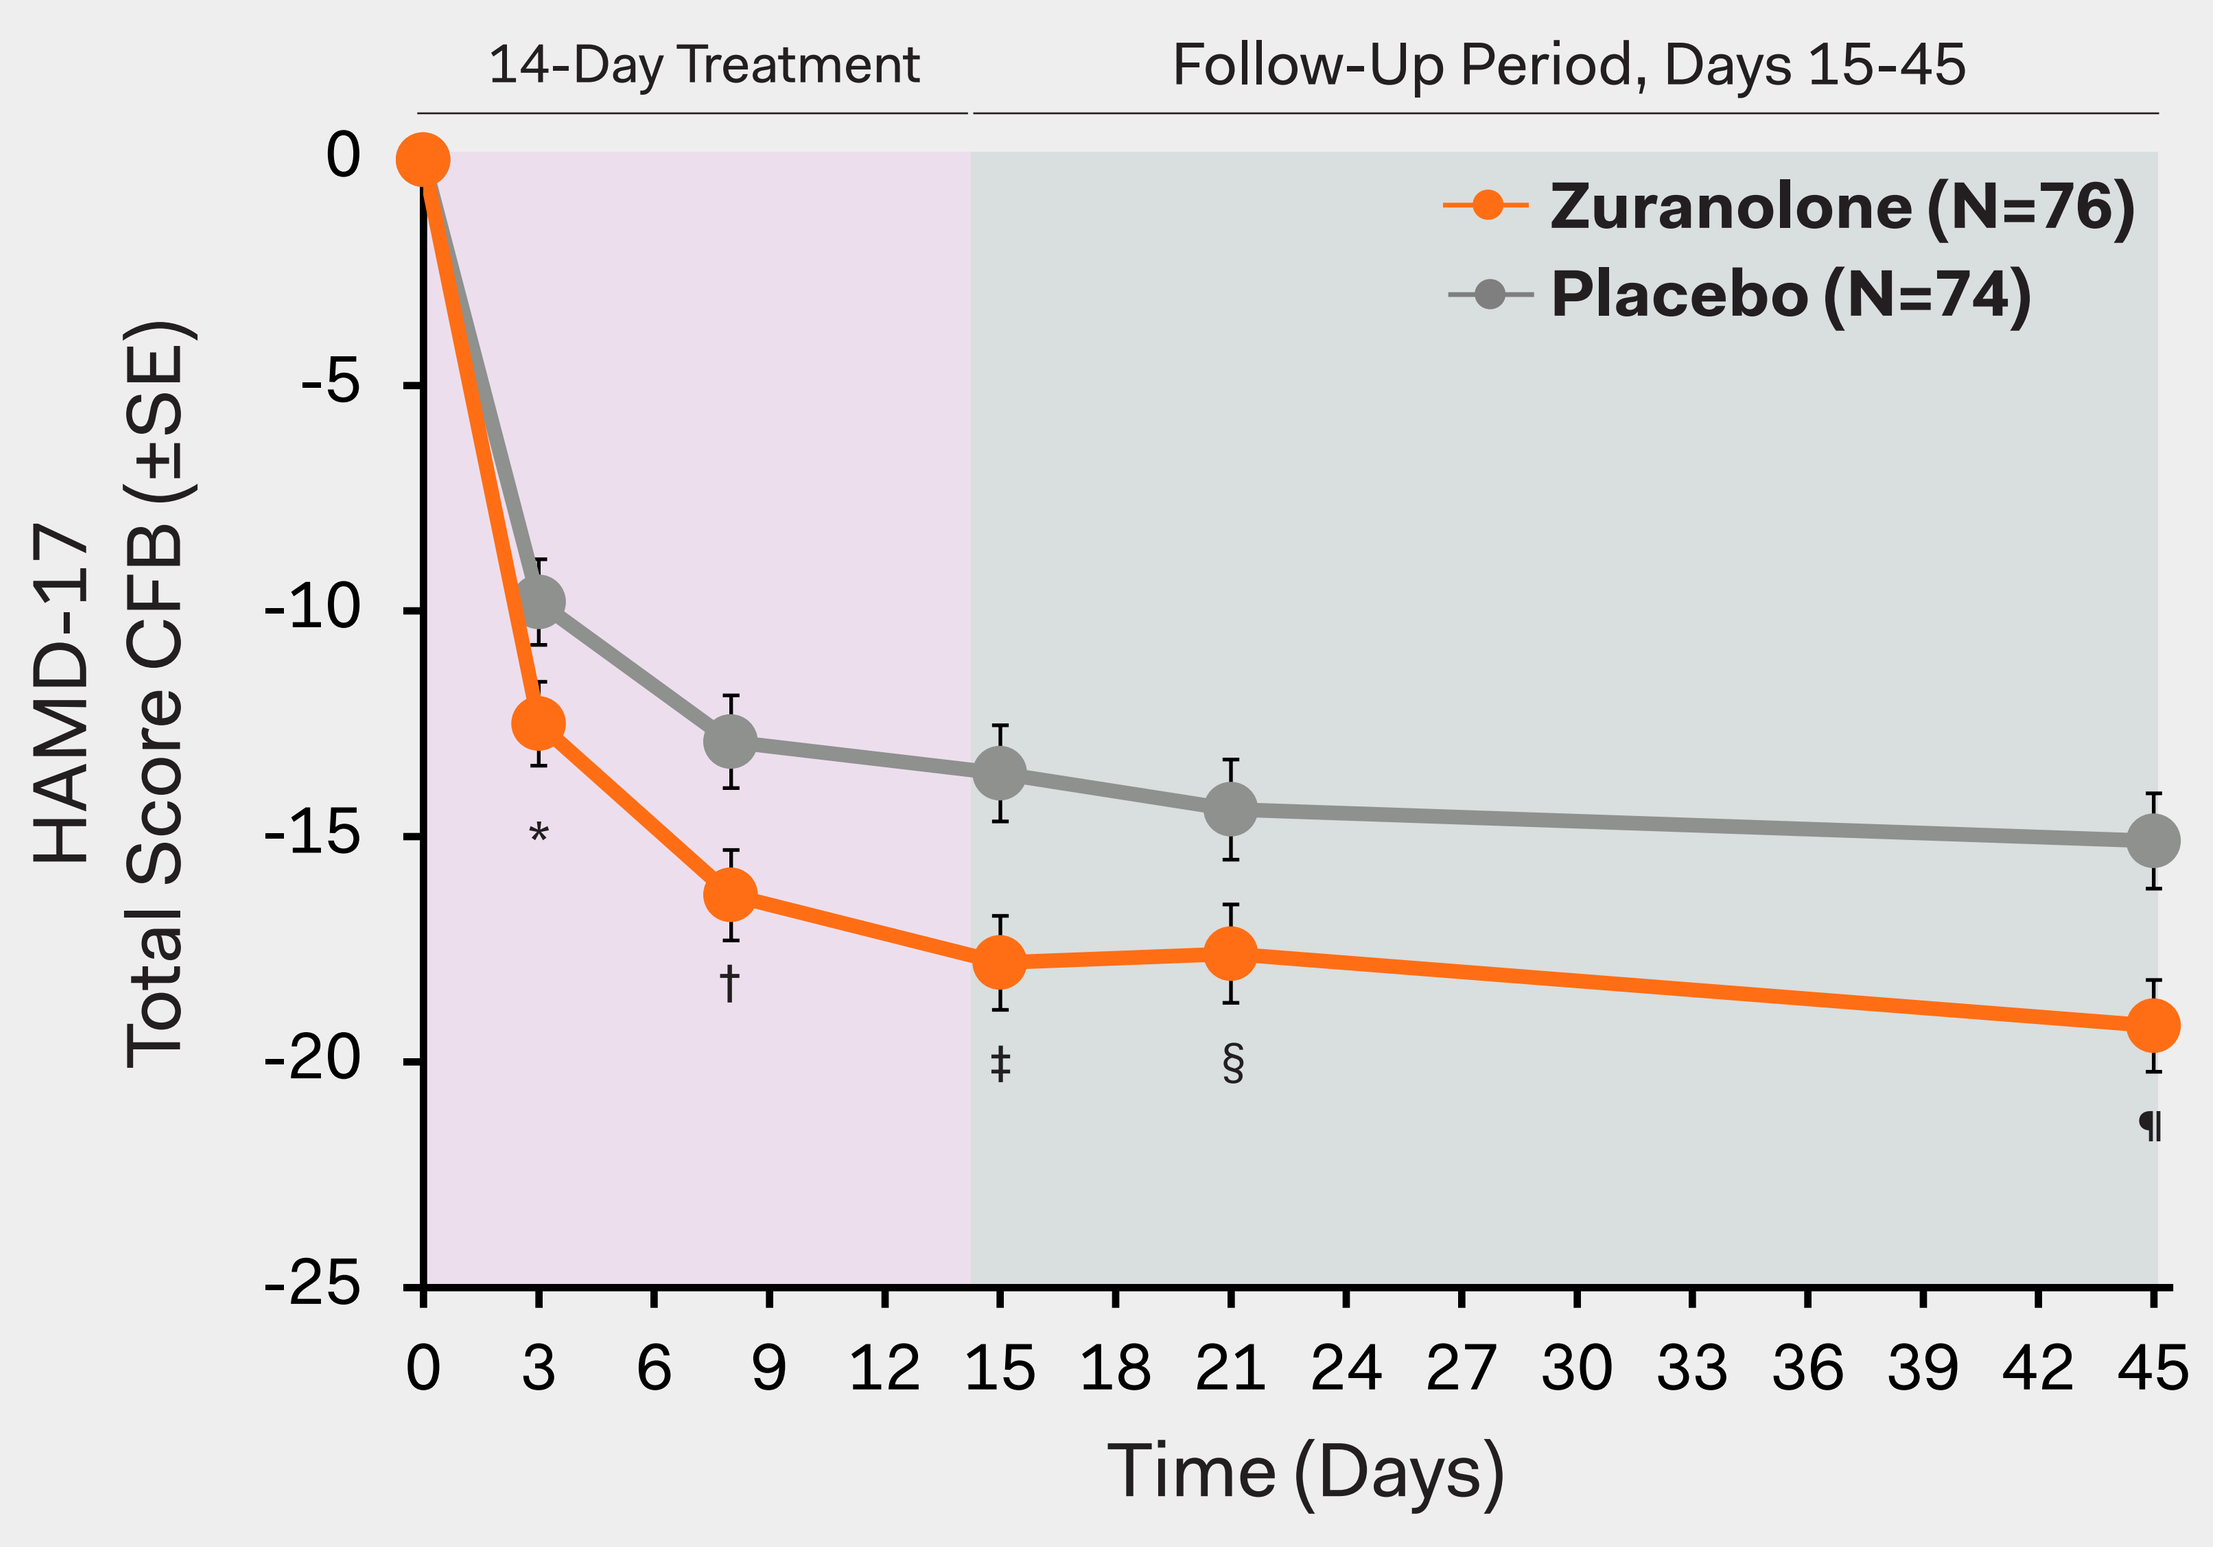
14-Day Treatment
Follow-Up Period, Days 15-45
Zuranolone (N=76)
Placebo (N=74)
HAMD-17 Total Score CFB (±SE)
0
-5
-10
-15
-20
-25
0 3 6 9 12 15 18 21 24 27 30 33 36 39 42 45
Time (Days)
*
†
‡
§
¶

*p=0.0252; †p=0.0106; ‡p=0.0028; §p=0.0321; ¶p=0.0027 vs placebo.

MADRS TOTAL SCORE

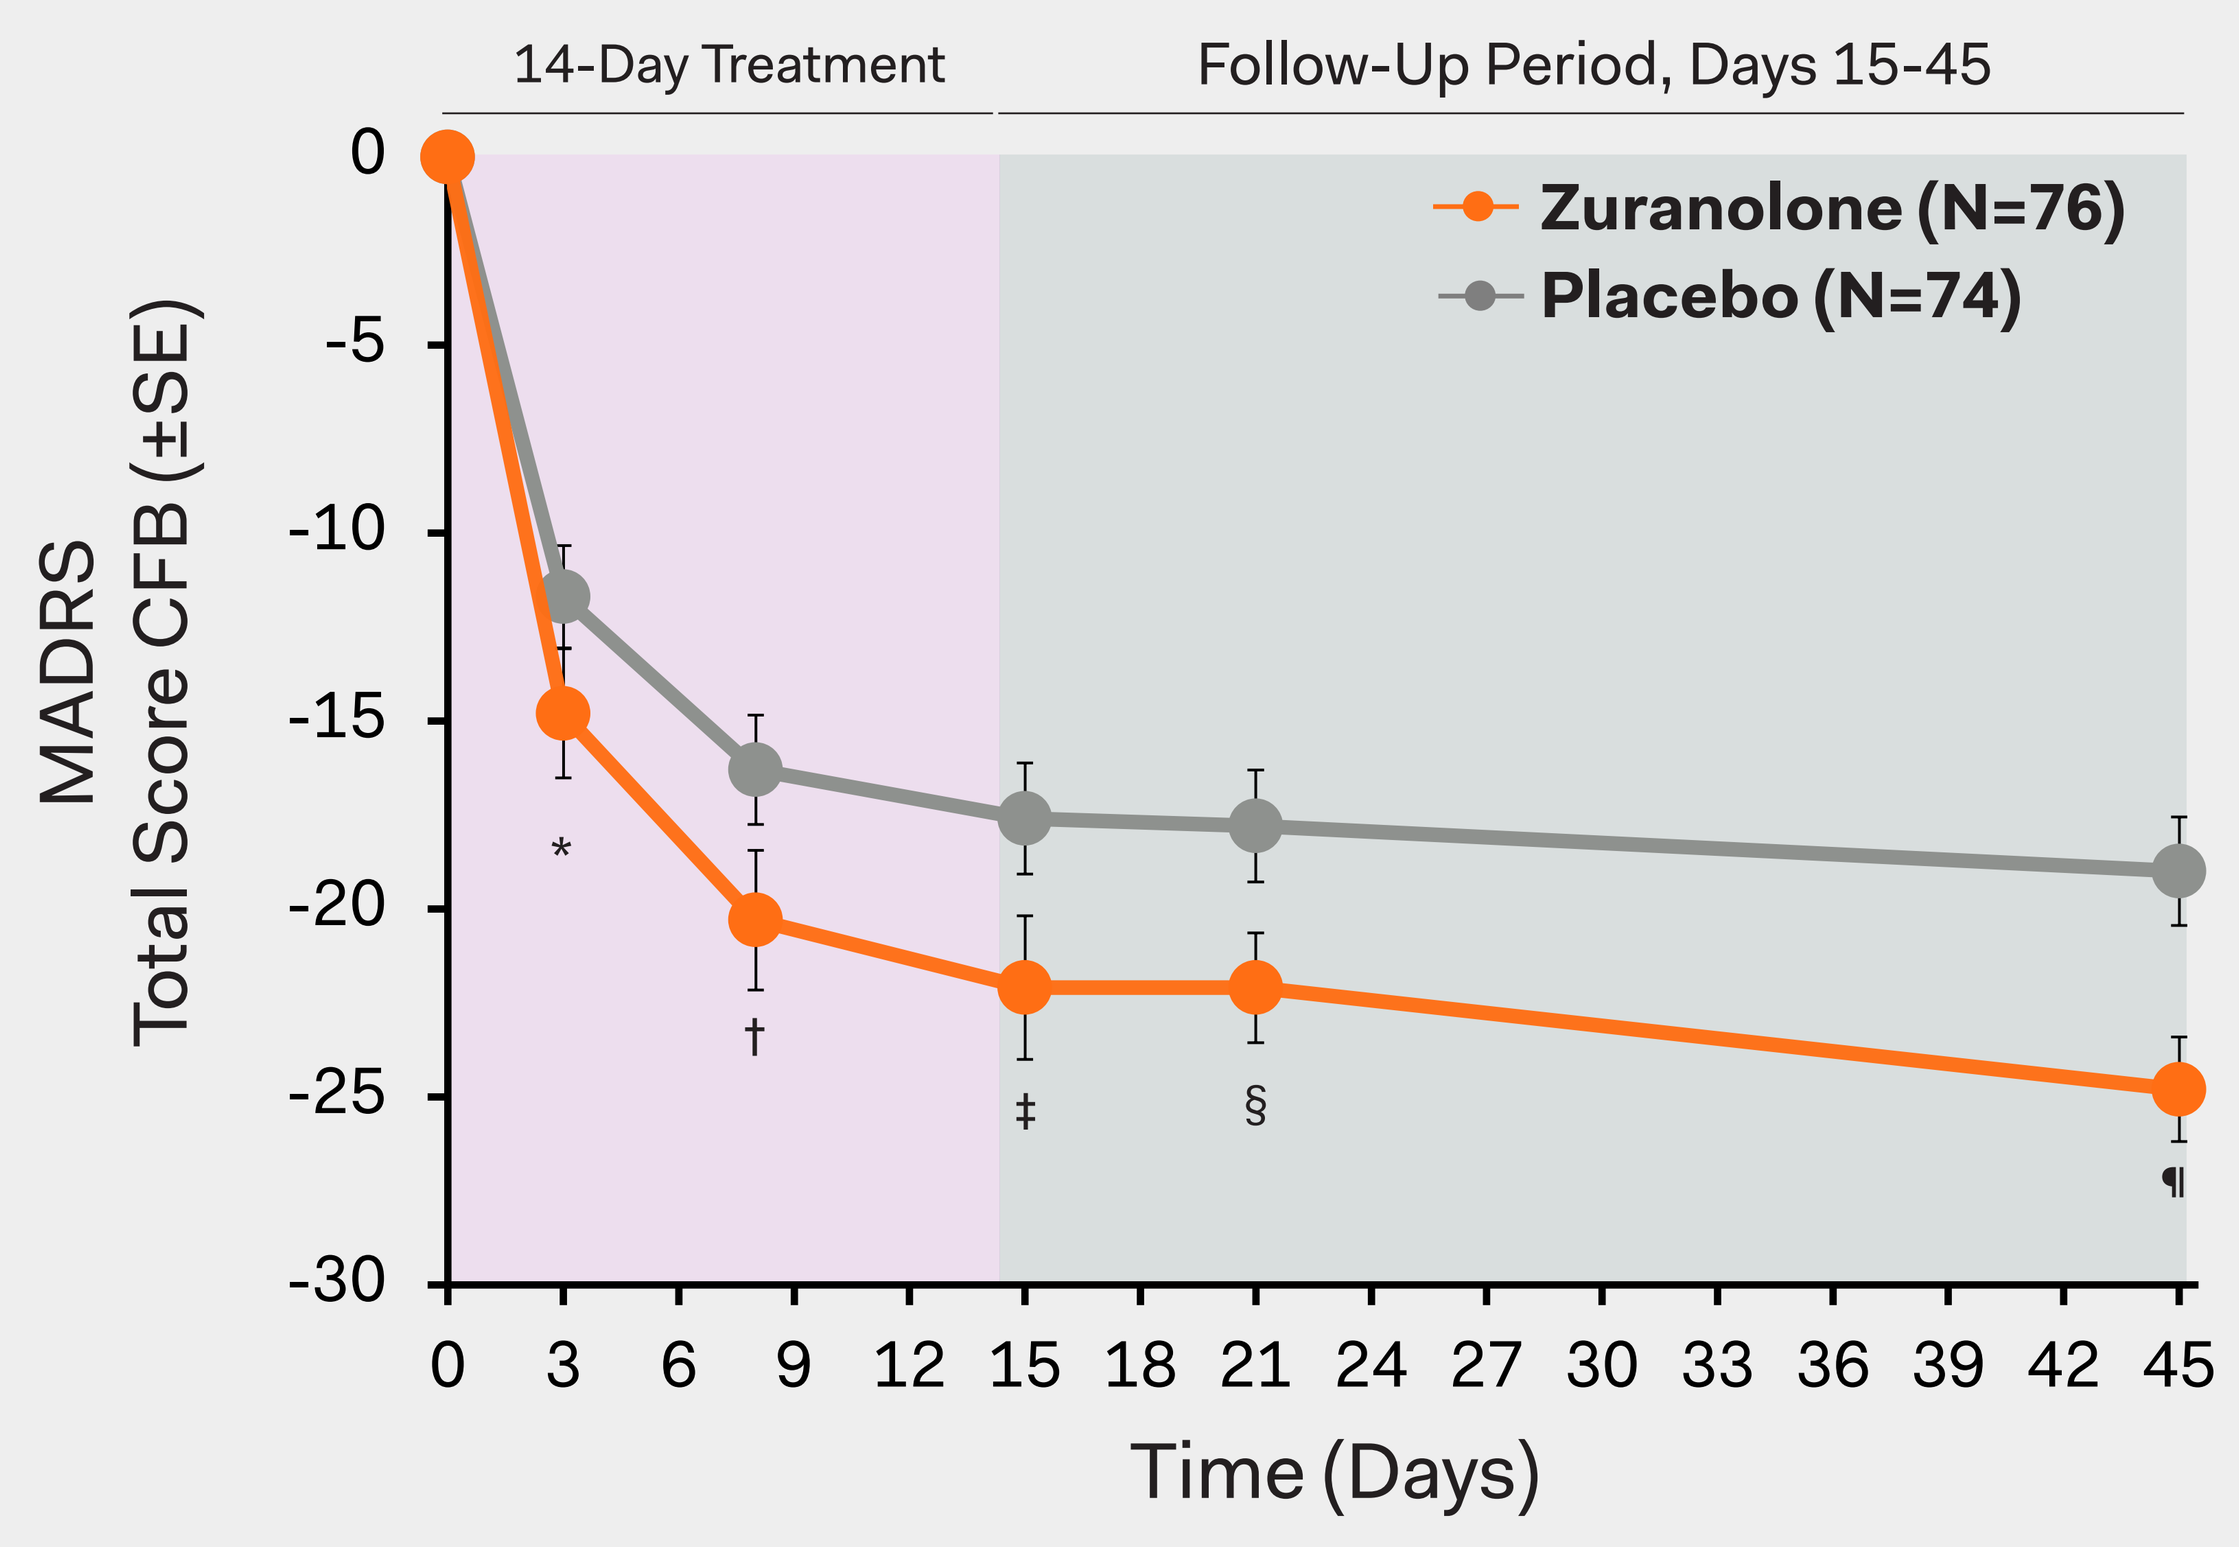
14-Day Treatment
Follow-Up Period, Days 15-45
Zuranolone (N=76)
Placebo (N=74)
MADRS Total Score CFB (±SE)
0
-5
-10
-15
-20
-25
-30
0 3 6 9 12 15 18 21 24 27 30 33 36 39 42 45
Time (Days)
*
†
‡
§
¶

*p=0.0791; †p=0.0322; ‡p=0.0180; §p=0.0271; ¶p=0.0018 vs placebo.

HAM-A TOTAL SCORE

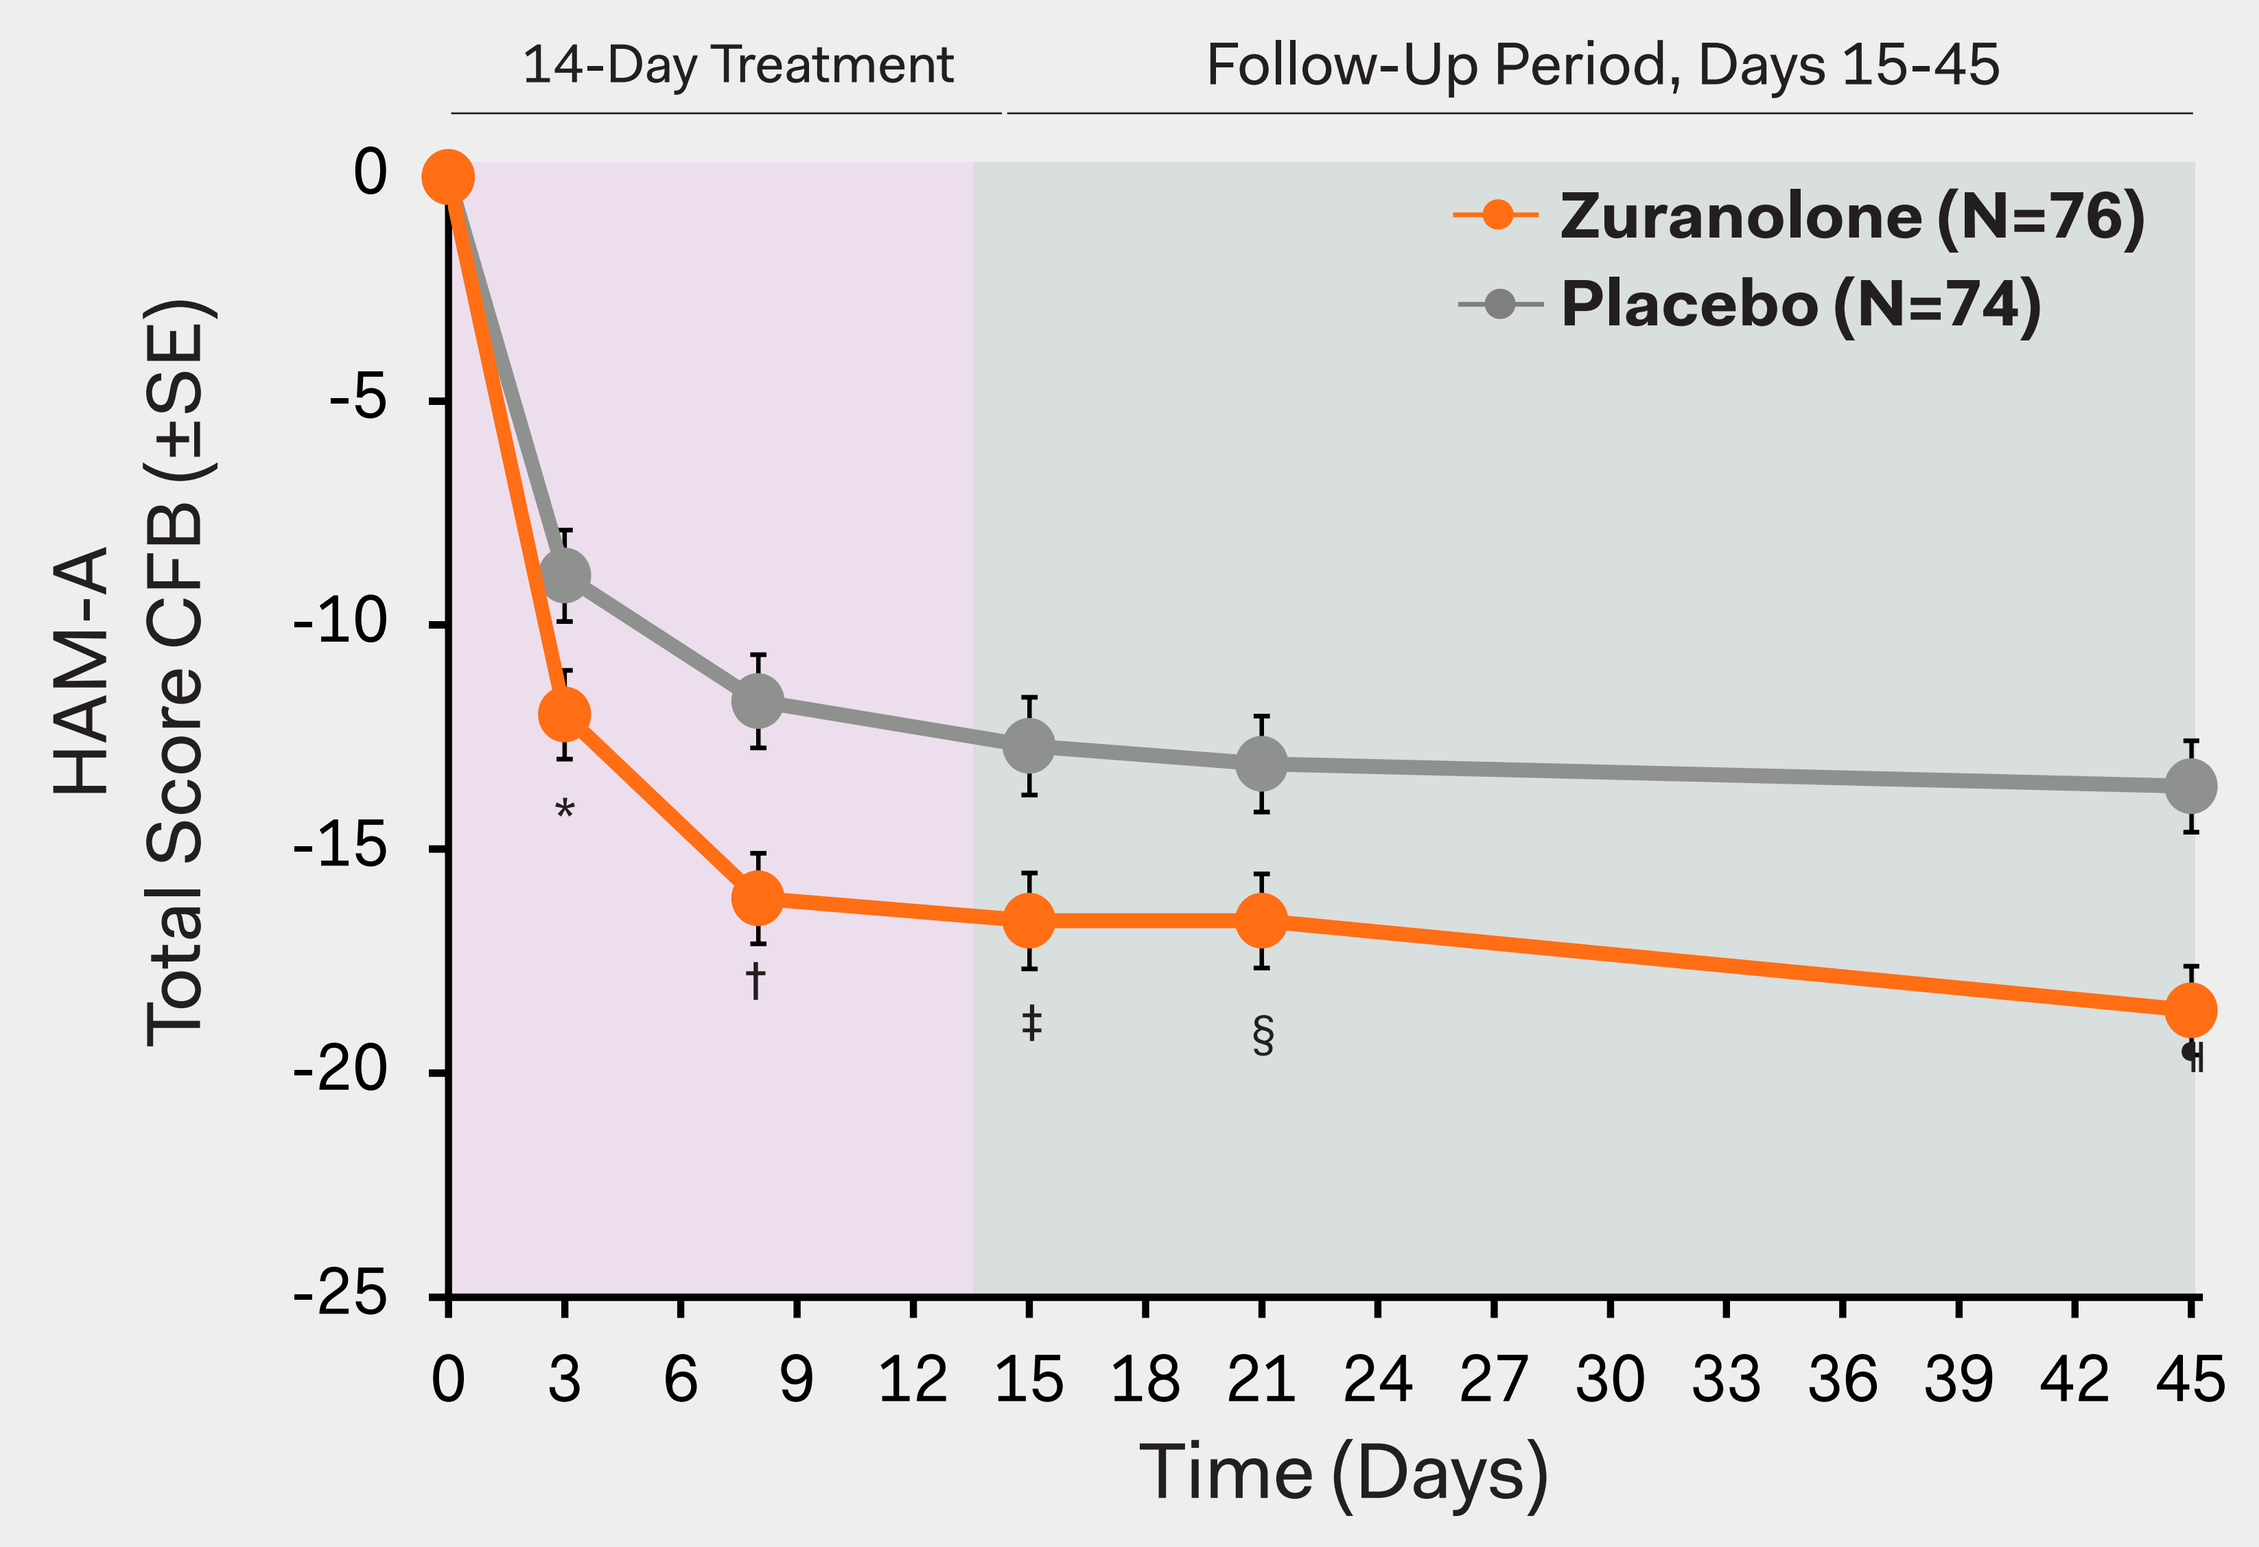
14-Day Treatment
Follow-Up Period, Days 15-45
Zuranolone (N=76)
Placebo (N=74)
HAM-A Total Score CFB (±SE)
0
-5
-10
-15
-20
-25
0 3 6 9 12 15 18 21 24 27 30 33 36 39 42 45
Time (Days)
*
†
‡
§
¶

*p=0.0169; †p=0.0010; ‡p=0.0063; §p=0.0119; ¶p=0.0002 vs placebo.

CONCURRENT IMPROVEMENT OF DEPRESSIVE AND ANXIETY SYMPTOMS WITH ZURANOLONE VS PLACEBO

HAMD-17 TOTAL SCORE ≤7 AND HAM-A TOTAL SCORE ≤7

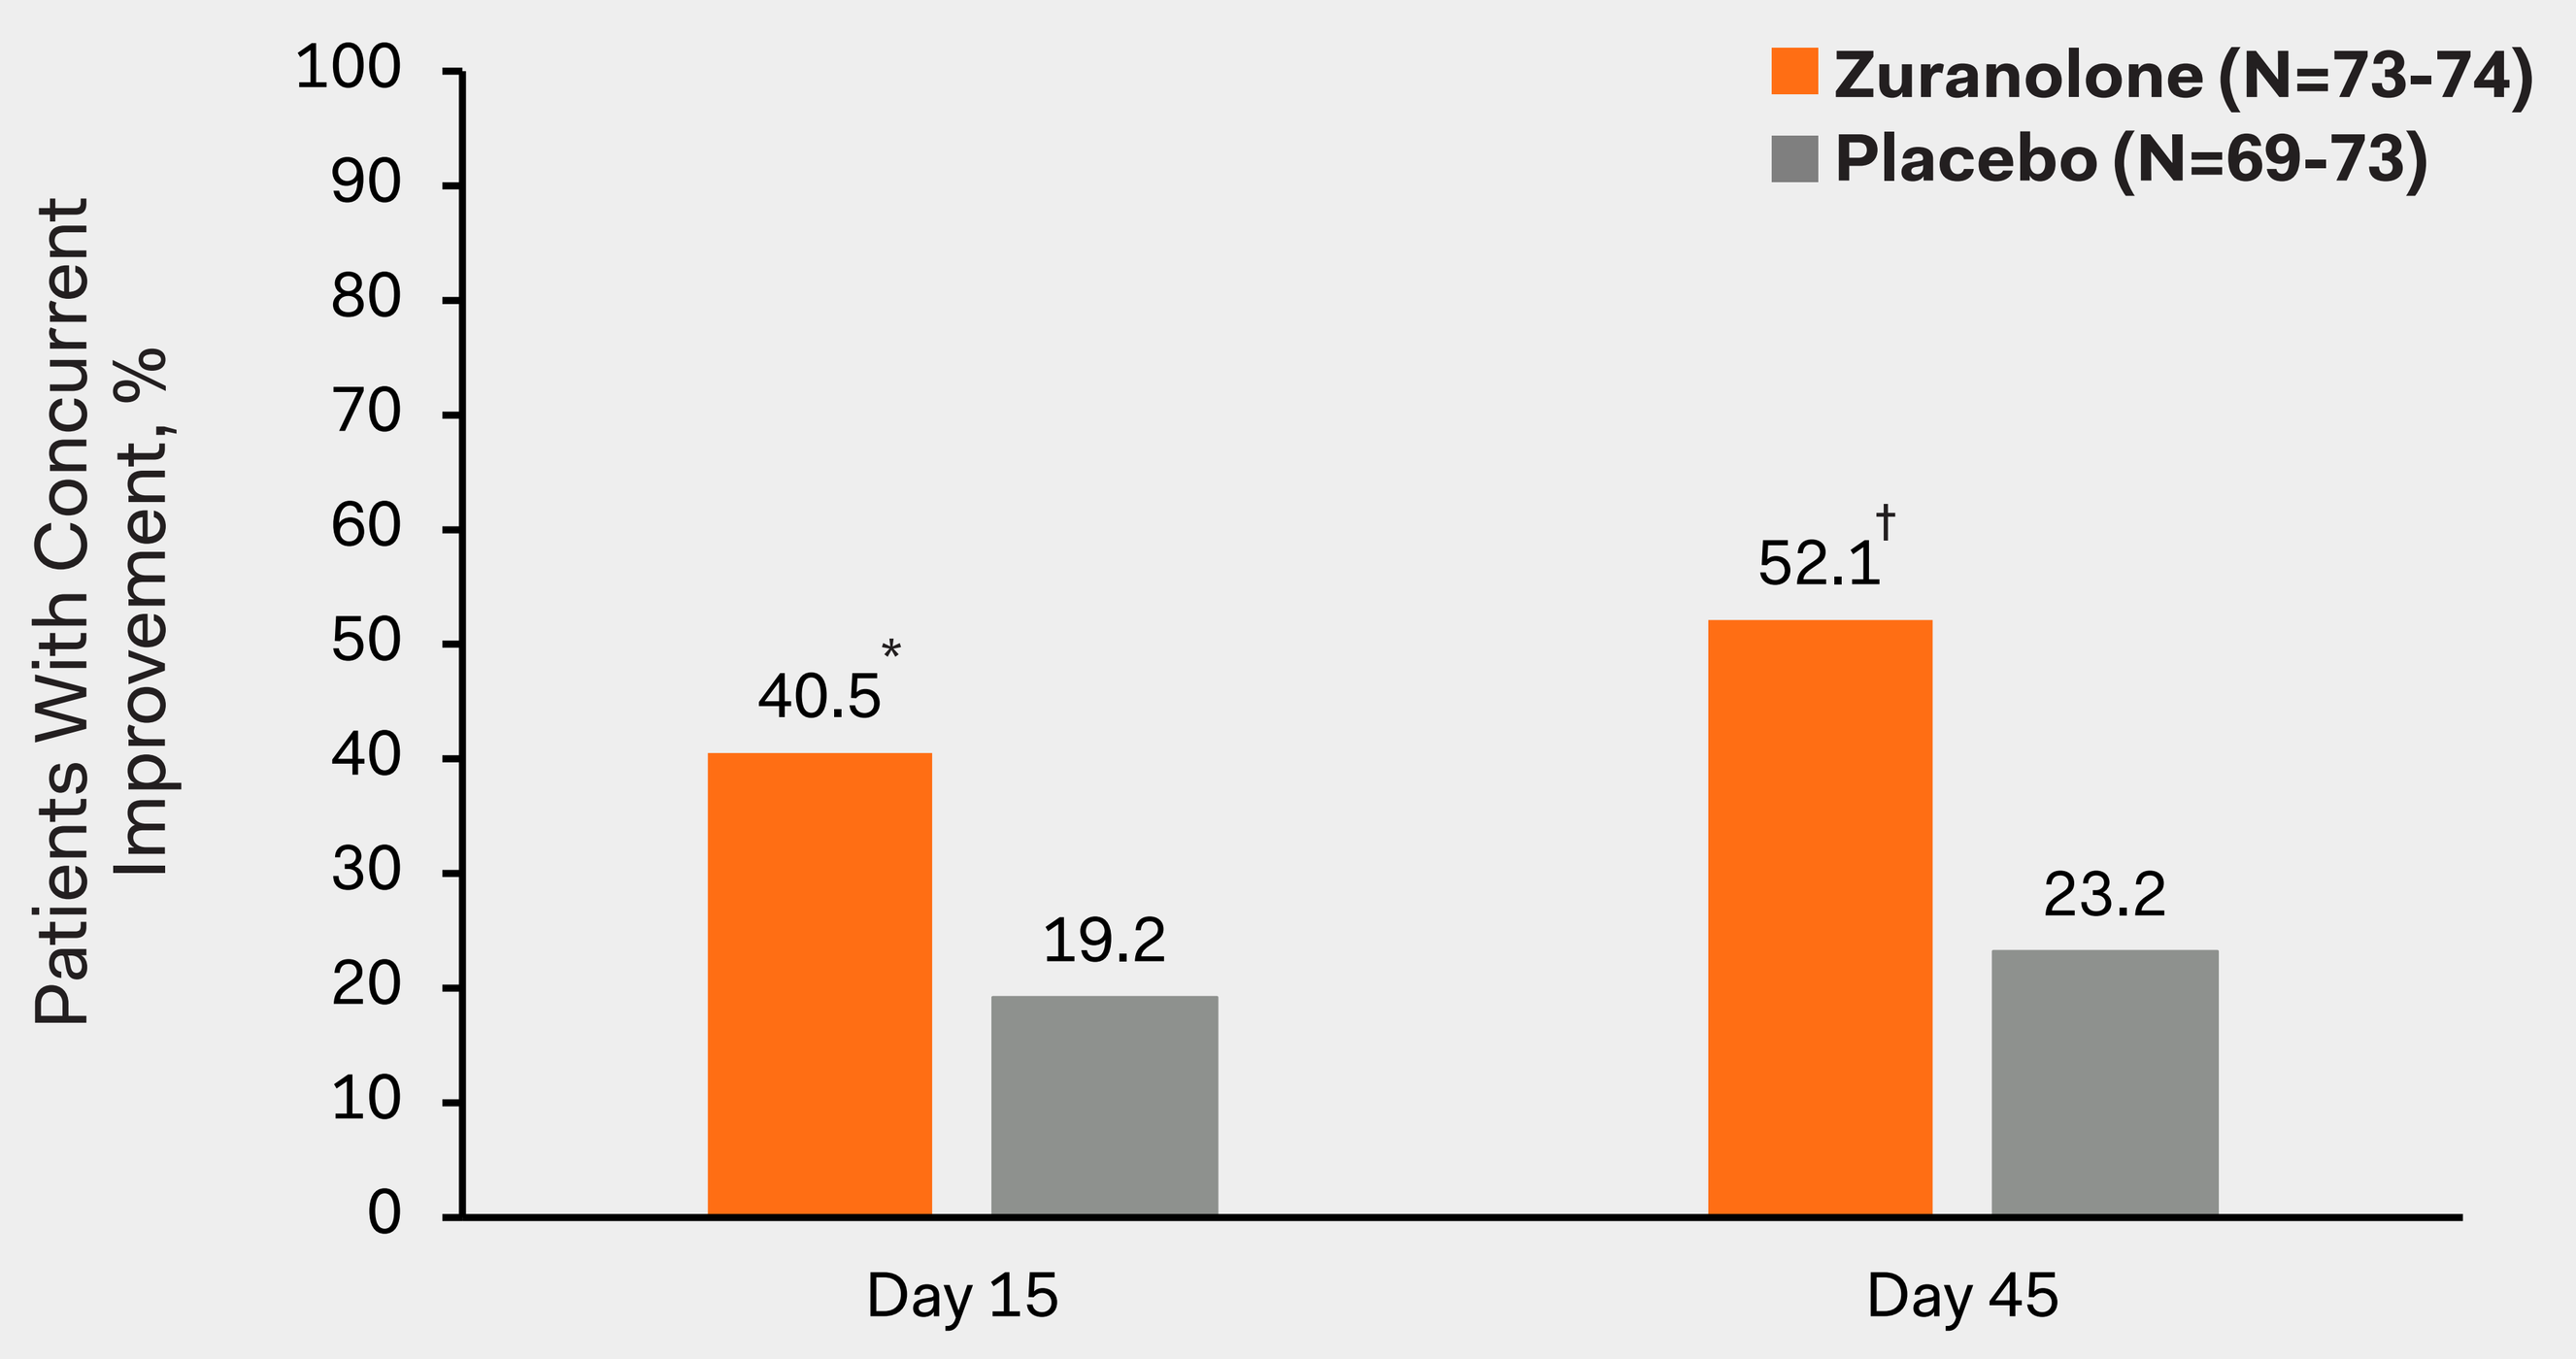
Zuranolone (N=73-74)
Placebo (N=69-73)
Patients With Concurrent Improvement, %
100
90
80
70
60
50
40
30
20
10
0
40.5*
19.2
52.1†
23.2
Day 15
Day 45

*p=0.0083; †p=0.0009 vs placebo.

- A significantly higher proportion of zuranolone-treated patients achieved concurrent improvement of depressive and anxiety symptoms at Day 15 (p=0.0083), and at Day 45 (p=0.0009) using the HAMD-17 and HAM-A scale combination.

MADRS TOTAL SCORE ≤10 AND HAM-A TOTAL SCORE ≤7

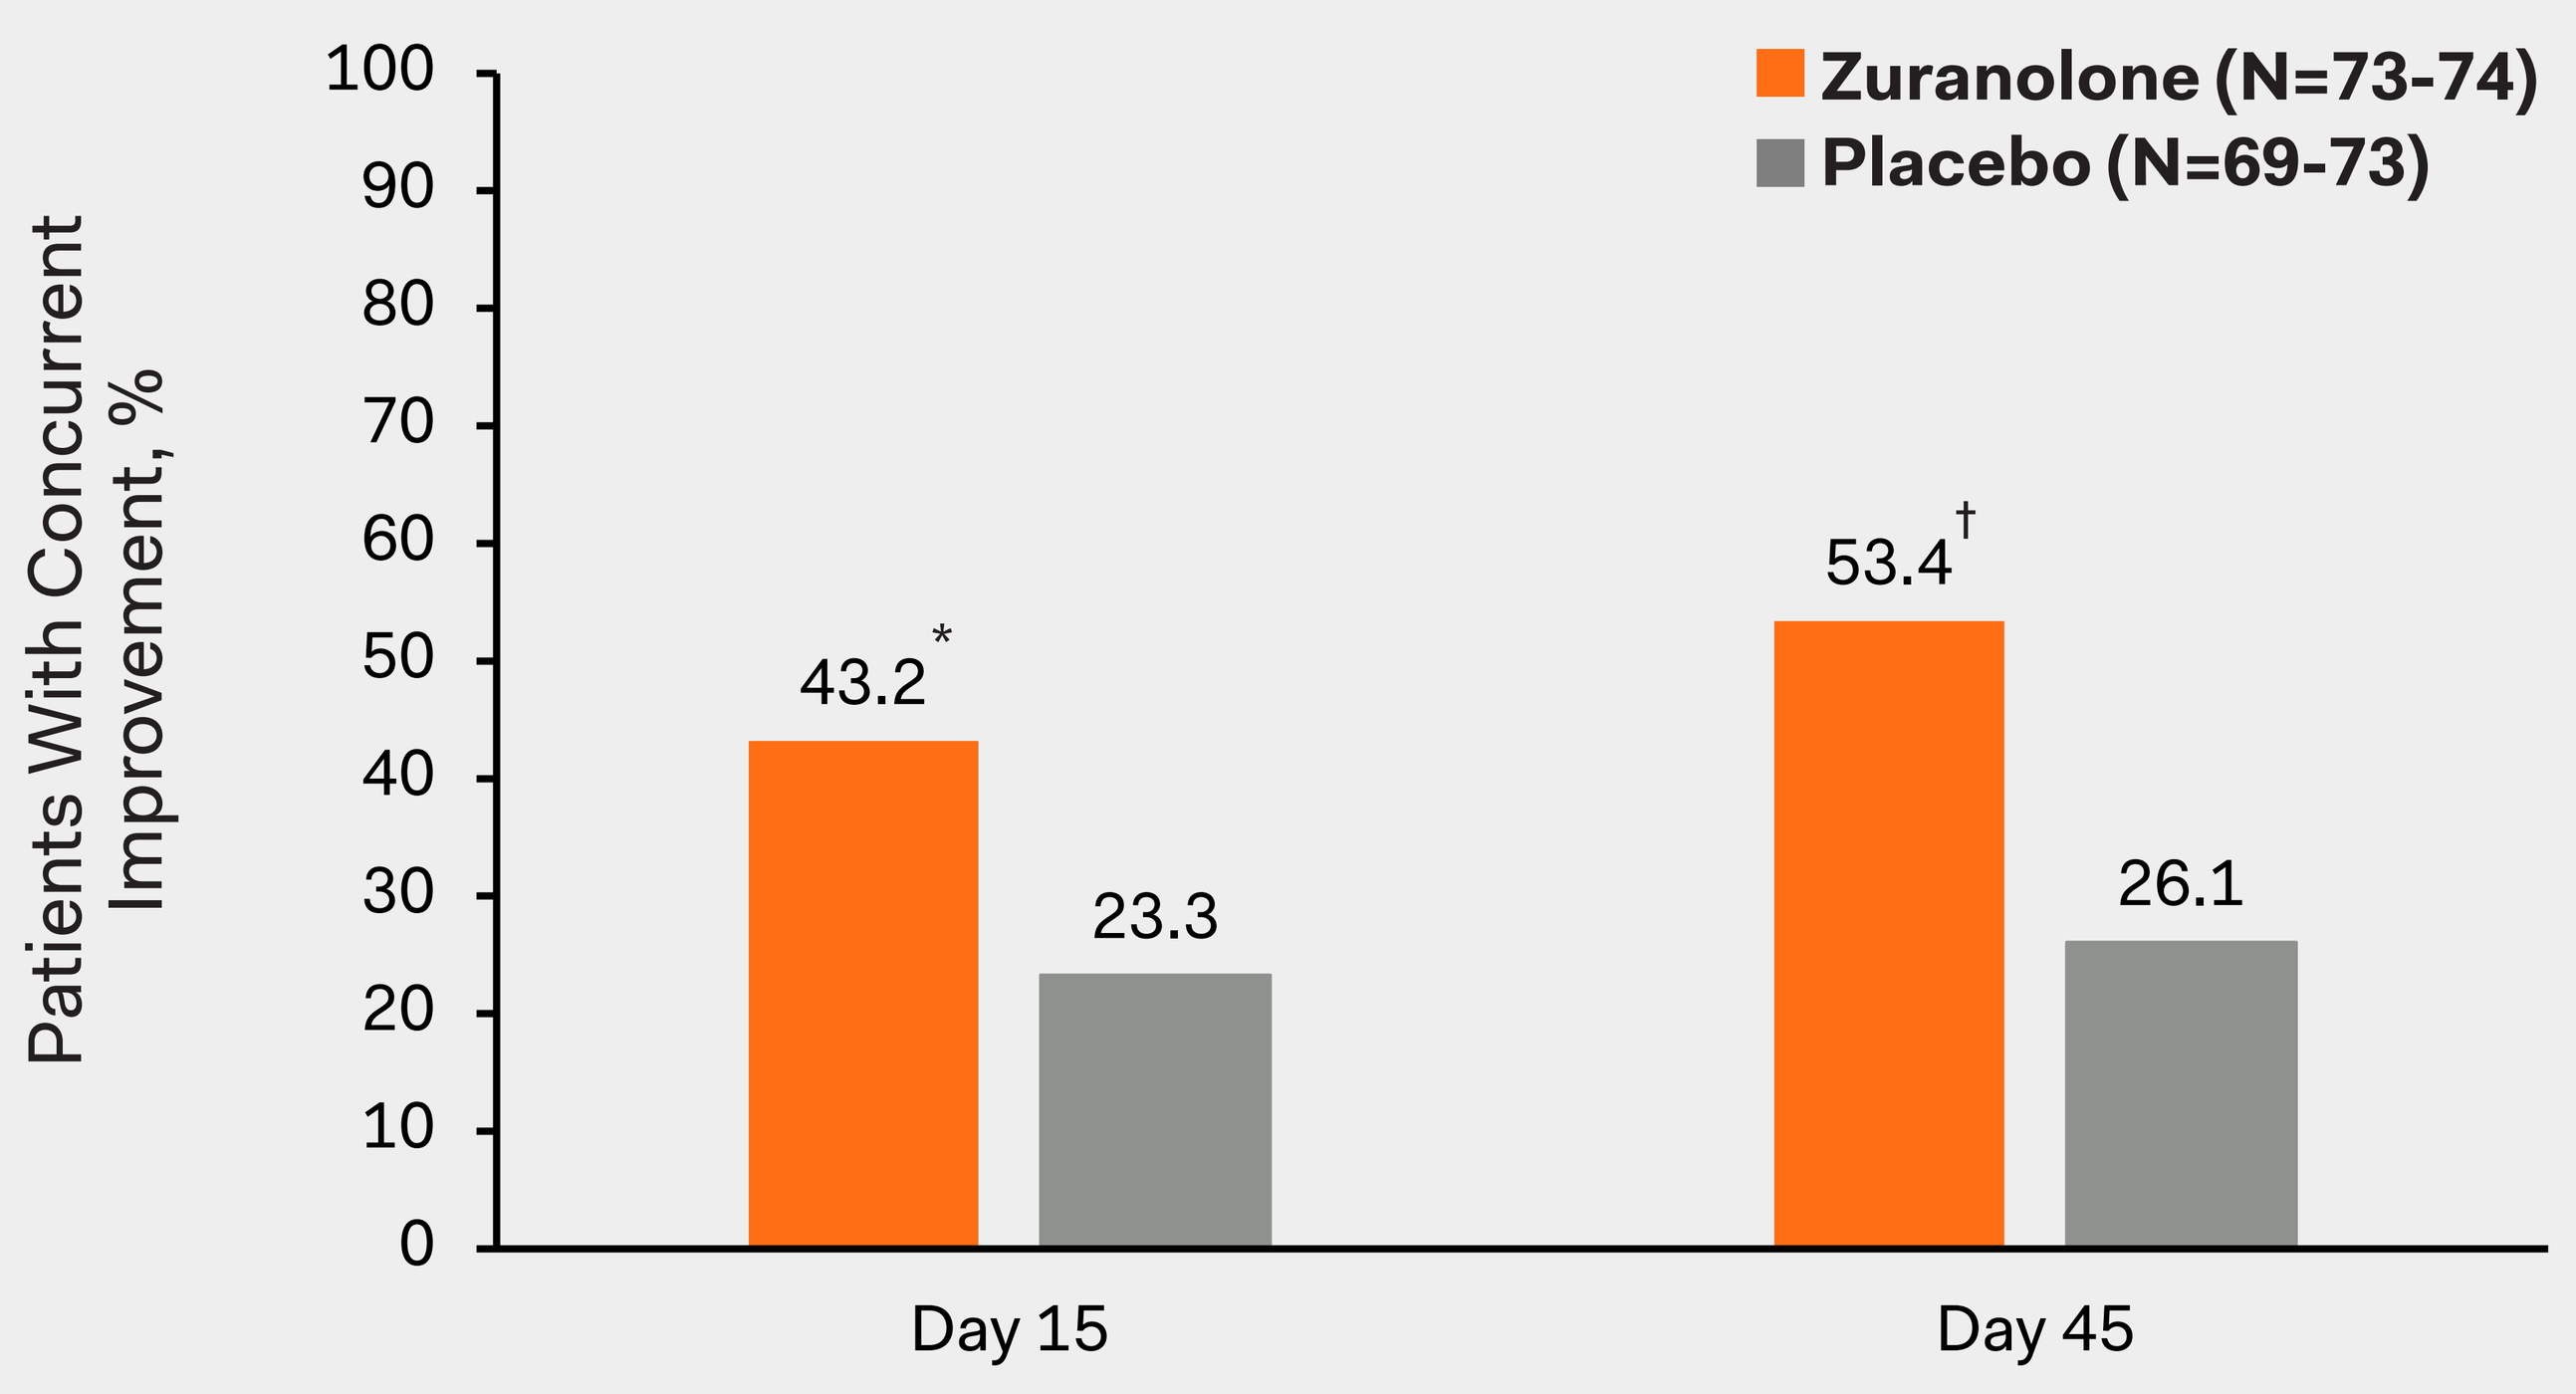
Zuranolone (N=73-74)
Placebo (N=69-73)
Patients With Concurrent Improvement, %
100
90
80
70
60
50
40
30
20
10
0
43.2*
23.3
53.4†
26.1
Day 15
Day 45

*p=0.0188; †p=0.0022 vs placebo.

- A significantly higher proportion of zuranolone-treated patients achieved concurrent improvement of depressive and anxiety symptoms at Day 15 (p=0.0188), and at Day 45 (p=0.0022) using the MADRS and HAM-A scale combination.

TREATMENT DIFFERENCES (ZURANOLONE VS PLACEBO)

HAMD-17/HAM-A	OR (95% CI)	p-value
Day 15	2.75 (1.30, 5.84)	0.0083
Day 45	3.45 (1.66, 7.16)	0.0009

MADRS/HAM-A	OR (95% CI)	p-value
Day 15	2.35 (1.15, 4.80)	0.0188
Day 45	3.09 (1.50, 6.36)	0.0022

OR: Odds Ratio; CI: Confidence Interval

- The odds of achieving concurrent improvement of depressive and anxiety symptoms were significantly higher for zuranolone-treated patients at Day 15 and at Day 45.

REFERENCES
1. Bauman BL, et al. *MMWR Morb Mortal Wkly Rep* (2020) 69:575-581.
2. Hamilton BE, et al. National Center for Health Statistics. Births: Provisional data for 2018. Vital Statistics Rapid Release; no 7. https://www.cdc.gov/nchs/data/vsrr/vsrr-007-508.pdf. Published May 2019. Accessed November 10, 2020.
3. DeSisto CL, et al. *Prev Chronic Dis* (2014) 11:E10.
4. Centers for Disease Control and Prevention. https://www.cdc.gov/reproductivehealthmaternalinfanthealth/diabetes-during-pregnancy.htm. Updated June 12, 2018. Accessed November 10, 2020.
5. Centers for Disease Control and Prevention. https://www.cdc.gov/reproductivehealth/maternalinfanthealth/pregnancy-complications-data.htm. Updated February 28, 2019. Accessed November 10, 2020.
6. Roberts JM, et al. American College of Obstetricians and Gynecologists Task Force on Hypertension in Pregnancy. https://www.acog.org/~/media/Task%20Force%20and%20Work%20Group%20Reportspublic/HypertensioninPregnancy.pdf. Published 2013. Accessed November 10, 2020.
7. Callaghan WM, et al. *Am J Obstet Gynecol* (2010) 202:353.e1-6.
8. Diagnostic and statistical manual of mental disorders: DSM-5. Washington, DC: American Psychiatric Association; 2013.
9. Kerstis B, et al. *Arch Womens Ment Health* (2016) 19:87.
10. Posmontier B, et al. *J Midwifery Womens Health* (2008) 53:310-318.
11. Barkin JL, et al. *J Womens Health* (Larchmt; 2016) 25:707-713.
12. Gagliardi L, et al. *Arch Dis Child* (2012) 97:355-357.
13. McLearn KT, et al. *Arch Pediatr Adolesc Med* (2006) 160:279-284.
14. Moore Simas TA, et al. *J Med Econ* (2019) 23:174-183.
15. Balbierz A, et al. *Matern Child Health J* (2015) 19:1212-1219.
16. Valla L, et al. *Infant Behav Dev* (2016) 45:83-90.
17. Koutra K, et al. *Soc Psychiatry Psychiatr Epidemiol* (2013) 48:1335-1345.
18. Robertson E, et al. *Gen Hosp Psychiatry* (2004) 26:289-295.
19. Biaggi A, et al. *J Affect Disord* (2016) 191:62-77.
20. Yim IS, et al. *Annu Rev Clin Psychol* (2015) 11:99-137.
21. Hoffmann E et al. *Clin Pharmacokinet* (2019) 59: 111-120.
22. Martinez Botella G, et al. *J Med Chem* (2017) 60:7810-7819.
23. Belfort GM, et al. Poster presented at American Epilepsy Society 2016.
24. Hamilton M. *J Neurol Neurosurg Psychiatry* (1960) 23: 56–62.
25. Deligiannidis KM, et al. Presented as ASCP 2019. Poster T74.
26. Cox JL, et al. *Br J Psychiatry* (1987) 150:782-6.
27. Deligiannidis KM, et al. Poster presentation at ACNP 2019. Poster W79.
28. Data on file MLIB-3497 Robin Safety Data. [217-PPD-201]. Sage Therapeutics, Inc. [2019].

SAGE THERAPEUTICS and the SAGE THERAPEUTICS logo are trademarks of Sage Therapeutics, Inc. All other trademarks referenced herein are the property of their respective owners.

SUPPORT & DISCLOSURES
BW, MYH, ES, RL, HGB, VB, and SJK are employees of Sage Therapeutics, Inc., with stock/stock options. KMD serves as a consultant to Sage Therapeutics, Inc., receives NIMH support and royalties from an NIH employee invention, and reports grants from Sage Therapeutics, Inc. awarded to Zucker Hillside Hospital during the conduct of the zuranolone clinical trials. SA and MF consulted for Sage Therapeutics, Inc. This study and medical writing support were funded by Sage Therapeutics, Inc. Writing and editorial support was provided by Bonnie L. Bullock, PhD and Kathryn P. Wall, PhD of Boston Strategic Partners, Inc.

American College of Neuropsychopharmacology 2020
Virtual Annual Meeting
December 6-9, 2020

MRC-PPD-00514 11/2020