Sage Therapeutics™

Insomnia Symptom Response in Patients with Postpartum Depression Treated with the Neuroactive Steroid Brexanolone Injection

Samantha Meltzer-Brody, MD, MPH[1], Brian Werneburg, PhD[2], Robert Riesenberg, MD[3], C. Neill Epperson, MD[4], Kristina M. Deligiannidis, MD[5], David R. Rubinow, MD[1], Kemi Bankole, MD[2], Ming-Yi Huang, PhD[2], Haihong Li, PhD[2], Stephen J. Kanes, MD, PhD[2]

[1] Department of Psychiatry, University of North Carolina School of Medicine, Chapel Hill, NC, USA
[2] Sage Therapeutics, Inc., Cambridge, MA, USA
[3] Atlanta Center for Medical Research, Atlanta, GA, USA
[4] Department of Psychiatry, University of Colorado School of Medicine, Aurora, CO, USA
[5] Department of Psychiatry, Women's Behavioral Health, Zucker Hillside Hospital, Glen Oaks, NY, USA

Introduction

- Postpartum depression (PPD) is one of the most common medical complications during and after pregnancy.[1-7]
- Altered sleep states are both a risk factor for and a symptom of PPD.[8-11]
 - Reduced sleep quality may be predictive of increased depressive symptomology.[12,13]
- Brexanolone injection (BRX),[14] a neuroactive steroid $GABA_A$ receptor positive allosteric modulator with a pharmacological profile distinct from benzodiazepines[15], is approved by the FDA for the treatment of PPD in adults.[14]
- The safety, tolerability, and efficacy of BRX in PPD were evaluated in three double-blind, randomized, placebo-controlled trials (RCTs) where patients treated with BRX90 met the primary efficacy endpoint at Hour 60, by achieving significantly greater Hamilton Rating Scale for Depression total score (HAMD-17)[16] change from baseline (CFB) vs patients receiving placebo (p<0.0001).[17]
- In addition, improvement from baseline in the core symptoms of depression using the predefined Bech-6 subscale of the HAMD-17 favored BRX90 compared with placebo beginning at Hour 24 (p=0.0173) and at all measured timepoints to Day 30 (p=0.0301).[17]
- Post hoc analyses from an integrated dataset of the three pivotal BRX RCTs assessed an effect on insomnia symptoms using the HAMD-17 insomnia subscale (Ins).

Conclusions

- BRX treatment in women with PPD has previously been shown to provide rapid (Hour 24 for HAMD-17 total score and Bech-6 score) and sustained improvement in depressive symptoms (HAMD-17 total score and Bech-6 score at all subsequent measured time points up to Day 30) compared with placebo.[17]
- In addition to its effects on core symptoms of depression in this trial, these post hoc analyses showed that BRX treatment also resulted in significantly greater improvement in reported symptoms of insomnia from the HAMD-17-Ins, and higher proportions of patients achieving HAMD-17-Ins response compared with placebo.

Methods

- An integrated dataset of three pivotal studies in PPD was used for these analyses.
- Women (N=247) ages 18-45, ≤6 months postpartum, with PPD (defined as a major depressive episode with onset in the 3rd trimester or ≤4 weeks postpartum) and a qualifying HAMD-17 total score (Studies A and B: HAMD-17≥26; Study C: HAMD-17 20-25) were enrolled.
- Studies A and C used 1:1 randomization to placebo and BRX 90 μg/kg/h (BRX90), and Study B used 1:1:1 randomization to placebo, BRX 60 μg/kg/h (BRX60), and BRX90.
- Patients received a 60-hour continuous infusion of placebo or BRX in a monitored setting, with four weeks follow-up.
- The primary endpoint in all studies was the least-squares mean change from baseline (CFB) in mean HAMD-17 total score at Hour 60.
- LSM HAMD-17 CFB data was analyzed using a mixed effects model for repeated measures.
- Secondary endpoints included HAMD-17 total score at all other time points.
- Treatment emergent adverse events (TEAEs) were assessed throughout the study.
- Post hoc analyses included the CFB in HAMD-17-Ins (sum of individual items for early, middle, and late insomnia).
- HAMD-17-Ins response was defined by either a ≥50% or ≥70% reduction in baseline score. Exploratory subgroup analyses of patients with HAMD-17-Ins ≥1, or HAMD-17-Ins ≥4 at baseline were assessed for a categorical response to below those values.
- Response assessments utilized the generalized estimating equation or logistic regression approaches.
- Secondary endpoints and post hoc analyses were not adjusted for multiplicity.

Results

SIGNIFICANTLY GREATER IMPROVEMENT IN DEPRESSIVE AND INSOMNIA-RELATED SYMPTOMS ACHIEVED WITH BRX VS PLACEBO

HAMD-17 TOTAL SCORE

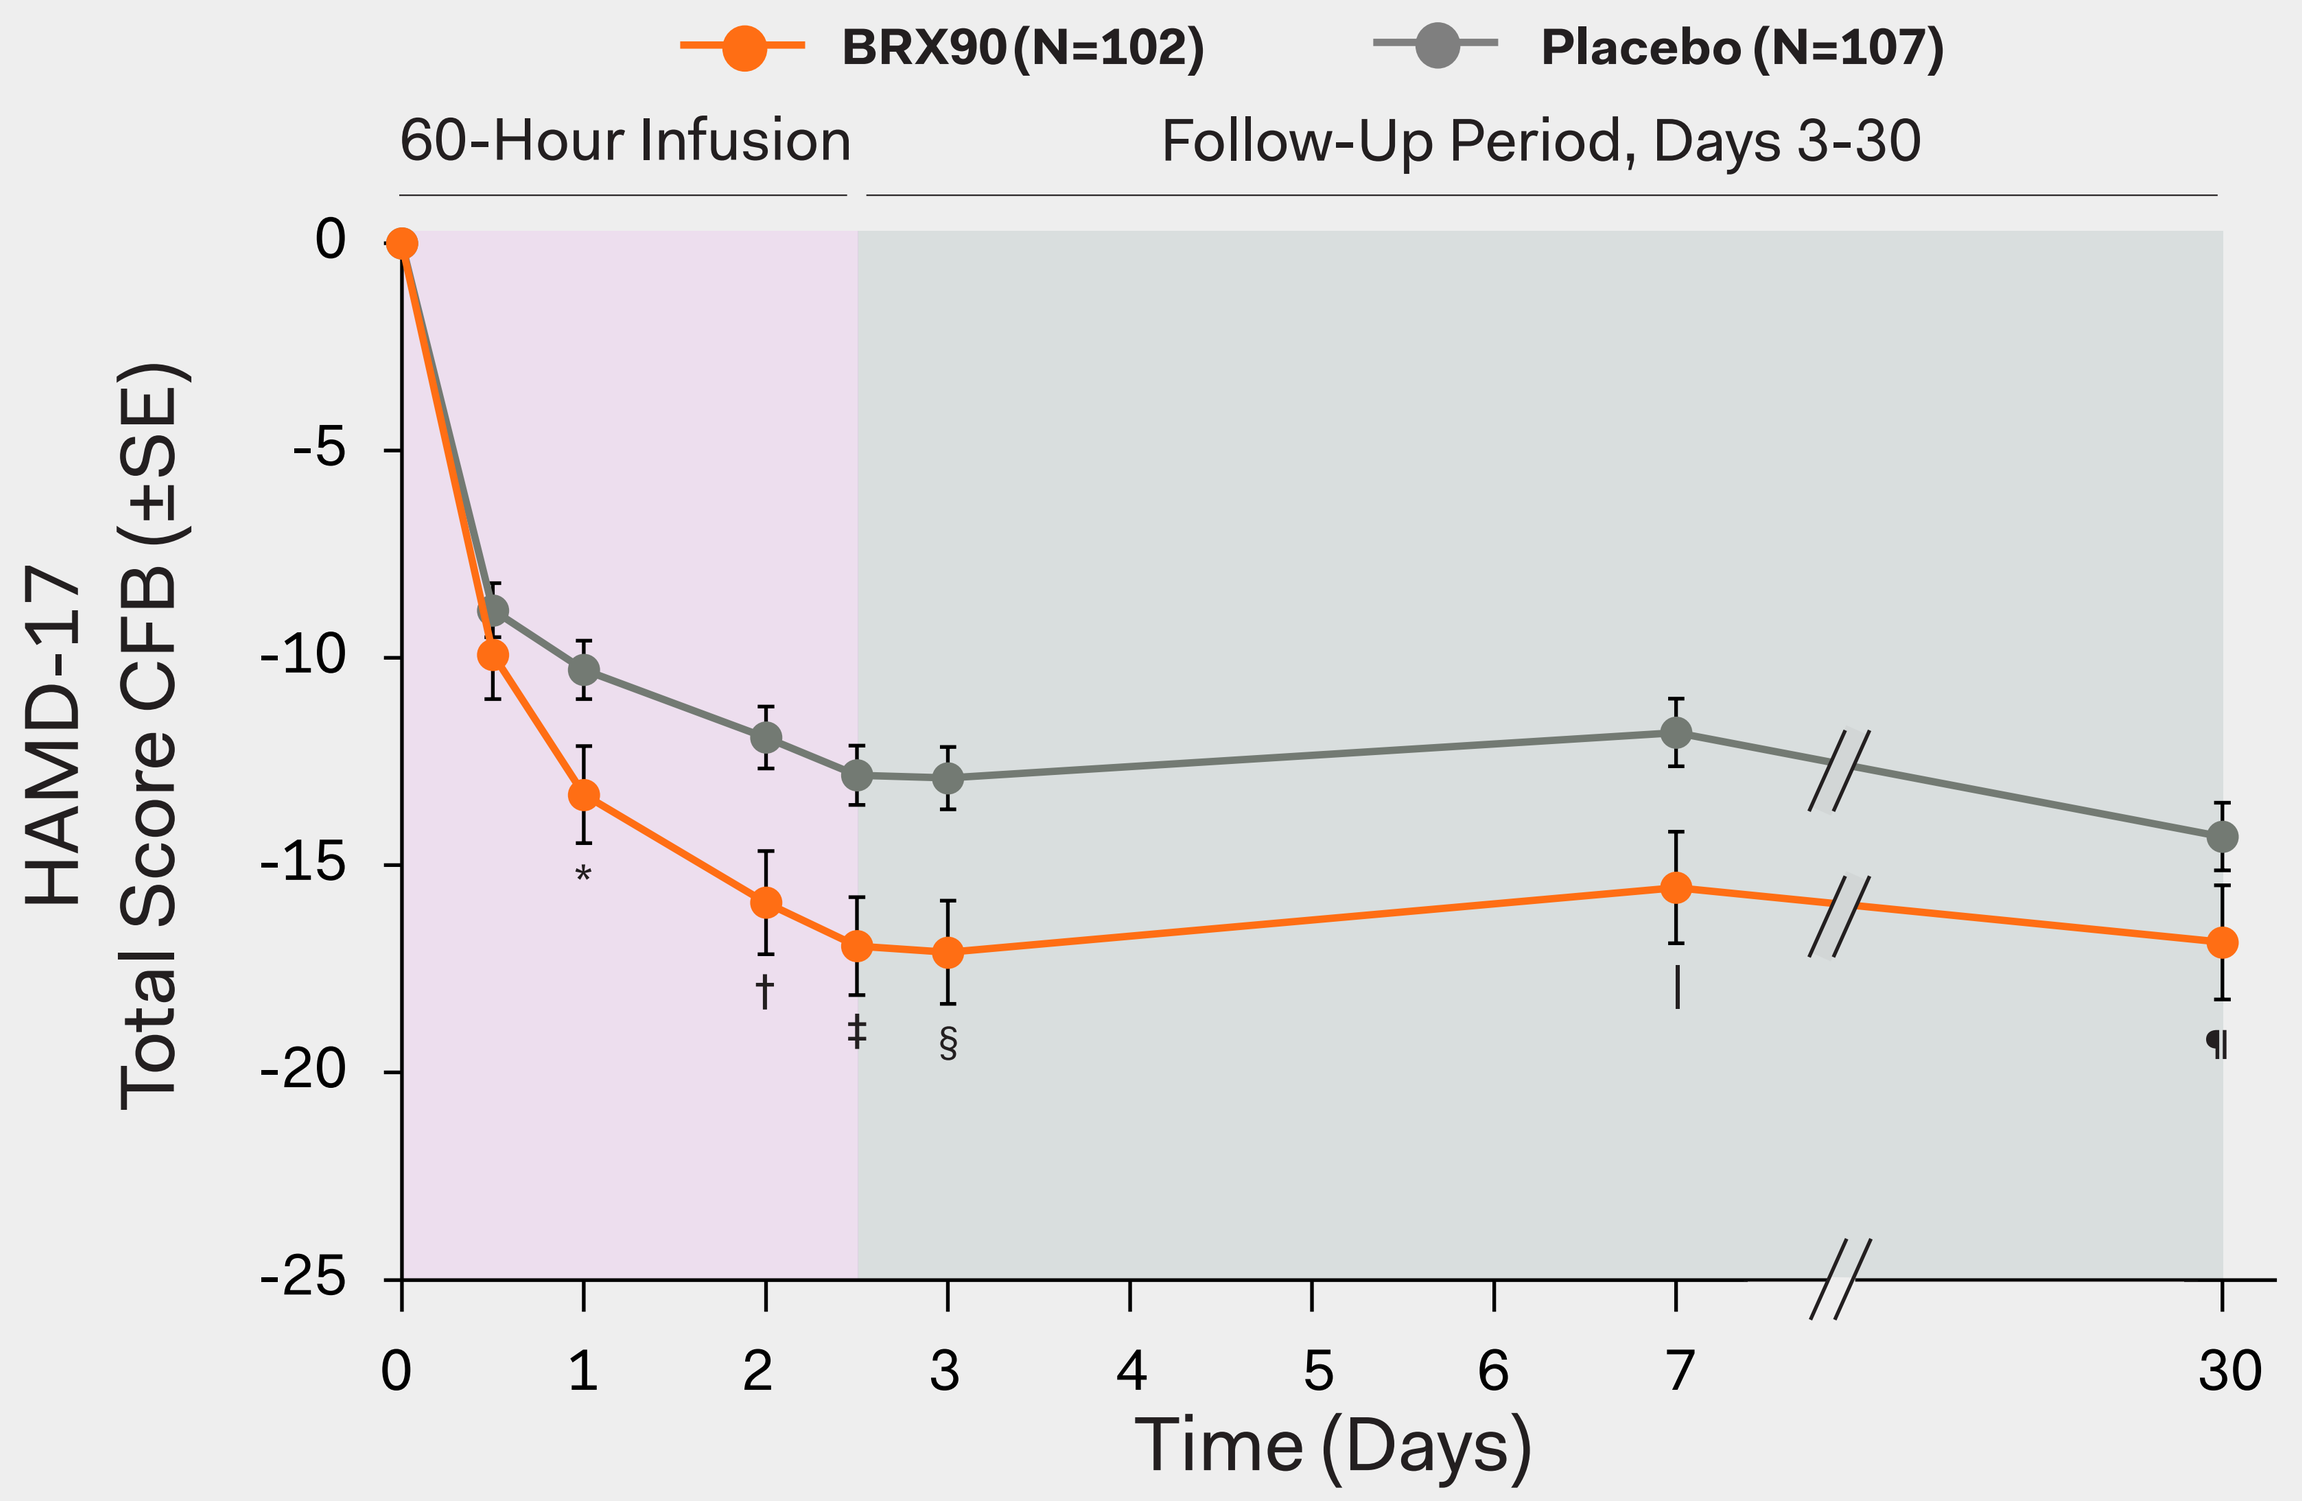
BRX90 (N=102)
Placebo (N=107)
60-Hour Infusion
Follow-Up Period, Days 3-30
HAMD-17
Total Score CFB (±SE)
0
-5
-10
-15
-20
-25
0
1
2
3
4
5
6
7
30
Time (Days)
*
†
‡
§
|
¶

*p=0.0012; †p<0.0001; ‡p<0.0001; §p=0.0001; |p=0.0007; ¶p=0.0213 vs placebo.

- Baseline demographics and patient characteristics were well balanced between the treatment arms and have been described in detail previously.[17]
- As previously presented, patients treated with BRX90 met the primary efficacy endpoint at Hour 60, by achieving significantly greater HAMD-17 total score CFB vs patients receiving placebo (p<0.0001).[17]
- Significant differences vs placebo in HAMD-17 CFB favoring BRX90 were observed at Hour 24 (p=0.0012) and were sustained at Day 30 (p=0.0213).

HAMD-17-INS SUBSCALE

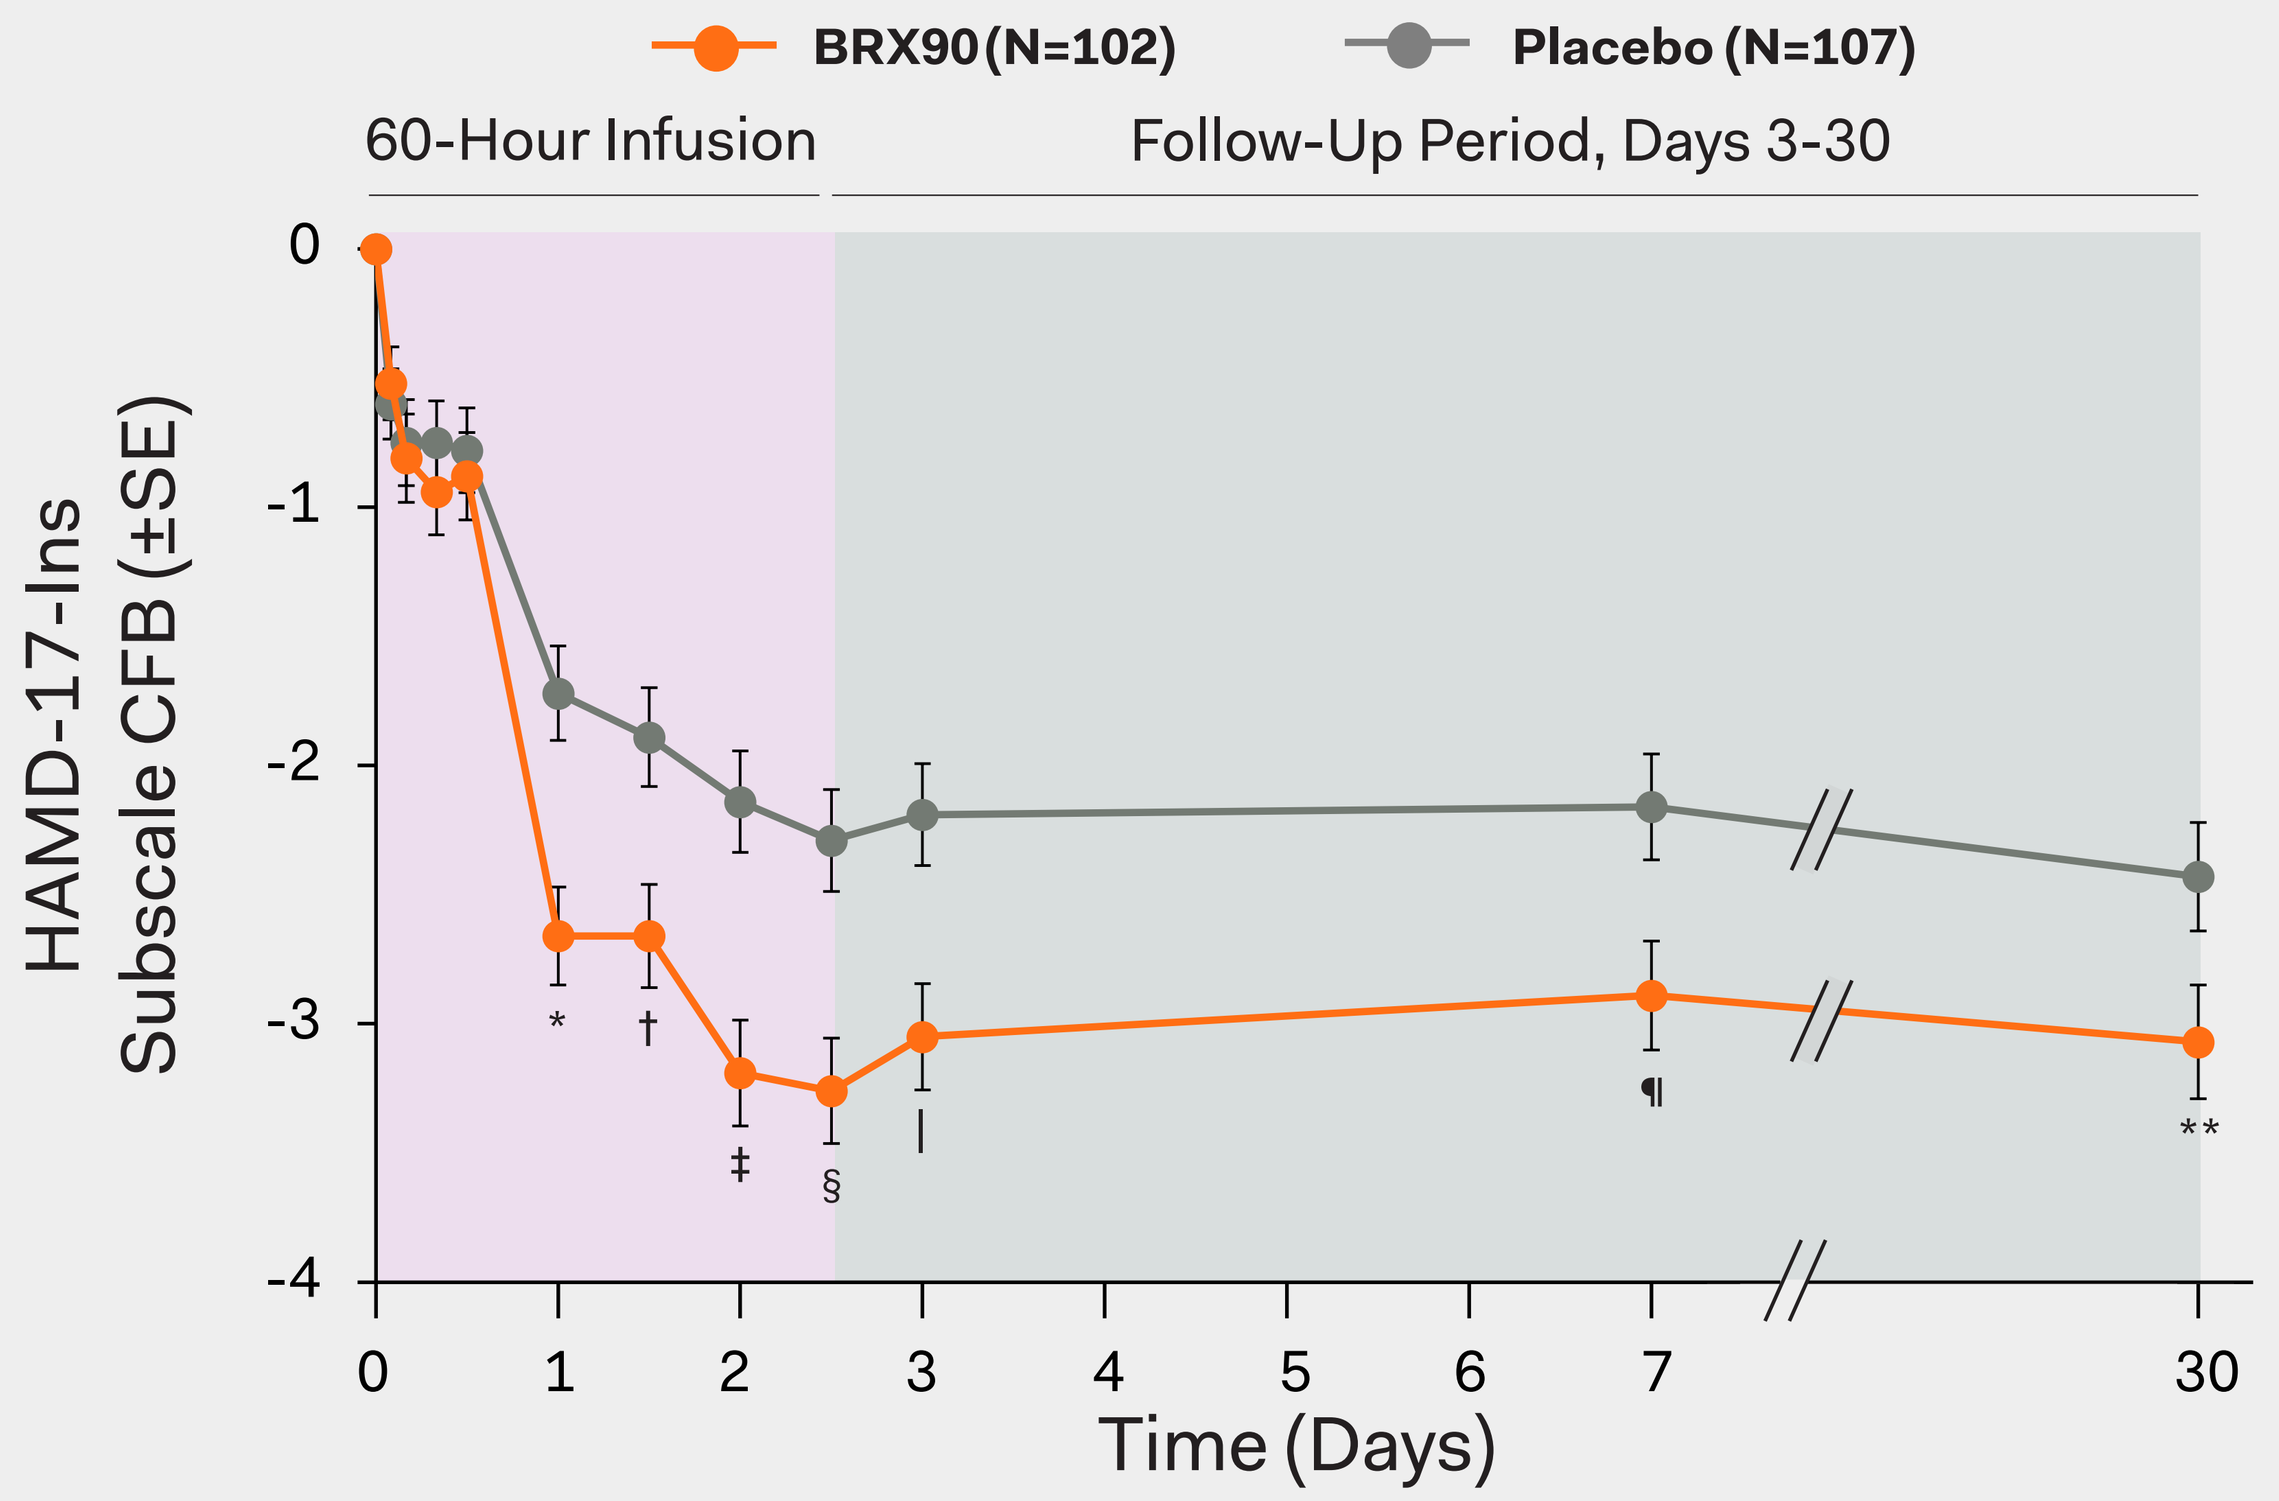
BRX90 (N=102)
Placebo (N=107)
60-Hour Infusion
Follow-Up Period, Days 3-30
HAMD-17-Ins
Subscale CFB (±SE)
0
-1
-2
-3
-4
0
1
2
3
4
5
6
7
30
Time (Days)
*
†
‡
§
|
¶
**

*p=0.0001; †p=0.0026; ‡p=0.0001; §p=0.0003; |p=0.0014; ¶p=0.0082; **p=0.0245 vs placebo.

- At baseline, the majority of patients reported insomnia (98% in both groups) and mean (±SD) HAMD-17-Ins scores were high (BRX90: 4.39±1.50; Placebo: 4.58±1.47).
- Significant differences vs placebo in HAMD-17-Ins CFB favoring BRX90 were sustained at Day 30 (p=0.0245).

SAFETY

- BRX common TEAEs (≥5% of BRX and ≥2x placebo rate) included sedation and/or somnolence, dry mouth, loss of consciousness, and flushing/hot flush.[17]
- Detailed adverse event rates including events of sedation/somnolence and loss of consciousness have been described in detail previously.[17]
- BRX caused sedation/somnolence requiring dose interruption/reduction in some patients (5% of BRX compared with 0% of placebo-treated patients). Some patients also had loss/altered state of consciousness (4% of BRX compared with 0% of placebo-treated patients). Time to recovery from loss/altered state of consciousness, after dose interruption, ranged from 15 to 60 minutes. All patients recovered with dose interruption.[17]

SIGNIFICANTLY GREATER IMPROVEMENTS IN HAMD-17-INS RESPONSE WITH BRX VS PLACEBO

ALL PATIENTS

≥50% HAMD-17-Ins Reduction From Baseline

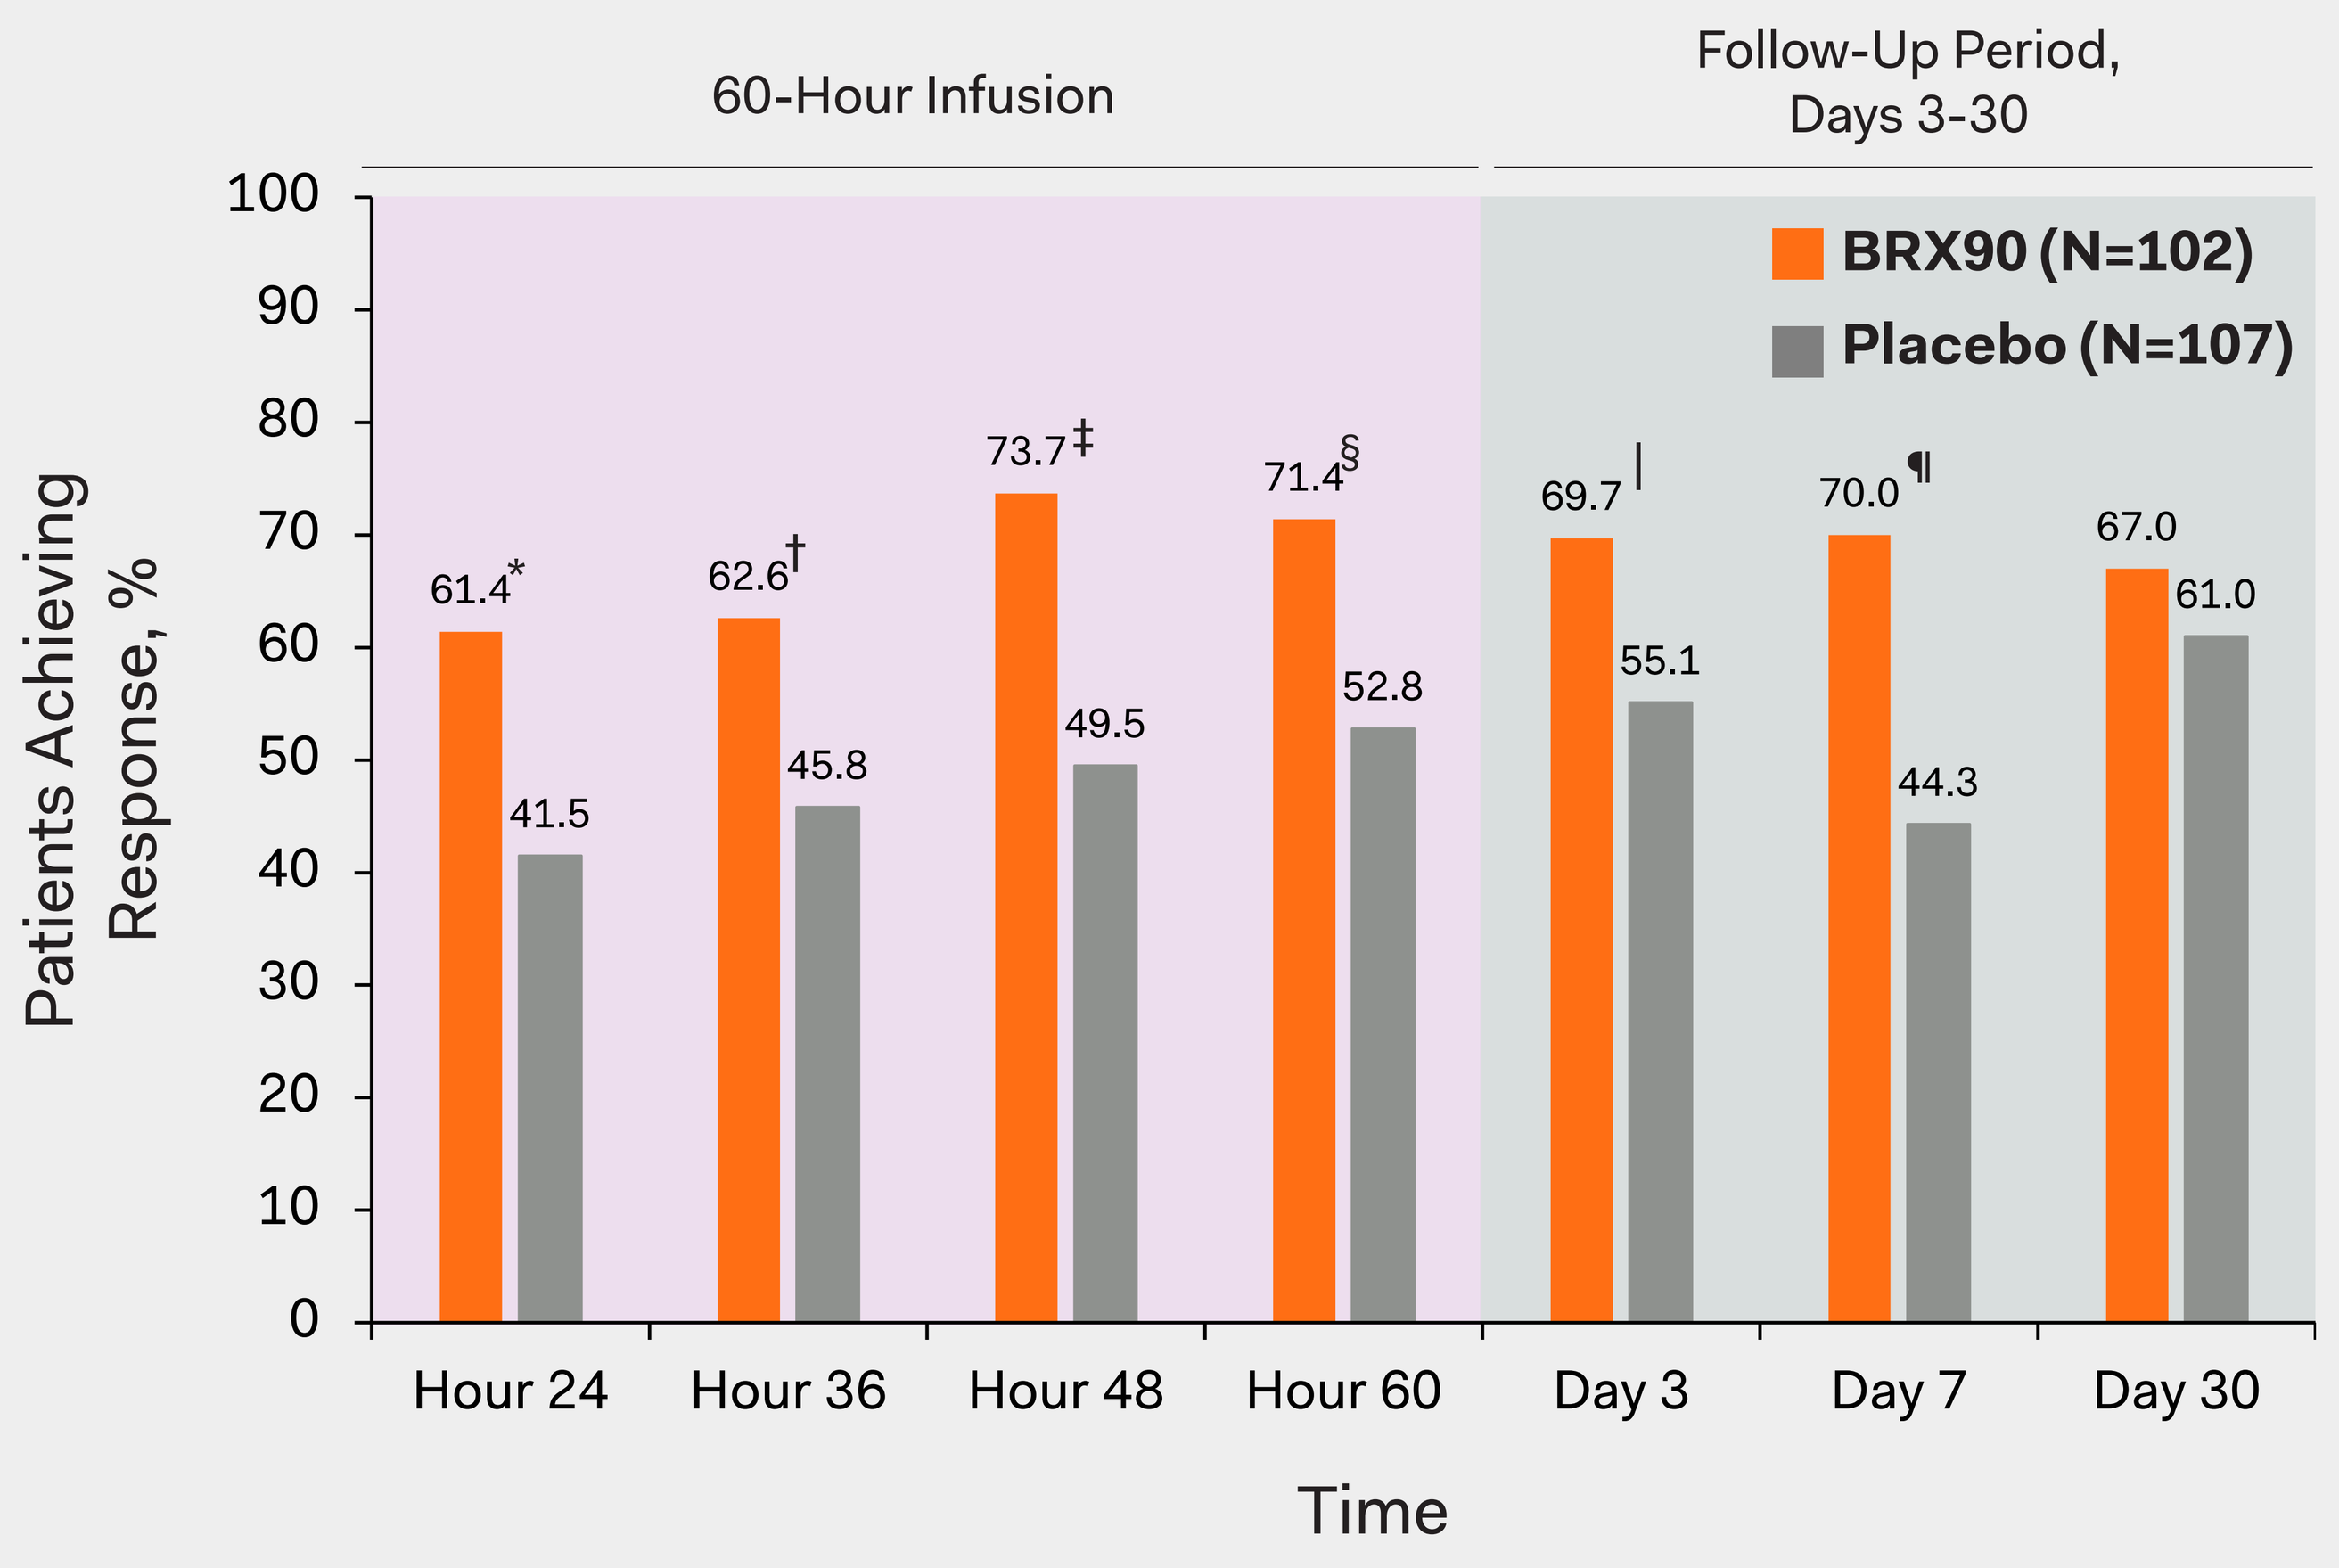
60-Hour Infusion
Follow-Up Period, Days 3-30
BRX90 (N=102)
Placebo (N=107)
Patients Achieving Response, %
61.4*
41.5
62.6†
45.8
73.7‡
49.5
71.4§
52.8
69.7|
55.1
70.0¶
44.3
67.0
61.0
Hour 24
Hour 36
Hour 48
Hour 60
Day 3
Day 7
Day 30
Time

*p=0.0007; †p=0.0062; ‡p<0.0001; §p=0.0013; |p=0.0104; ¶p<0.0001 vs placebo.

- A significantly greater proportion of BRX90 patients achieved HAMD-17-Ins response (≥50% reduction from baseline) compared with placebo at Hour 24, 36, 48, 60, Day 3 and 7.

≥70% HAMD-17-Ins Reduction From Baseline

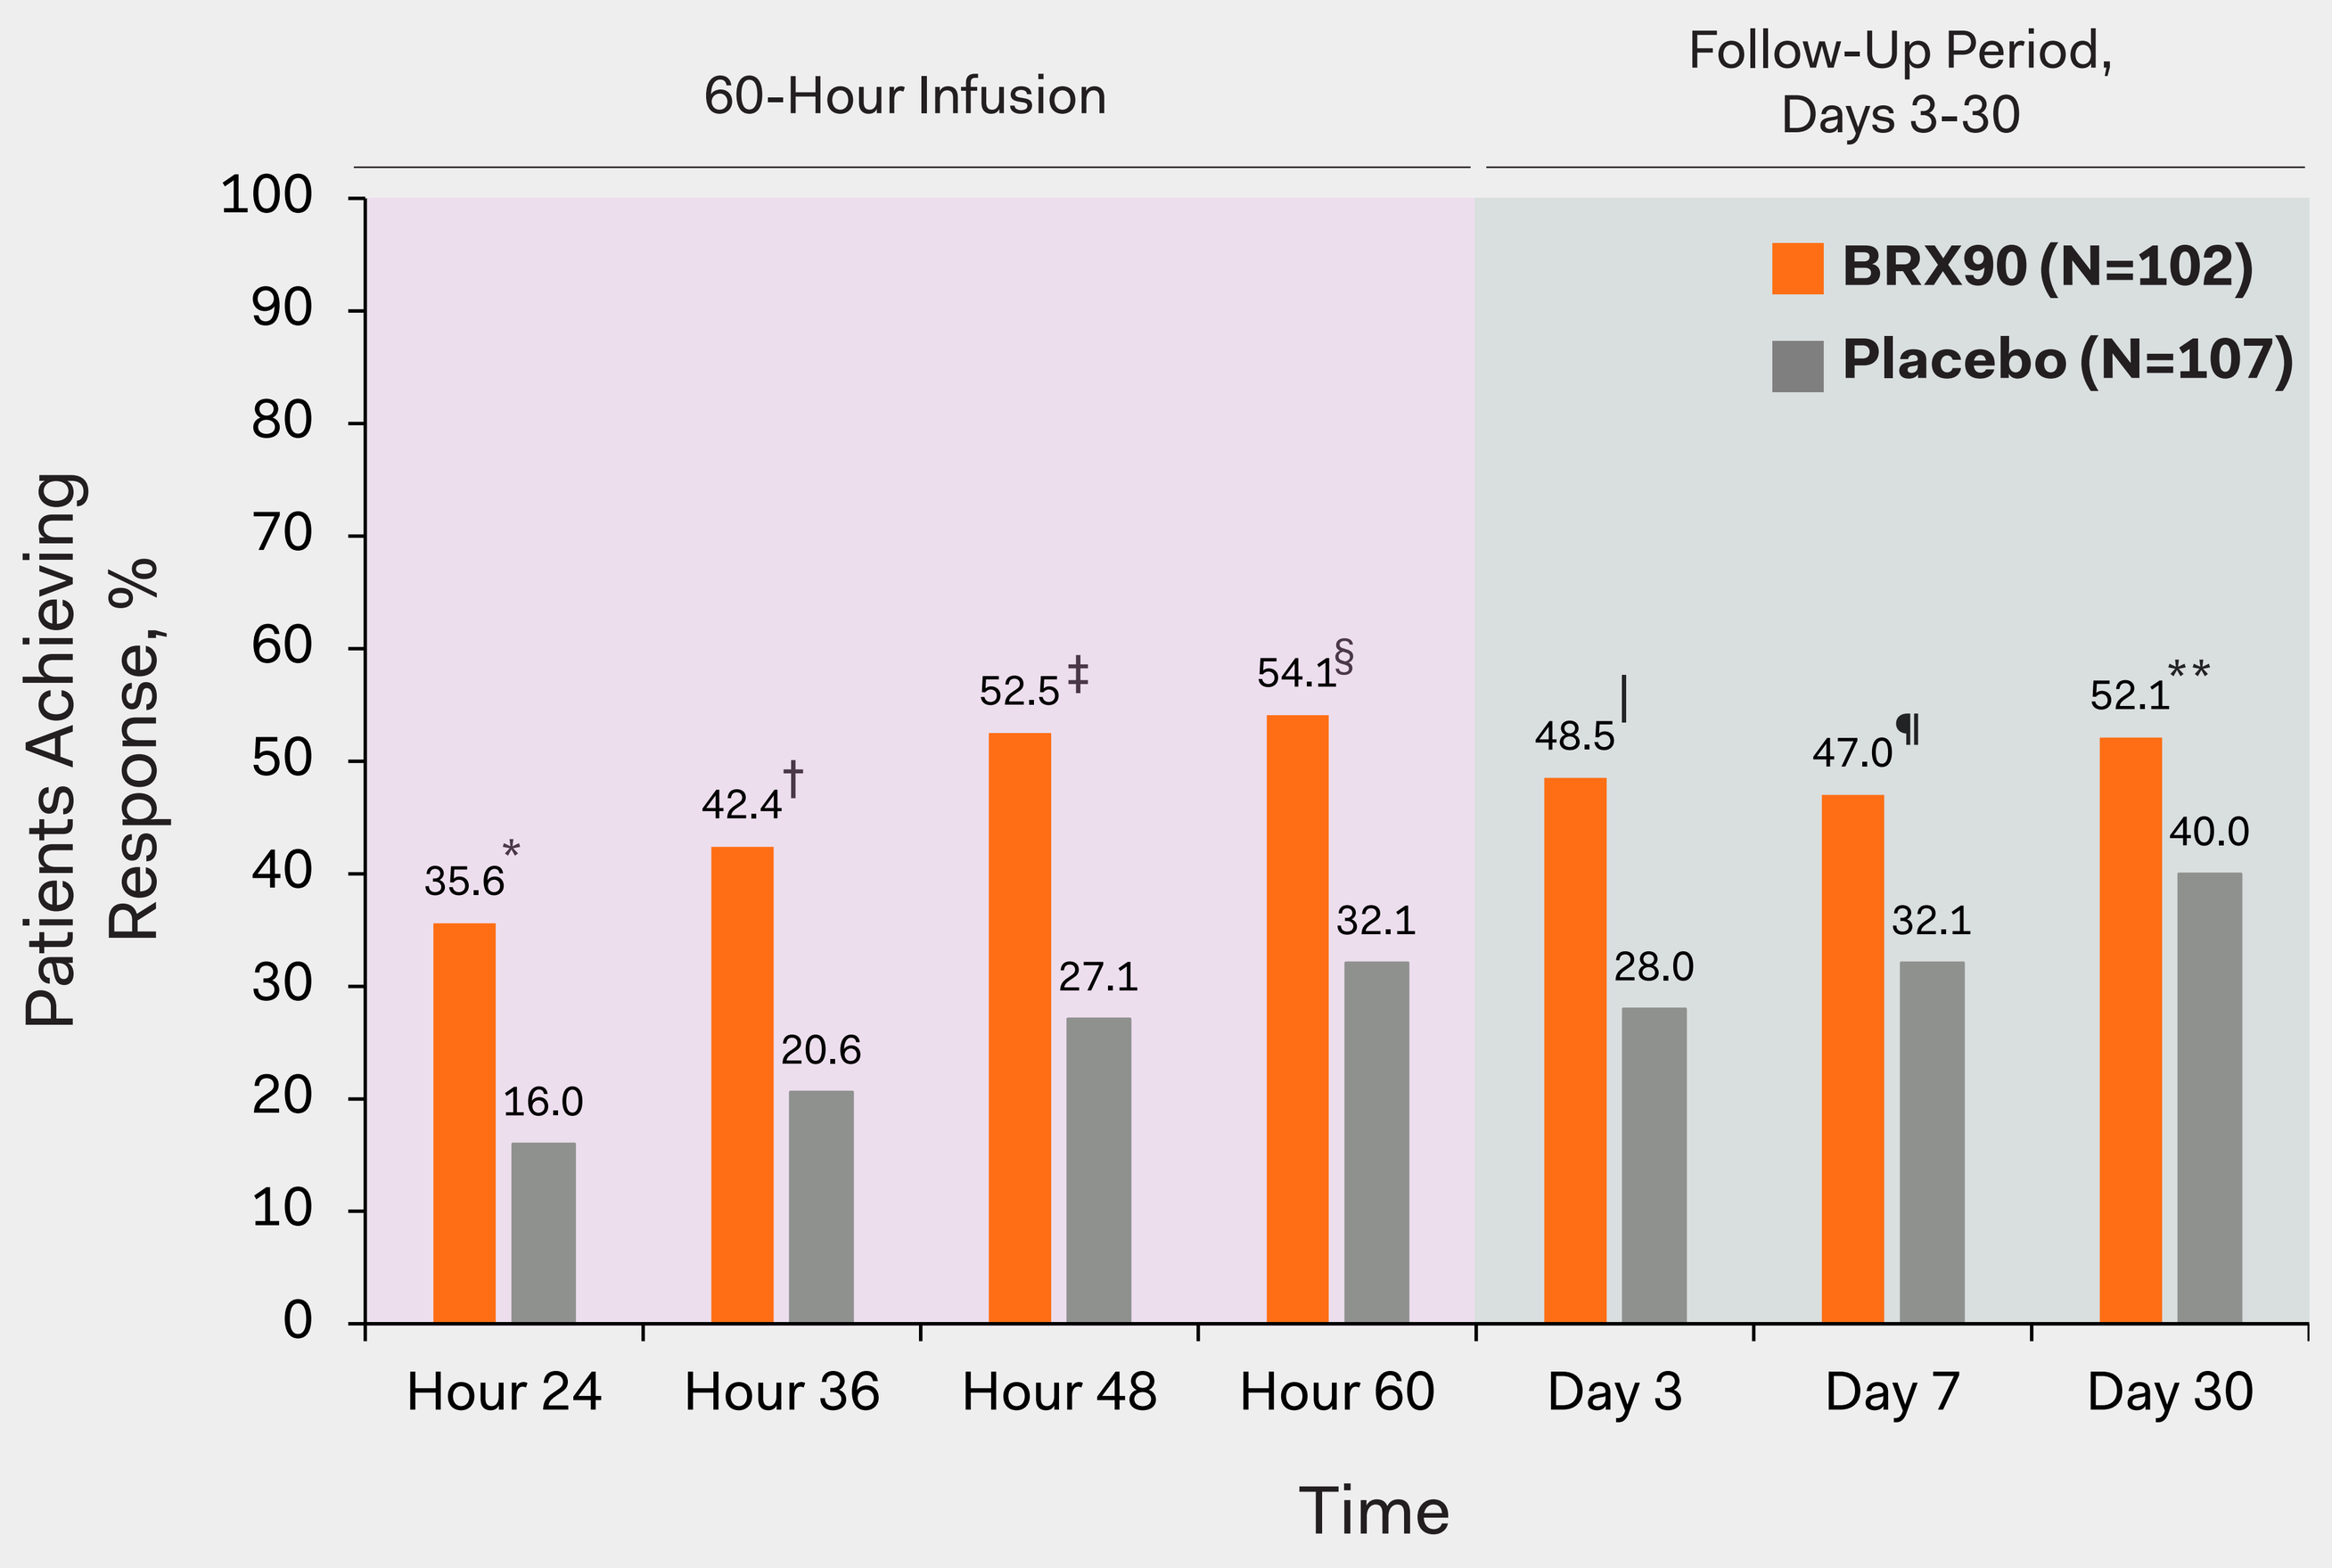
60-Hour Infusion
Follow-Up Period, Days 3-30
BRX90 (N=102)
Placebo (N=107)
Patients Achieving Response, %
35.6*
16.0
42.4†
20.6
52.5‡
27.1
54.1§
32.1
48.5|
28.0
47.0¶
32.1
52.1**
40.0
Hour 24
Hour 36
Hour 48
Hour 60
Day 3
Day 7
Day 30
Time

*p=0.0006; †p=0.0003; ‡p<0.0001; §p=0.0008; |p=0.0011; ¶p=0.0145; **p=0.0375 vs placebo.

- A significantly greater proportion of BRX90 patients achieved HAMD-17-Ins response (≥70% reduction from baseline) compared with placebo at all time points measured.

SUBGROUP CATEGORICAL RESPONSE ANALYSIS BY BASELINE SEVERITY OF INSOMNIA-RELATED SYMPTOMS

PATIENTS WITH SYMPTOMS OF INSOMNIA (HAMD-17-INS SCORE ≥1) AT BASELINE

Percentage of Patients Reporting Complete Absence of Symptoms of Insomnia

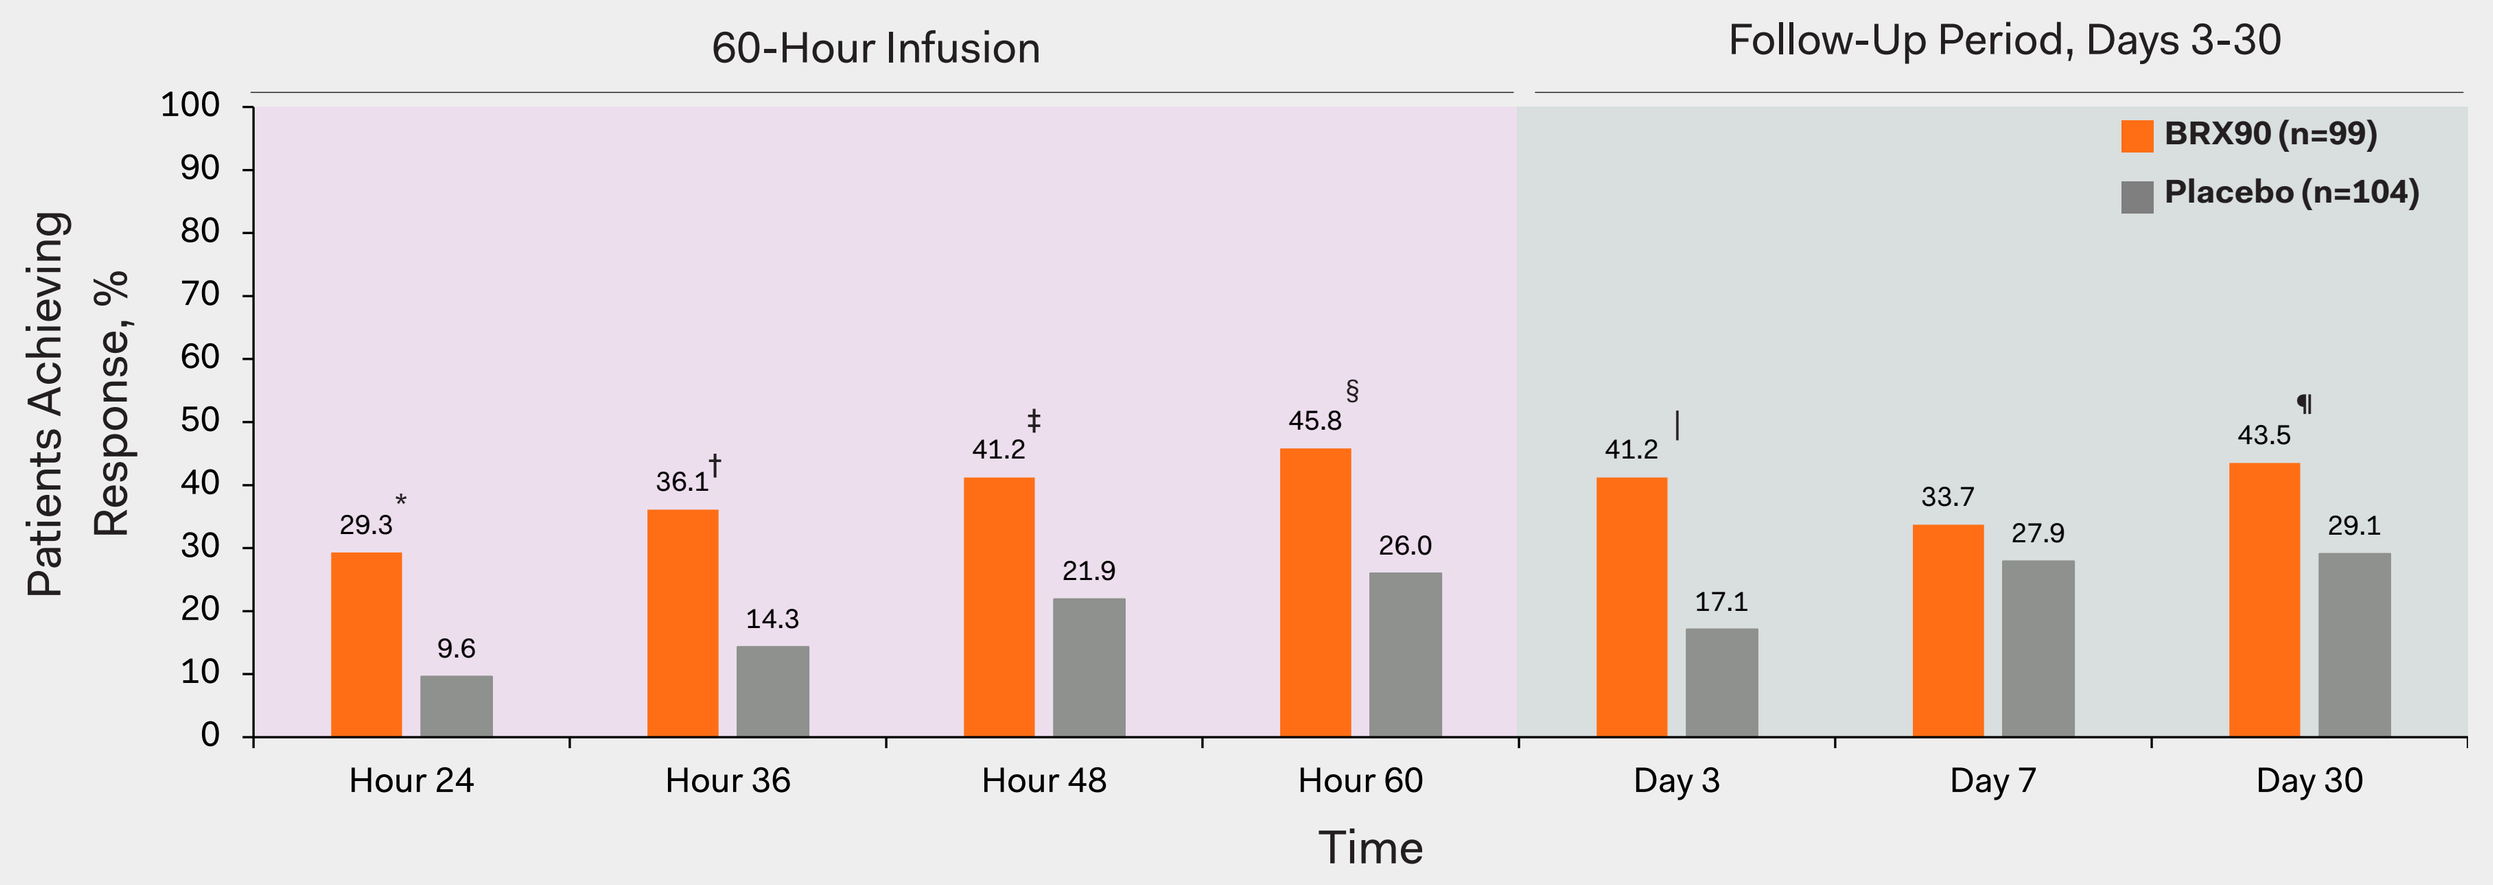
60-Hour Infusion
Follow-Up Period, Days 3-30
BRX90 (n=99)
Placebo (n=104)
Patients Achieving Response, %
29.3*
9.6
36.1†
14.3
41.2‡
21.9
45.8§
26.0
41.2|
17.1
33.7
27.9
43.5¶
29.1
Hour 24
Hour 36
Hour 48
Hour 60
Day 3
Day 7
Day 30
Time

*p=0.0004; †p=0.0003; ‡p=0.0027; §p=0.0031; |p=0.0001; ¶p=0.0254 vs placebo.

- A significantly greater proportion of BRX90 patients achieved HAMD-17-Ins response (with a baseline score ≥1) compared with placebo at Hour 24, 36, 48, 60, Day 3 and 30.

PATIENTS WITH SEVERE SYMPTOMS OF INSOMNIA (HAMD-17-INS SCORE ≥4) AT BASELINE

Percentage of Patients Achieving a HAMD-17-Ins Score <4

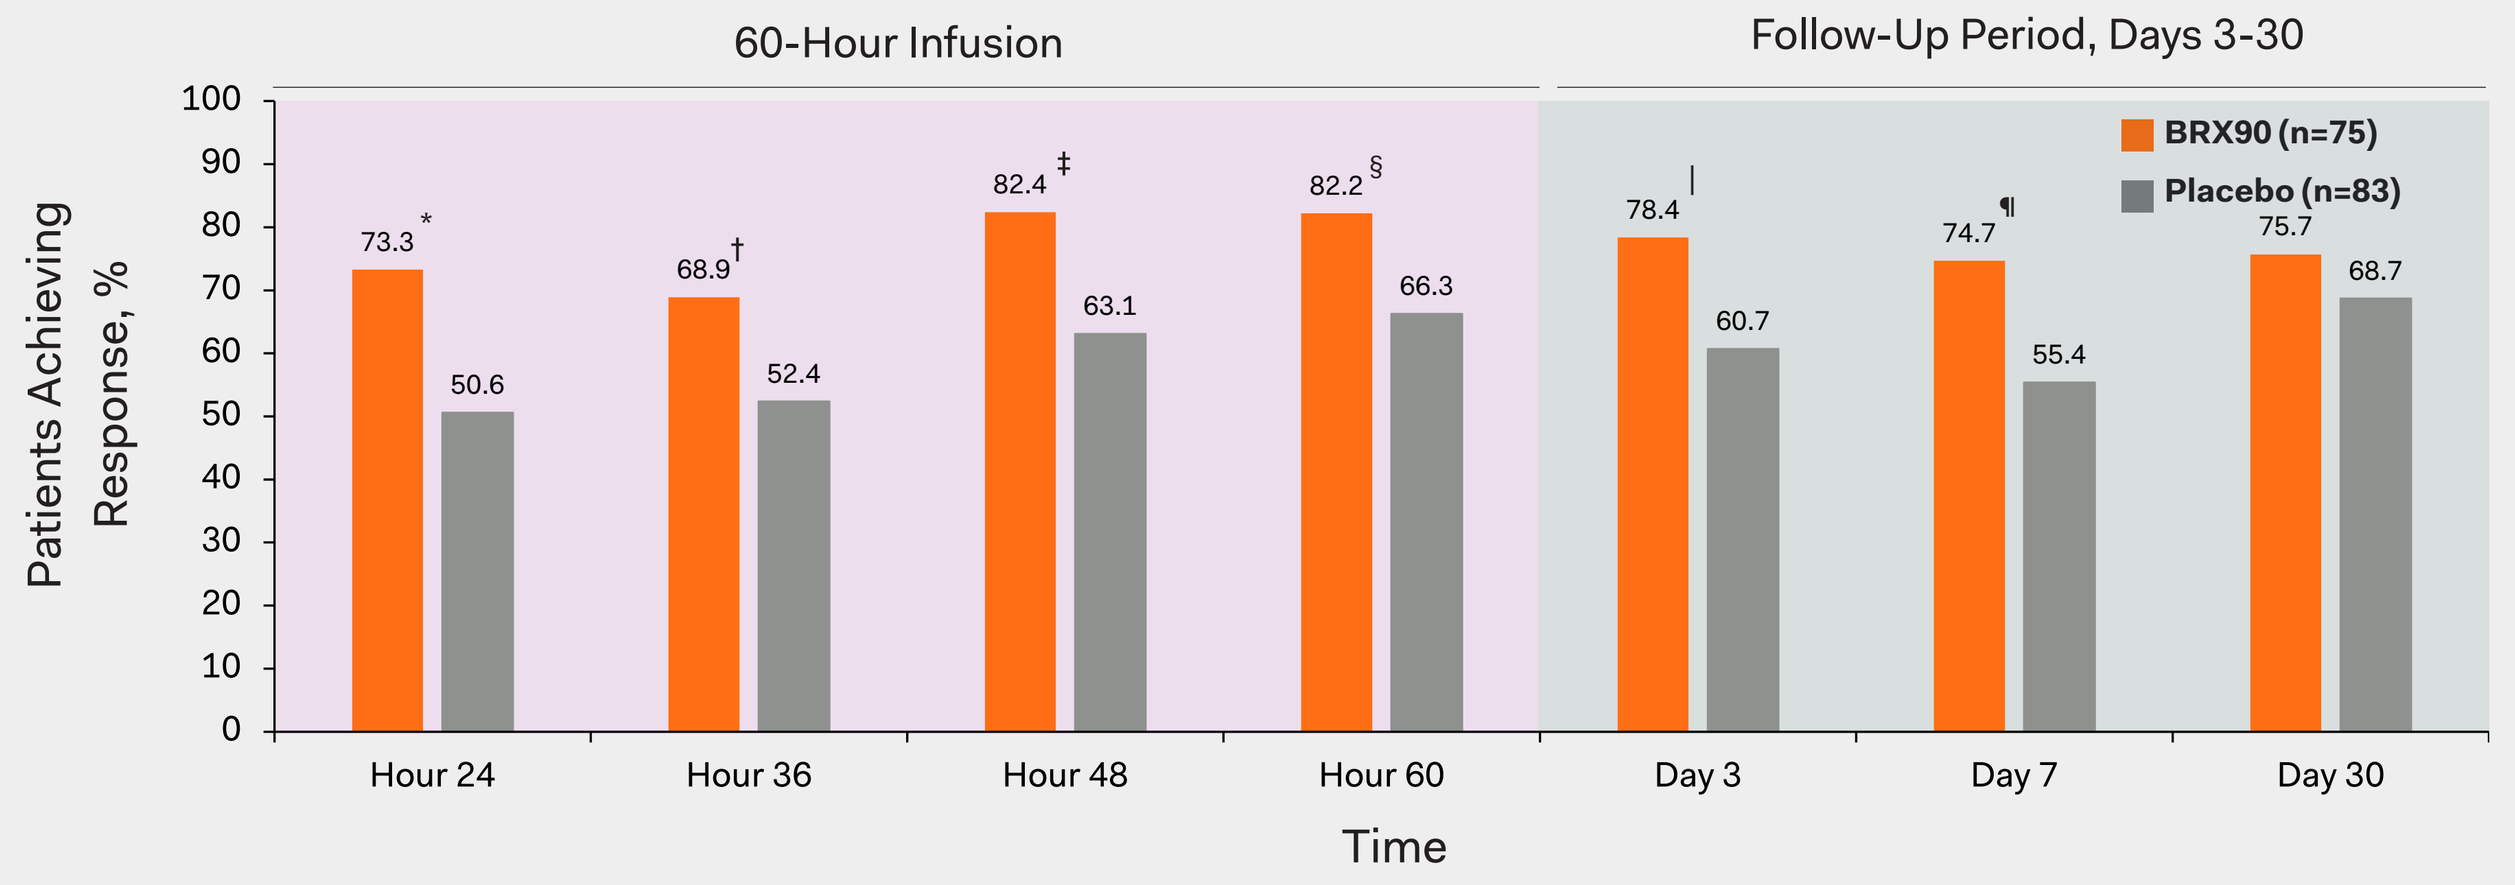
60-Hour Infusion
Follow-Up Period, Days 3-30
BRX90 (n=75)
Placebo (n=83)
Patients Achieving Response, %
73.3*
50.6
68.9†
52.4
82.4‡
63.1
82.2§
66.3
78.4|
60.7
74.7¶
55.4
75.7
68.7
Hour 24
Hour 36
Hour 48
Hour 60
Day 3
Day 7
Day 30
Time

*p=0.0010; †p=0.0217; ‡p=0.0022; §p=0.0109; |p=0.0066; ¶p=0.0050 vs placebo.

- A significantly greater proportion of BRX90 patients achieved HAMD-17-Ins response (with a baseline score ≥4) compared with placebo at Hour 24, 36, 48, 60, Day 3 and 7.

REFERENCES

1. Bauman BL et al. *MMWR Morb Mortal Wkly Rep* (2020) 69:575-581.
2. Hamilton BE, et al. National Center for Health Statistics. Births: Provisional data for 2018. Vital Statistics Rapid Release; no 7. https://www.cdc.gov/nchs/data/vsrr/vsrr-007-508.pdf. Published May 2019. Accessed November 10, 2020.
3. DeSisto CL, et al. *Prev Chronic Dis* (2014) 11:E10.
4. Centers for Disease Control and Prevention. https://www.cdc.gov/reproductivehealth/maternalinfanthealth/diabetes-during-pregnancy.htm. Updated June 12, 2018 Accessed November 10, 2020.
5. Centers for Disease Control and Prevention. https://www.cdc.gov/reproductivehealth/maternalinfanthealth/pregnancy-complications-data.htm. Updated February 28, 2019. Accessed November 10, 2020.
6. Roberts JM, et al. American College of Obstetricians and Gynecologists Task Force on Hypertension in Pregnancy. https://www.acog.org/~/media/Task%20Force%20and%20Work%20Group%20Reportspublic/HypertensioninPregnancy.pdf. Published 2013. Accessed November 10, 2020.
7. Callaghan WM, et al. *Am J Obstet Gynecol* (2010) 202:353.e1-6.
8. Diagnostic and statistical manual of mental disorders: DSM-5. Washington, DC: American Psychiatric Association; 2013.
9. Okun, Michele L. *J Affect Disord* (2011) 130:378-384.
10. Dorheim SK, et al. *PLoS ONE* (2014) 9:e94674.
11. Wolfson AR, et al. *Behav Sleep Med* (2003) 1:54-67.
12. Marques M, et al. Psychiatry Res. (2011) 186(2-3):272-280.
13. Emamian, F, et al. J Sleep Res. (2019) 28:e12858.
14. ZULRESSO™ (brexanolone) injection, for intravenous use. Sage Therapeutics, Inc., Cambridge, MA. Revised: 06/2019.
15. Martinez Botella G, et al. *J Med Chem* (2015) 58:3500-3511.
16. Hamilton M. J *Neurol Neurosurg Psychiatry* (1960) 23:56-62.
17. Meltzer-Brody S, et al. *Lancet* (2018) 392:1058-1070.

SAGE THERAPEUTICS and the SAGE THERAPEUTICS logo are trademarks of Sage Therapeutics, Inc. All other trademarks referenced herein are the property of their respective owners.

SUPPORT & DISCLOSURES

BW, KB, MYH, HL, and SJK are employees of Sage Therapeutics, Inc., with stock/stock options. RR has nothing to disclose. CNE has received consultation fees from Sage Therapeutics, Inc. KMD serves as a consultant to Sage Therapeutics, Inc., receives NIMH support and royalties from an NIH employee invention, and reports grants from Sage Therapeutics, Inc. awarded to Zucker Hillside Hospital during the conduct of the brexanolone injection clinical trials. DRR is a member of the Sage Therapeutics, Inc. Clinical Advisory Board and has stock/stock options. SM-B reports personal fees from MedScape and grants from Sage Therapeutics, Inc. awarded to the University of North Carolina (Chapel Hill, NC, USA) during the conduct of the brexanolone injection clinical trials and grants from Janssen, PCORI, and the NIH outside of the submitted work. This study and medical writing support were funded by Sage Therapeutics, Inc. Writing and editorial support was provided by Bonnie Bullock, PhD and Kathryn P. Wall, PhD of Boston Strategic Partners, Inc.

American College of Neuropsychopharmacology 2020
Virtual Annual Meeting
December 6-9, 2020

MRC-ZUL-00149 11/2020