Sage Therapeutics™

Clinical Global Impression Scores and Number Needed to Treat Outcomes in Patients with Postpartum Depression Treated with the Oral Neuroactive Steroid Zuranolone

Kristina M. Deligiannidis, MD[1], Brian Werneburg, PhD[2], Ming-Yi Huang, PhD[2], Ellison Suthoff, BS, MBA[2], Robert Lasser, MD, MBA[2], Handan Gunduz-Bruce, MD, MBA[2], Sarah Acaster, MSc[3], Moshe Fridman, PhD[4], Vijayveer Bonthapally, PhD[2], Stephen J. Kanes, MD, PhD[2]

[1] Department of Psychiatry, Women's Behavioral Health, Zucker Hillside Hospital, Glen Oaks, NY
[2] Sage Therapeutics, Inc., Cambridge, MA
[3] Acaster Lloyd Consulting Ltd., London, UK
[4] AMF Consulting, Los Angeles, CA

Introduction

— Postpartum depression (PPD) is one of the most common medical complications during and after pregnancy.[1-7]

— In the United States, estimates of new mothers experiencing symptoms of PPD[8] each year vary by state from 9.7% to 23.5%, with an average of 13.2%.[1]

— PPD is associated with a significant impairments in mother-infant bonding[9] and maternal function,[10, 11] including breastfeeding[12] and caring for the child,[11,13] with implications for the child's health and development.[14-17]

— Multiple environmental and biological risk factors have been proposed to play a role in the development of PPD.[18-20]

— Zuranolone (SAGE-217) is an investigational oral neuroactive steroid $GABA_A$ receptor positive allosteric modulator.[21-23]

— Zuranolone binds to both synaptic and extrasynaptic $GABA_A$ receptors, distinct from benzodiazepines, which bind to synaptic $GABA_A$ receptors.[21-23]

— In a double-blind, randomized, placebo-controlled Phase 3 trial in adult women with PPD (NCT02978326), zuranolone achieved the primary endpoint of a statistically significant reduction in symptoms of depression compared with placebo assessed by the clinician-rated 17-item Hamilton Rating Scale for Depression (HAMD-17) total score at Day 15.[24,25]

- Post hoc analyses examined patients achieving categorical response and remission as measured by the Clinical Global Impression-Improvement (CGI-I) and Clinical Global Impression-Severity (CGI-S) scales.[26]
- Number needed to treat (NNT) outcomes for the CGI-I and the CGI-S were also examined.

Conclusions

— Zuranolone treatment in patients with PPD has previously been shown to provide rapid (HAMD-17 total score by Day 3) and sustained (HAMD-17 total score at all measured timepoints through Day 45) improvement in core symptoms of depression.[25]

— Post hoc analyses show that zuranolone achieved significantly greater improvements and rates of response and remission as measured by CGI-I response (Day 15) CGI-I remission (Day 45) and CGI-S (at both Day 15 and Day 45, respectively) in women with PPD.

— Single-digit NNTs support the robustness of these effects compared to placebo.

Methods

— Women (n=151), ages 18-45, ≤6 months postpartum, diagnosed with PPD (defined as a major depressive episode with onset in the 3rd trimester or ≤4 weeks postpartum), and a baseline HAMD-17 ≥26, were administered study drug in an outpatient setting.

— Patients were randomized 1:1 to receive zuranolone 30 mg or placebo once daily for 14 days, with 4 weeks follow-up.

— The change from baseline (CFB) in HAMD-17 at Day 15 was the primary endpoint.

— Secondary endpoints included the CFB in HAMD-17 throughout the trial, CGI-I scores, which measure overall improvement post-treatment, and CGI-S scores, which reflect severity at the time of assessment.

— Treatment emergent adverse events (TEAEs) were assessed throughout the study.

— Post hoc analyses assessed response and remission rates (see table in the "CGI-I and CGI-S Response and Remission Criteria" section below) defined as scores ≤2 ('much improved'/'very much improved' for CGI-I; 'borderline mentally ill'/'normal, not at all ill' for CGI-S) and scores ≤1 ('very much improved' for CGI-I, 'normal, not at all ill' for CGI-S), respectively, for both the CGI-I and CGI-S scales.

— NNT was calculated using the proportion of CGI-I and CGI-S responders or remitters in each treatment arm at Day 15 and Day 45.

— CFB in HAMD-17 total scores were evaluated using the least-squares mean from a mixed-effect model for repeated measures. CGI-I and CGI-S response and remission rates were assessed using generalized estimating equations models for repeated measures, adjusting for baseline covariates.

— Secondary endpoints and post hoc analyses were not adjusted for multiplicity.

Results

— The zuranolone and placebo arms included 76 and 74 patients, respectively, who were randomized and included in the efficacy analyses.

— Baseline demographics and patient characteristics were well balanced between the two treatment arms and have been described in detail previously.[25]

— Zuranolone was generally well tolerated, as previously described.[25] The most common TEAEs occurring in ≥5% of patients who received zuranolone were somnolence, headache, dizziness, upper respiratory tract infection, diarrhea, and sedation.[25]

— One subject experienced a serious adverse event (SAE) in the zuranolone arm that resolved after dose reduction, and one subject experienced an SAE in the placebo arm.[27] There were no reports of loss of consciousness or syncope in either arm.[28]

IMPROVEMENT IN DEPRESSIVE SYMPTOMS ACHIEVED WITH ZURANOLONE VS PLACEBO

HAMD-17 TOTAL SCORE

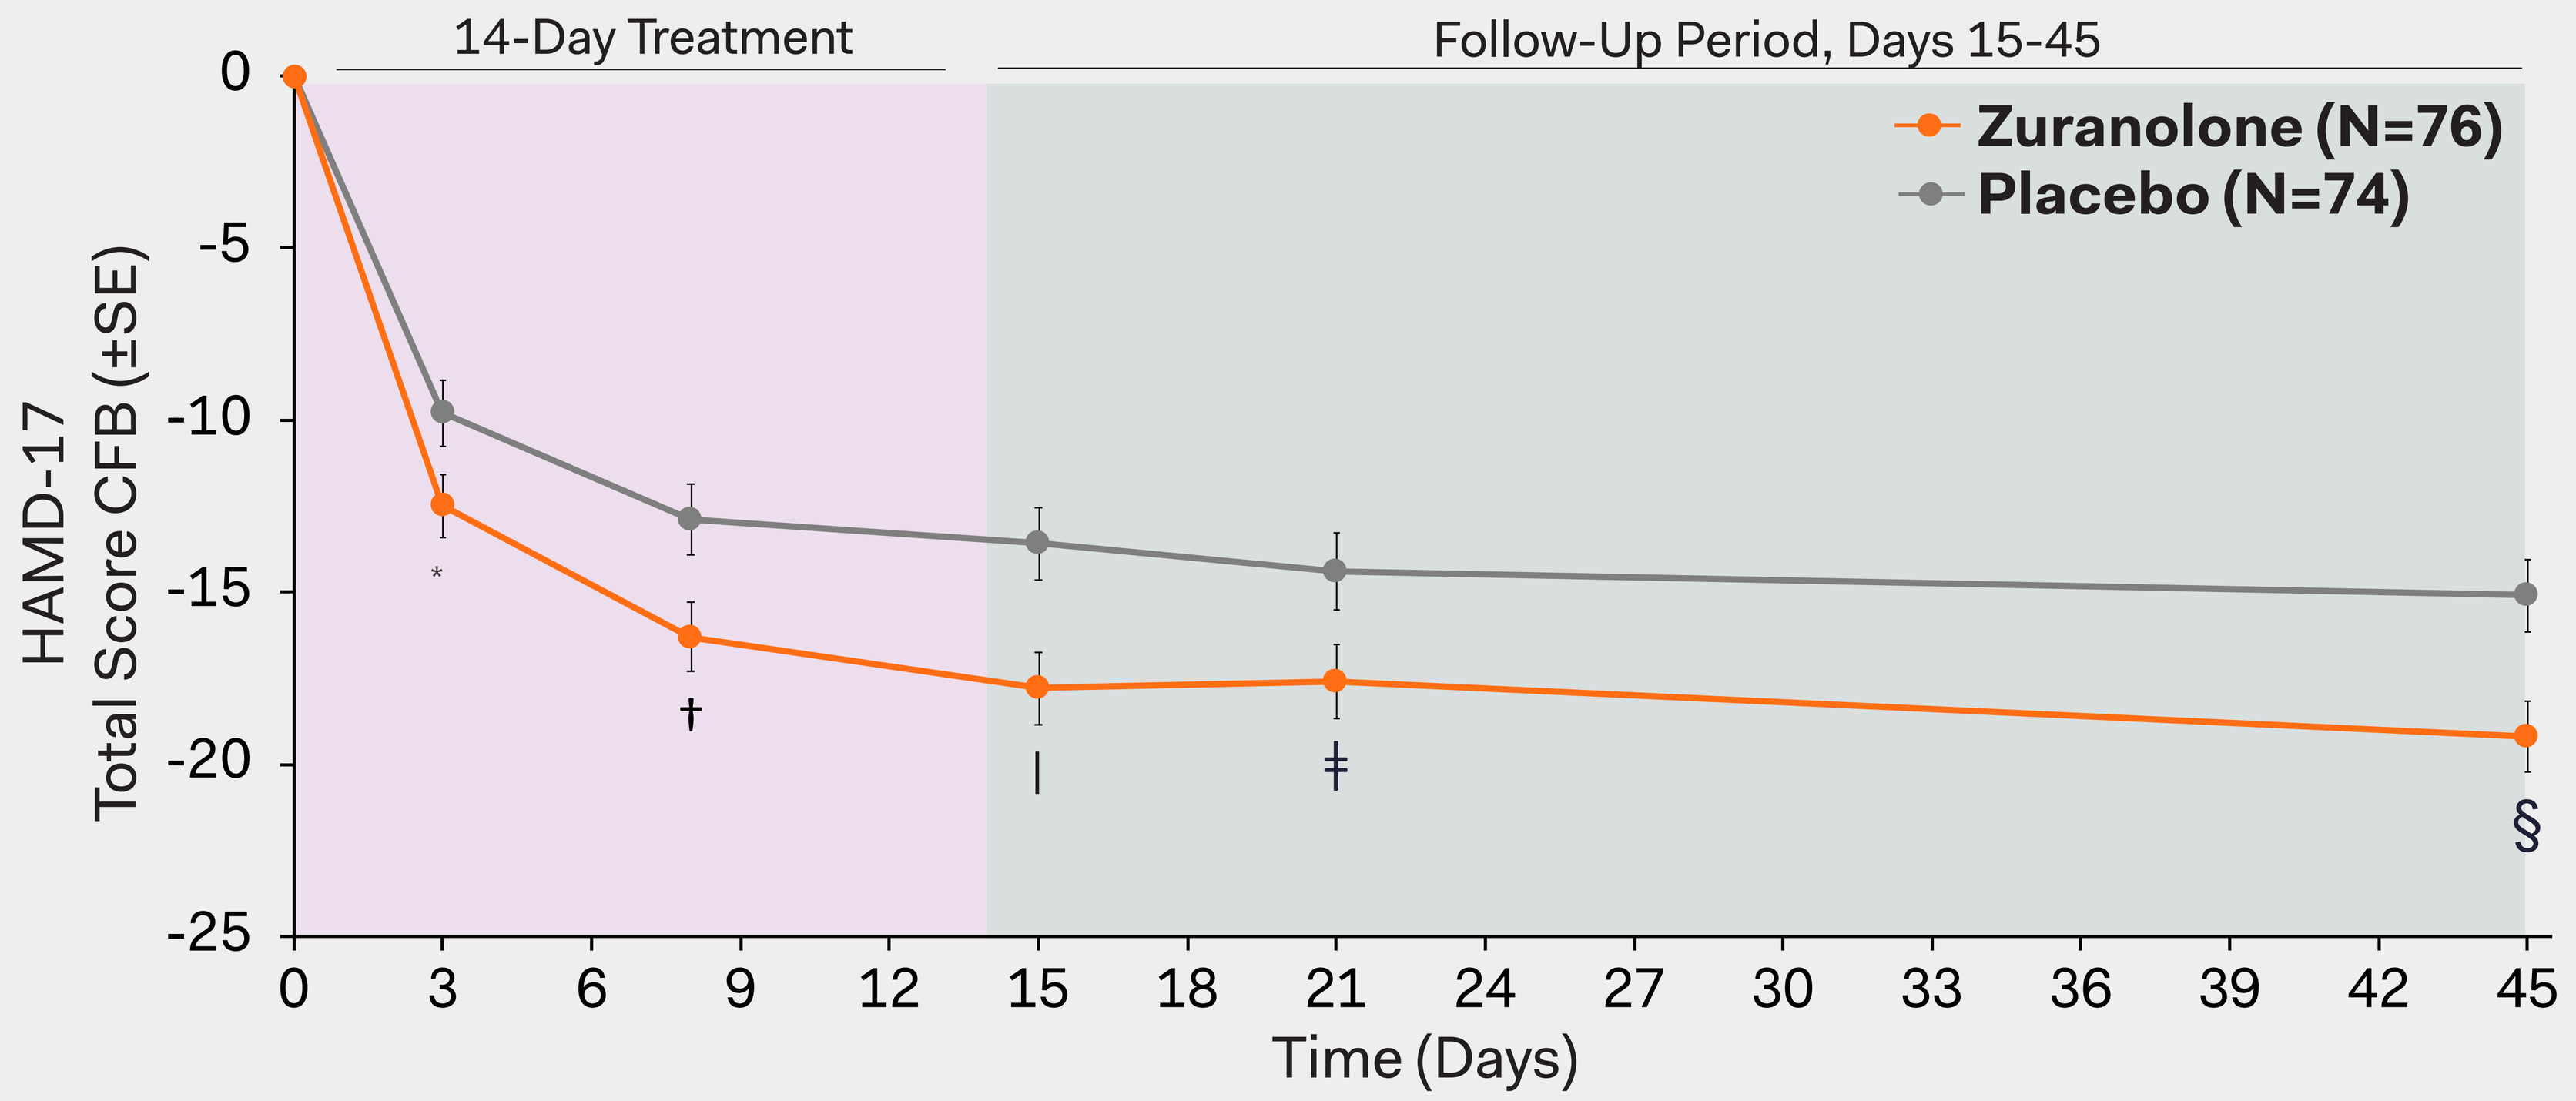
14-Day Treatment
Follow-Up Period, Days 15-45
Zuranolone (N=76)
Placebo (N=74)
HAMD-17
Total Score CFB (±SE)
0
-5
-10
-15
-20
-25
0 3 6 9 12 15 18 21 24 27 30 33 36 39 42 45
Time (Days)
*
†
|
‡
§

*p=0.0252; †p=0.0106; |p=0.0028; ‡p=0.0321; §p=0.0027 vs placebo.

— As previously presented, patients treated with zuranolone met the primary efficacy endpoint at Day 15 by achieving a significantly greater HAMD-17 total score CFB vs those who received placebo (p=0.0028).[25]

CGI-I AND CGI-S RESPONSE AND REMISSION CRITERIA

	CRITERION
CGI-I Response	CGI-I score ≤2 ('much improved'/'very much improved')
CGI-I Remission	CGI-I score ≤1 ('very much improved')
CGI-S Response	CGI-S score ≤2 ('borderline mentally ill'/'normal, not at all ill')
CGI-S Remission	CGI-S score ≤1 ('normal, not at all ill')

NNT FOR CGI-I AND CGI-S RESPONSE AND REMISSION, DAY 15 AND DAY 45

	DAY 15 NNT (95% CI)	DAY 45 NNT (95% CI)
CGI-I Response	6 (3, 26)	7 (ns)
CGI-I Remission	8 (ns)	5 (3, 13)
CGI-S Response	5 (3, 13)	5 (3, 26)
CGI-S Remission	6 (4, 18)	4 (3, 7)

Placebo: N= 68-73; Zuranolone: N=73-74; ns: not statistically significant at the p<0.05 threshold

— Single-digit NNTs for CGI-I response at Day 15 and remission at Day 45, and for CGI-S response and remission at Day 15 and Day 45, support the robustness of the observed zuranolone effect.

SIGNIFICANTLY HIGHER PROPORTION OF PATIENTS ACHIEVED CGI-I RESPONSE OR REMISSION WITH ZURANOLONE VS PLACEBO

Zuranolone (N=73-74) Placebo (N=68-73)

CGI-I RESPONSE

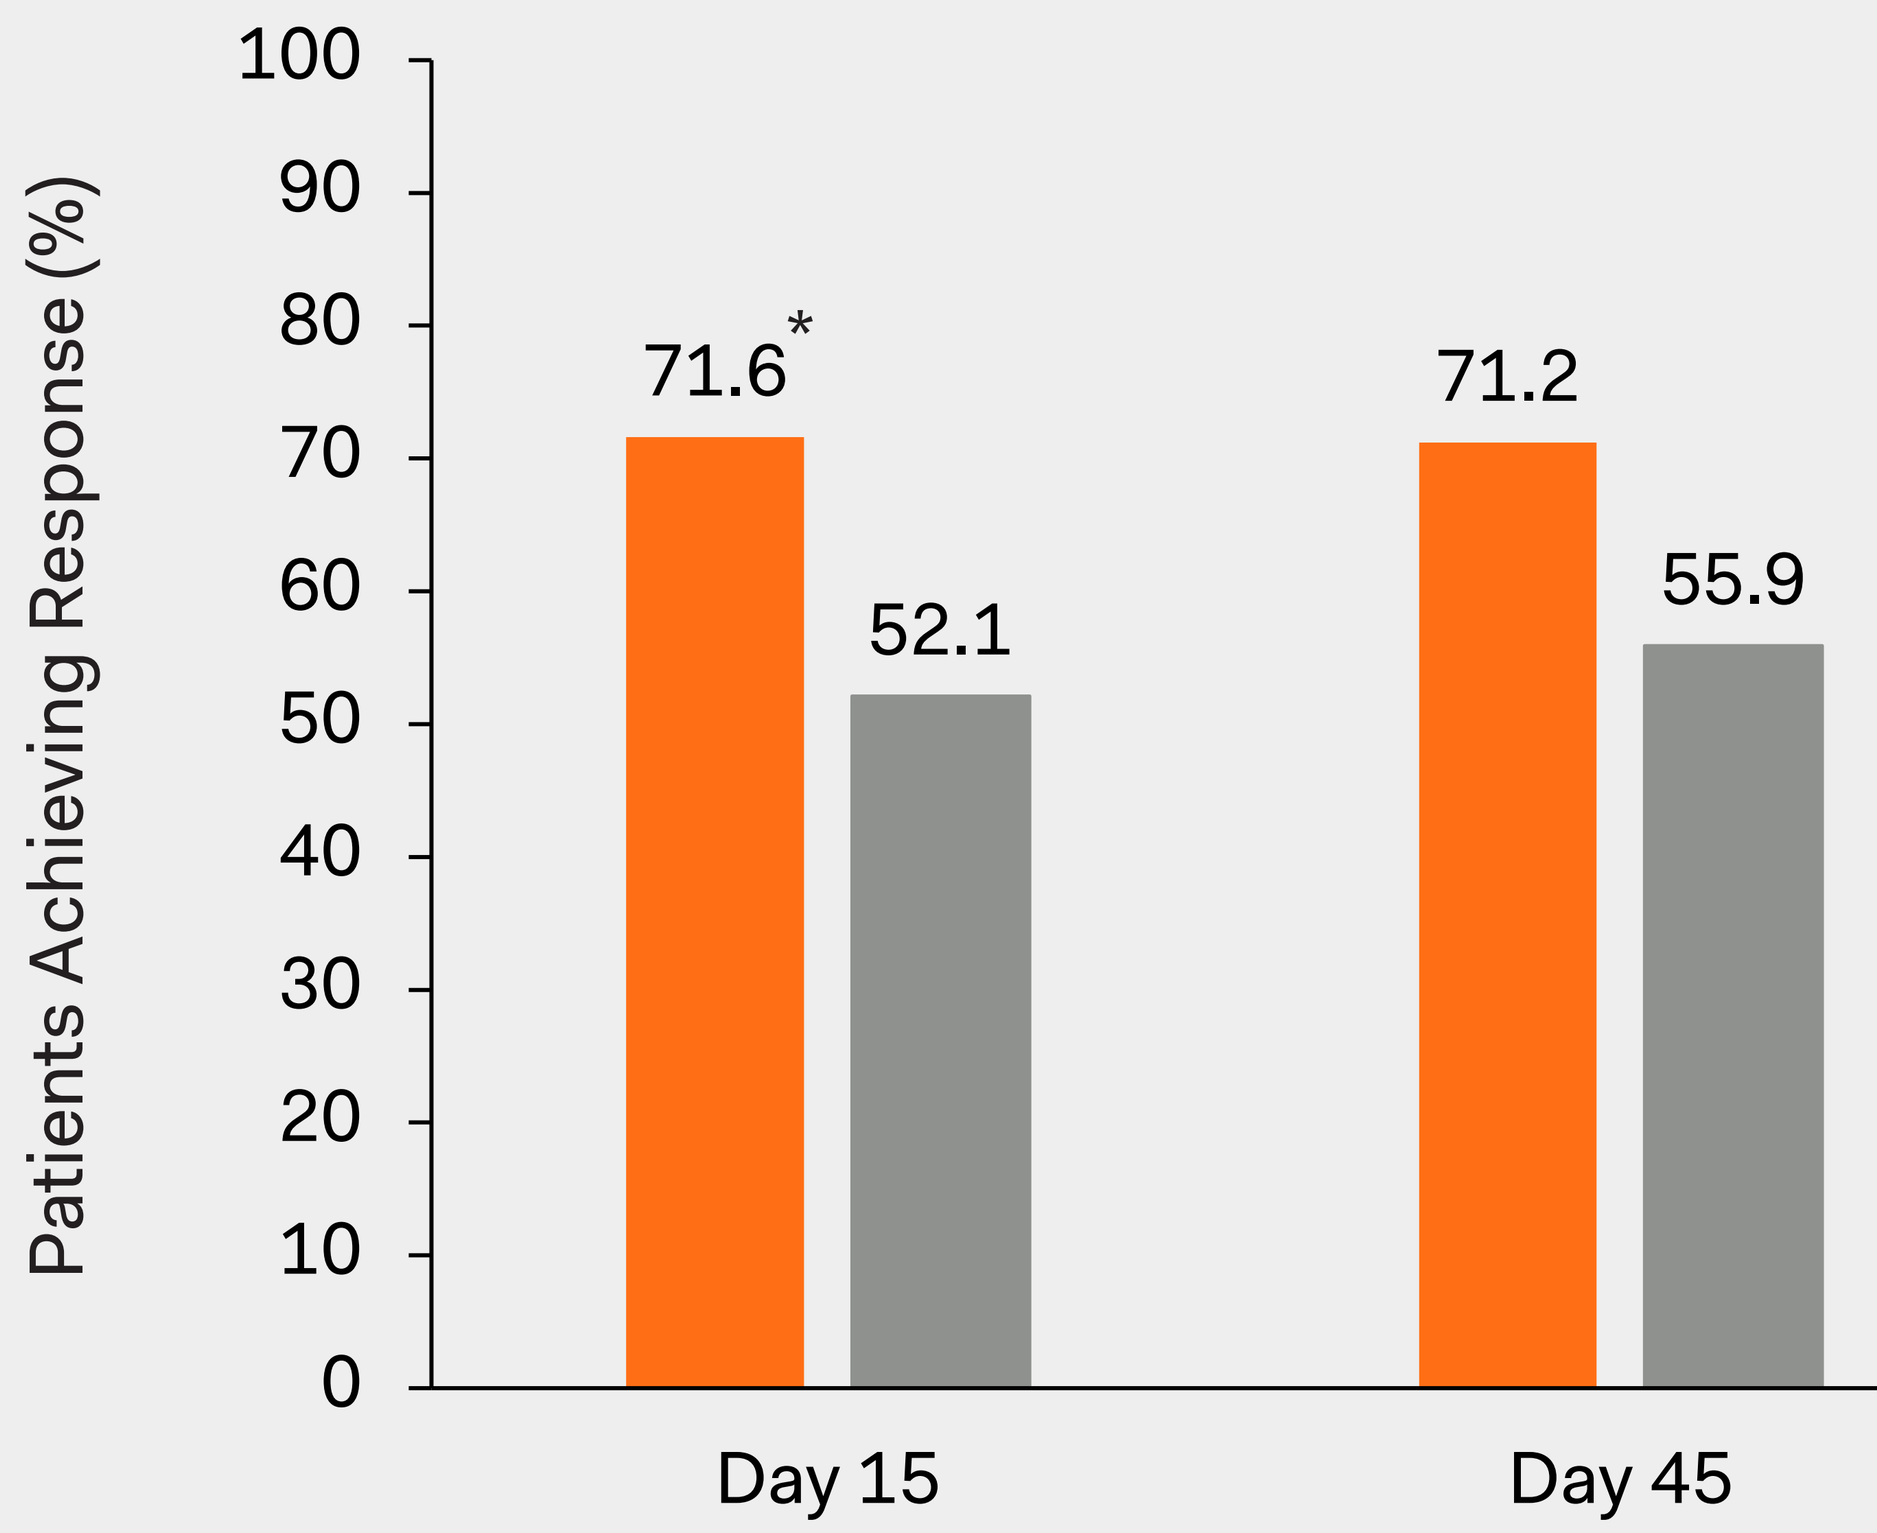
Patients Achieving Response (%)
100
90
80
70
60
50
40
30
20
10
0
71.6*
52.1
71.2
55.9
Day 15
Day 45

*p=0.0276; p-values compare adjusted rates from the models (not the raw rates presented in the charts).

— A significantly higher proportion of zuranolone-treated patients achieved CGI-I response at Day 15 compared with placebo (p=0.0276).

CGI-I REMISSION

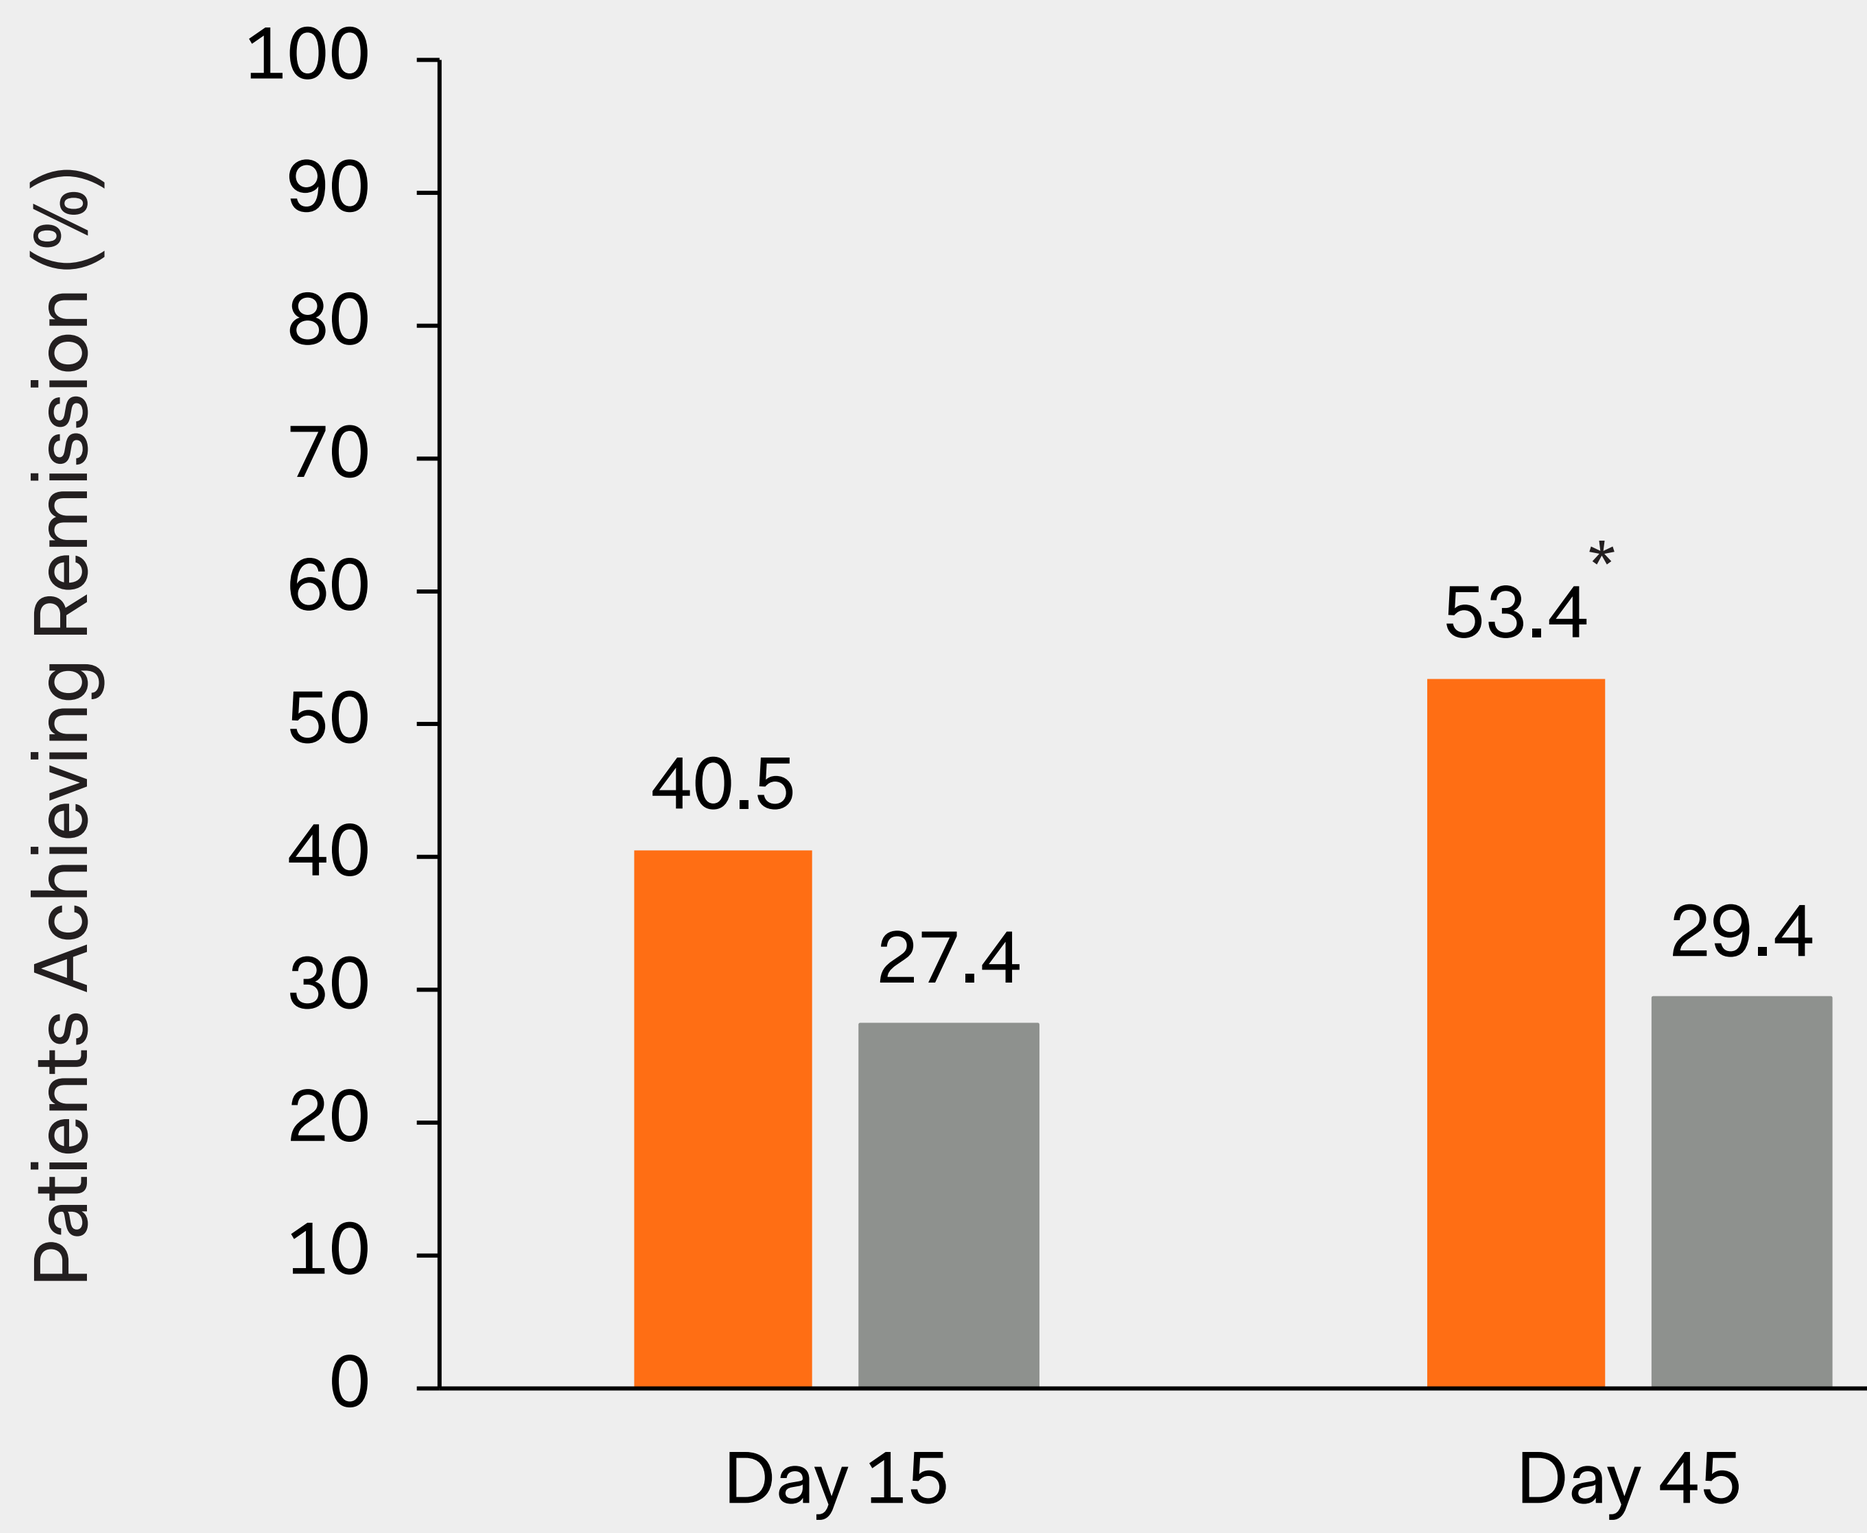
Patients Achieving Remission (%)
100
90
80
70
60
50
40
30
20
10
0
40.5
27.4
53.4*
29.4
Day 15
Day 45

*p=0.0111; p-values compare adjusted rates from the models (not the raw rates presented in the charts).

— A significantly higher proportion of zuranolone-treated patients achieved CGI-I remission at Day 45 compared with placebo (p=0.0111).

SIGNIFICANTLY HIGHER PROPORTION OF PATIENTS ACHIEVED CGI-S RESPONSE OR REMISSION WITH ZURANOLONE VS PLACEBO

CGI-S RESPONSE

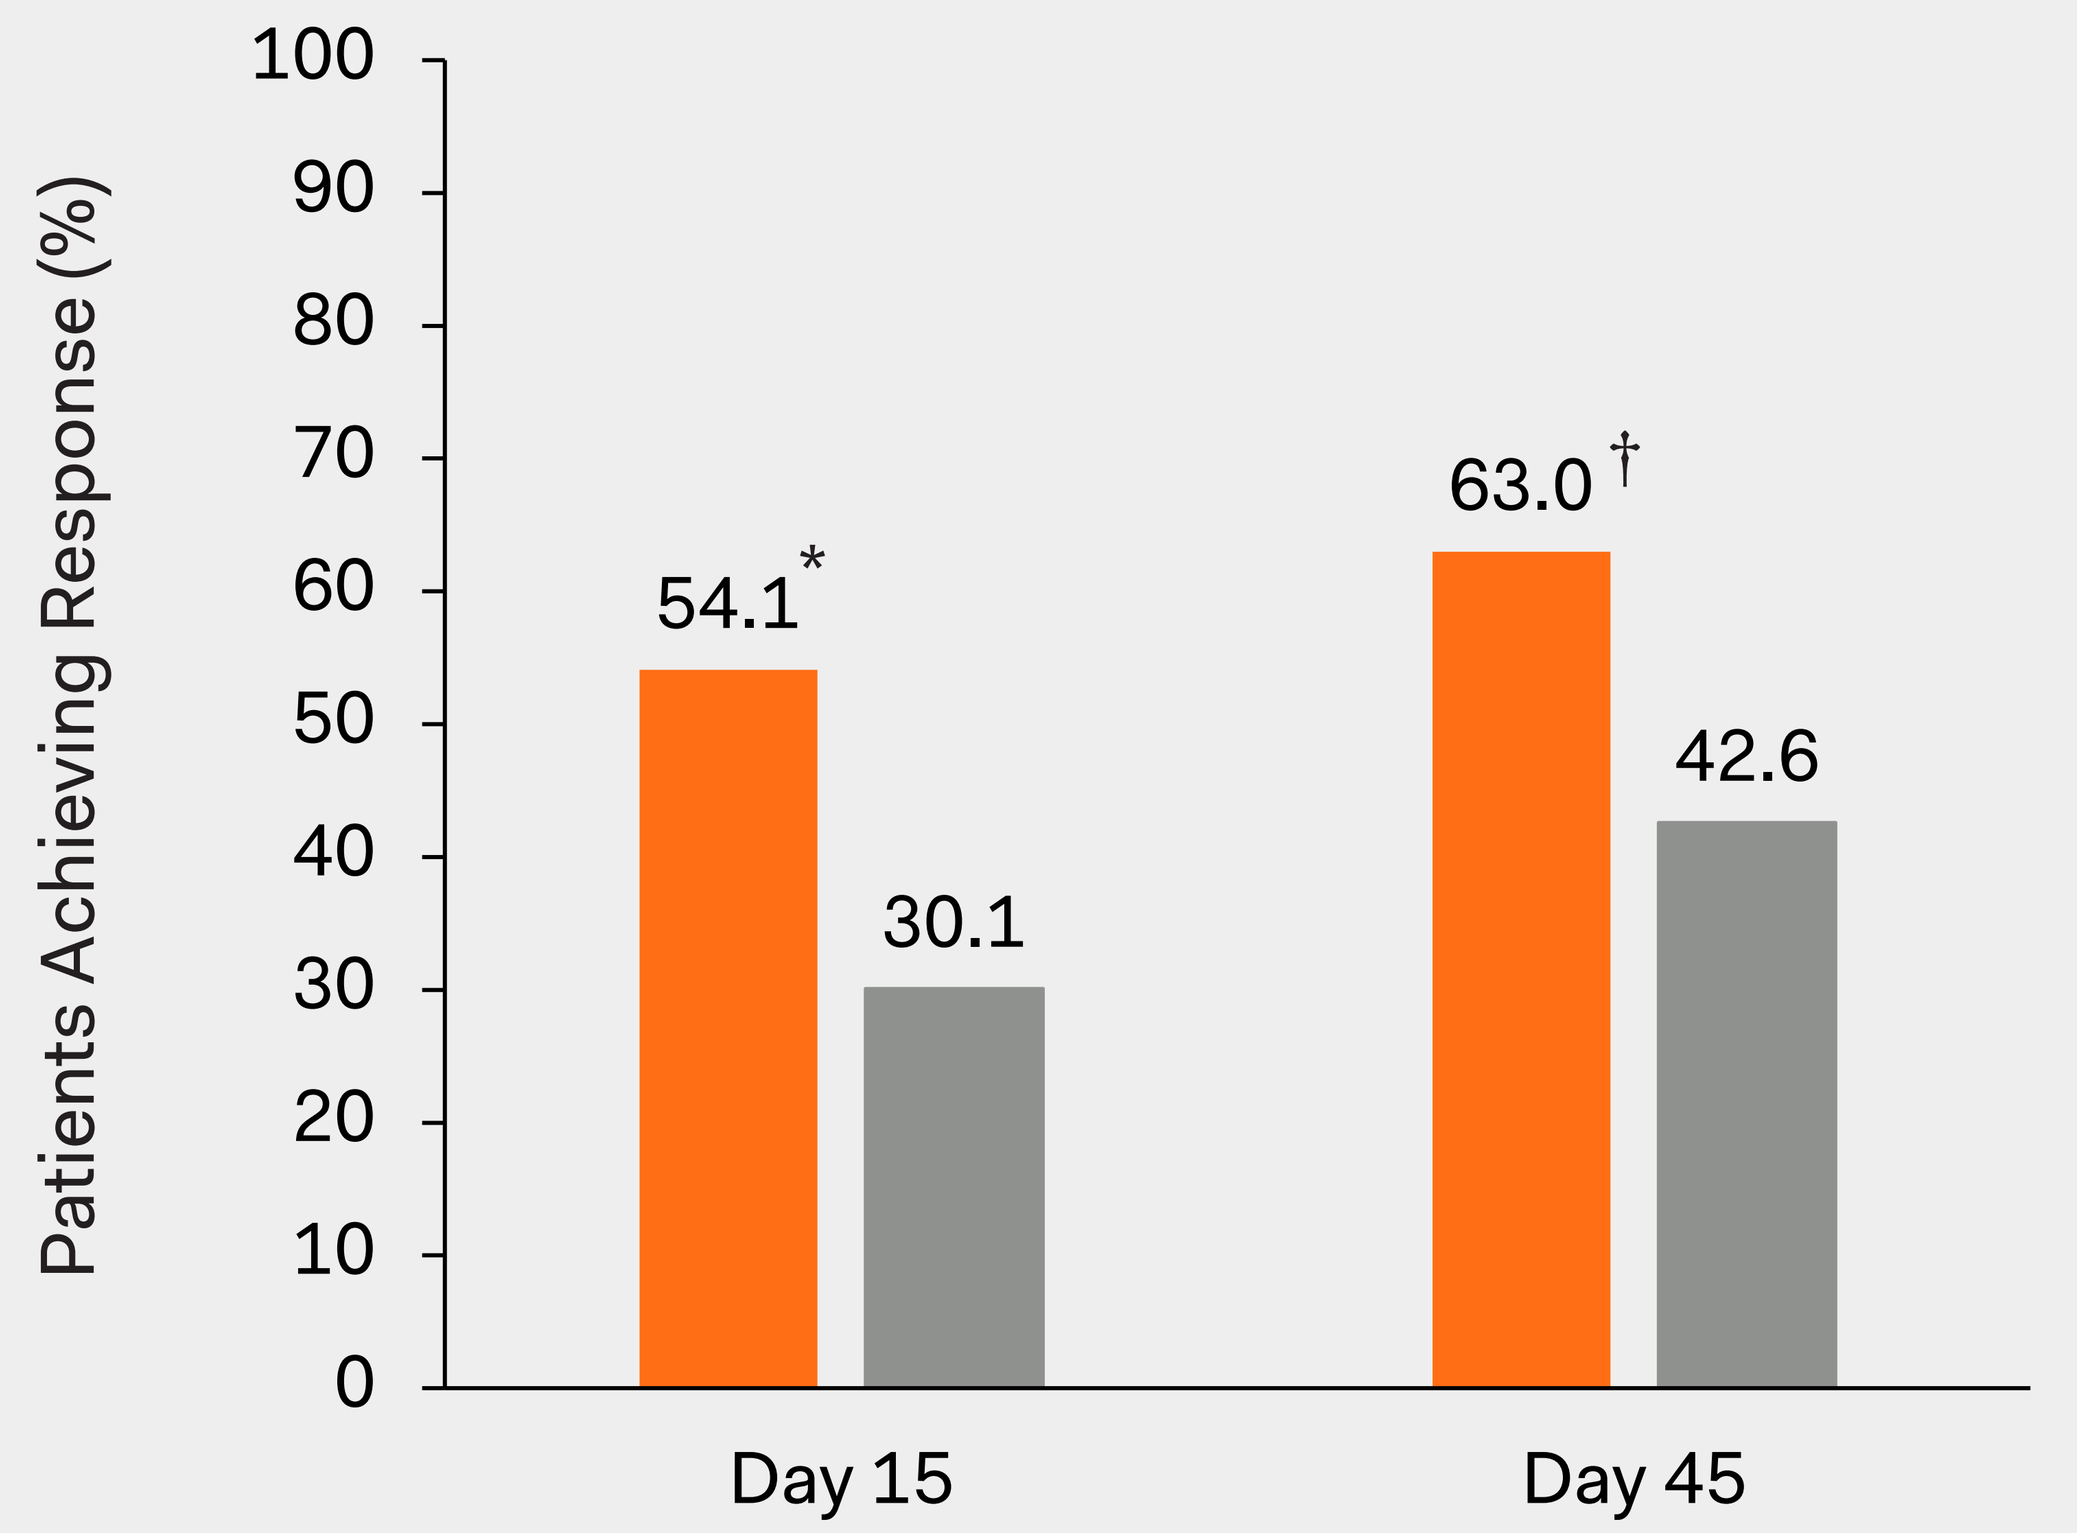
Patients Achieving Response (%)
100
90
80
70
60
50
40
30
20
10
0
54.1*
30.1
63.0†
42.6
Day 15
Day 45

*p=0.0070, †p=0.0334; p-values compare adjusted rates from the models (not the raw rates presented in the charts).

— A significantly higher proportion of zuranolone-treated patients achieved CGI-S response at Day 15 (p=0.0070) and at Day 45 (p=0.0334) compared with placebo.

CGI-S REMISSION

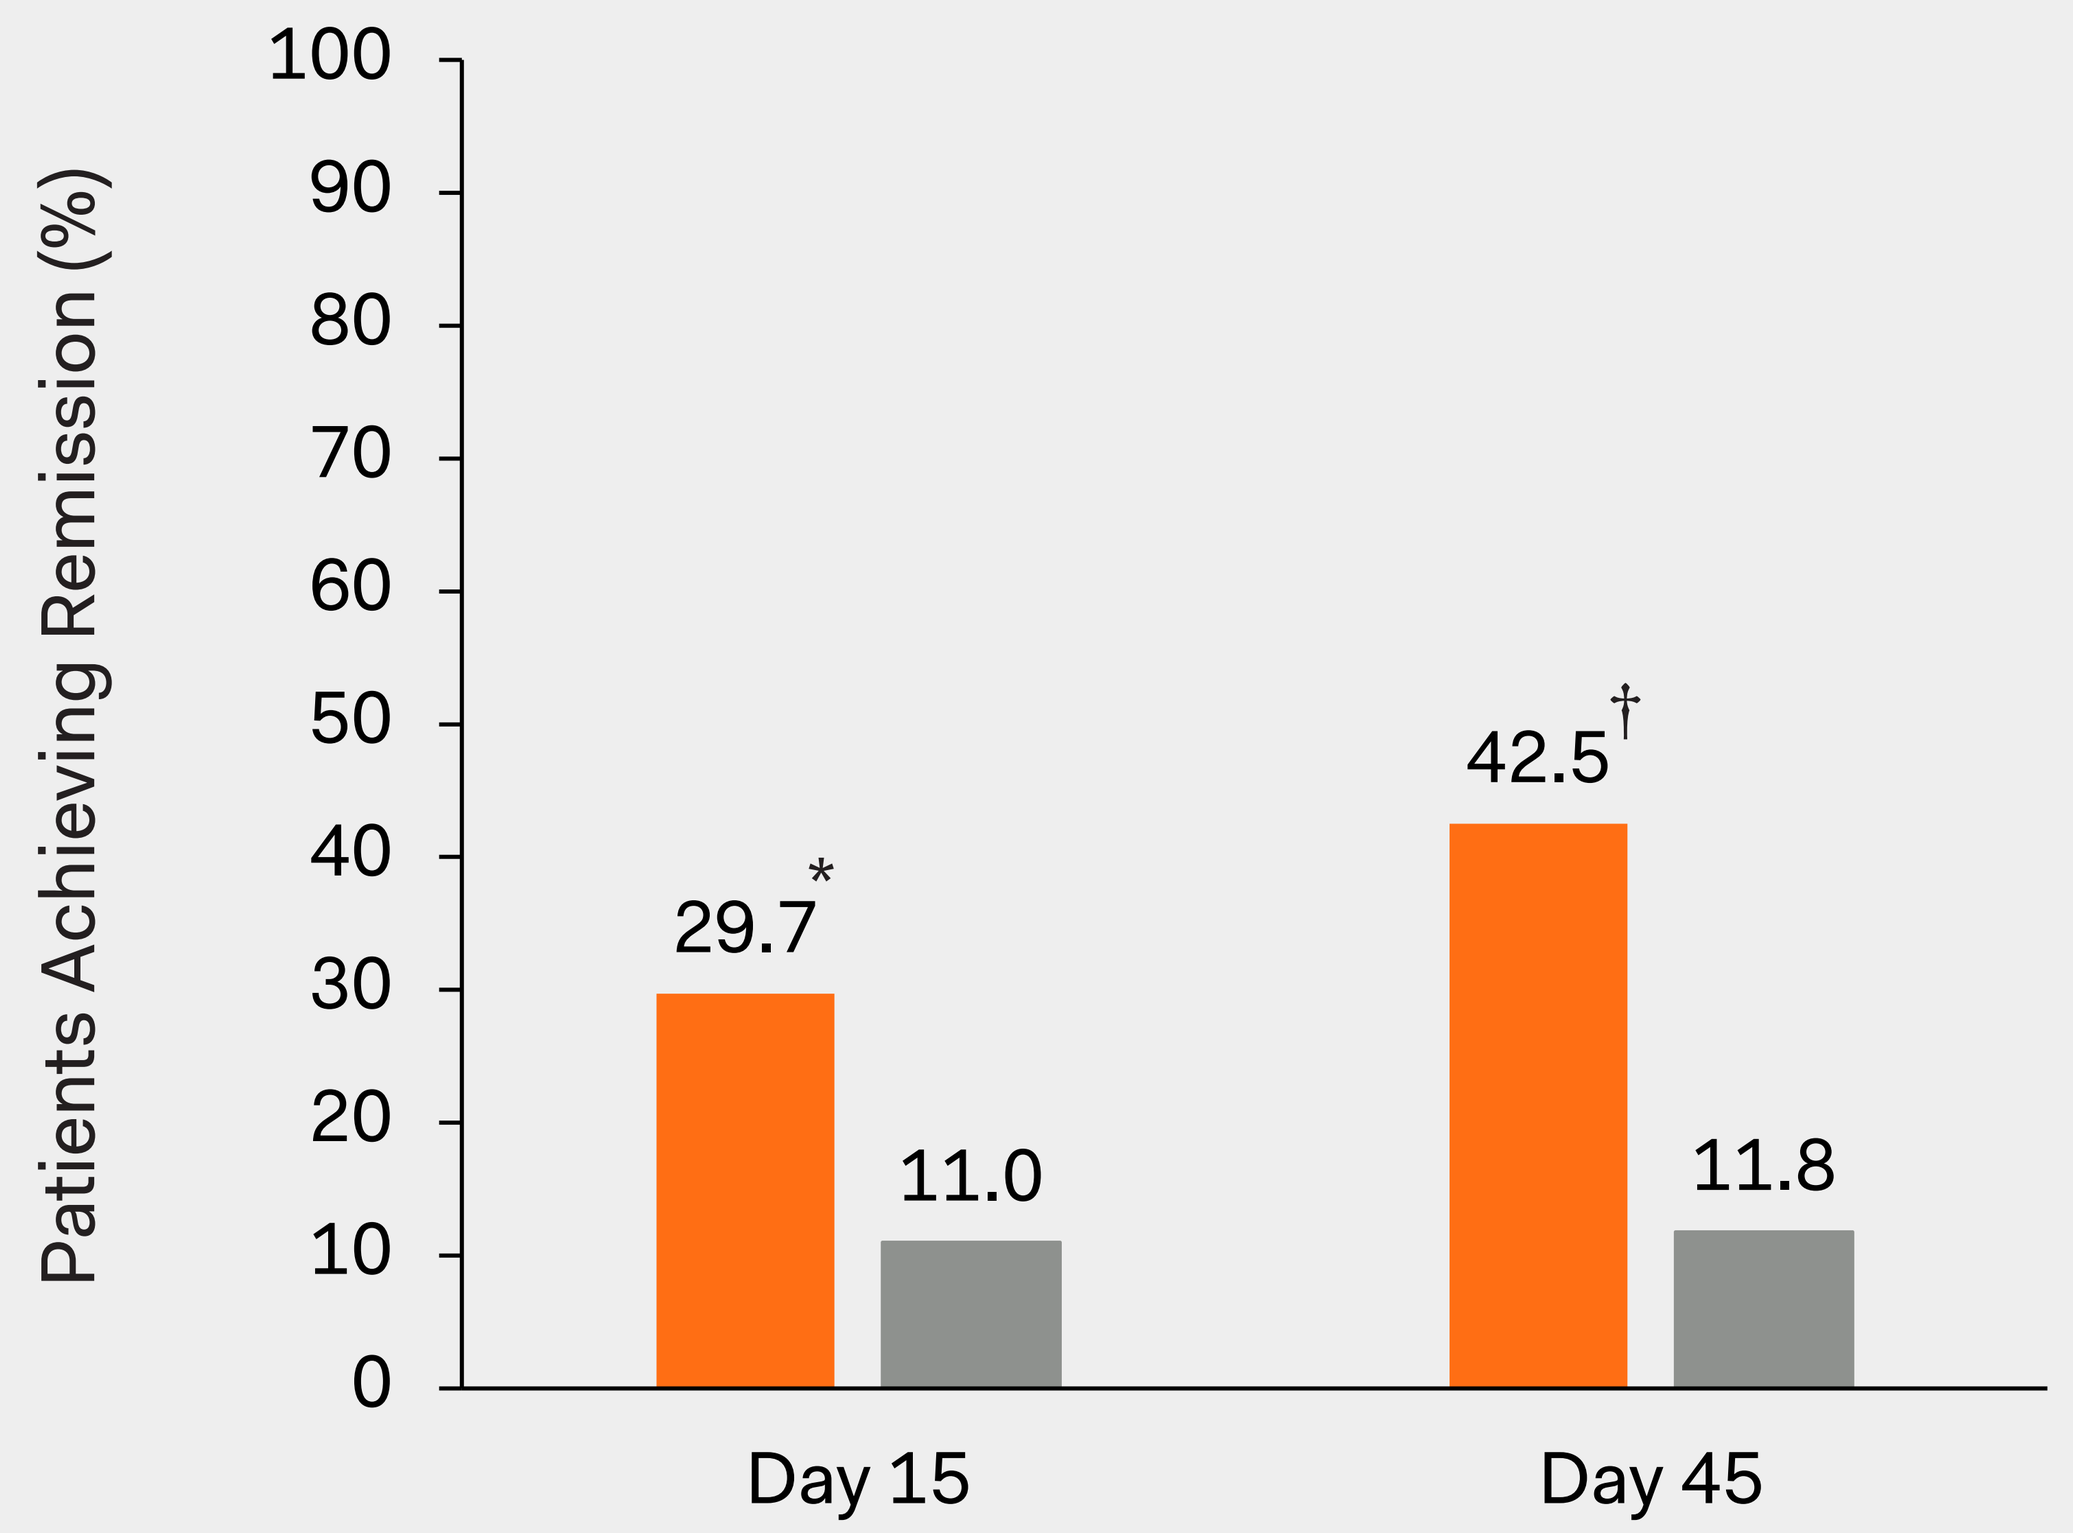
Patients Achieving Remission (%)
100
90
80
70
60
50
40
30
20
10
0
29.7*
11.0
42.5†
11.8
Day 15
Day 45

*p=0.0117, †p=0.0007; p-values compare adjusted rates from the models (not the raw rates presented in the charts).

— A significantly higher proportion of zuranolone-treated patients achieved CGI-S remission at Day 15 (p=0.0117) and at Day 45 (p=0.0007) compared with placebo.

REFERENCES
1. Bauman BL, et al. *MMWR Morb Mortal Wkly Rep* (2020) 69:575-581.
2. Hamilton BE, et al. National Center for Health Statistics. Births: Provisional data for 2018. Vital Statistics Rapid Release; no 7. https://www.cdc.gov/nchs/data/vsrr/vsrr-007-508.pdf. Published May 2019. Accessed November 10, 2020.
3. DeSisto CL, et al. *Prev Chronic Dis* (2014) 11:E10.
4. Centers for Disease Control and Prevention. https://www.cdc.gov/reproductivehealthmaternalinfanthealth/diabetes-during-pregnancy.htm. Updated June 12, 2018 Accessed November 10, 2020.
5. Centers for Disease Control and Prevention. https://www.cdc.gov/reproductivehealth/maternalinfanthealth/pregnancy-complications-data.htm. Updated February 28, 2019. Accessed November 10, 2020.
6. Roberts JM, et al. American College of Obstetricians and Gynecologists Task Force on Hypertension in Pregnancy. https://www.acog.org/~/media/Task%20Force%20and%20Work%20Group%20Reportspublic/HypertensioninPregnancy.pdf. Published 2013. Accessed November 10, 2020.
7. Callaghan WM, et al. *Am J Obstet Gynecol* (2010) 202:353.e1-6.
8. Diagnostic and statistical manual of mental disorders: DSM-5. Washington, DC: American Psychiatric Association; 2013.
9. Kerstis B, et al. *Arch Womens Ment Health* (2016) 19:87.
10. Posmontier B, et al. *J Midwifery Womens Health* (2008) 53:310-318.
11. Barkin JL, et al. *J Womens Health* (Larchmt; 2016) 25:707-713.
12. Gagliardi L, et al. *Arch Dis Child* (2012) 97:355-357.
13. McLearn KT, et al. *Arch Pediatr Adolesc Med* (2006) 160:279-284.
14. Moore Simas TA, et al. *J Med Econ* (2019) 23:174-183.
15. Balbierz A, et al. *Matern Child Health J* (2015) 19:1212-1219.
16. Valla L, et al. *Infant Behav Dev* (2016) 45:83-90.
17. Koutra K, et al. *Soc Psychiatry Psychiatr Epidemiol* (2013) 48:1335-1345.
18. Robertson E, et al. *Gen Hosp Psychiatry* (2004) 26:289-295.
19. Biaggi A, et al. *J Affect Disord* (2016) 191:62-77.
20. Yim IS, et al. *Annu Rev Clin Psychol* (2015) 11:99-137.
21. Hoffmann E et al. *Clin Pharmacokinet* (2019) 59: 111-120.
22. Martinez Botella G, et al. *J Med Chem* (2017) 60:7810-7819.
23. Belfort GM, et al. Poster presented at American Epilepsy Society 2016.
24. Hamilton M. *J Neurol Neurosurg Psychiatry* (1960) 23: 56–62.
25. Deligiannidis KM, et al. Presented as ASCP 2019. Poster T74.
26. Busner J and Targum SD. *Psychiatry* (2007) July:29-37.
27. Deligiannidis KM, et al. Poster presentation at ACNP 2019. Poster W79.
28. Data on file MLIB-3497 Robin Safety Data. [217-PPD-201]. Sage Therapeutics, Inc. [2019].

SAGE THERAPEUTICS and the SAGE THERAPEUTICS logo are trademarks of Sage Therapeutics, Inc. All other trademarks referenced herein are the property of their respective owners.

SUPPORT & DISCLOSURES
BW, MYH, ES, RL, HGB, VB, and SJK are employees of Sage Therapeutics, Inc., with stock/stock options. KMD serves as a consultant to Sage Therapeutics, Inc., receives NIMH support and royalties from an NIH employee invention, and reports grants from Sage Therapeutics, Inc. awarded to Zucker Hillside Hospital during the conduct of the zuranolone clinical trials. SA and MF consulted for Sage Therapeutics, Inc. This study and medical writing support were funded by Sage Therapeutics, Inc. Writing and editorial support was provided by Bonnie L. Bullock, PhD and Kathryn P. Wall, PhD of Boston Strategic Partners, Inc.

American College of Neuropsychopharmacology 2020
Virtual Annual Meeting
December 6-9, 2020

MRC-PPD-00515 11/2020